Update on Phase 1/2 Clinical Trials for MPS I and MPS II Using ZFN-mediated *in vivo* Genome Editing

Paul Harmatz[a], Joseph Muenzer[b], Barbara K Burton[c], Can Ficicioglu[d], Heather A Lau[e], Nancy D Leslie[f], Edward Conner[g], Cheryl Wong Po Foo[g], Sagar A Vaidya[g], Thomas Wechsler[g], Chester B Whitley[h]

[a]UCSF Benioff Children's Hospital Oakland, Oakland CA, USA; [b]University of North Carolina, Chapel Hill, NC, USA; [c]Lurie Children's Hospital, Chicago, IL, USA; [d]Children's Hospital of Philadelphia, PA, Unites States; [e]NYU Langone Medical Center, New York City, NY, United States; [f]Cincinnati Children's Hospital Medical Center, Cincinnati, OH, United States; [g]Sangamo Therapeutics, Inc.. Richmond, CA, United States; [h]University of Minnesota, Minneapolis, MN, United States

Abstract

Mucopolysaccharidosis (MPS) type I and type II are inherited monogenic lysosomal storage diseases characterized by progressive neurodegeneration, respiratory complications, skeletal abnormalities, and premature death. Sangamo Therapeutics is developing two new therapies for the treatment of MPS I and MPS II, named SB-318 and SB-913, respectively. These therapies are designed to use Zinc Finger Nuclease (ZFN)-mediated *in vivo* genome editing to insert a therapeutic copy of the gene for the missing enzyme into liver cells. The high precision and specificity of ZFNs allows for integration at a defined genomic location and is intended to generate high levels of constant enzyme expression. This one-time therapy aims for lifetime production of the missing enzyme, with the goal of eliminating the need for enzyme replacement therapy (ERT) to improve therapeutic benefit and quality of life for patients. Two Phase 1/2 clinical trials, ***EMPOWERS*** and ***CHAMPIONS*** (clinicaltrials.gov NCT02702115 and NCT03041324), are ongoing in the U.S. to determine if these investigational therapies are safe and tolerable in patients with MPS I or MPS II. Both studies are multicenter, open-label, dose ranging trials with one-time peripheral intravenous infusion of the study drug, SB-318 or SB-913, followed by three years of observation and testing. Enzyme activity and other clinical endpoints will also be measured. A total of nine subjects are planned for enrollment in each trial. An update is provided on the progress of the clinical trials.

Background

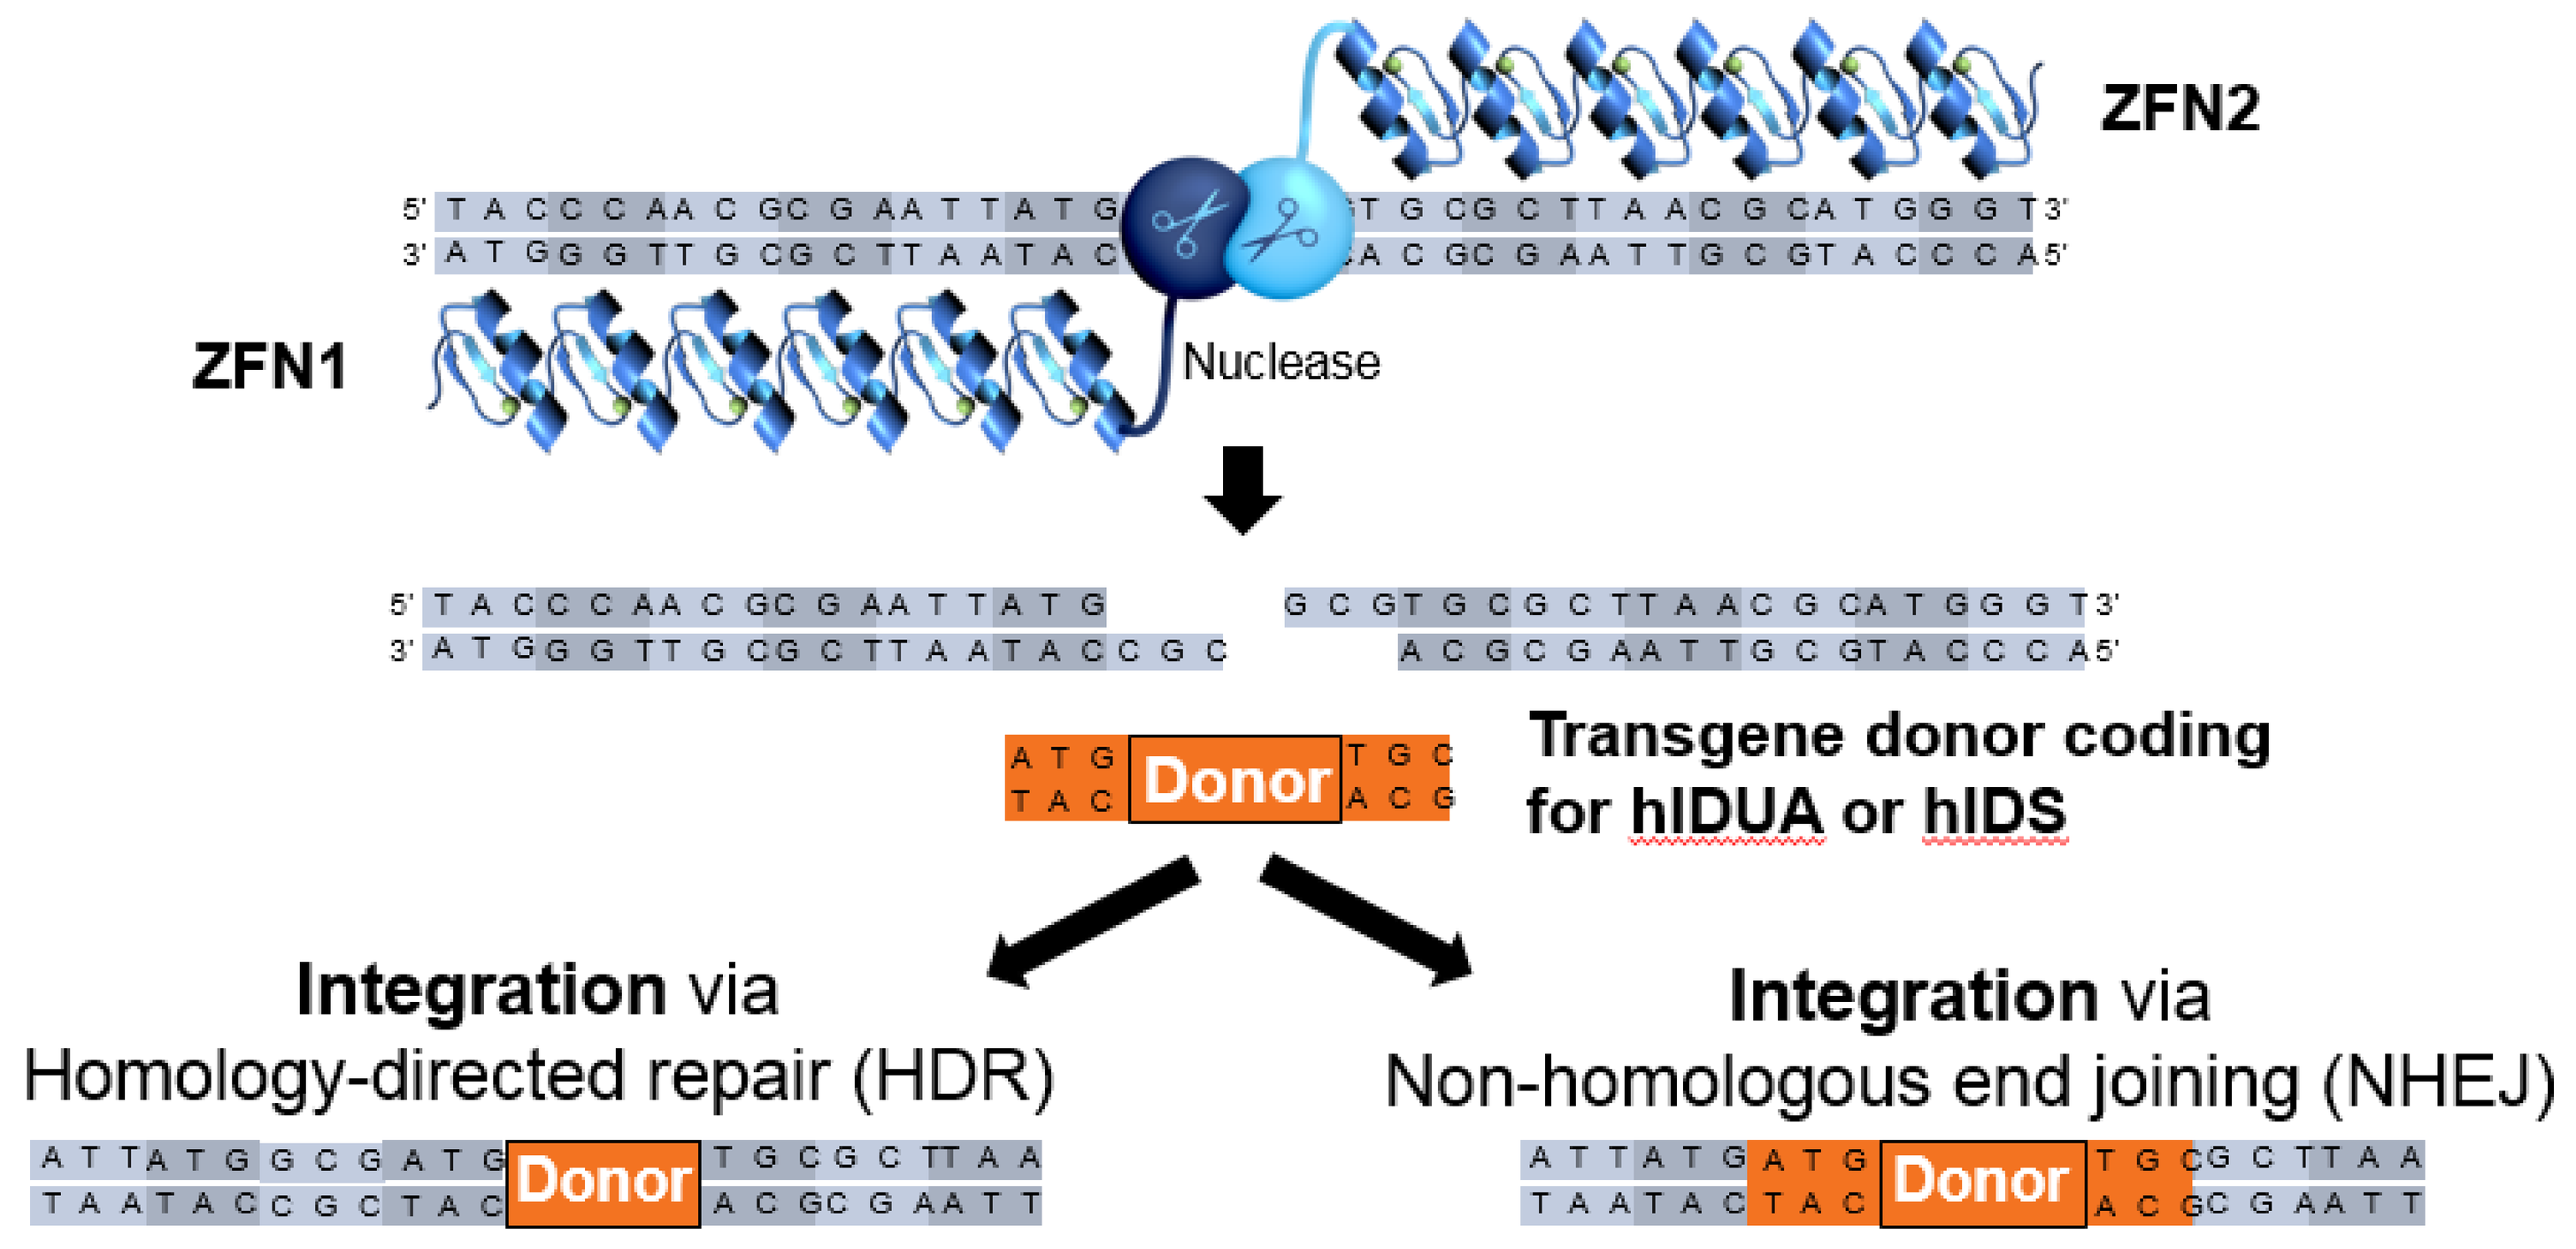

ZFN2
ZFN1
Nuclease
Donor
Transgene donor coding for hIDUA or hIDS
Integration via Homology-directed repair (HDR)
Integration via Non-homologous end joining (NHEJ)

Figure 1. Sangamo's engineered ZFNs can precisely and accurately target and edit any desired location in the human genome. When a transgene is present, the donor DNA is integrated into the targeted location in the human genome.

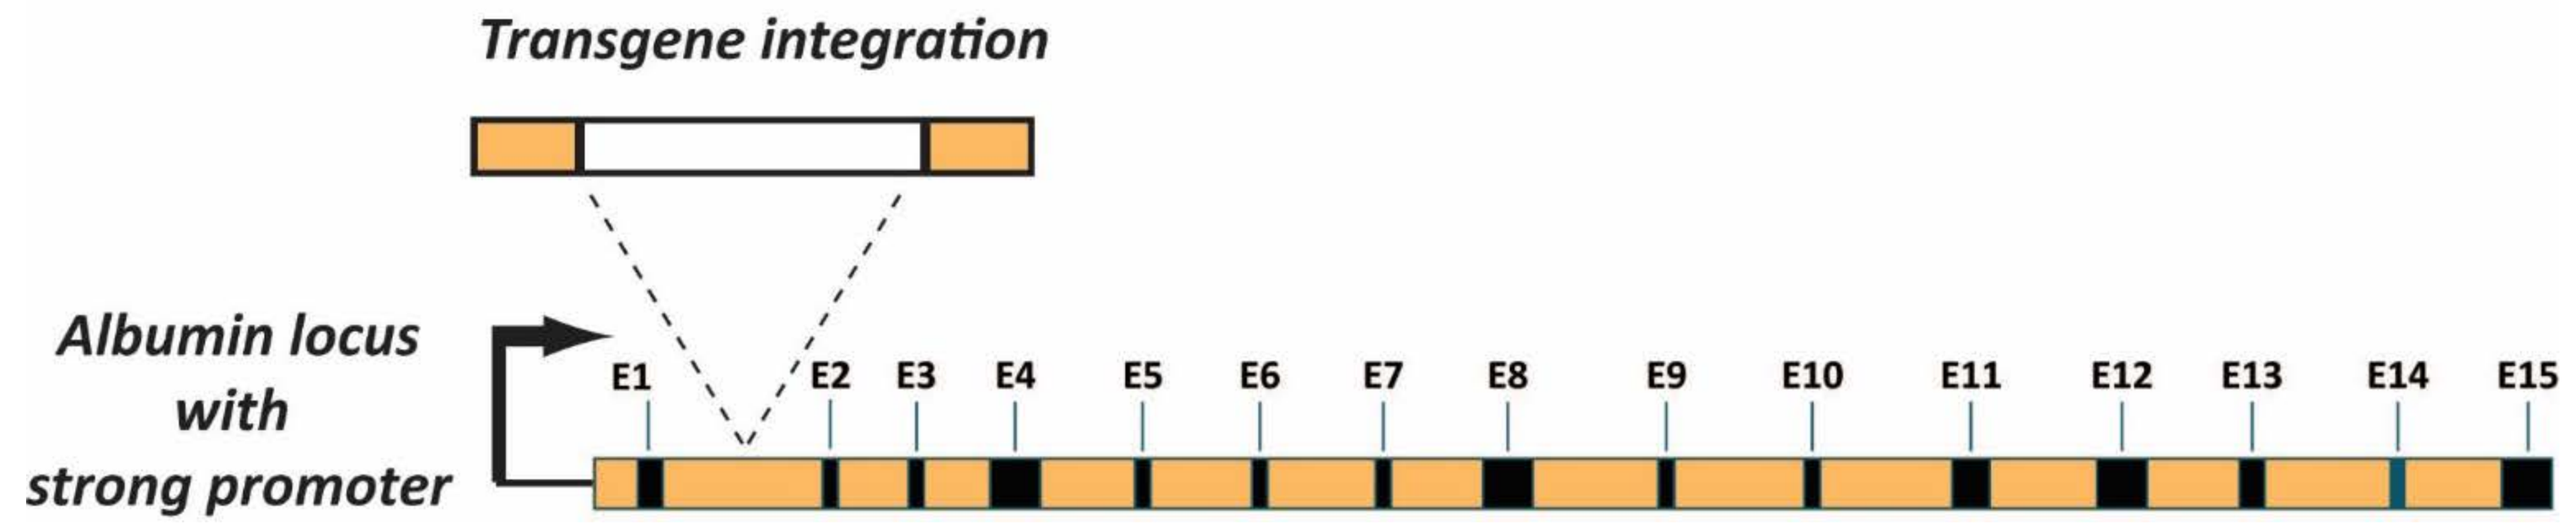

Transgene integration
Albumin locus with strong promoter
E1 E2 E3 E4 E5 E6 E7 E8 E9 E10 E11 E12 E13 E14 E15

Figure 2. Targeted insertion into the albumin locus has many advantages, including (1) tissue-specific and only produced in the liver, (2) redundant as no appreciable effects on albumin levels after integration, (3) the powerful promoter leads to high levels of constant protein production relative to ERT

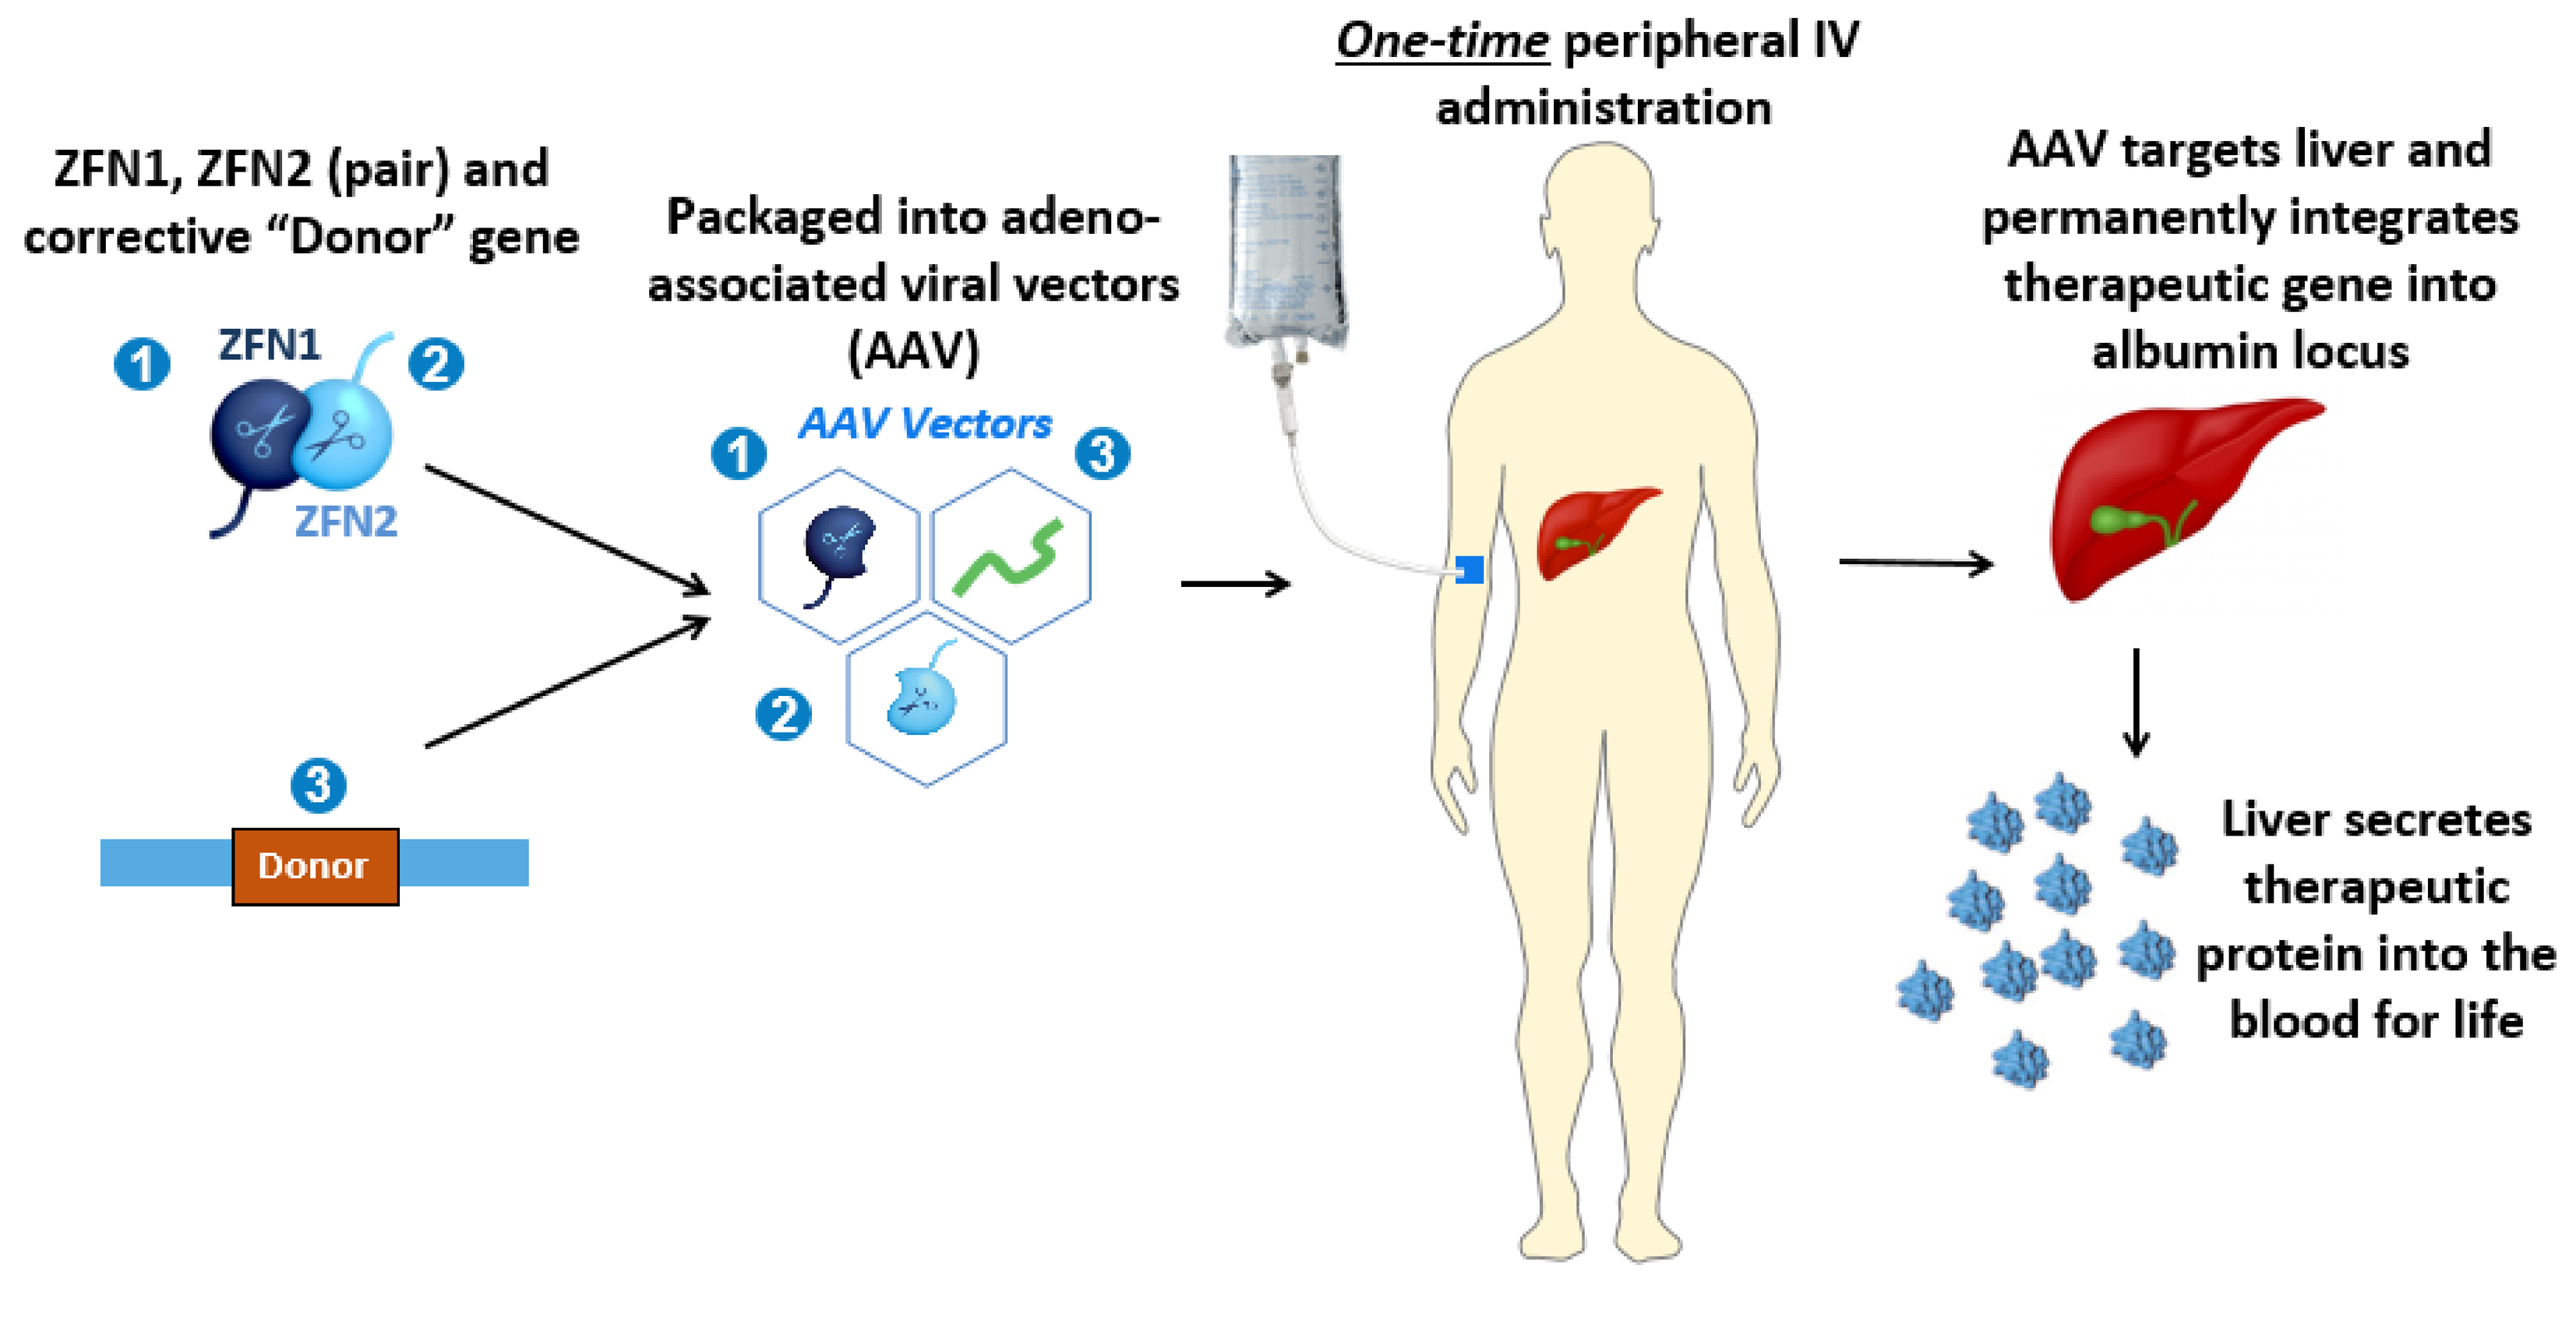

ZFN1, ZFN2 (pair) and corrective "Donor" gene
ZFN1
ZFN2
Donor
Packaged into adeno-associated viral vectors (AAV)
AAV Vectors
One-time peripheral IV administration
AAV targets liver and permanently integrates therapeutic gene into albumin locus
Liver secretes therapeutic protein into the blood for life

Figure 3. Schematic of Sangamo's *in vivo* genome editing platform using rAAV2/6 to deliver two ZFNs and a donor transgene with the aim of providing lifelong production of the therapeutic protein after a single intravenous dose.

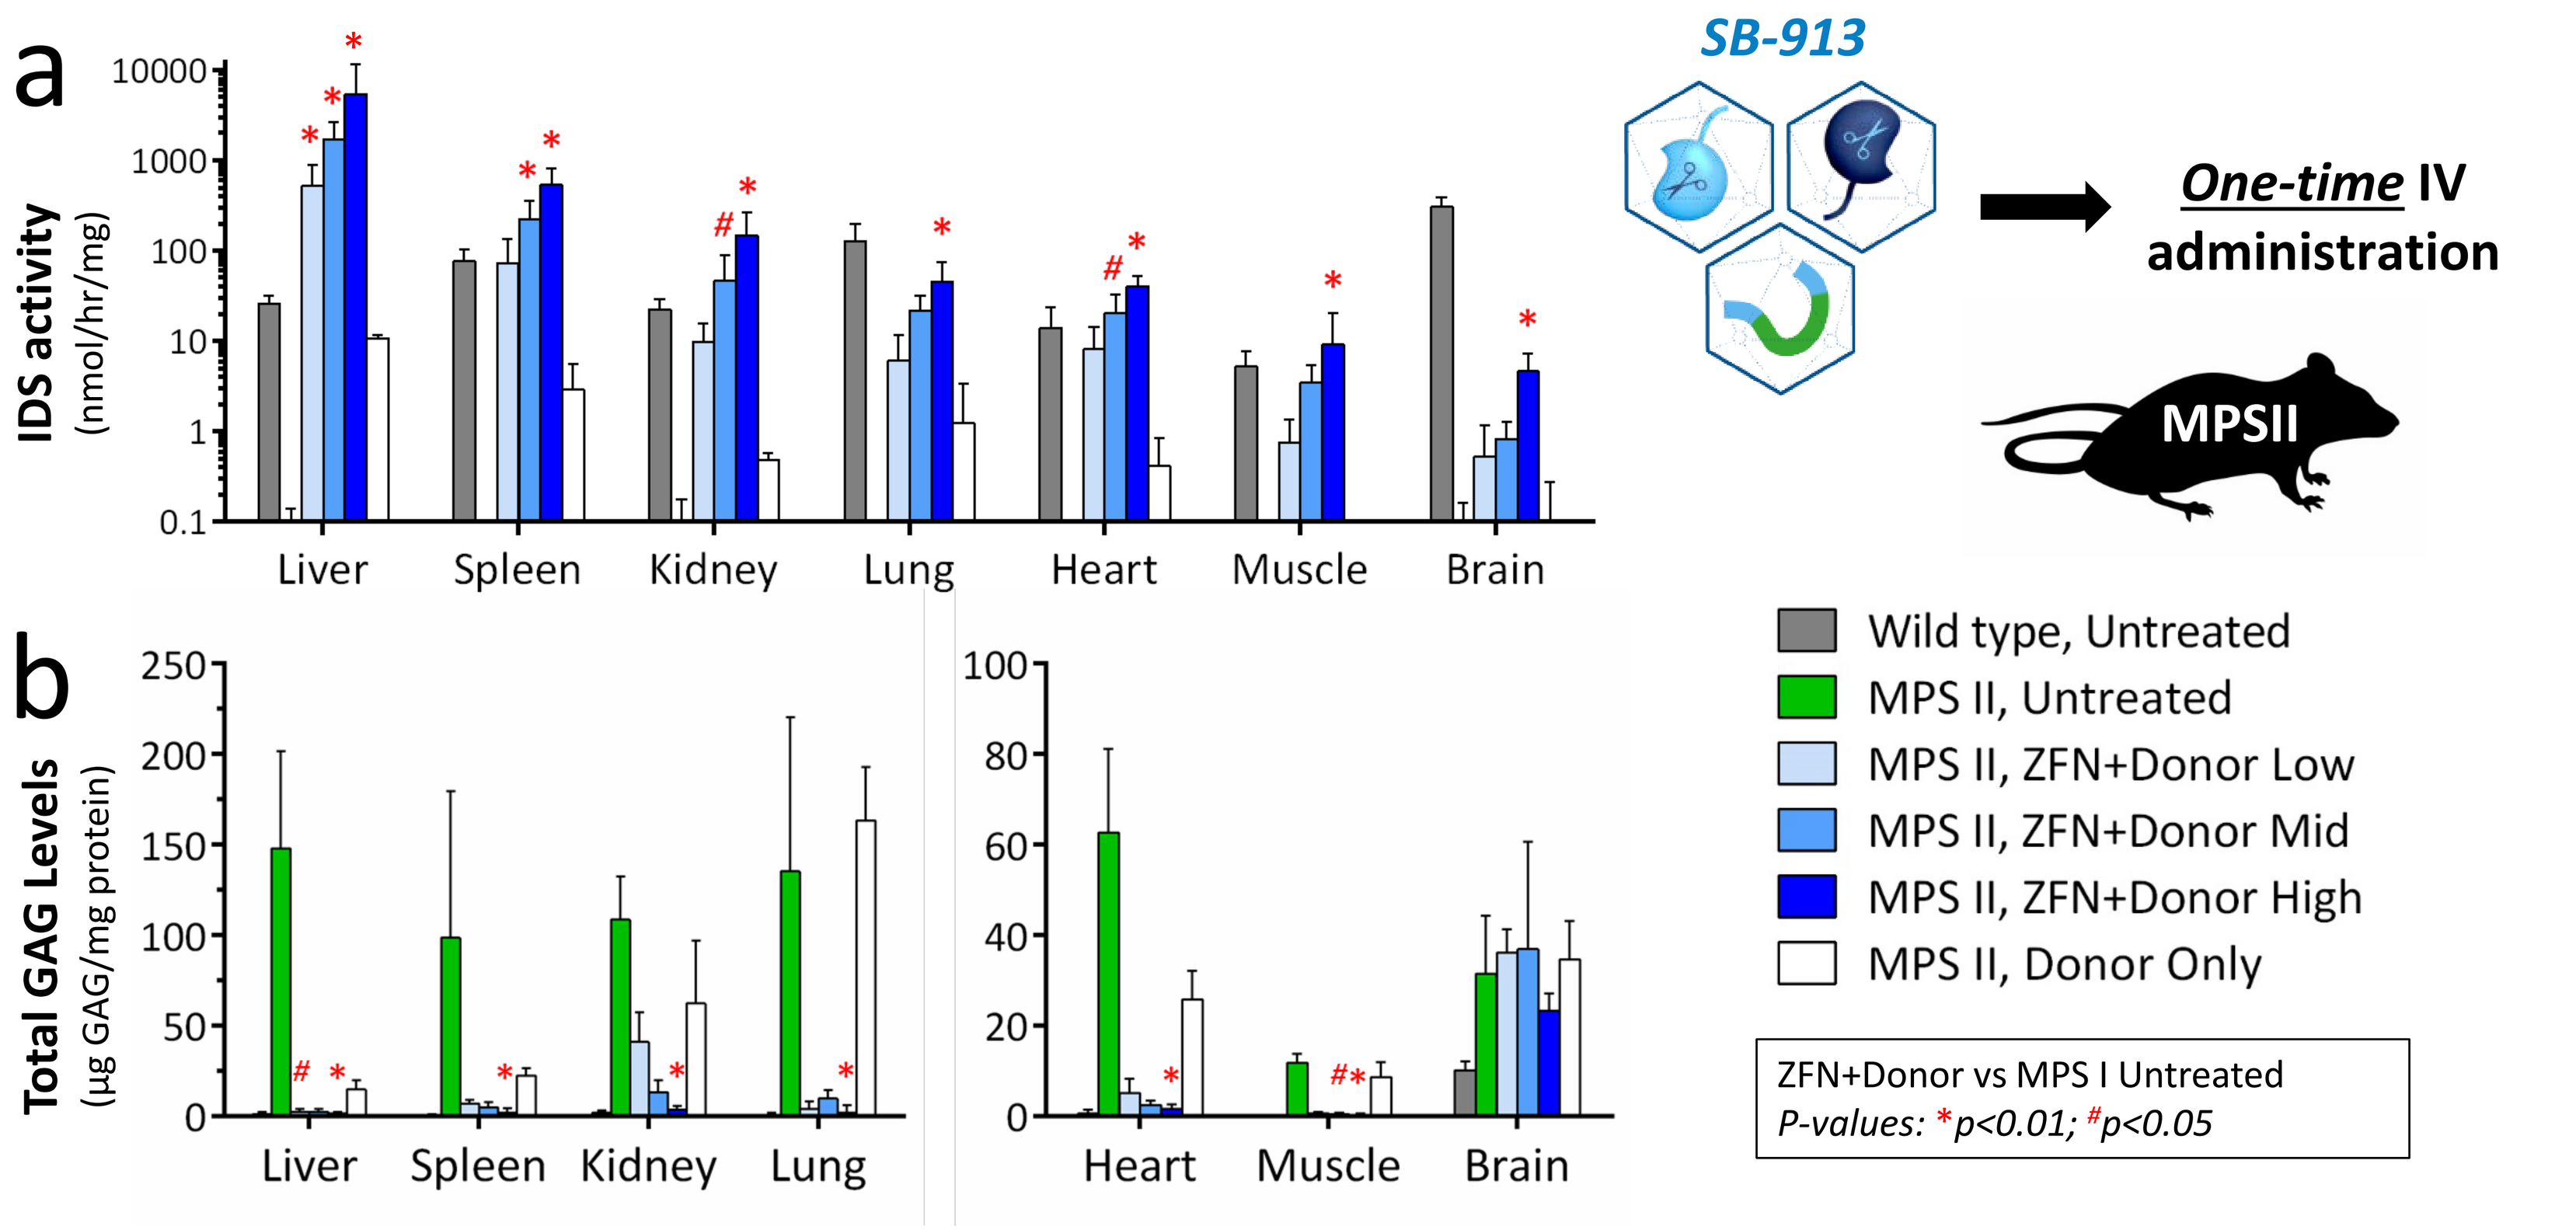

a
IDS activity (nmol/hr/mg)
Liver Spleen Kidney Lung Heart Muscle Brain
SB-913
One-time IV administration
MPSII
b
Total GAG Levels (µg GAG/mg protein)
Liver Spleen Kidney Lung
Heart Muscle Brain
Wild type, Untreated
MPS II, Untreated
MPS II, ZFN+Donor Low
MPS II, ZFN+Donor Mid
MPS II, ZFN+Donor High
MPS II, Donor Only
ZFN+Donor vs MPS I Untreated
P-values: *p<0.01; #p<0.05

Figure 4. In a mouse model of MPS II, male mice received SB-913 murine-specific surrogate drug product at 1-2 months of age and the indicated tissues were analyzed for **(a)** iduronate 2-sulfatase (IDS) enzyme activity and **(b)** glycosaminoglycans (GAG) storage levels at 4 months post-dose. Similar results were observed in a mouse model of MPS I (data not shown)

Methods

We describe a Phase 1, multi-center, open-label, single-dose, dose-ranging study which is the first-in-human experience with SB-913. SB-913 is a combination of 3 recombinant adeno-associated virus serotype 2/6 (rAAV2/6) vectors that encode:

1. ***ZFN 1 (SB-47171): Left-side ZFN that targets the albumin locus***
2. ***ZFN 2 (SB-47898): Right-side ZFN that targets the albumin locus***
3. ***hIDS Donor (SB-IDS): DNA template encoding the corrective transgene***

Subjects with MPS II receive a one-time intravenous infusion of SB-913 with three years of follow up. Three dose levels will be administered (5.00E+12, 1.00E+13, and 5.00E+13 vg/kg) with two subjects per dose cohort and potential expansion to 9 total subject at the maximally-tolerated dose. All subjects will receive prophylactic oral steroid taper for the first 20 weeks after dosing. The study has oversight from an independent external Safety Monitoring Committee (SMC).

Primary Objective

- To evaluate the safety and tolerability of SB-913

Secondary Objectives

- To evaluate change from baseline over time in:
 - Plasma IDS activity
 - Urine GAG levels
 - AAV2/6 clearance

Key Eligibility Criteria

- Age ≥18 years old
- Clinical diagnosis of attenuated MPS II
- Lack of neutralizing antibodies to AAV2/6
- Subjects with lack of tolerance to ERT or significant infusion reactions excluded
- Subjects with significant liver dysfunction or liver mass excluded
- Subject who have received prior treatment with a gene therapy product excluded

Results

As of 27 DEC 2017, one MPS II subject has been enrolled and received SB-913 at a dose of 5.00E+12 vg/kg. The subject is a 44-year old male with a history of Hunter Syndrome, COPD, and tracheobronchomalacia. Adverse events since the time of infusion are shown in **Table 1**.

Adverse Event *(Preferred Term)*	Grade	Serious	Study Drug-Related	Resolved
Cold sweat	1-Mild	No	**Yes**	Yes
Dizziness	1-Mild	No	**Yes**	Yes
Asthenia	1-Mild	No	**Yes**	Yes
Pollakiuria	1-Mild	No	No	Yes
Upper respiratory tract infection	2-Mod	No	No	Yes
Bronchitis	3-Sev	**Yes**	No	Yes
Gastrooesophageal reflux disease	2-Mod	No	No	Yes
Pneumothorax	1-Mild	No	No	No

Table 1. Summary of adverse events (AEs) in the first subject after SB-913 administration. The subject developed a URI post-dosing and was subsequently hospitalized for acute bronchitis. This was reported as a serious adverse event (SAE) and the subject made a full recovery after receiving medical treatment. The event was considered not related to SB-913 as the patient has chronic pulmonary disease and was previously hospitalized for a similar respiratory illness.

- No clinically-significant increases from baseline were observed in liver function tests of aspartate aminotransferase (AST) or alanine aminotransferase (ALT) at any time points following SB-913 administration for the first subject.
- A second subject received SB-913 at a dose of 5.00E+12 vg/kg in Jan 2018 and tolerated dosing well (personal communication, P. Harmatz)

Conclusions

- SB-913 was administered to two subjects with MPSII at a dose of 5.00+E12 vg/kg
- No SAEs related to SB-913 were reported in the first subject 1 month post-dosing
- Both the ***EMPOWERS*** and ***CHAMPIONS*** clinical studies are currently recruiting adult patients with MPS I and MPS II

Acknowledgements

We would like to thank the clinical sites, Principal Investigators and coordinating staff for their participation in the EMPOWERS and CHAMPIONS clinical trials and their hard work in enrolling MPS I and MPS II subjects. We would further like to thank the MPS I and MPS II subjects who have graciously agreed to participate in these clinical trials.