Engineering of Factor IX-secreting B cells using ZF-Nuclease / AAV6 editing technology in a GMP-compatible medium

Poster #P130

Marion David, Davide Monteferrario, Gaëlle Saviane, Caroline Jeanneau, Irène Marchetti, Coralie Dupont, Céline Dumont, Jason Fontenot, Maurus de la Rosa,* and David Fenard*
*Co-senior authors

Sangamo Therapeutics France, Allée de la Nertière, Les Cardoulines HT1, 06560 VALBONNE, France

Introduction

- After antigen engagement, B cells, key effectors of adaptative immune response, differentiate into plasmablast/plasma cells.
- The long-lived plasma cells can survive for decades in the bone marrow and secrete large quantities of antibodies.
- Recent developments around precise editing technologies open the possibility of engineering plasmablast/plasma cells to act as autologous cell factories, capable of delivering sustained and high doses of therapeutic proteins.
- In this study, using a serum-free Good Manufacturing Practice (GMP)-compatible medium, we developed optimal culture conditions for B cell differentiation into plasmablast/plasma cells in vitro and site-specific transgene insertion into distinct loci. For that, B cell engineering was performed by combining electroporation of zinc finger nuclease (ZFN) mRNA with adeno-associated virus type 6 (AAV6) donor DNA template delivery.
- As a proof of concept, B cells were engineered for the expression of the human factor IX (FIX)-Padua. FIX deficiency is the leading cause of hemophilia B, a genetic disease characterized by blood clotting defects.

Methods

Autologous engineered B-cell therapy concept

Patient-derived B cells can be engineered to express a therapeutic protein, expanded and differentiated into plasmablasts/plasma cells ex vivo.

Upon re-infusion, cells are expected to engraft and serve as an in vivo source for sustained circulation of the therapeutic factor.

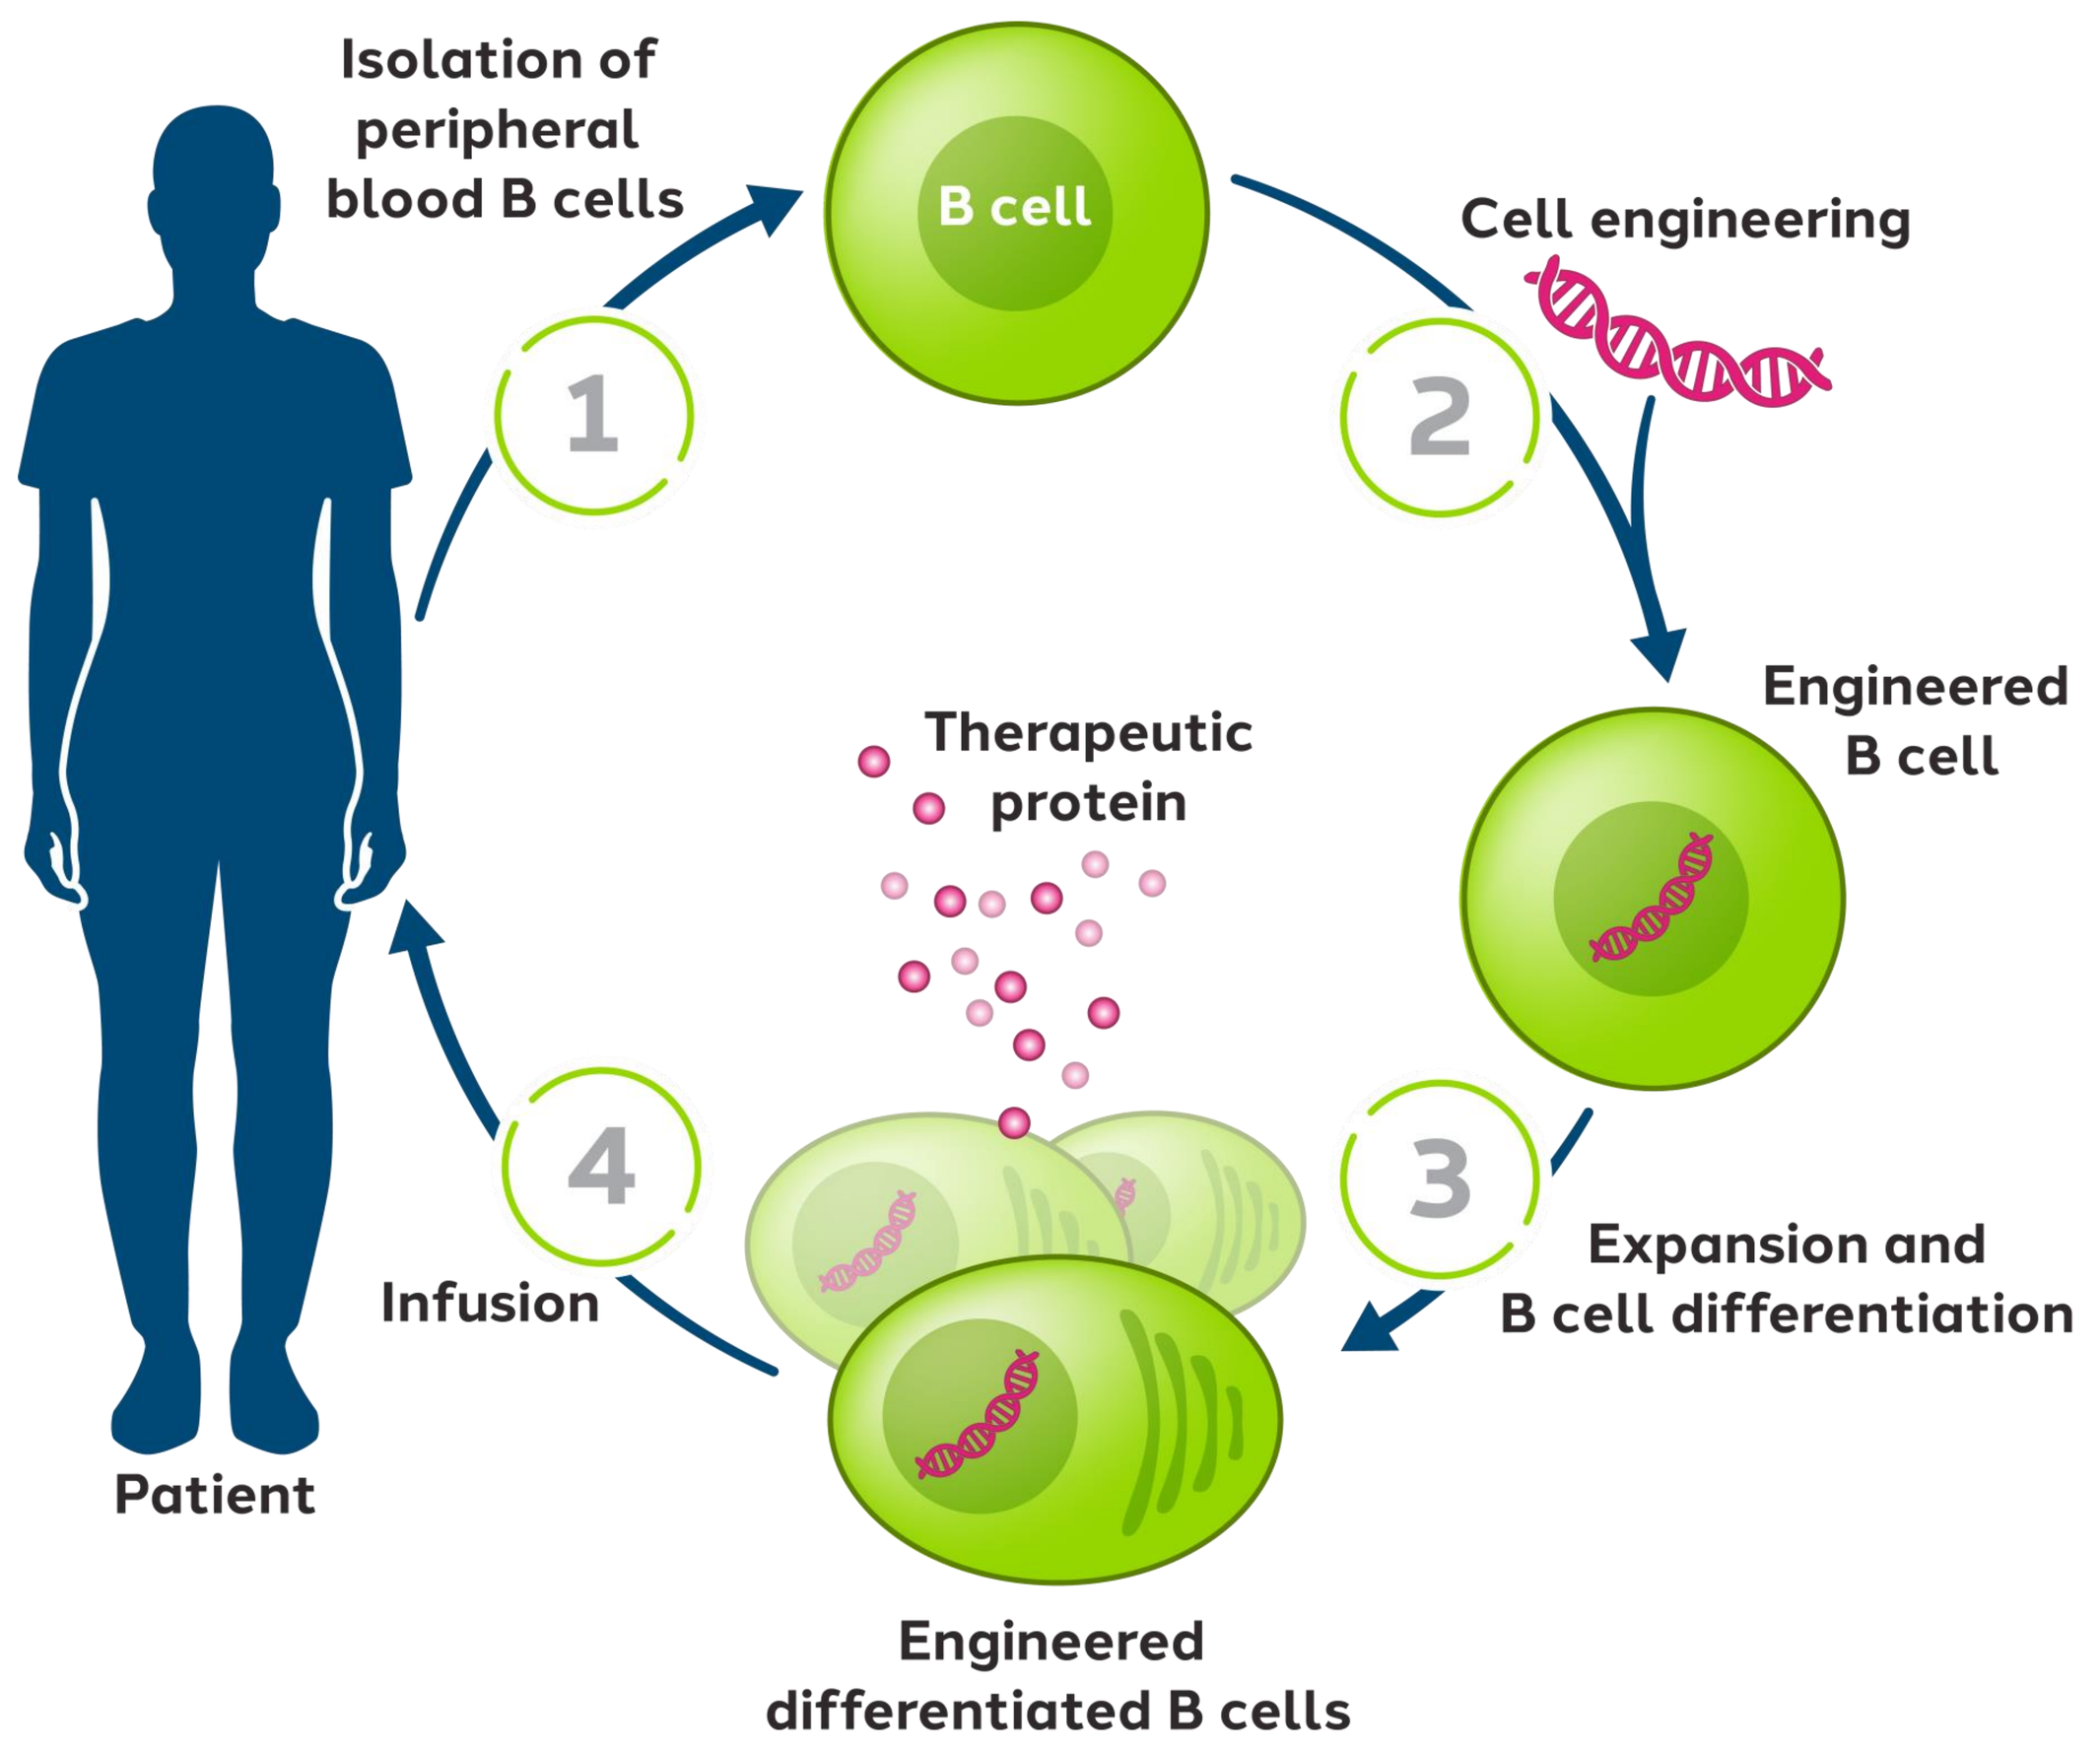
Isolation of peripheral blood B cells
B cell
Cell engineering
Engineered B cell
Therapeutic protein
Expansion and B cell differentiation
Engineered differentiated B cells
Infusion
Patient
1
2
3
4

B-cell engineering using ZFN/AAV6 technology

The ZFN mRNA electroporation generates a double-strand break on a targeted gene locus.

Next, the transduction with an AAV6 vector, harboring the transgene flanked by genomic sequences homologous to the target site, triggers the insertion of the transgene into the targeted locus by homologous recombination.[1]

The target genes are a safe harbor (*AAVS1*) and silent genes (*TRAC*, *CCR5*).

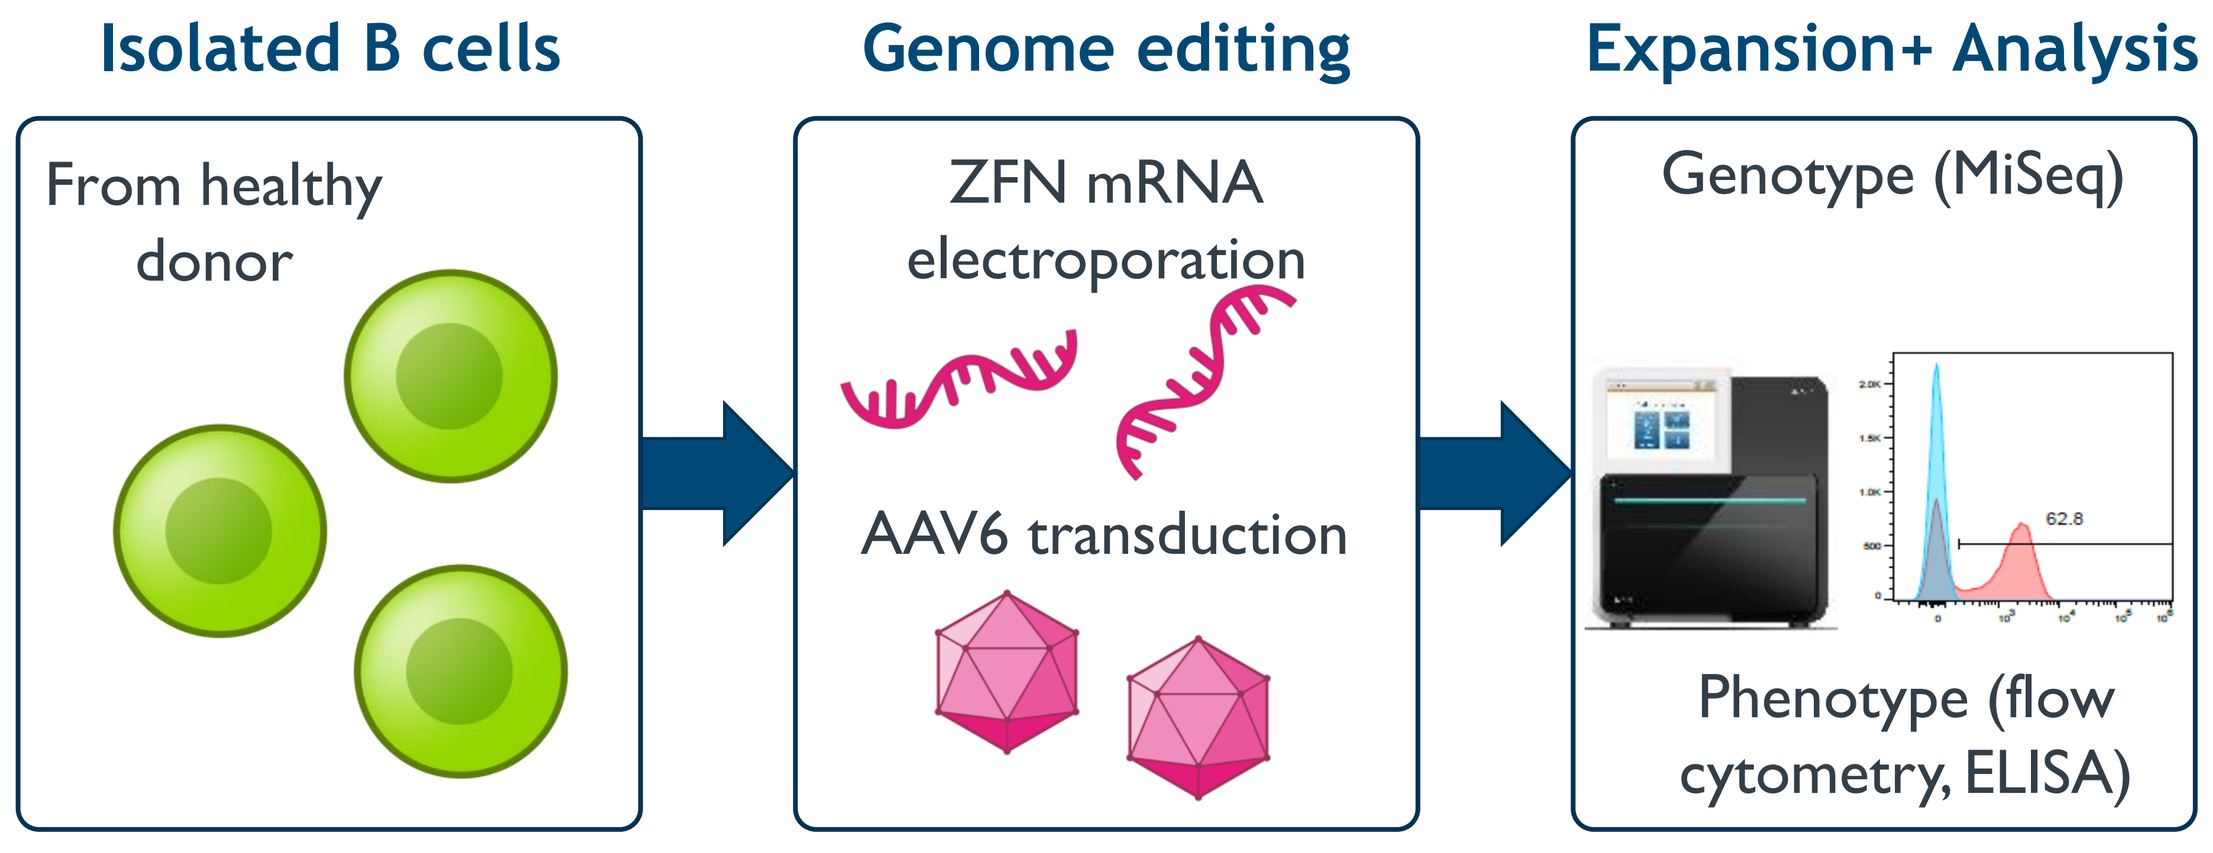
Isolated B cells
Genome editing
Expansion+ Analysis
From healthy donor
ZFN mRNA electroporation
AAV6 transduction
Genotype (MiSeq)
Phenotype (flow cytometry, ELISA)

ELISA, enzyme-linked immunosorbent assay.

Acknowledgments

We thank Sangamo's collaborators, in particular the animal care facility.

Author Disclosures

All authors are or were employees of Sangamo Therapeutics

Results

Efficient B cell differentiation was achieved in vitro using a GMP-compatible serum-free medium

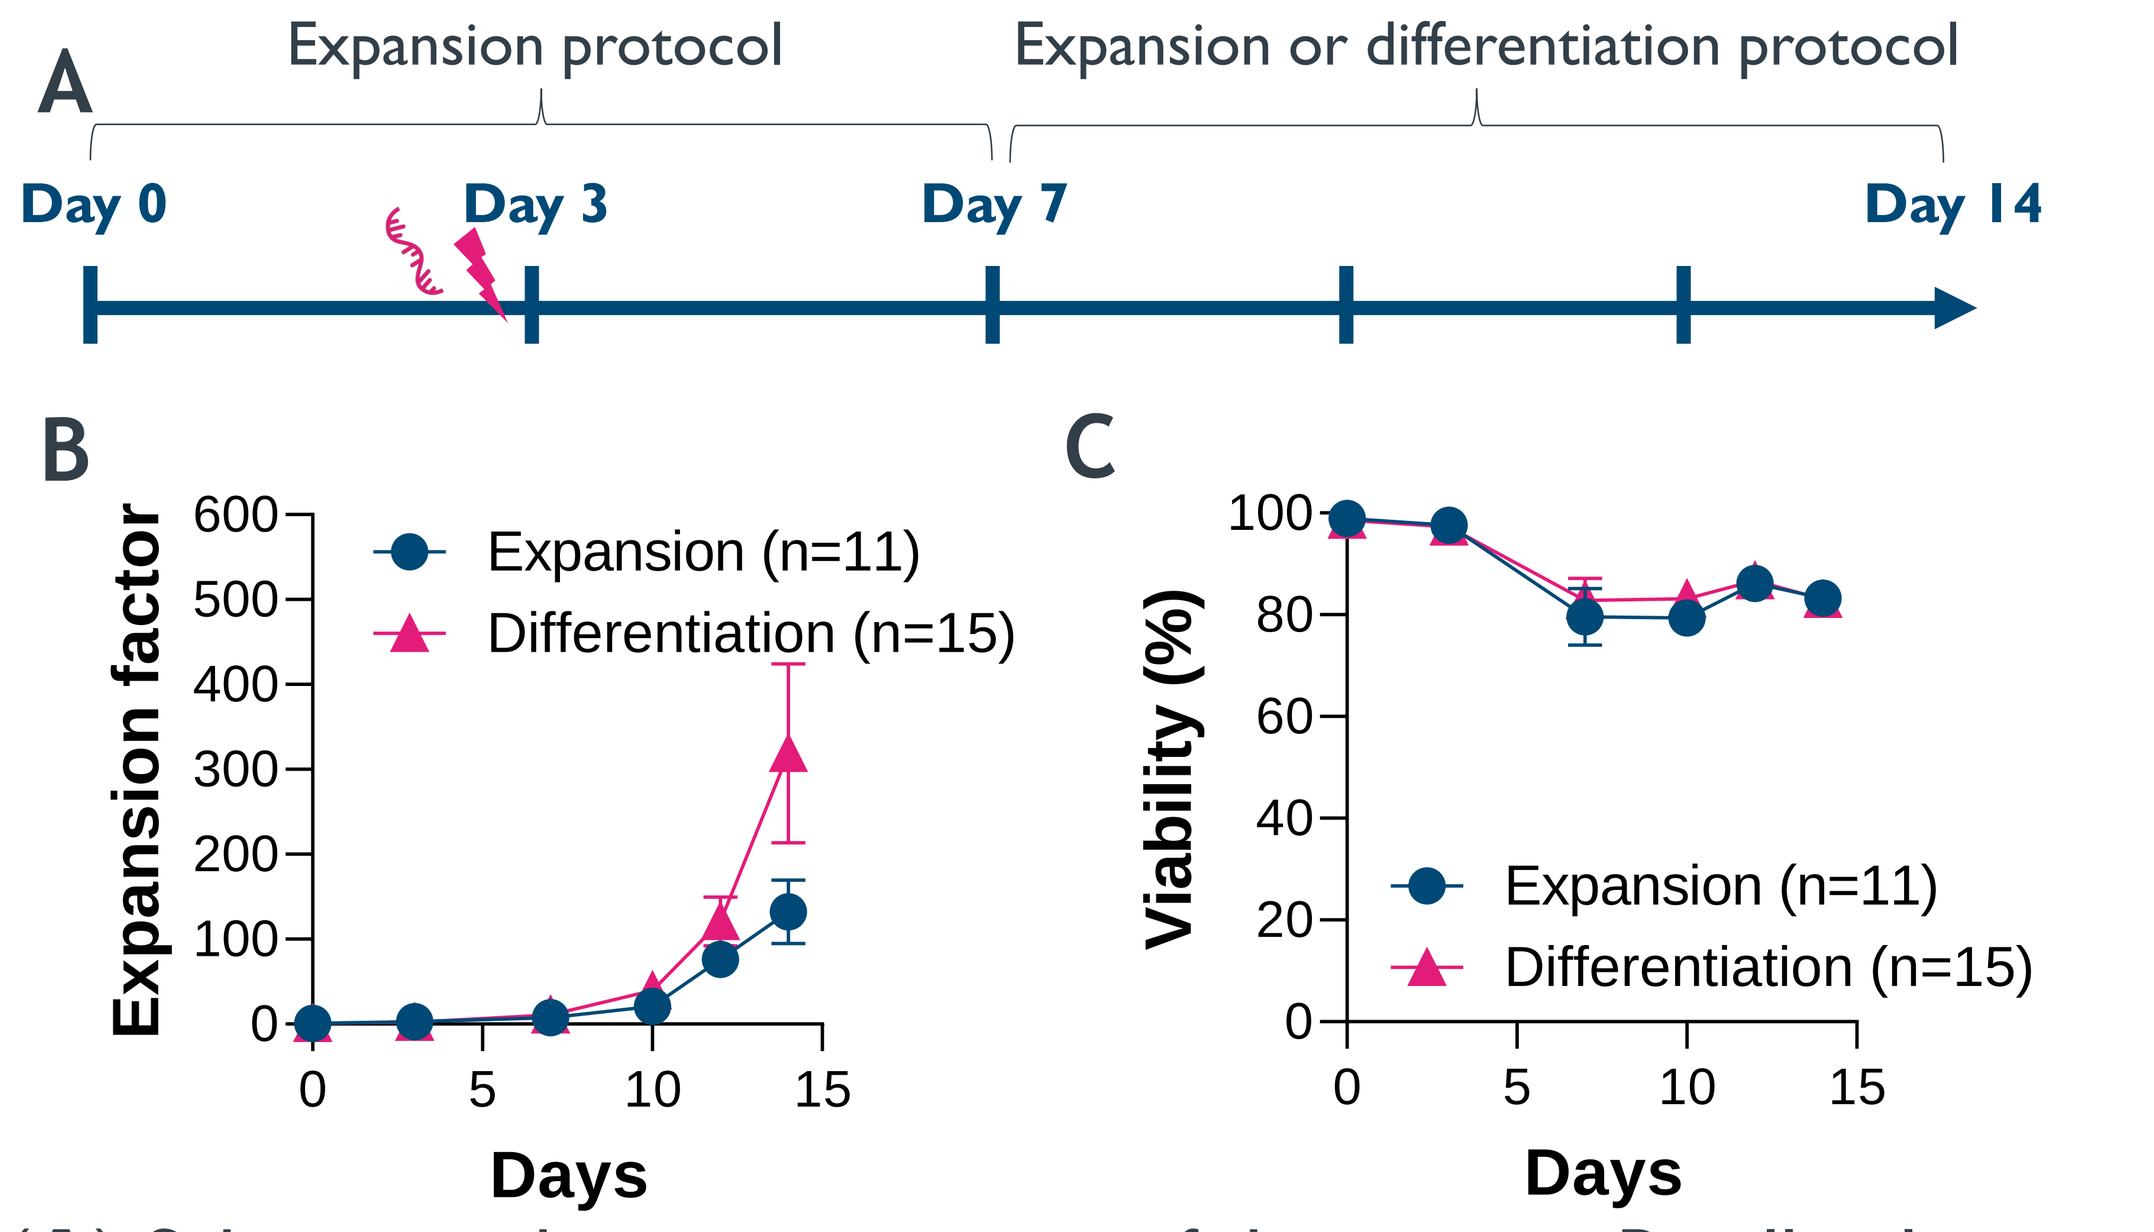
A
Expansion protocol
Expansion or differentiation protocol
Day 0
Day 3
Day 7
Day 14
B
Expansion factor
Expansion (n=11)
Differentiation (n=15)
Days
C
Viability (%)
Expansion (n=11)
Differentiation (n=15)
Days

(**A**) Schematic showing overview of the in vitro B cell culture protocol. The first week, B cells were expanded in presence of sCD40L, ODN, and IL-2, IL-10, and IL-15 cocktail.[2] The second week, B cells were continuously expanded or differentiated. Every 2-4 days, cell expansion fold and cell viability were monitored (**B** and **C**).

IL, interleukin; ODN, oligodeoxynucleotide; sCD40L, soluble CD40 ligand.

The B cell culture protocol promotes differentiation mainly in plasmablasts and to a lesser extent in plasma cells

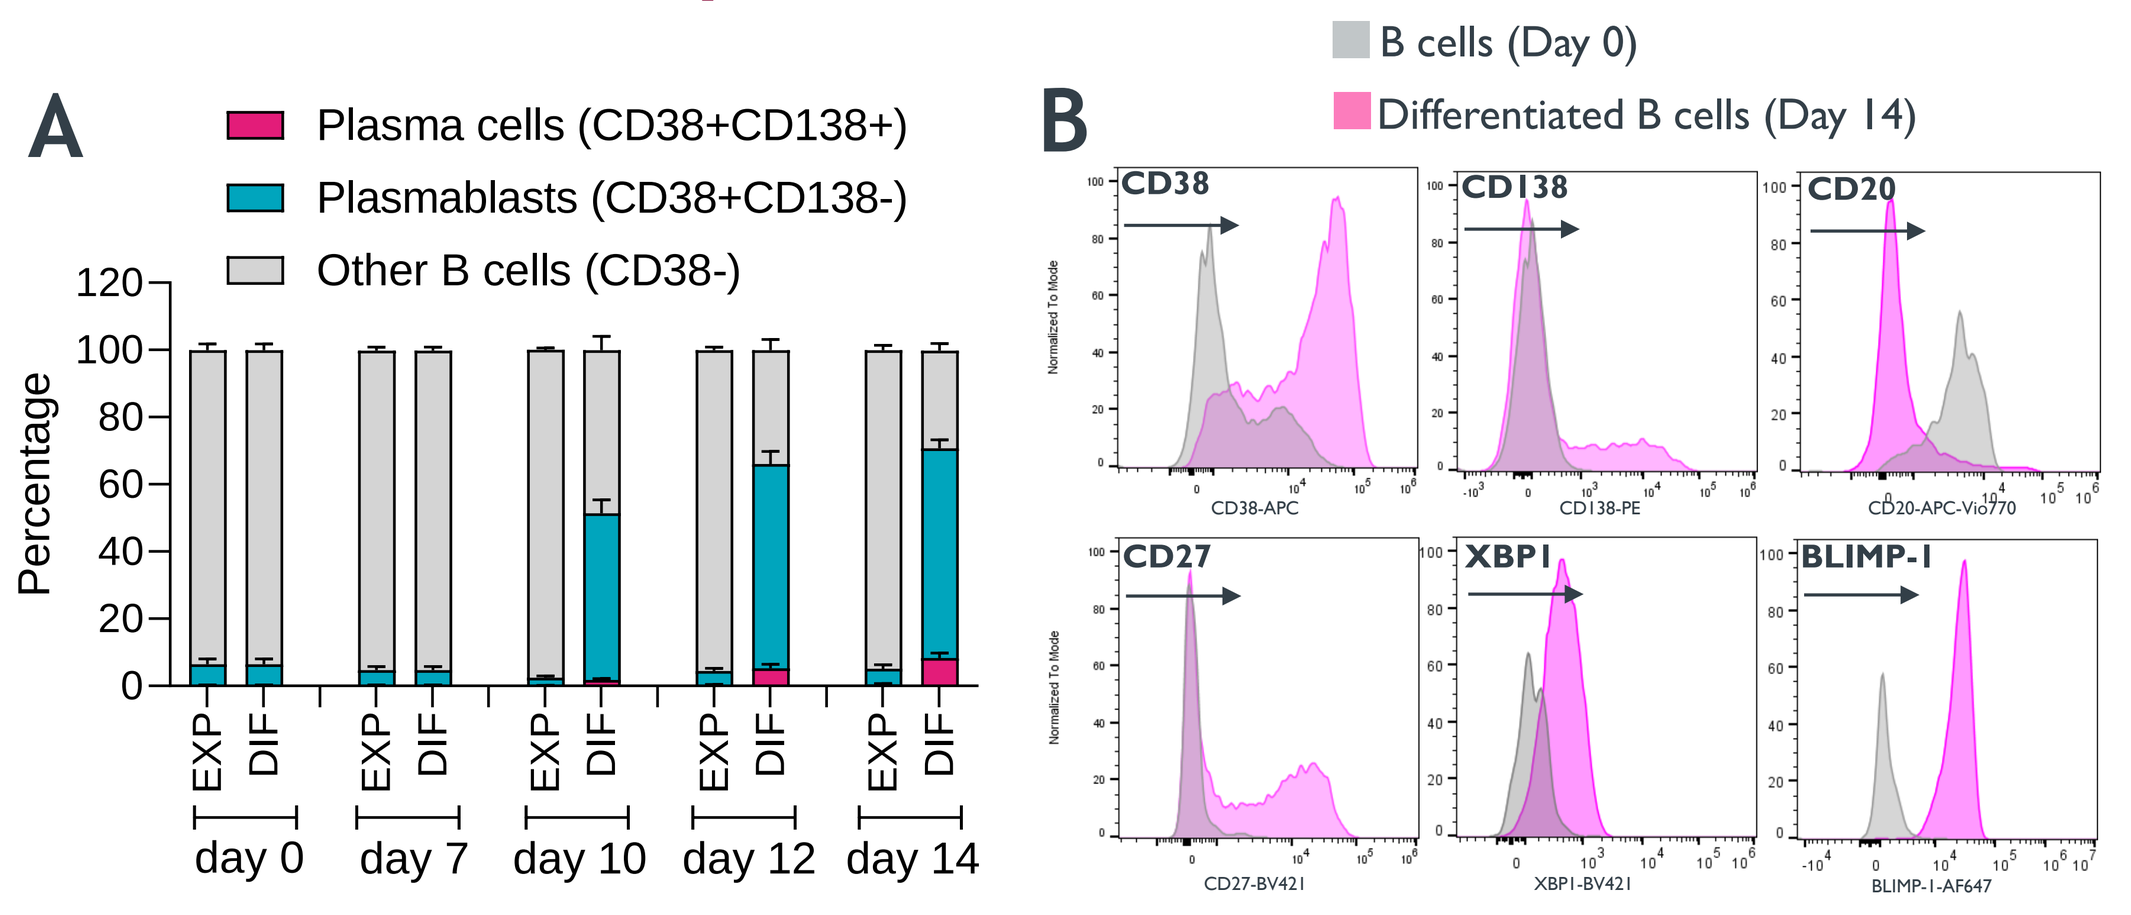
A
Plasma cells (CD38+CD138+)
Plasmablasts (CD38+CD138-)
Other B cells (CD38-)
Percentage
EXP DIF
day 0
day 7
day 10
day 12
day 14
B
B cells (Day 0)
Differentiated B cells (Day 14)
CD38
CD138
CD20
CD27
XBP1
BLIMP-1

(**A**) Monitoring of plasmablast (CD38+CD138-) and plasma cell (CD38+CD138+) production in expansion (EXP, n=11) versus differentiation (DIF, n=15) protocol. (**B**) Cells were stained for multiple B cell differentiation markers (CD38++, CD138+, CD20-, CD27++, XBP1+, and BLIMP-1+). Gray and pink curves correspond respectively to B cells obtained at day 0 or day 14 in DIF protocol.

The use of ZFN/AAV6 editing technology allows highly efficient engineering of B cells at multiple genomic loci

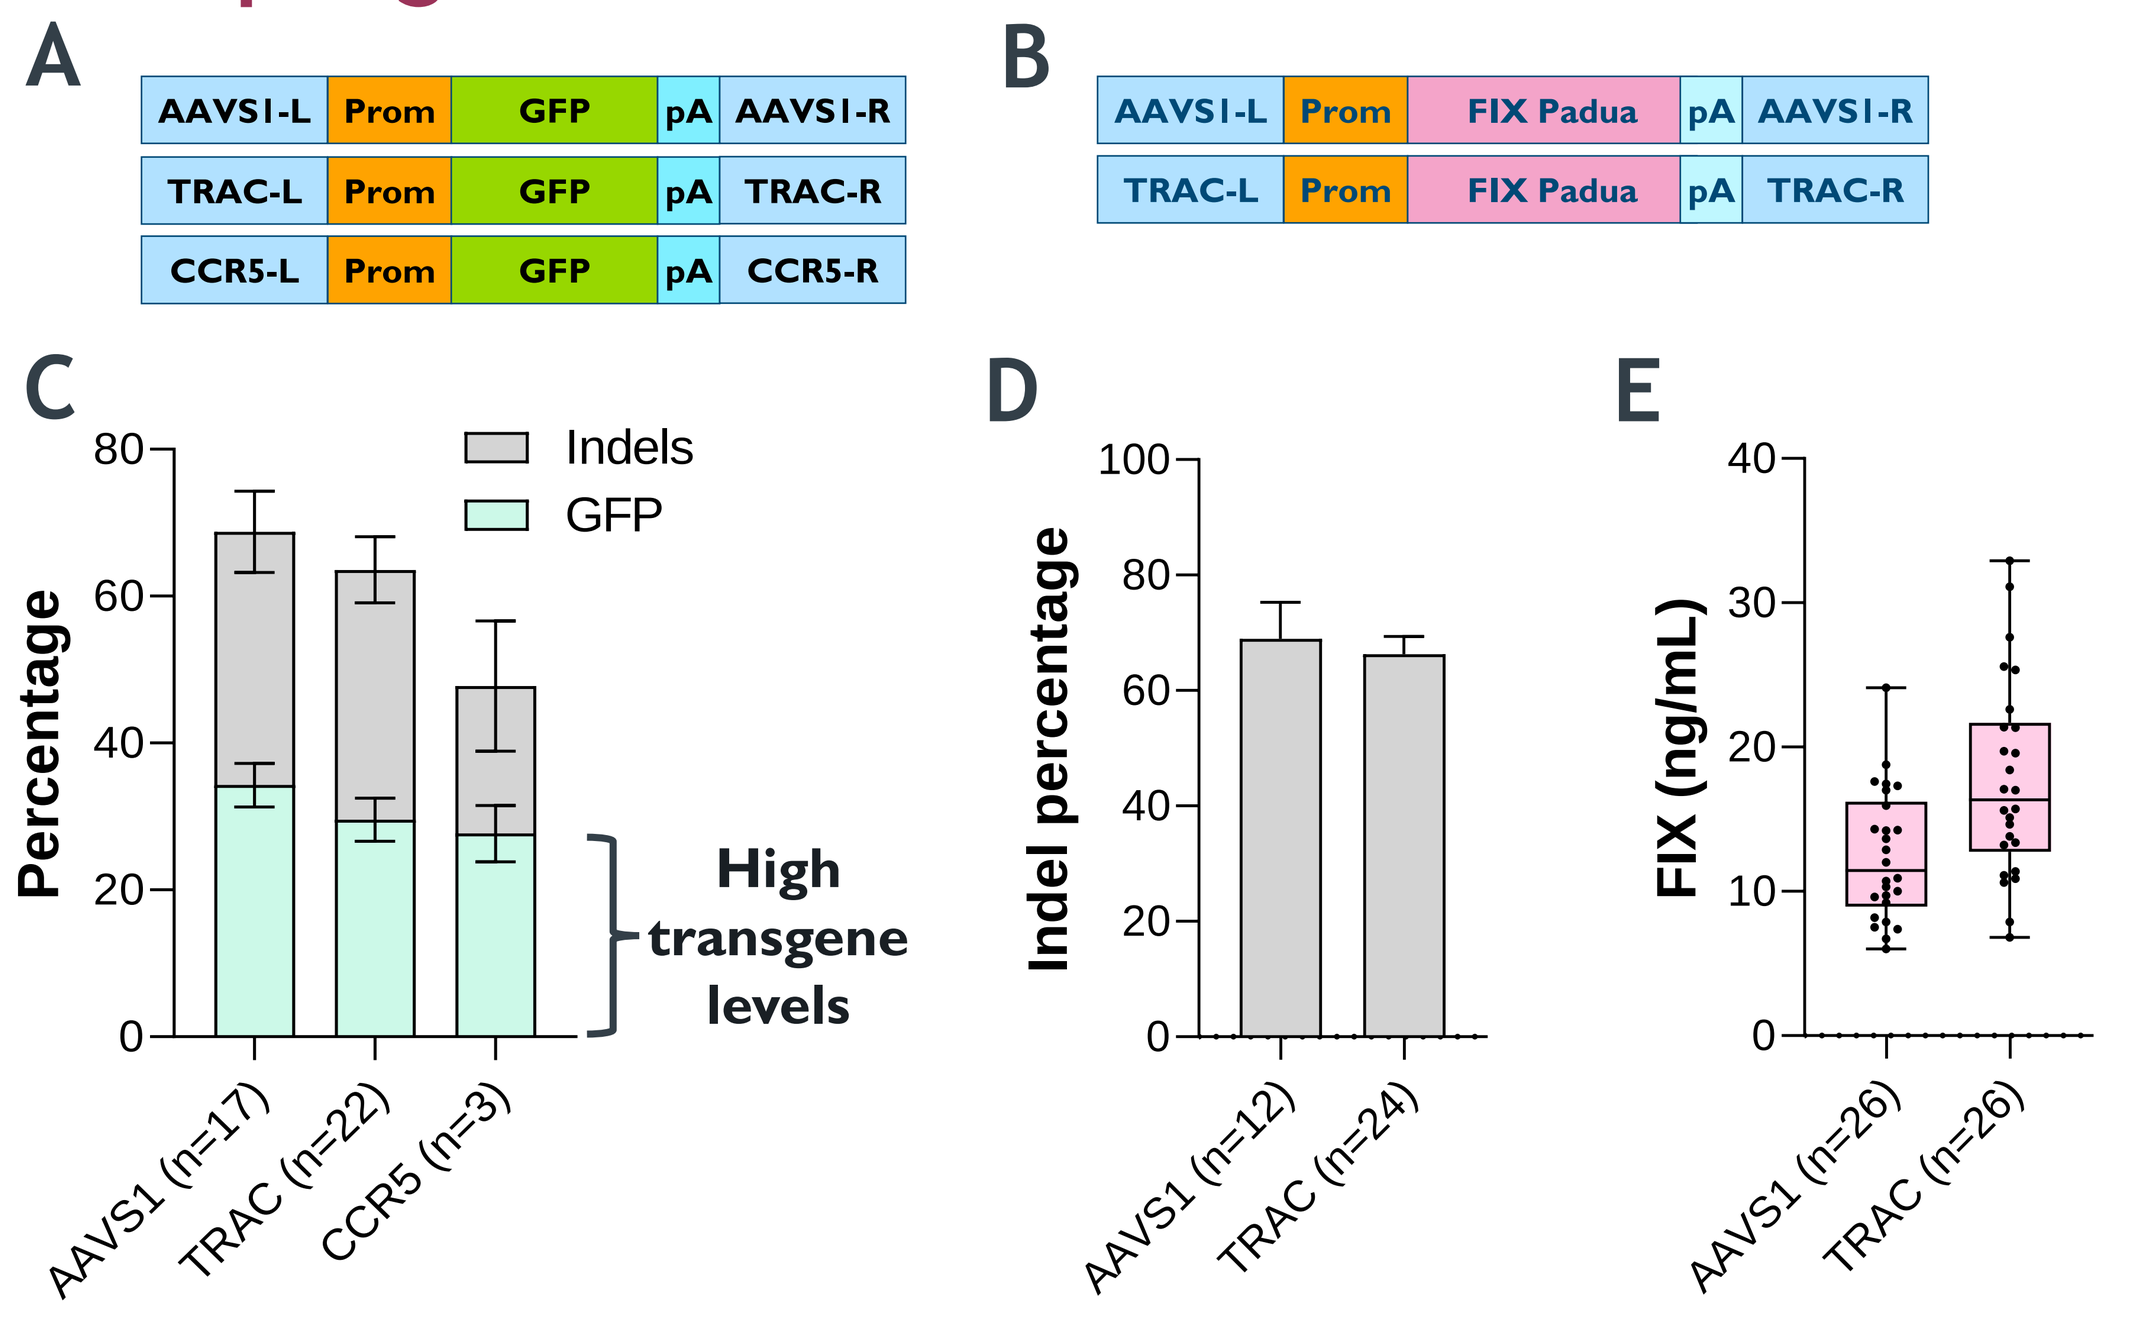
A
AAVS1-L Prom GFP pA AAVS1-R
TRAC-L Prom GFP pA TRAC-R
CCR5-L Prom GFP pA CCR5-R
B
AAVS1-L Prom FIX Padua pA AAVS1-R
TRAC-L Prom FIX Padua pA TRAC-R
C
Percentage
Indels
GFP
High transgene levels
AAVS1 (n=17)
TRAC (n=22)
CCR5 (n=3)
D
Indel percentage
AAVS1 (n=12)
TRAC (n=24)
E
FIX (ng/mL)
AAVS1 (n=26)
TRAC (n=26)

Cells were electroporated with locus-specific ZFN mRNA followed by AAV6 transduction using the indicated vectors expressing green fluorescent protein (GFP) (**A**) or human FIX-Padua (**B**). Cells were analyzed 4 days post editing to quantify indels associated with GFP-engineered B cells (**C**) or FIX-engineered B cells (**D**). (**E**) Supernatants were collected 1 day post seeding, and total FIX antigen was quantified by ELISA for FIX-engineered B cells.

An in vivo NSG/Tm mouse model was used for optimal engraftment of differentiated/engineered human B cells

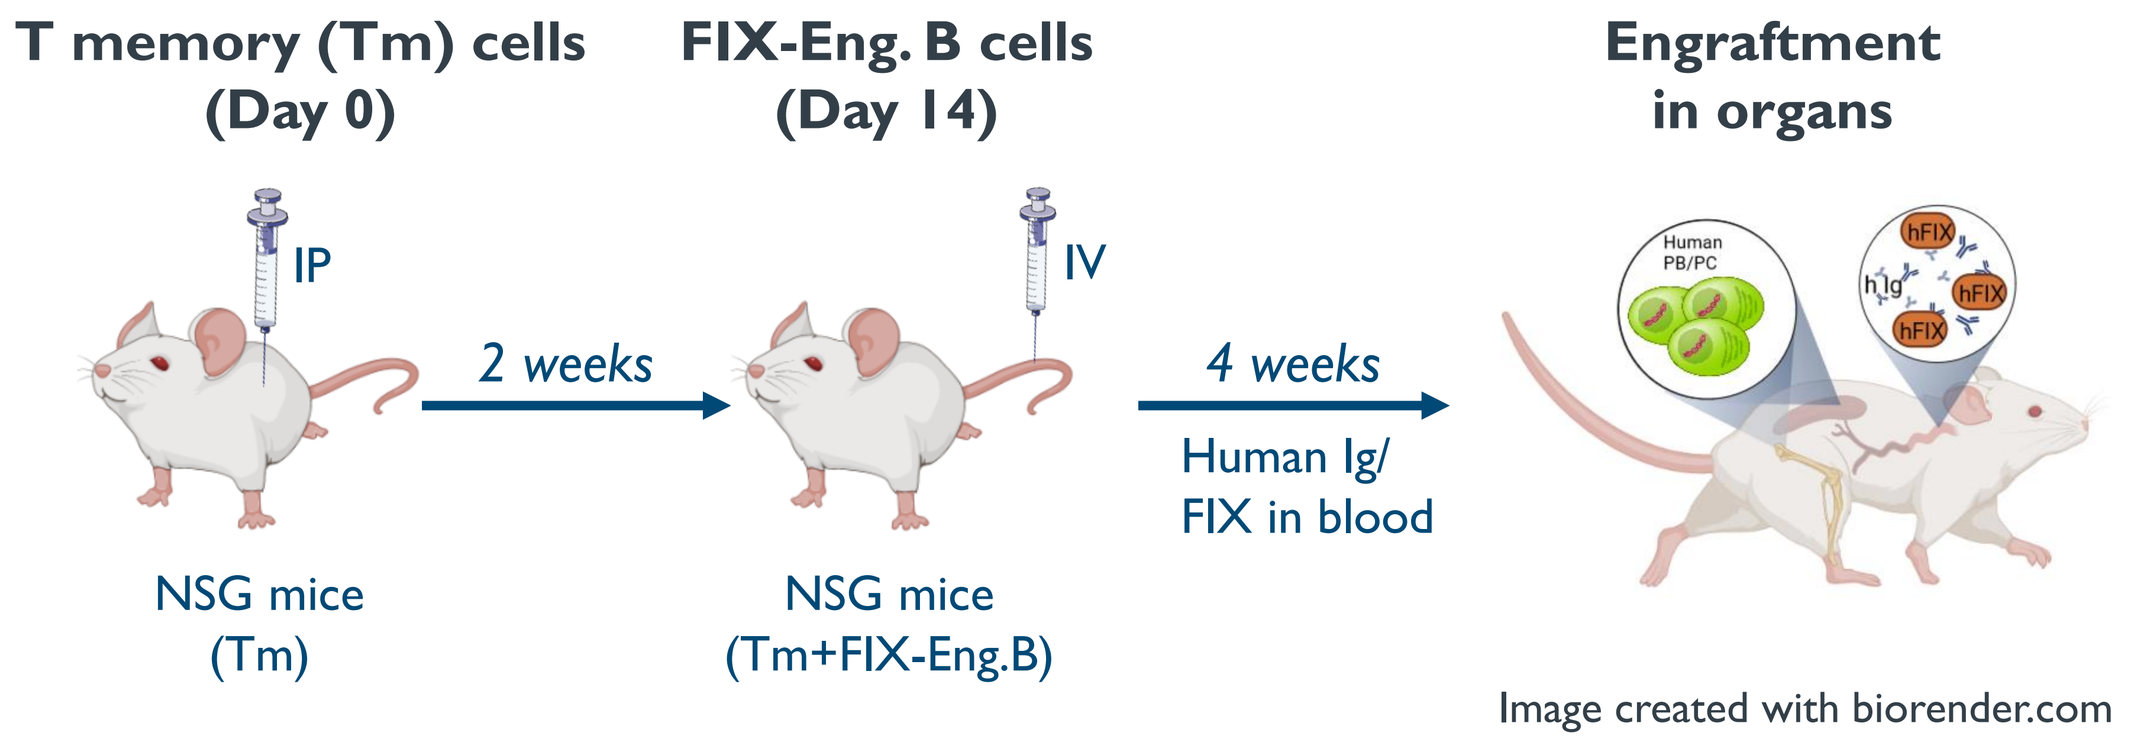
T memory (Tm) cells (Day 0)
FIX-Eng. B cells (Day 14)
Engraftment in organs
IP
IV
2 weeks
4 weeks
Human Ig/ FIX in blood
NSG mice (Tm)
NSG mice (Tm+FIX-Eng.B)
Image created with biorender.com

In vivo mouse model for differentiated human B cell engraftment. Memory T (Tm) cells were administered via the intraperitoneal (IP) route into 8-week-old NSG mice,[3] 2 weeks prior to intravenous (IV) administration of donor-matched, differentiated, and engineered B cells (FIX-Padua cassette into the *TRAC* locus). Every week post injection, blood was drawn for monitoring of human immunoglobulins (HuIg) and FIX-Padua production. At day 41, mice were euthanized, and spleen and bone marrow were recovered to evaluate the efficiency of human B cell engraftment.

Eng, engineered.

FIX-engineered B cells engrafted in mice are strongly differentiated in plasma cells and plasmablasts

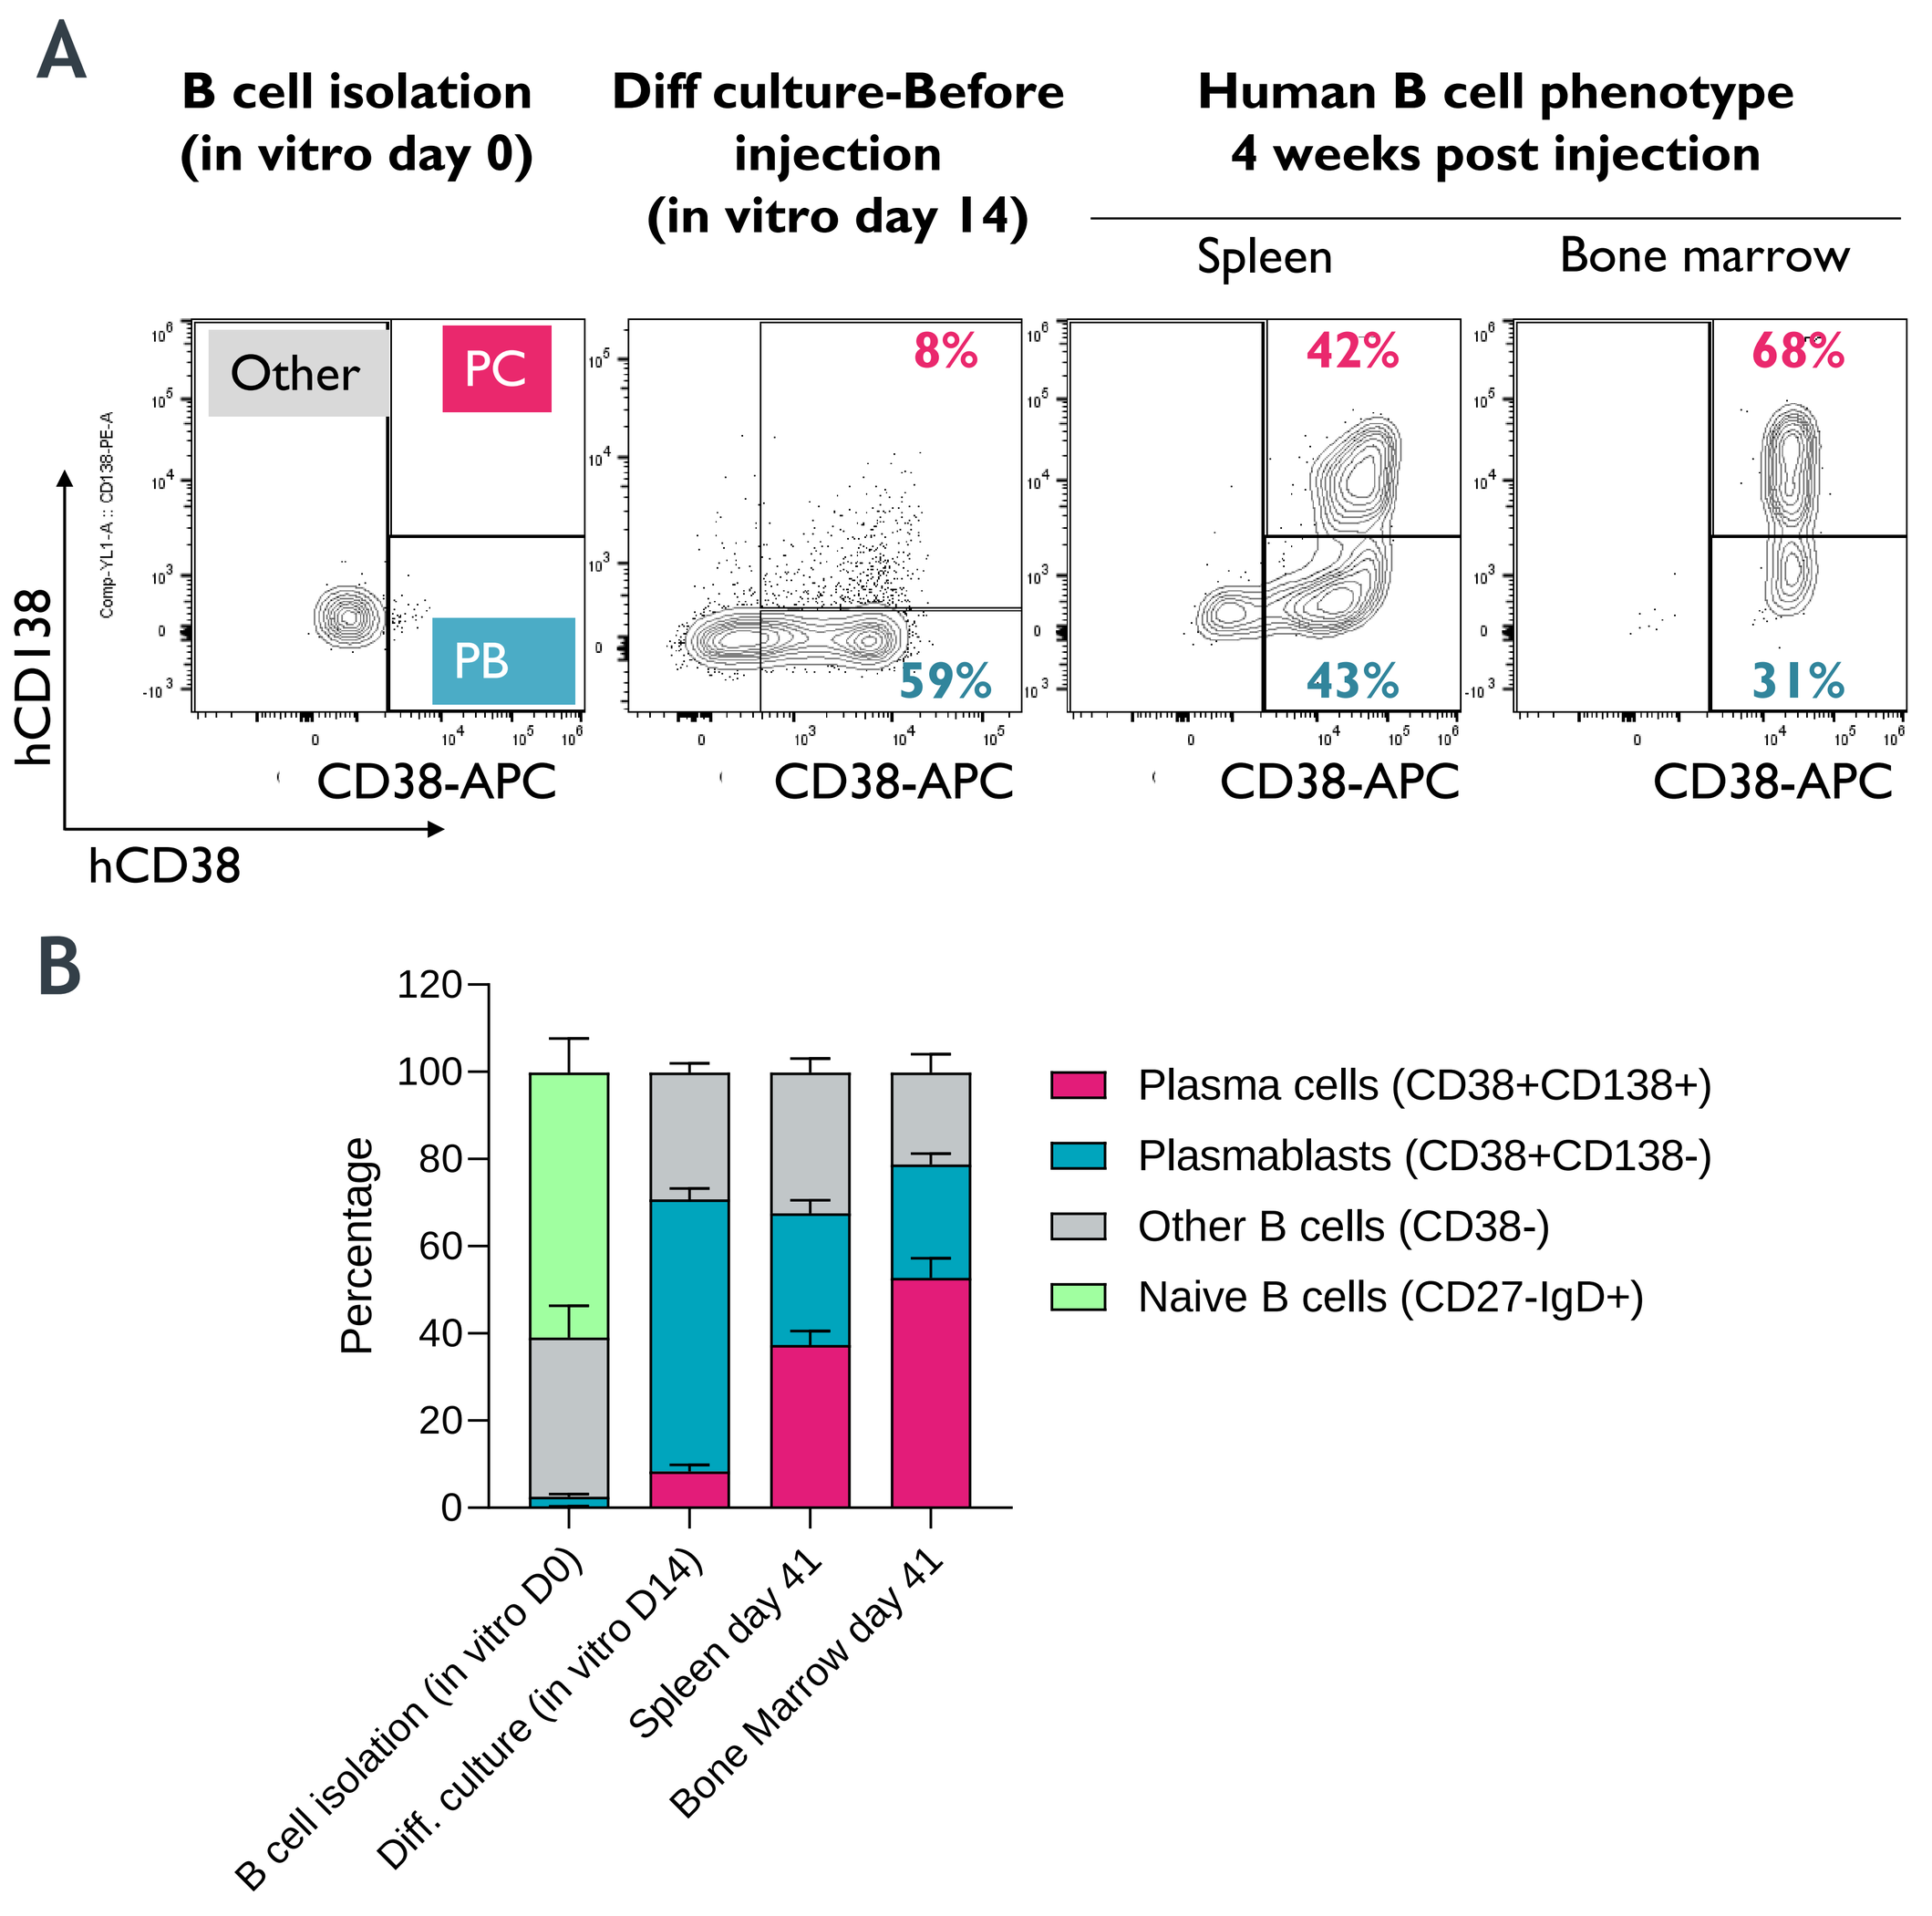
A
B cell isolation (in vitro day 0)
Diff culture-Before injection (in vitro day 14)
Human B cell phenotype 4 weeks post injection
Spleen
Bone marrow
Other
PC
PB
8%
59%
42%
43%
68%
31%
CD38-APC
hCD138
hCD38
B
Percentage
Plasma cells (CD38+CD138+)
Plasmablasts (CD38+CD138-)
Other B cells (CD38-)
Naive B cells (CD27-IgD+)
B cell isolation (in vitro D0)
Diff. culture (in vitro D14)
Spleen day 41
Bone Marrow day 41

(**A**) The percentage of human plasmablasts (PB) and plasma cells (PC) was evaluated in organs after labeling of CD38 and CD138 markers and flow cytometry analysis. (**B**) Phenotype of engrafted B cells. Monitoring of PB (CD38+CD138-), PC (CD38+CD138+), naïve B cells (CD27-IgD+), and other CD19+ B cells just after B cell isolation (in vitro day 0), differentiation protocol (in vitro day 14), or in vivo in murine spleen and bone marrow (day 41 post injection).

Human plasma cells are highly engrafted in mouse bone marrow

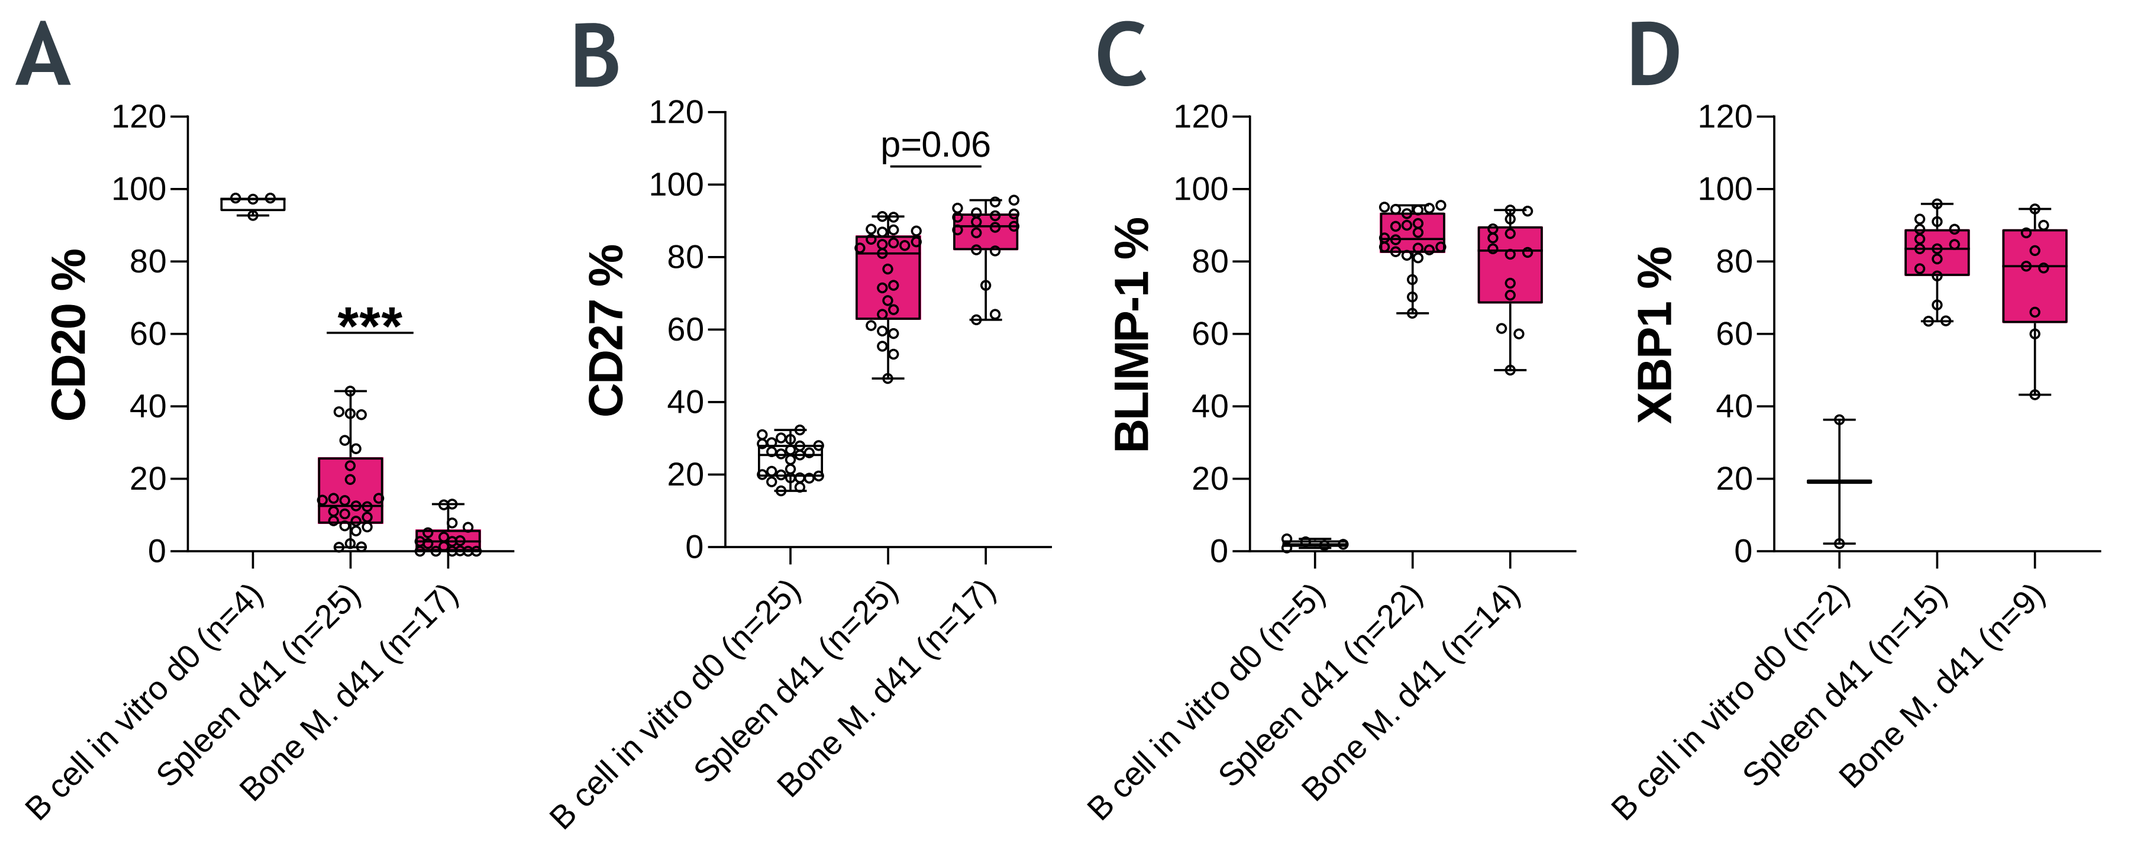
A
CD20 %

B cell in vitro d0 (n=4)
Spleen d41 (n=25)
Bone M. d41 (n=17)
B
CD27 %
p=0.06
B cell in vitro d0 (n=25)
Spleen d41 (n=25)
Bone M. d41 (n=17)
C
BLIMP-1 %
B cell in vitro d0 (n=5)
Spleen d41 (n=22)
Bone M. d41 (n=14)
D
XBP1 %
B cell in vitro d0 (n=2)
Spleen d41 (n=15)
Bone M. d41 (n=9)

Phenotyping (day 41) of engrafted human B cells by monitoring the expression of CD20 (**A**), CD27 (**B**), BLIMP-1 (**C**), or XBP1 (**D**). As expected, human B cells engrafted in the bone marrow (Bone M) are mainly plasma cells (CD20-CD27++BLIMP-1+XBP1+).

B cells engineered with ZFN/AAV6 technology secrete a therapeutical level of human FIX-Padua in vivo

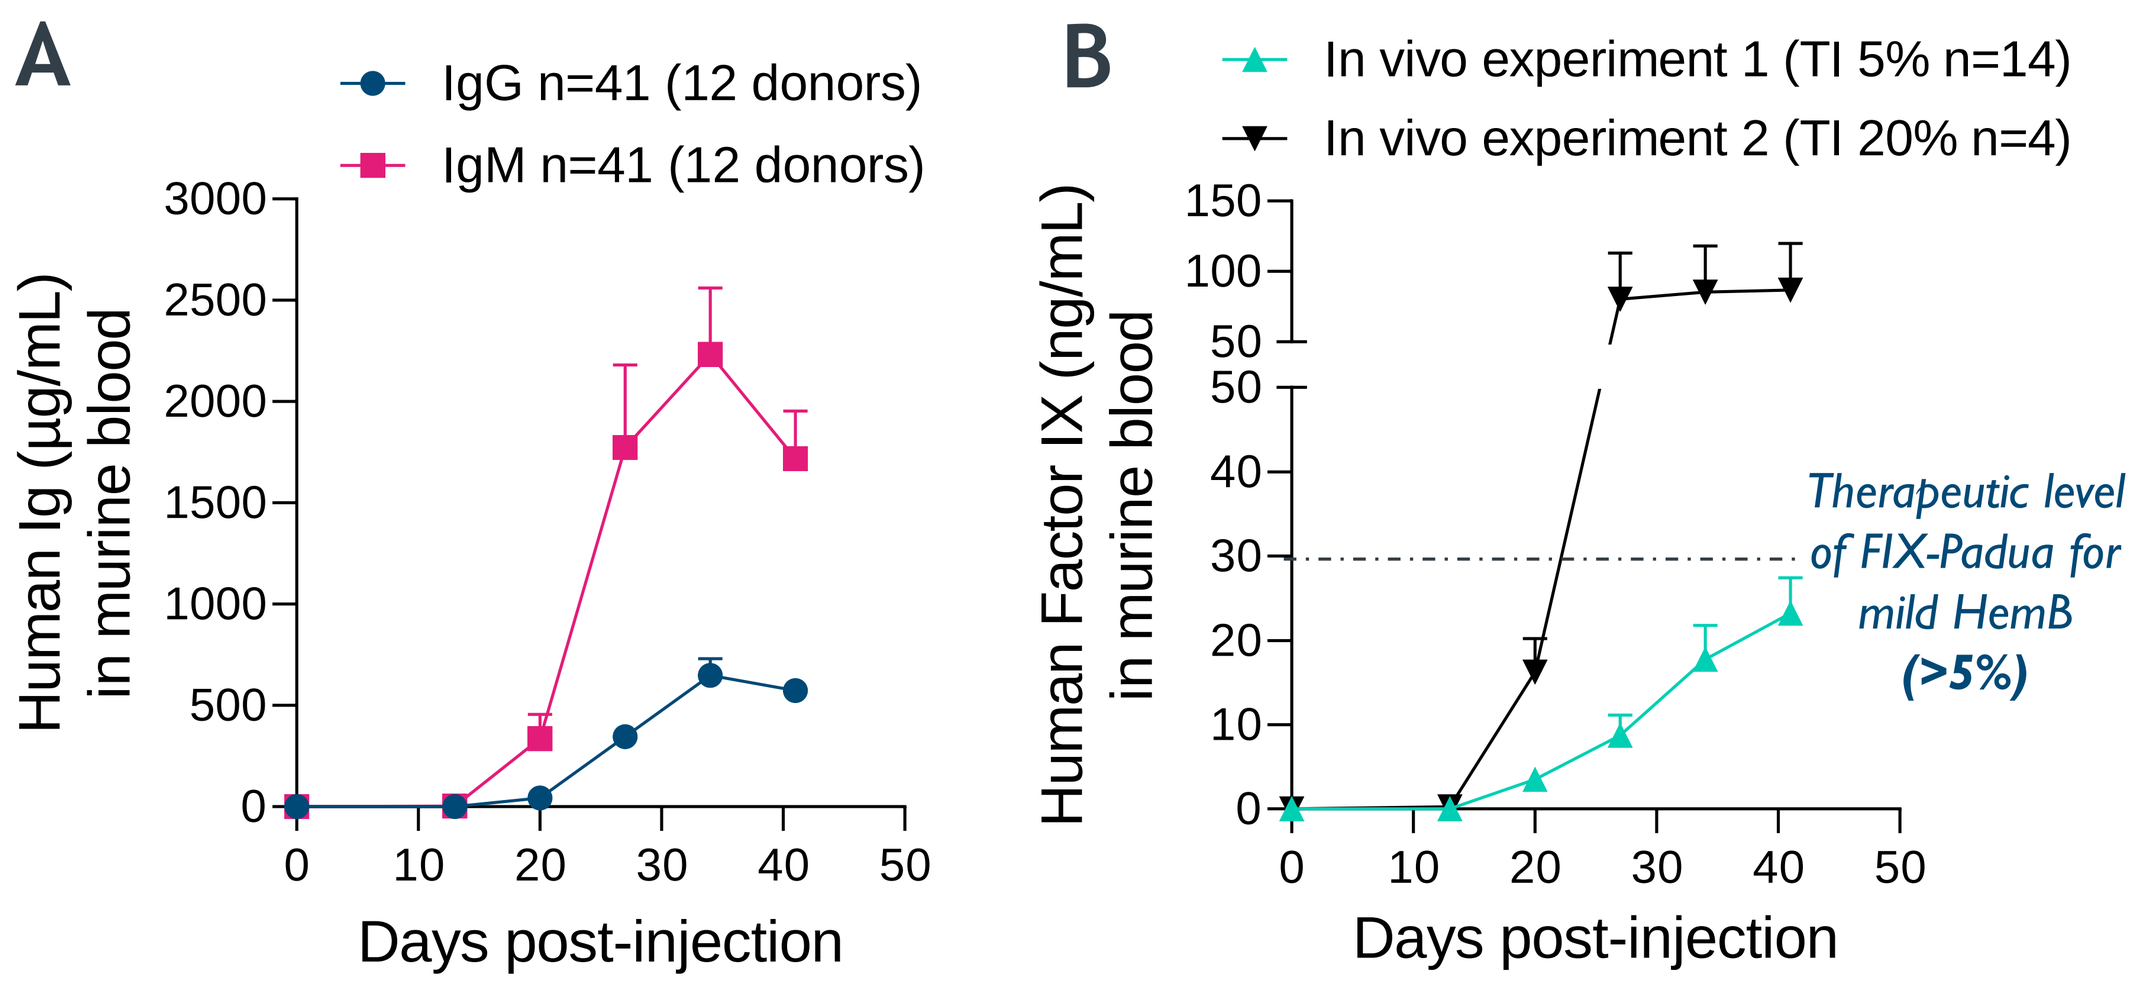
A
IgG n=41 (12 donors)
IgM n=41 (12 donors)
Human Ig (µg/mL) in murine blood
Days post-injection
B
In vivo experiment 1 (TI 5% n=14)
In vivo experiment 2 (TI 20% n=4)
Human Factor IX (ng/mL) in murine blood
Therapeutic level of FIX-Padua for mild HemB (>5%)
Days post-injection

Every week post injection, blood was drawn from NSG mice, and presence of human immunoglobulins (Ig) and FIX-Padua was quantified. (**A**) High levels of circulating human IgG and IgM in mouse blood reflect the engraftment of differentiated human B cells. (**B**) Efficient secretion of human FIX-Padua from FIX-engineered B cells in vivo. A targeted integration (TI) efficiency ranging from 5% to 20% allows a therapeutic FIX expression from 5% to 15% of normal wild-type FIX, respectively.

HemB, hemophilia B.

Conclusions

- In summary, we demonstrated that ZFN mRNA combined with rAAV6 donor DNA allows efficient site-specific insertion of human FIX transgene into the B cell *TRAC* locus, resulting in therapeutic levels of FIX-Padua expression in vivo.
- This study highlights the therapeutic potential of engineering B cells as a new technological platform to treat a variety of protein deficiencies, blood disorders, infectious diseases, or cancers.

References

1. Wang J, et al. *Nat Biotechnol*. 2015;33(12):1256-1263.
2. Hung KL, et al. *Mol Ther*. 2018;26(2):456-467.
3. Ishikawa Y, et al. *Eur J Immunol*. 2014;44:3453-3463.

Presented at the ESGCT 29th Annual Meeting, 2022