Engineering of Allogeneic Regulatory T Cells Expressing a Chimeric Antigen Receptor (Allo-CAR-Tregs) Using Zinc Finger Nuclease/AAV6-Mediated Editing

Poster P447

Davide Monteferrario*, Satish Tadi*, Coralie Dupont, Caroline Jeanneau, Gaëlle Saviane, Irène Marchetti, Gregory Lara, Sandrine Rouquier, Marion Ayrault, Julie Gertner-Dardenne, Maurus de la Rosa,# David Fenard#
*Co-first authors; #Co-senior authors

Sangamo Therapeutics France, Allée de la Nertière, Les Cardoulines HT1, 06560 VALBONNE, France

Introduction

- Conversion of polyclonal regulatory T cells (Tregs) into antigen-specific Tregs by the introduction of a chimeric antigen receptor (CAR) has gained increasing attention as a potential treatment option for autoimmune diseases.
- Designed to recognize a clinically relevant antigen, CAR-Tregs can be specifically activated through the CAR upon binding to a specific antigen, proliferate, and acquire their full immunomodulatory capacities. Therefore, these genetically modified CAR-Tregs are potentially able to prevent or dampen autoimmune reactions in patients.
- Allogeneic CAR-Tregs were engineered using a donor DNA consisting of a CAR-expressing adeno-associated virus type 6 (CAR-AAV6) and zinc finger nuclease (ZFN) editing technology to insert the CAR into the beta-2 microglobulin (*B2M*) locus, leading to the knockout of cell-surface expression of the major histocompatibility complex I (MHC-I). MHC-I plays a major role in allogeneic rejection, and by deleting it, we aim to limit the elimination of allogeneic CAR-Tregs by the patient's immune system.
- Here we generated CAR-Tregs using ZFN/AAV6 technology to screen various expression cassettes for optimal CAR expression and functionality. The tested CAR-Tregs were designed to recognize the human interleukin-23 (IL-23) receptor (IL23R), a key player in the pathogenesis of inflammatory bowel disease (IBD), and against the human myelin oligodendrocyte glycoprotein (MOG), an autoimmune target in multiple sclerosis (MS).

Tregs: a new class of cell-based therapeutics

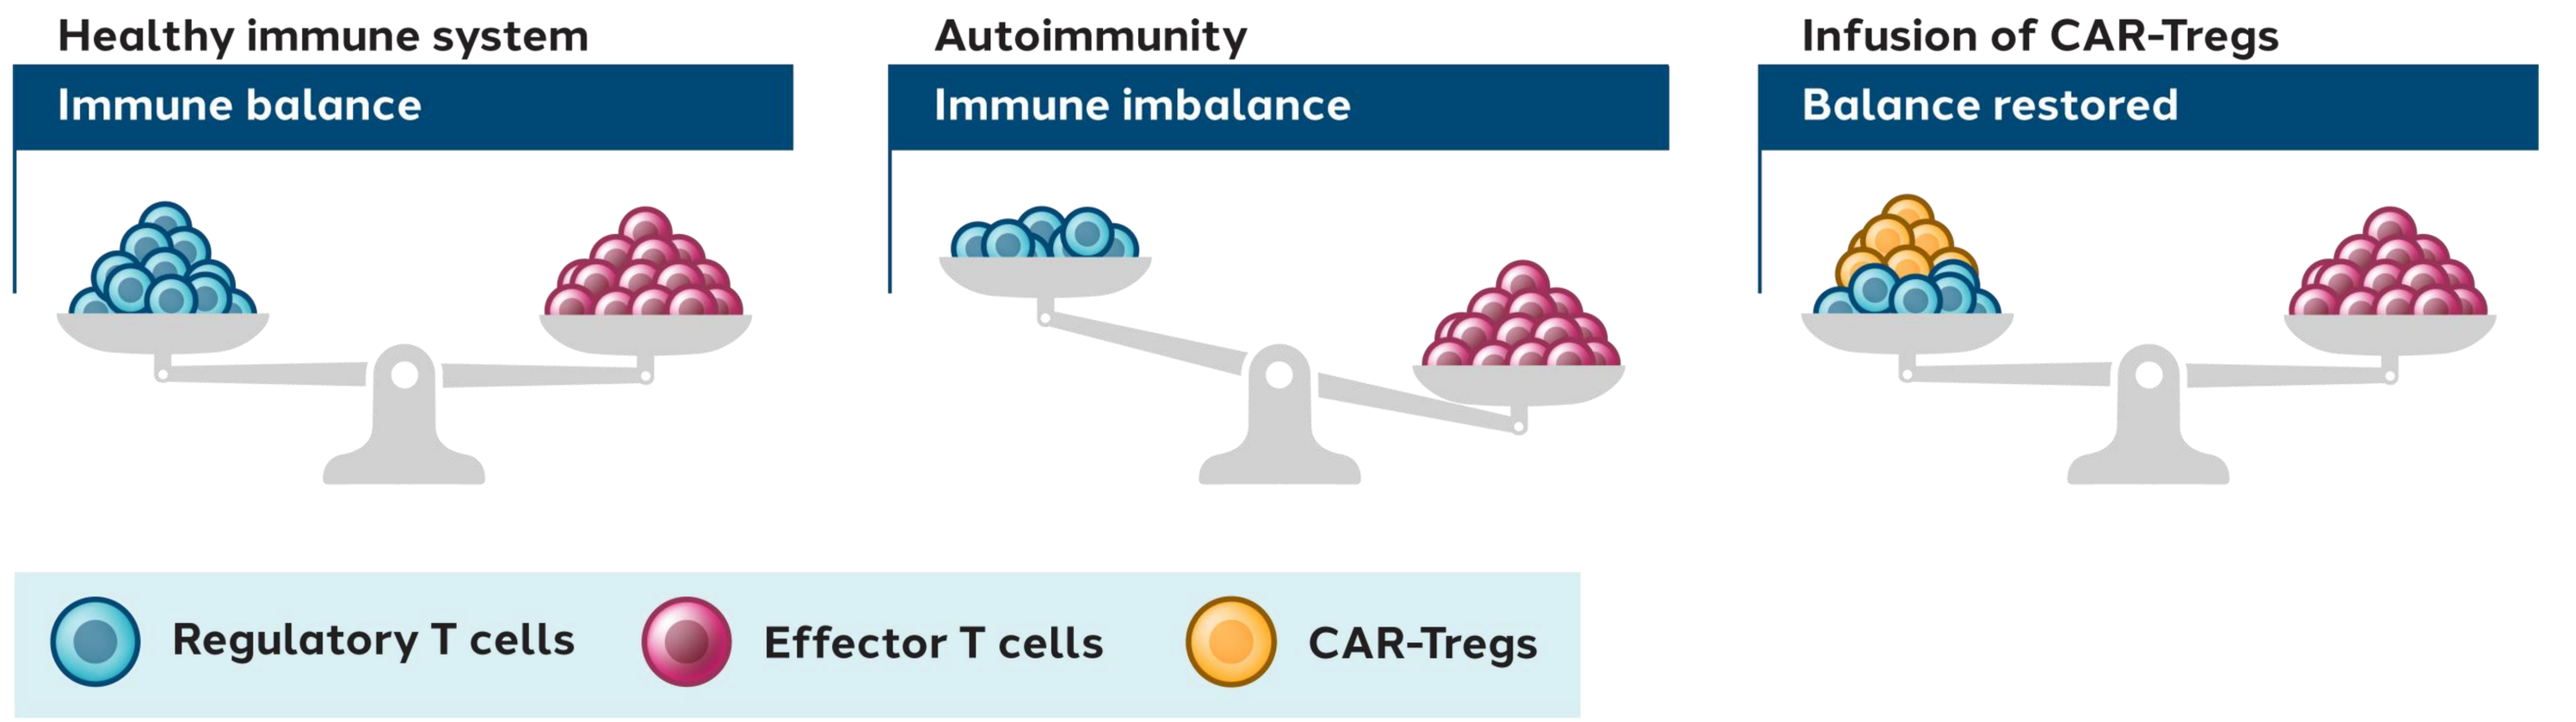
Healthy immune system
Immune balance
Autoimmunity
Immune imbalance
Infusion of CAR-Tregs
Balance restored
Regulatory T cells
Effector T cells
CAR-Tregs

- Tregs maintain immune homeostasis at various tissues.
- The suppressive function of Tregs inhibits the mounting of inflammatory responses, i.e., Tregs confer tolerance.
- Tregs can be used as a cell-based therapy across various applications where induction of immune tolerance can restore homeostasis and counter the disease state (e.g., prevention of transplant rejection, treatment of a multitude of autoimmune diseases).

Advantages of off-the-shelf allogeneic CAR-Treg cell therapies

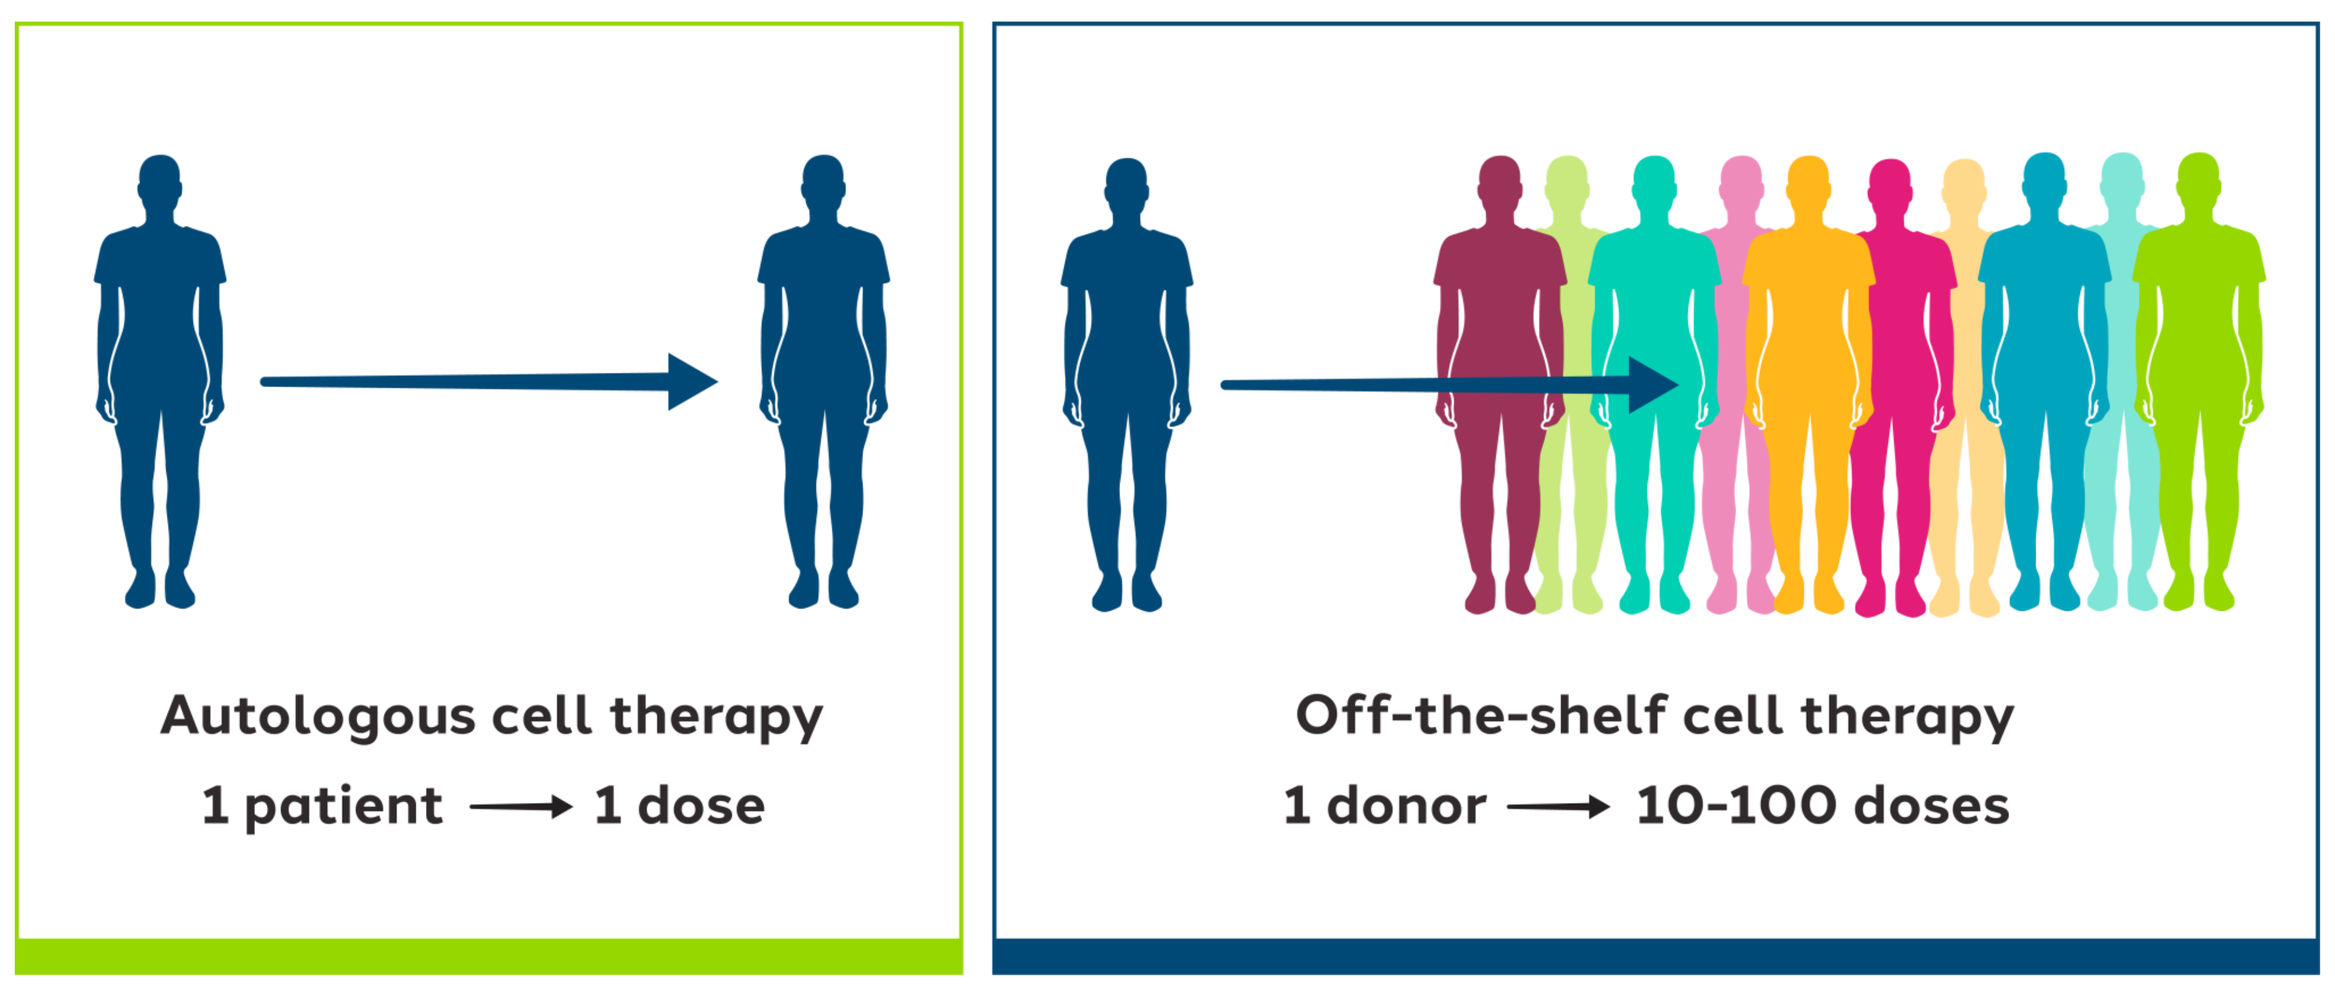
Autologous cell therapy
1 patient ⟶ 1 dose
Off-the-shelf cell therapy
1 donor ⟶ 10-100 doses

- Allogeneic CAR-Treg cell product confers an advantage when autologous Tregs cannot be isolated from the patient.
- Universal off-the-shelf allogeneic CAR-Treg product will significantly cut costs and time.
- ZFN technology allows the development of allogeneic CAR-Tregs.

Methods

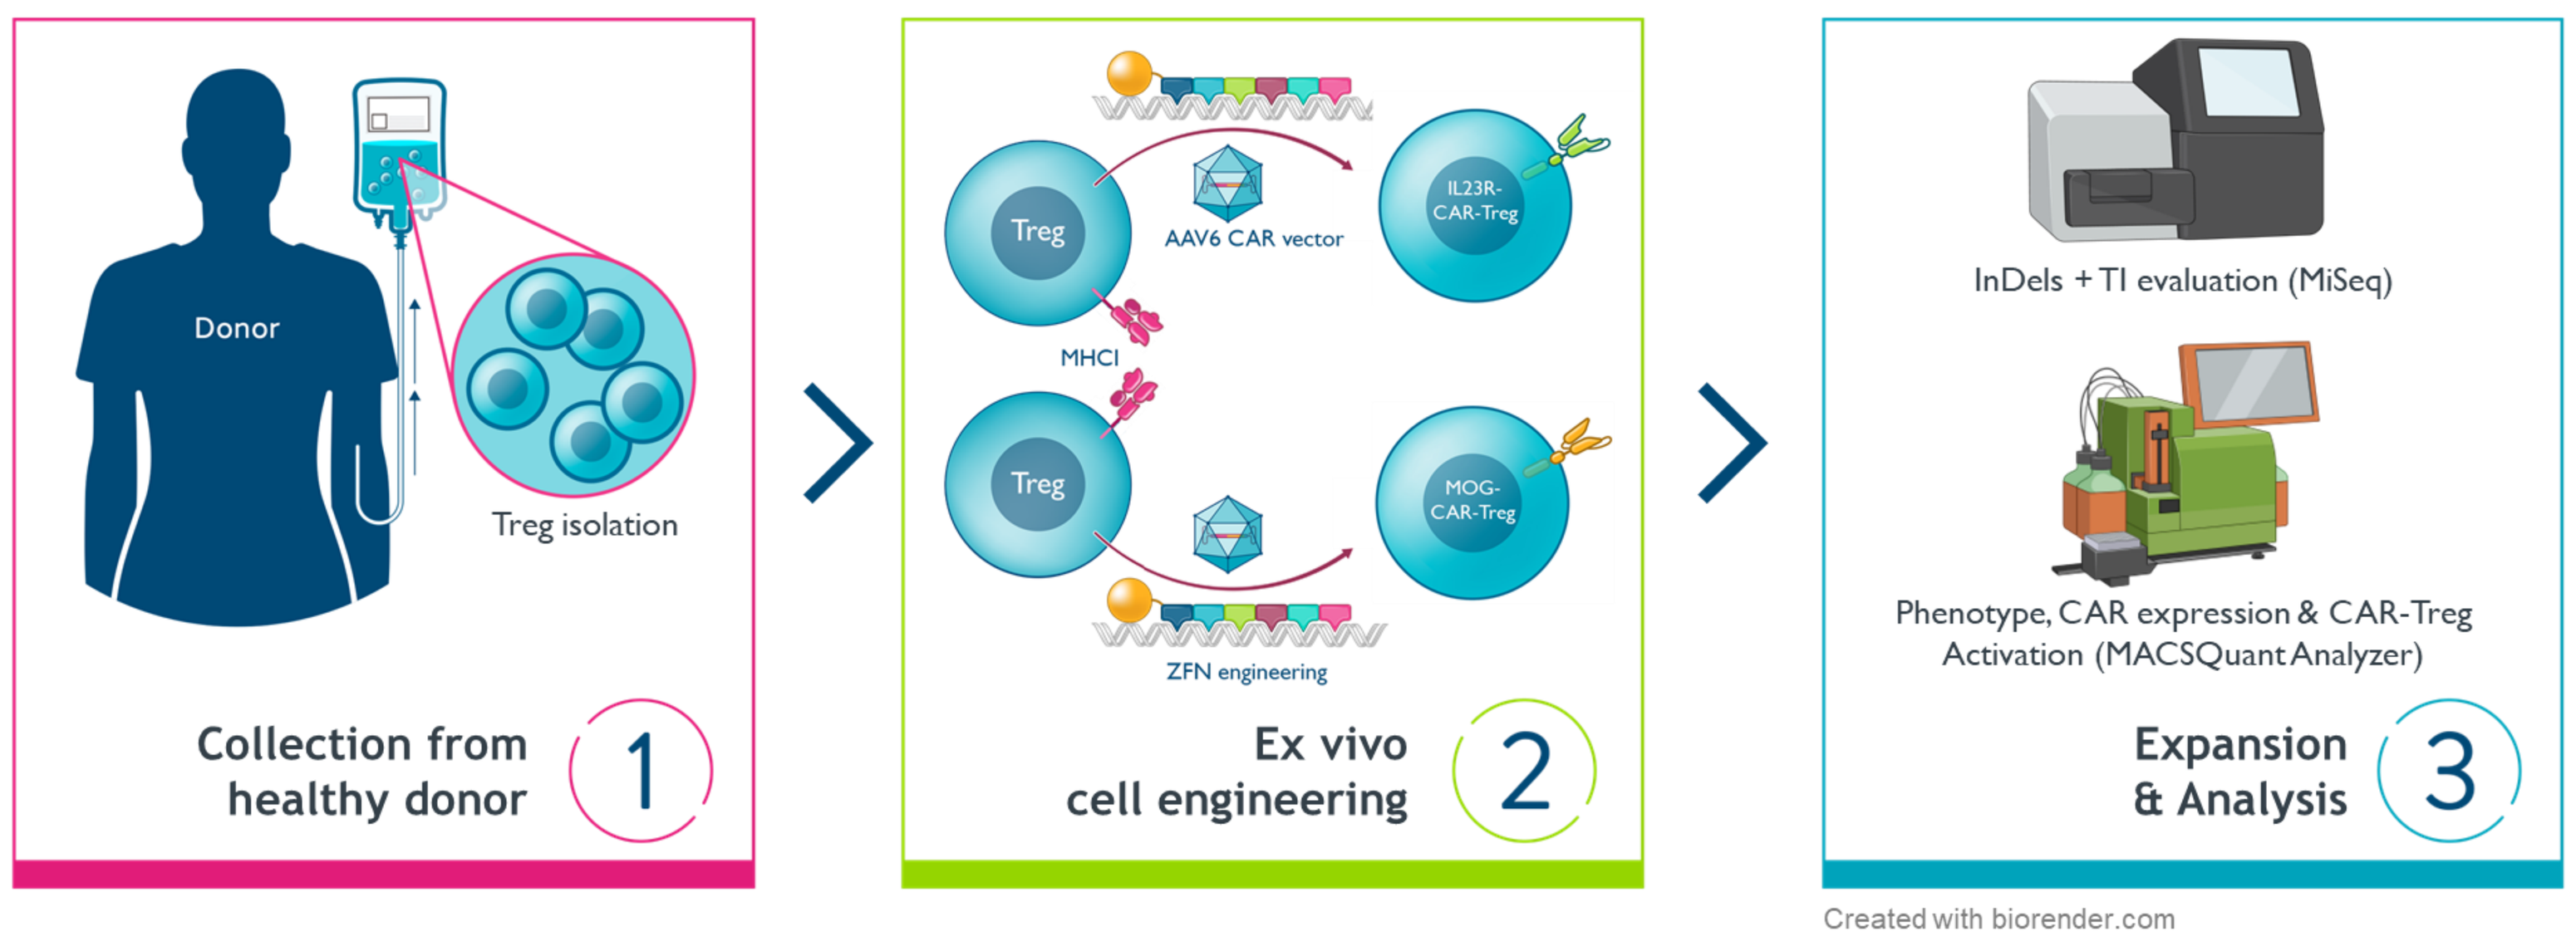
Donor
Treg isolation
Collection from healthy donor
1
Treg
AAV6 CAR vector
IL23R-CAR-Treg
MHCI
Treg
MOG-CAR-Treg
ZFN engineering
Ex vivo cell engineering
2
InDels + TI evaluation (MiSeq)
Phenotype, CAR expression & CAR-Treg Activation (MACSQuant Analyzer)
Expansion & Analysis
3
Created with biorender.com

Schematic showing the experimental design. Tregs are isolated from a healthy donor, followed by ex vivo cell engineering to knock out the expression of MHC-I and to introduce CAR into the *B2M* locus.

InDels, insertions/deletions; TI, targeted integration

AAV6 CAR expression vectors with different promoters

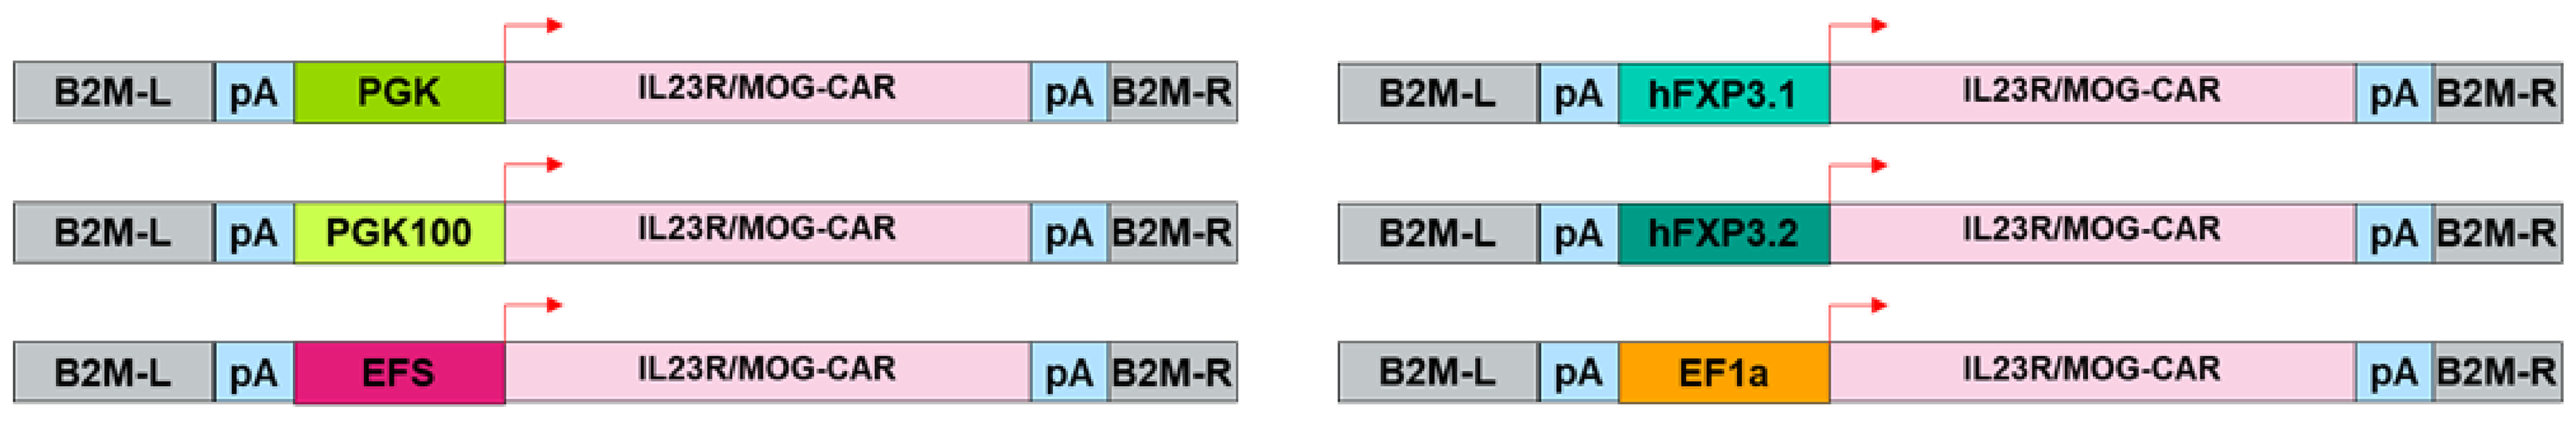
B2M-L pA PGK IL23R/MOG-CAR pA B2M-R
B2M-L pA PGK100 IL23R/MOG-CAR pA B2M-R
B2M-L pA EFS IL23R/MOG-CAR pA B2M-R
B2M-L pA hFXP3.1 IL23R/MOG-CAR pA B2M-R
B2M-L pA hFXP3.2 IL23R/MOG-CAR pA B2M-R
B2M-L pA EF1a IL23R/MOG-CAR pA B2M-R

Tregs were electroporated with *B2M* ZFN mRNA followed by AAV6 transduction (1) using the indicated vectors expressing either IL23R-CAR or MOG-CAR under the control of various promoters (human phosphoglycerate kinase [PGK], short PGK [PGK100], human elongation factor 1 alpha [EF1a], short EF1a [EFS], and short human FOXP3-derived promoters [hFXP3.1 and hFXP3.2]).

Results

B2M ZFN efficiently knocks out MHC-I expression in Tregs

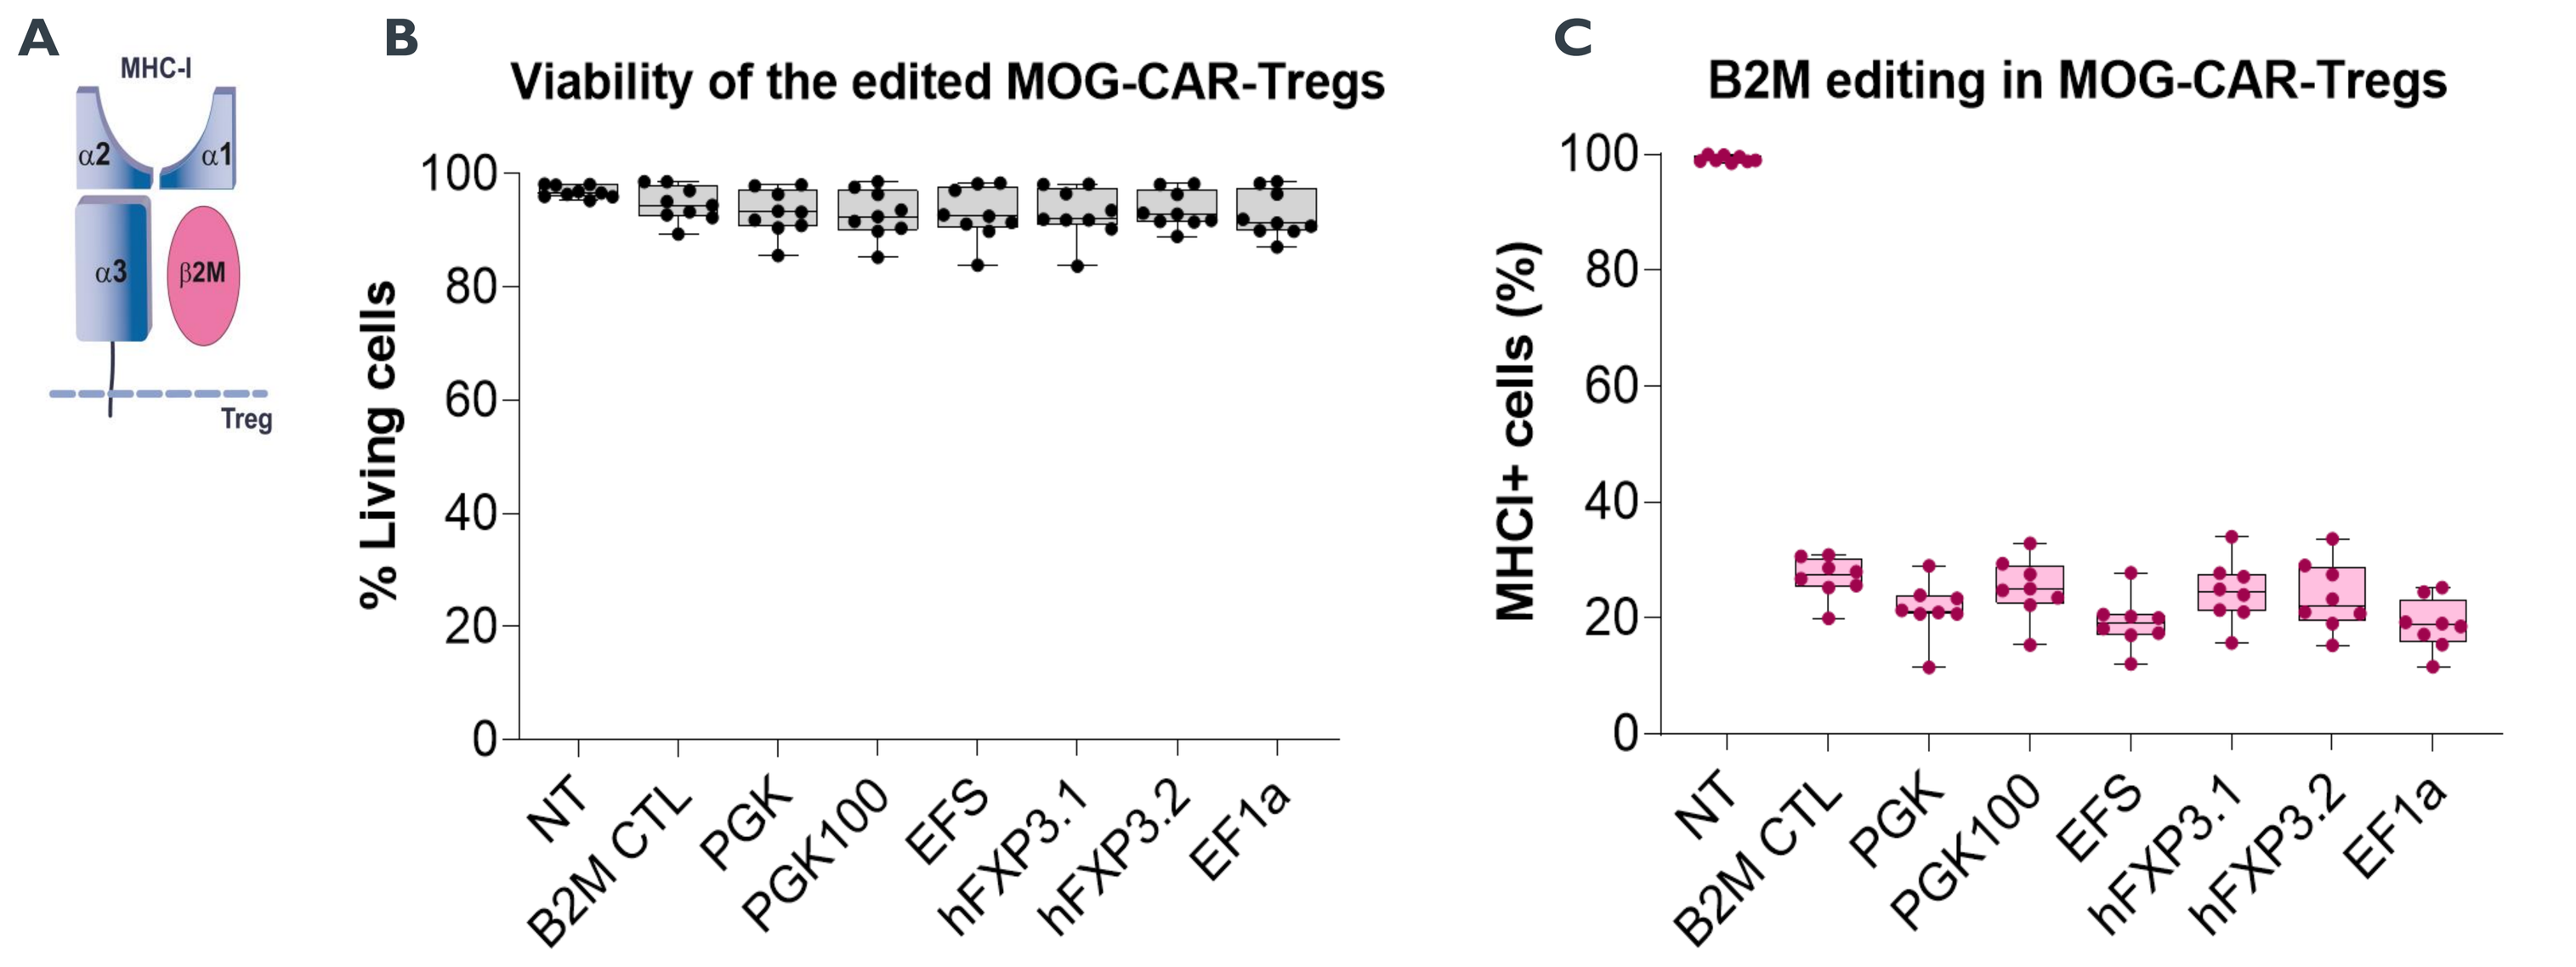
A
MHC-I
α2
α1
α3
β2M
Treg
B
Viability of the edited MOG-CAR-Tregs
% Living cells
NT
B2M CTL
PGK
PGK100
EFS
hFXP3.1
hFXP3.2
EF1a
C
B2M editing in MOG-CAR-Tregs
MHCI+ cells (%)

Knockout of MHC-I expression in Tregs using a ZFN directed against the *B2M* gene locus. (**A**) Schematic representation of the MHC-I molecule composed of alpha 1, 2, and 3, and B2M proteins. (**B**) Viability of edited and non-edited Treg cells with B2M ZFN was assessed by fluorescent activated cell sorter analysis with propidium iodide staining at day 4 post electroporation. (**C**) Knockout of MHC-I expression after editing using B2M ZFN in human Tregs. Results are presented as bars and whiskers (n=8).

Tregs engineered with ZFN/AAV6 technology at *B2M* locus efficiently express IL23R-CAR and MOG-CAR

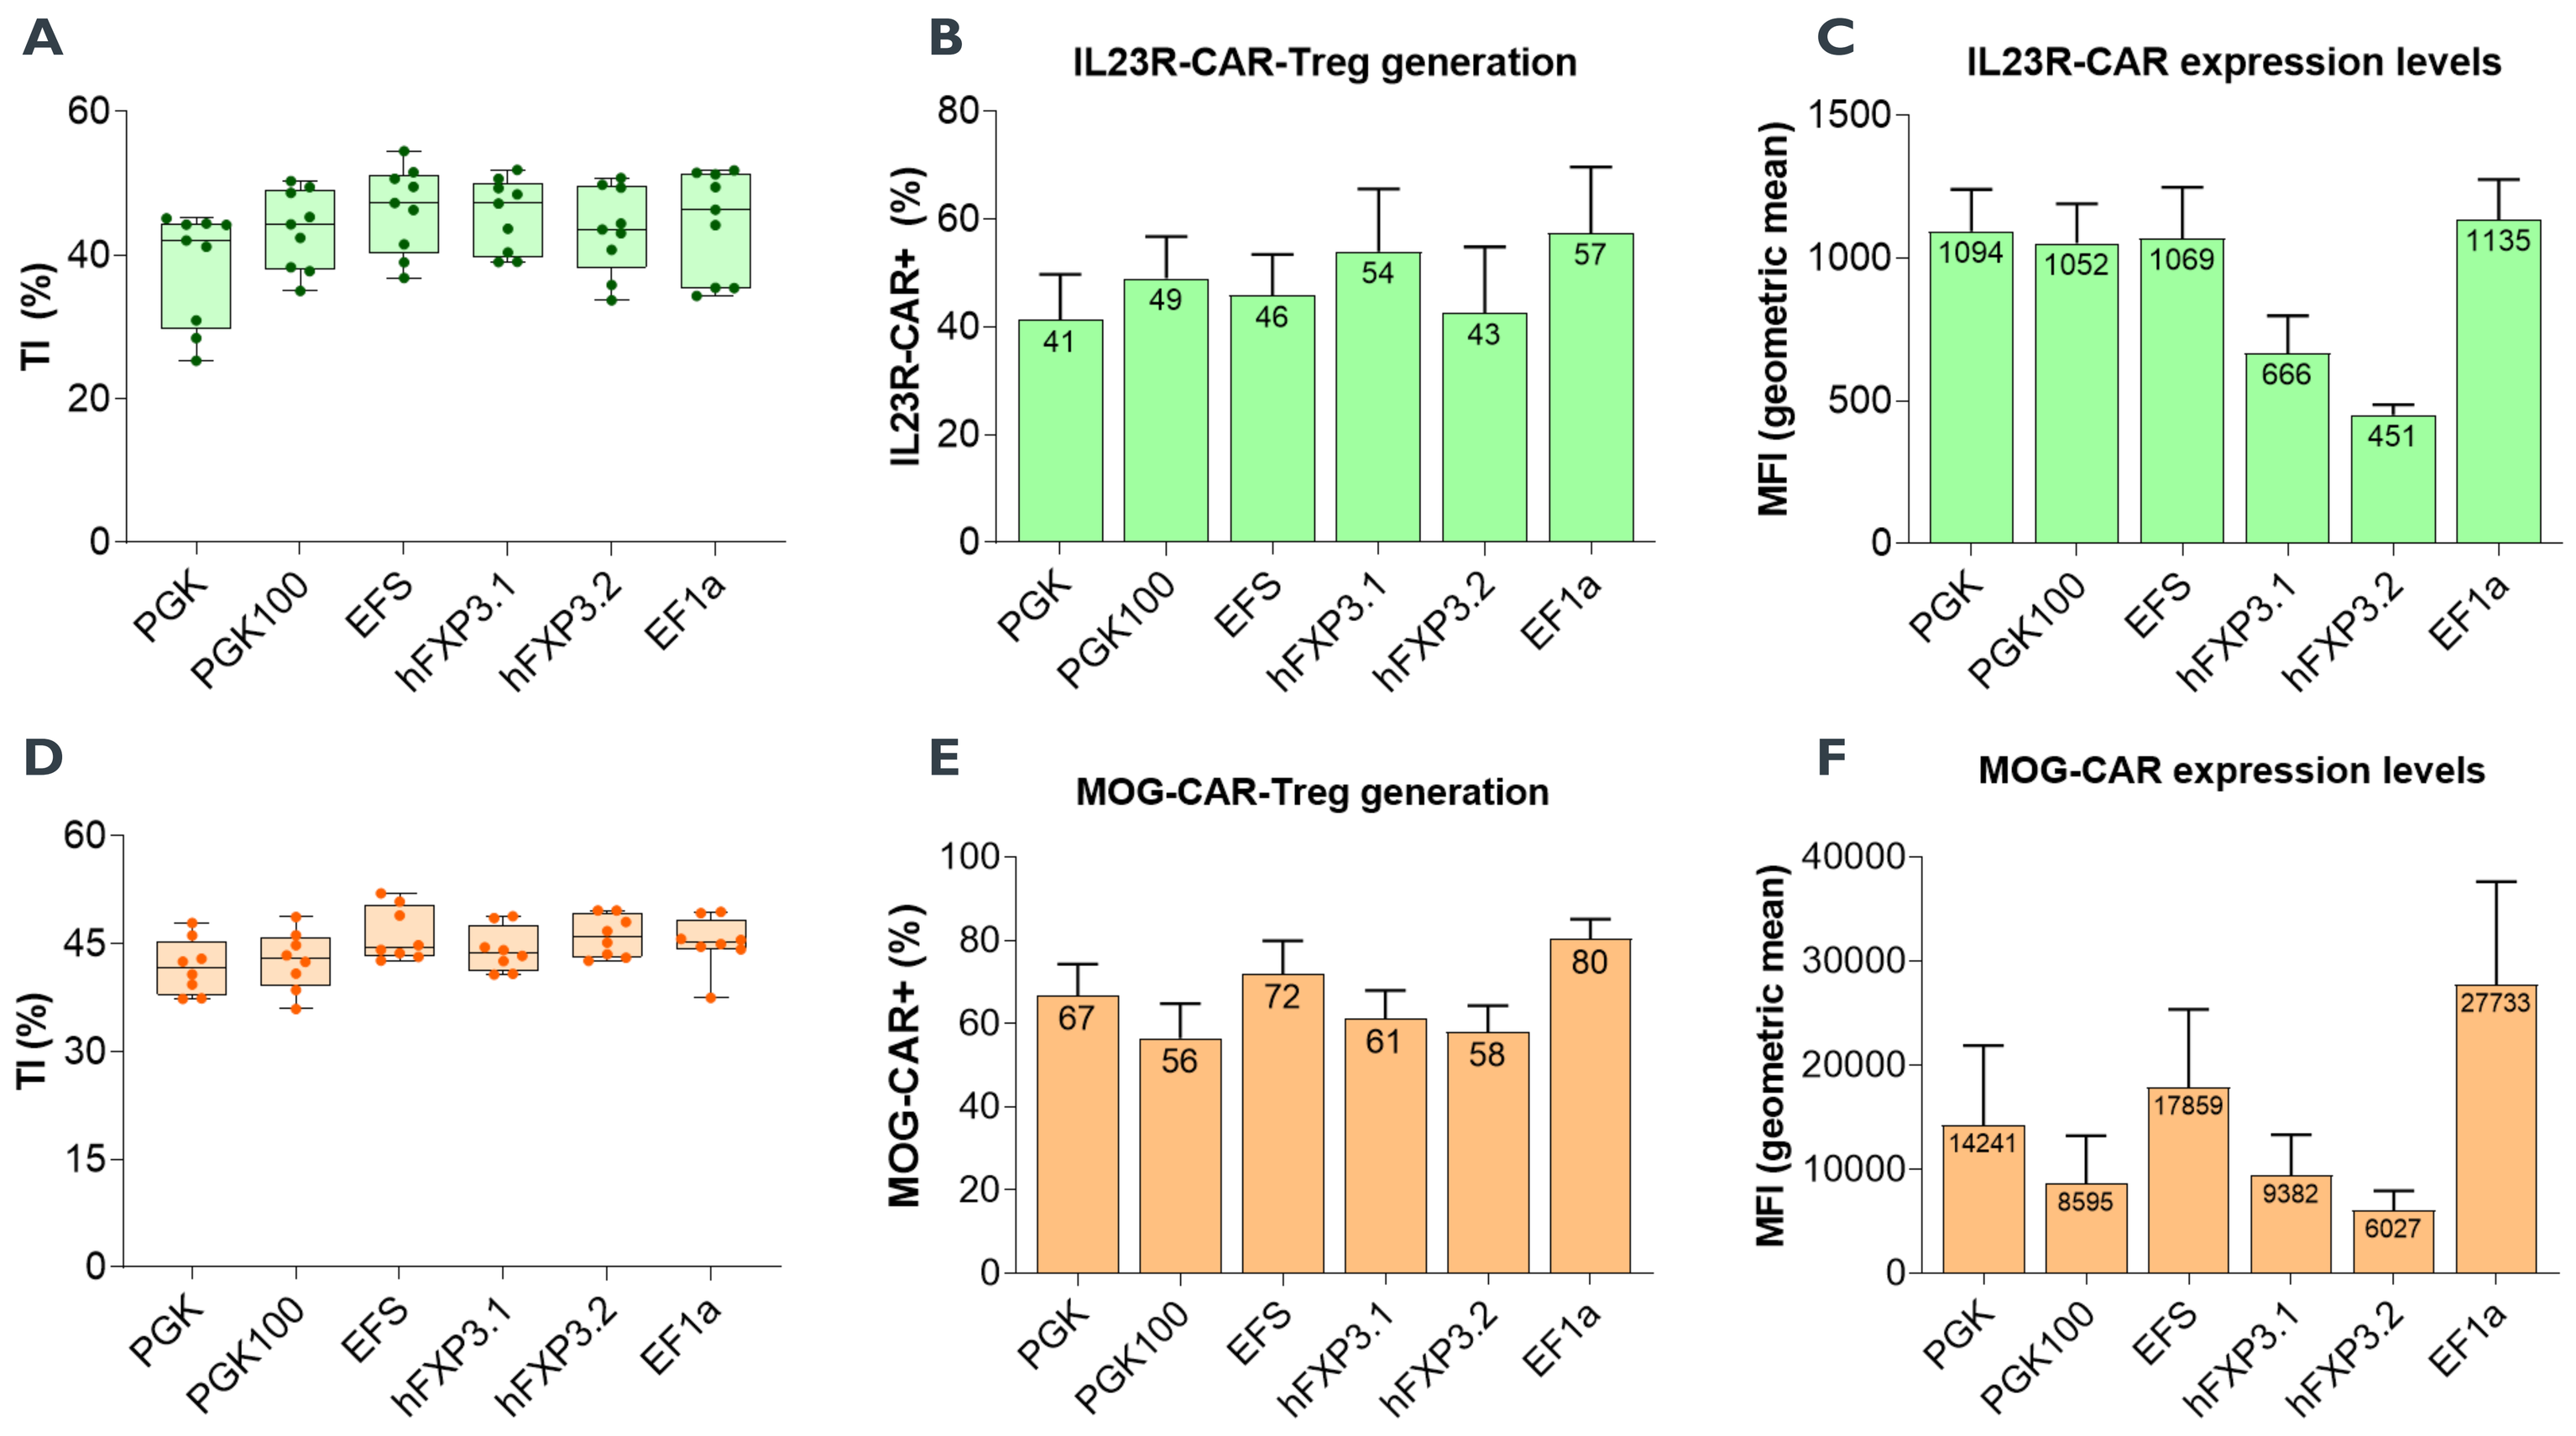
A
TI (%)
B
IL23R-CAR-Treg generation
IL23R-CAR+ (%)
41
49
46
54
43
57
C
IL23R-CAR expression levels
MFI (geometric mean)
1094
1052
1069
666
451
1135
D
TI (%)
E
MOG-CAR-Treg generation
MOG-CAR+ (%)
67
56
72
61
58
80
F
MOG-CAR expression levels
MFI (geometric mean)
14241
8595
17859
9382
6027
27733
PGK
PGK100
EFS
hFXP3.1
hFXP3.2
EF1a

Screening of different promoters to modulate IL23R-CAR and MOG-CAR expression in Tregs. (**A**, **D**) Targeted integration (TI) of IL23R-CAR and MOG-CAR sequences into the *B2M* locus was quantified by next-generation sequencing (MiSeq). (**B**, **E**) The impact of the different promoters on CAR-Treg generation efficiency was assessed by measuring the percentage of CAR+/MHC-I(-) cells using flow cytometry. (**C**, **F**) IL23R-CAR and MOG-CAR expression levels on the Treg cell surface were determined by measuring the mean fluorescence intensity (MFI) of the CARs.

Data are from 3 independent experiments (n=9 and 8 Treg donors, respectively).

Highly efficient CAR-mediated activation of CAR-Tregs generated using ZFN/AAV6 technology

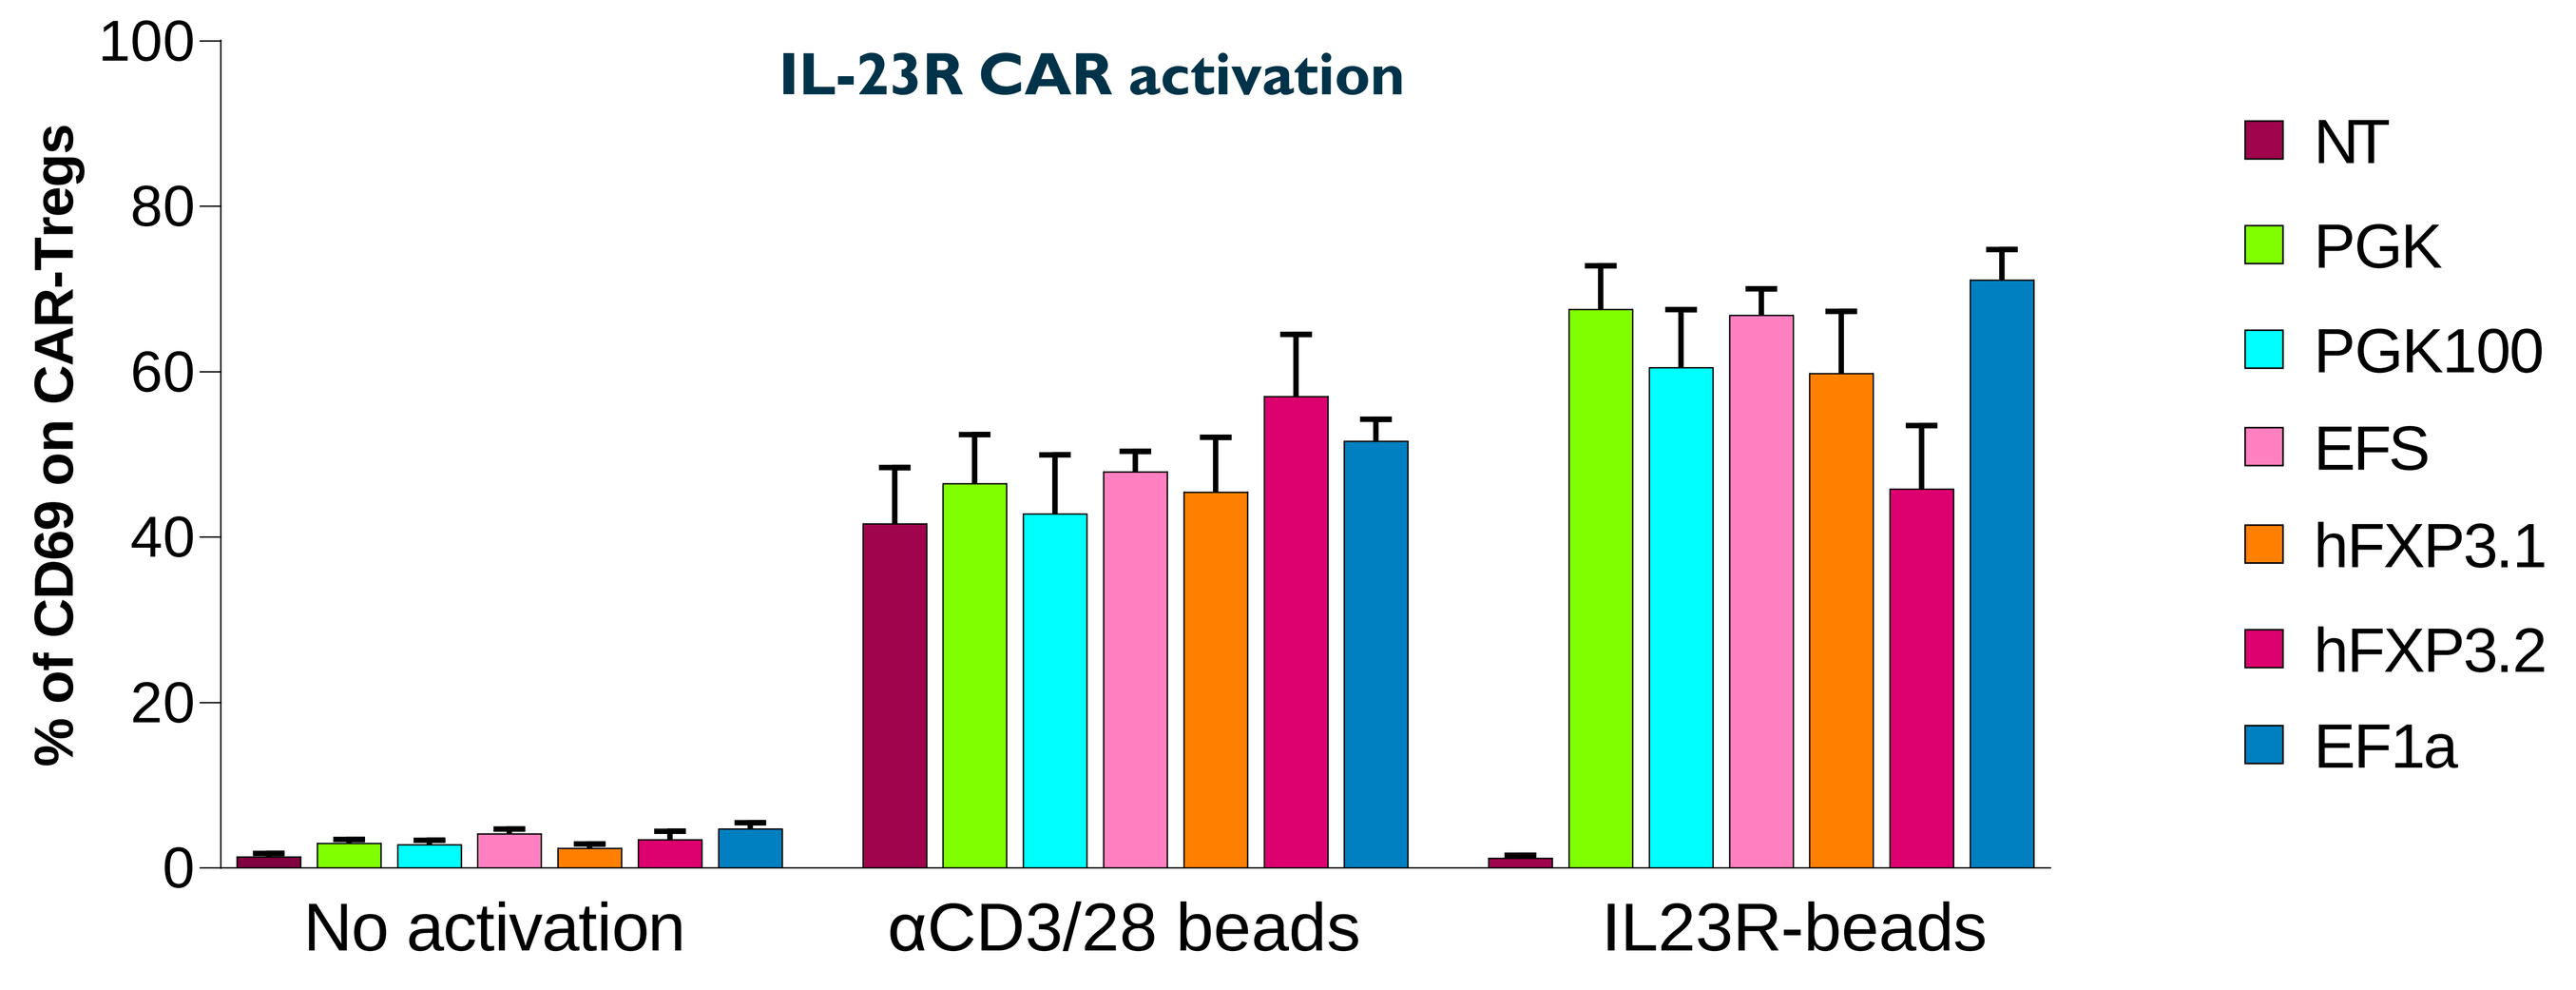
IL-23R CAR activation
% of CD69 on CAR-Tregs
No activation
αCD3/28 beads
IL23R-beads
NT
PGK
PGK100
EFS
hFXP3.1
hFXP3.2
EF1a

The CAR-mediated activation assay monitors the expression of the CD69 activation marker at the cell surface of CAR-Tregs. Activation of CAR-Tregs was measured 24 hours after incubation either with anti-CD3/CD28 beads to stimulate the T-cell receptor (positive control) or with IL23R-coated beads to stimulate the CAR (n=4).

The ranking from optimal to minimal CAR-mediated activation is as follows: EF1a=PGK=EFS>PGK100=hFXP3.1>hFXP3.2.

Conclusions

- Our results show high MHC-I knockout efficiency ranging between 70%-90% with no impact on Treg viability.
- The targeted integration efficiency was similar overall (40% to 50%) between all the AAV6-CAR donor DNA regardless of the expression cassette used.
- Tregs engineered with ZFN/AAV6 technology at the *B2M* locus efficiently express IL23R-CAR and MOG-CAR.
- CAR-mediated activation was highly efficient, especially for CAR-AAV6 harboring the EF1a or PGK promoters.
- Overall, our results provide a robust strategy for genome engineering in Tregs using ZFN in combination with AAV6-CAR donor DNA.

References

1. Wang J. *et al.* (2015) Nat. Biotechnol. 33(12):1256-1263.

Author Disclosures

All authors are or were employees of Sangamo Therapeutics

Presented at the ESGCT 29th Annual Meeting, 2022