Abstract #1186

Strategic Formulation Development for AAV-Delivered Gene Therapies - A Case Study

Madhura Som, Michelle Jones, Katelyn Esposto, Benson Gikanga, Momina Mahmood, Saba Ghazvini, Sandeep Yadav

Sangamo Therapeutics, 7000 Marina Blvd., Brisbane, CA 94005, USA

Abstract

Formulation optimization (e.g., choice of buffer, pH, excipients) is crucial to develop and stabilize viral vectors as delivery vehicles for genomic medicines. Phosphate buffers have been widely used for formulating viral vectors for gene therapy due to their well-known clinical compatibility and desired pH range for biopharmaceuticals. However, phosphate buffer is known to show a considerable pH drop in frozen state, which might scale with volume, irrespective of the buffer concentration. For frozen AAV-based drug product (DP), the freeze/thaw cycling during manufacturing and/or storage and handling with associated pH drop may negatively impact the product critical quality attributes (CQAs). To protect AAV-based DPs from freeze/thaw stress, sucrose can be used as a cryoprotectant.

In this study, we explored varying sucrose concentrations and their effectiveness in preventing pH drop mediated degradation of AAVs in a phosphate and a non-phosphate buffer formulation, respectively. This work demonstrates strategic selection of buffer (phosphate/non-phosphate) and other excipients (e.g., sucrose) to enable optimal stability of the DP. Further, we present an analytical method (free DNA assay) that can be utilized in optimizing excipient concentration to manage formulation characteristics (e.g., osmolality, density, and endotoxin) to meet ocular and central nervous system (CNS) routes of administration requirements.

pH drift in formulations during freeze-thaw stress

Buffers react differently to various stresses during manufacturing, storage and use, e.g., freeze-thaw (FT) stress. Phosphate buffers show pH drift in frozen condition due to precipitation of sodium phosphate component[1]. Using a pH sensitive dye, we have shown that frozen pH drift scales with volume as a function of buffer type (phosphate vs. non-phosphate). Our results indicate that within a 2-500ml scale, phosphate formulation undergoes up to a 3.5-unit drop in frozen pH compared to room temperature (RT) while a comparable non-phosphate formulation shows only up to a 1-unit pH increase. Low pH is known to affect AAV product quality, e.g., capsid protein conformation changes/lysis[2,3], and reduction in transduction efficiency[4].

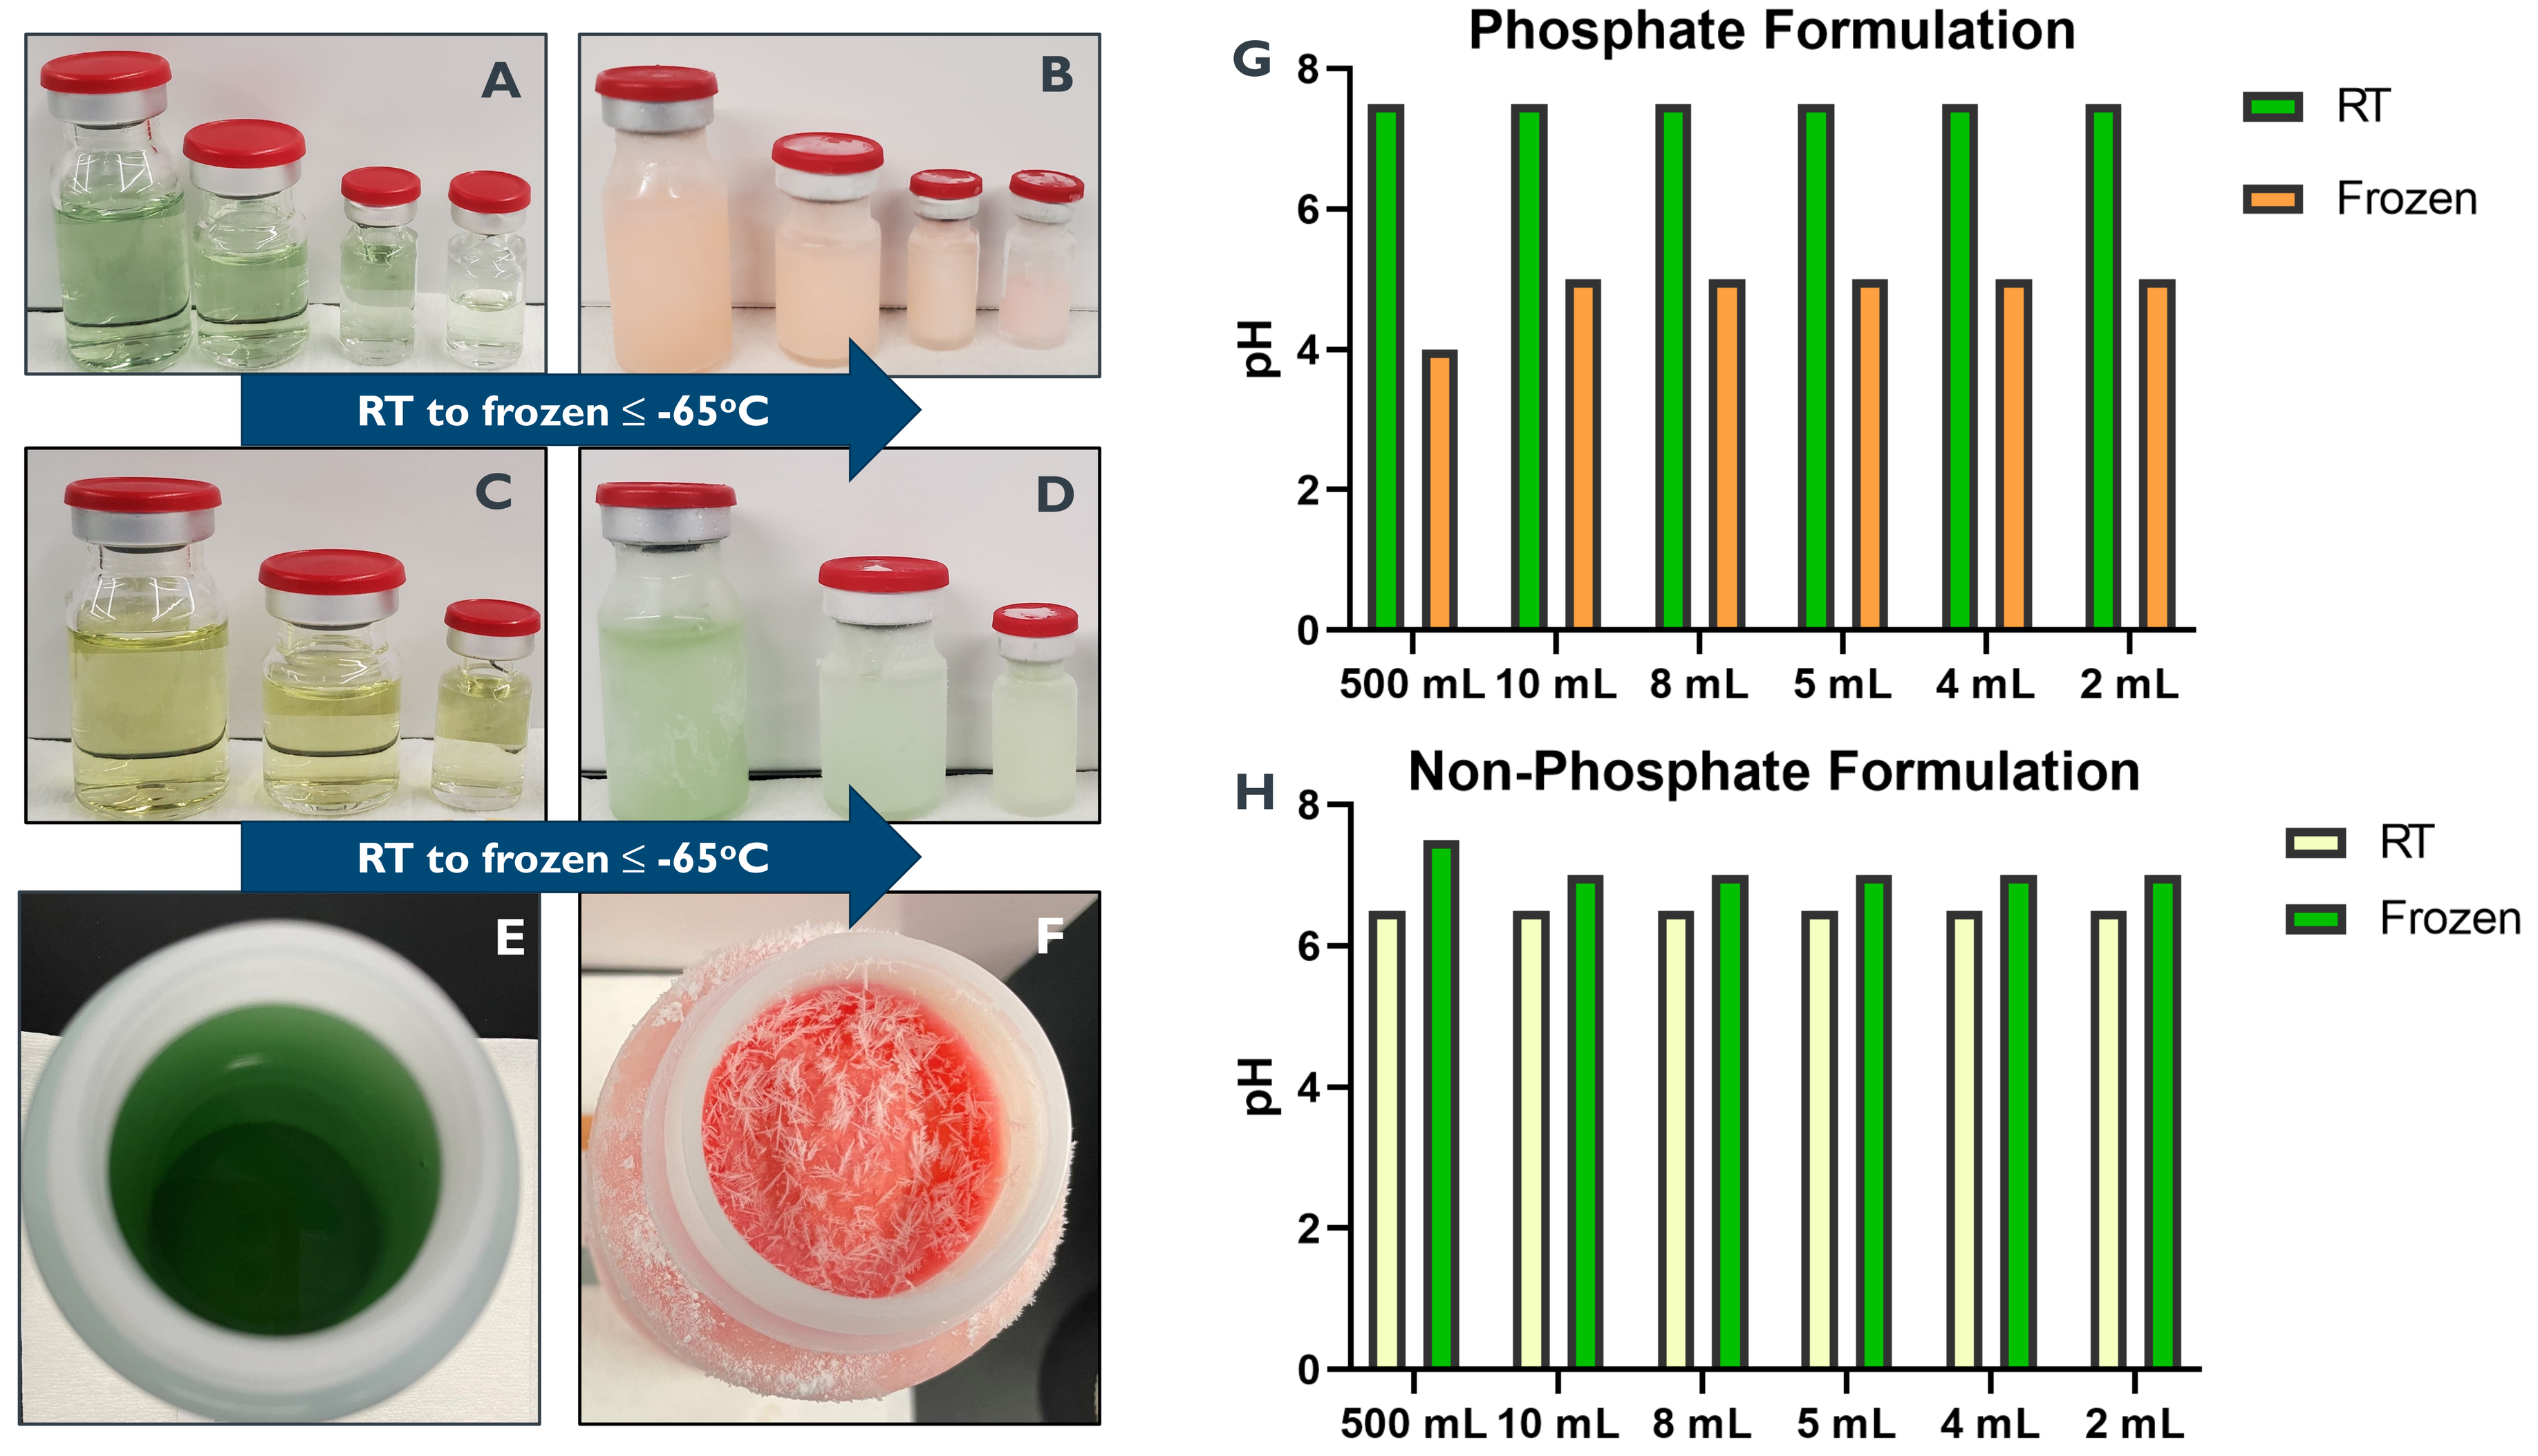

A
B
RT to frozen ≤ -65°C
C
D
RT to frozen ≤ -65°C
E
F
G
Phosphate Formulation
pH
RT
Frozen
500 mL 10 mL 8 mL 5 mL 4 mL 2 mL
H
Non-Phosphate Formulation
pH
RT
Frozen
500 mL 10 mL 8 mL 5 mL 4 mL 2 mL

Figure 1. 10 mL, 5 mL, 2 mL and 1 mL volumes of phosphate formulation (pH 7.4) before **(A)** and after freezing **(B)**. 10 mL, 5 mL, and 1 mL volumes of non-phosphate formulation (pH 6.8) before at room temperature (RT) **(C)** and after freezing **(D)**. Top view of 500 ml phosphate formulation before **(E)** and after freezing **(F)**. Color changes reflect change in pH after freezing as indicated in bar graphs **(G)** Phosphate formulation and **(H)** Non-phosphate formulation

Phosphate buffered formulations show higher free DNA after multi-cycle freeze-thaw stress

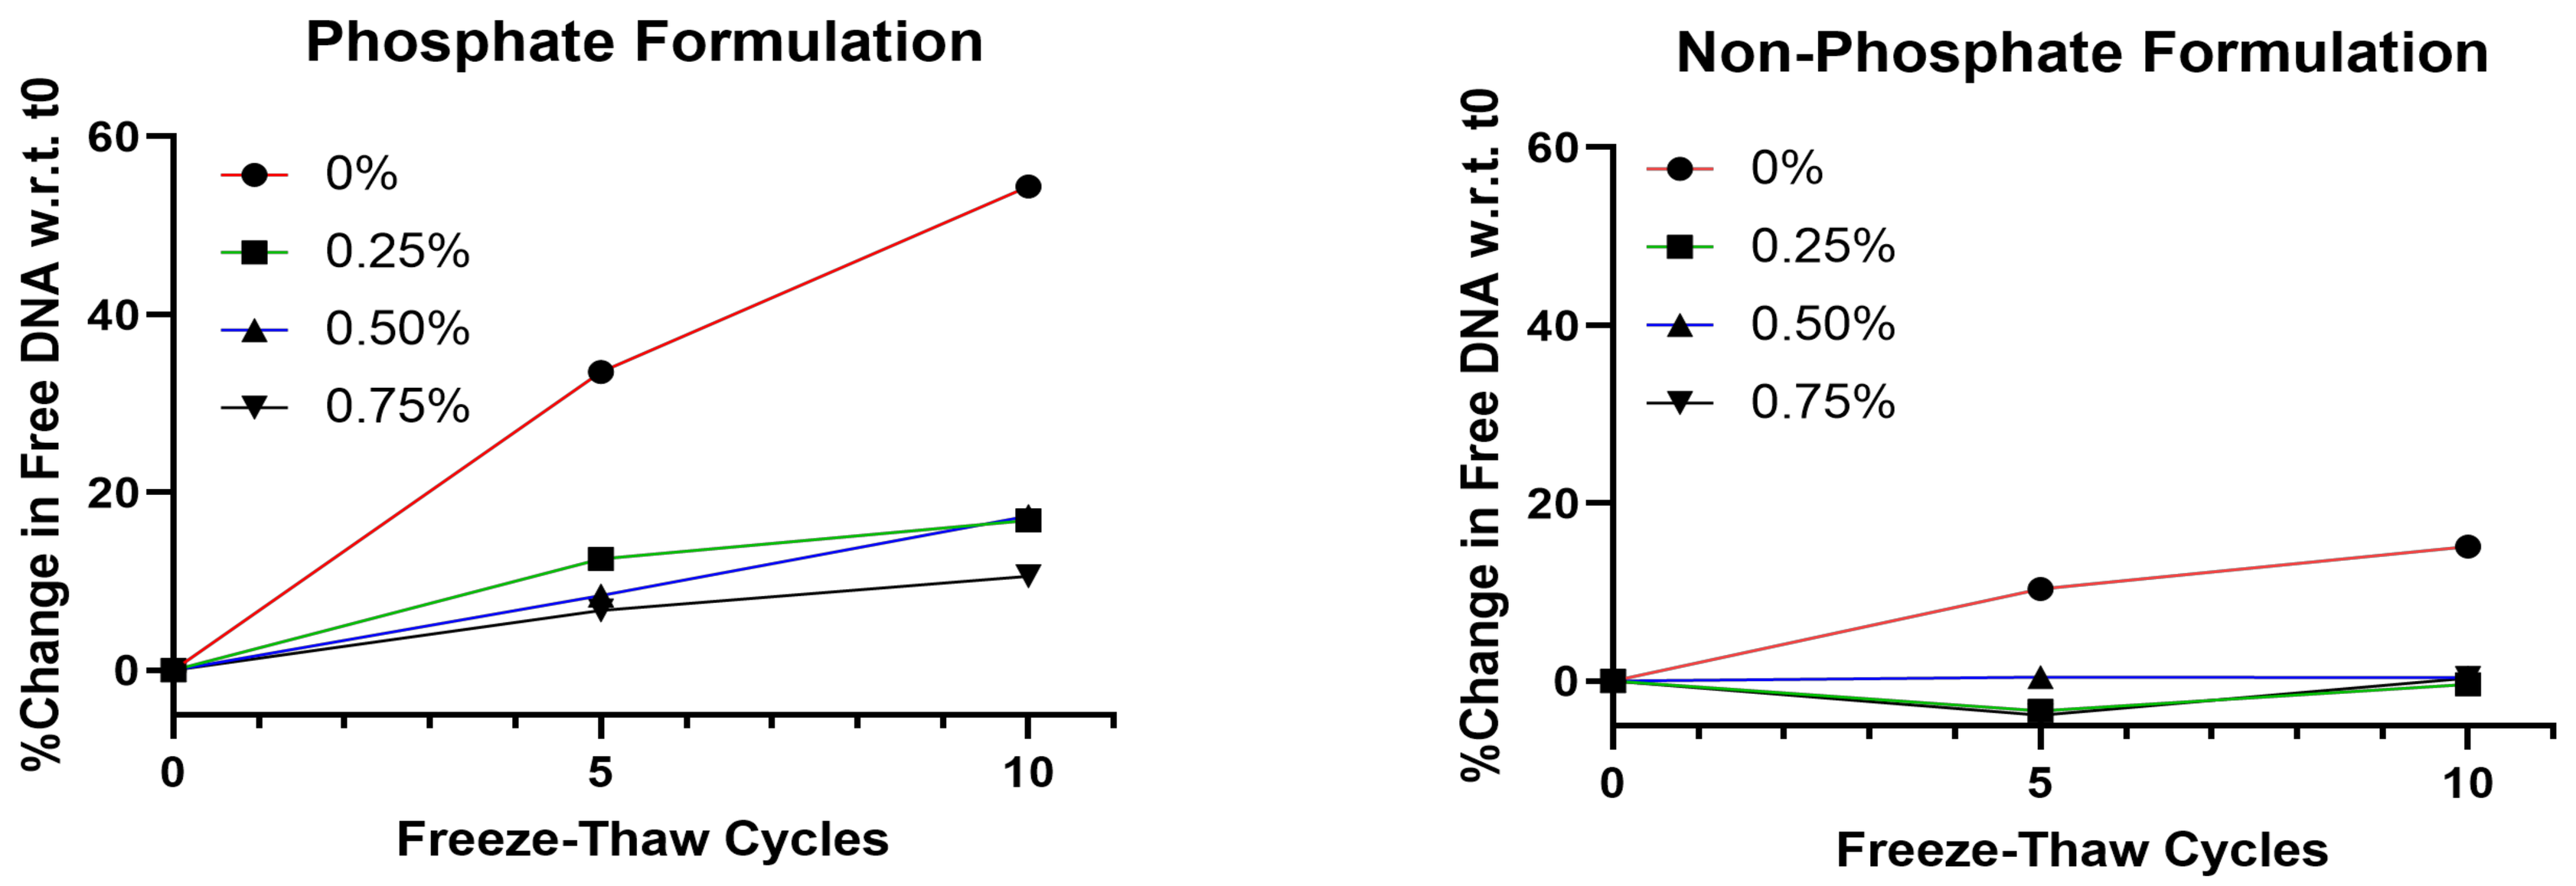

Phosphate Formulation
%Change in Free DNA w.r.t. t0
0%
0.25%
0.50%
0.75%
Freeze-Thaw Cycles
Non-Phosphate Formulation
%Change in Free DNA w.r.t. t0
0%
0.25%
0.50%
0.75%
Freeze-Thaw Cycles

Figure 2. Amount of free DNA measured was normalized to t0 for each formulation and plotted as a %change for phosphate **(left)** and non-phosphate **(right)** formulations with various levels of sucrose 0% (w/v) to 0.75% (w/v), used as a cryoprotectant

Although not a typical critical quality attribute (CQA), free DNA quantification can indicate effect of formulation stress on capsid integrity in the drug product. This assay uses a DNA-binding fluorescent dye to measure DNA that is not encapsulated by the AAV capsid and, therefore, not part of the therapeutic payload. Thus, increasing free DNA with formulation stress like FT cycling could indicate potential leakage of previously encapsulated DNA into the formulation. Here, we observe that:

- % Free DNA increases with increasing FT cycles 5x-10x
- Without sucrose, both formulations exhibit highest increase in free DNA (compared to w/ sucrose) and the non-phosphate formulation has lower % increase in free DNA over 5-10x FT cycles
- Phosphate formulation requires more sucrose to reduce %free DNA over 5-10x FT cycles
- Free DNA change is a valuable attribute to assess differences in formulations as a function of stress

Phosphate buffered formulations show higher subvisible particulates after multi-cycle freeze-thaw stress

Subvisible particulates (SvP) are an important CQA for AAV drug products (DP) and are related to the safety of the DP surrounding which there are compendial limits/container. Here, SvP was measured using a background membrane imaging (BMI) method which shows that phosphate formulations show higher SvP/container when exposed to 5x-10x freeze thaw cycles compared to non-phosphate formulation. Addition of sucrose reduces SvP formation for the phosphate formulation.

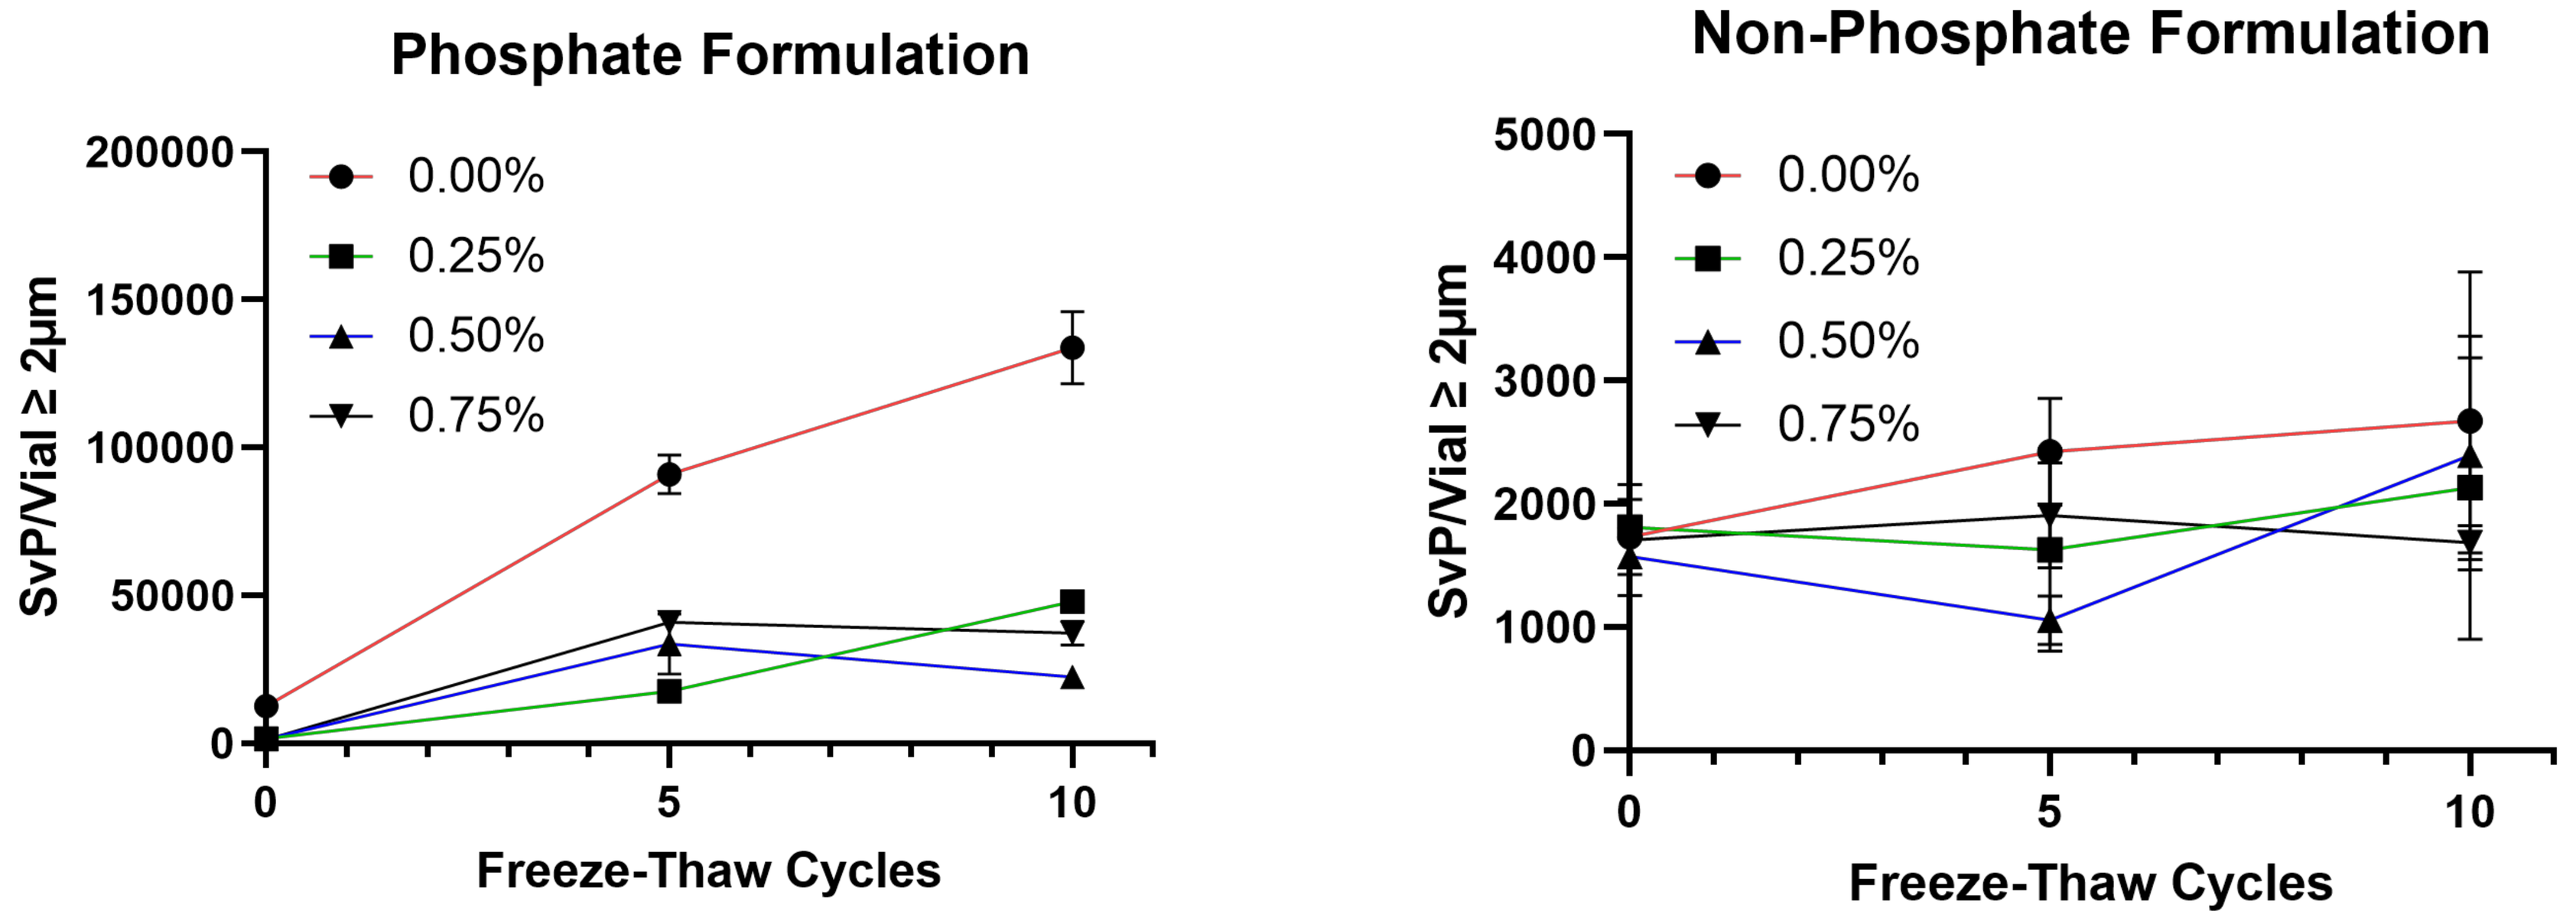

Phosphate Formulation
SvP/Vial ≥ 2μm
0.00%
0.25%
0.50%
0.75%
Freeze-Thaw Cycles
Non-Phosphate Formulation
SvP/Vial ≥ 2μm
0.00%
0.25%
0.50%
0.75%
Freeze-Thaw Cycles

Figure 3. Number of SvP ≥ 2μm measured per vial with a BMI method are plotted for each formulation : Phosphate **(left)** and Non-phosphate **(right)** with various levels of sucrose 0% (w/v) to 0.75% (w/v), used as a cryoprotectant.

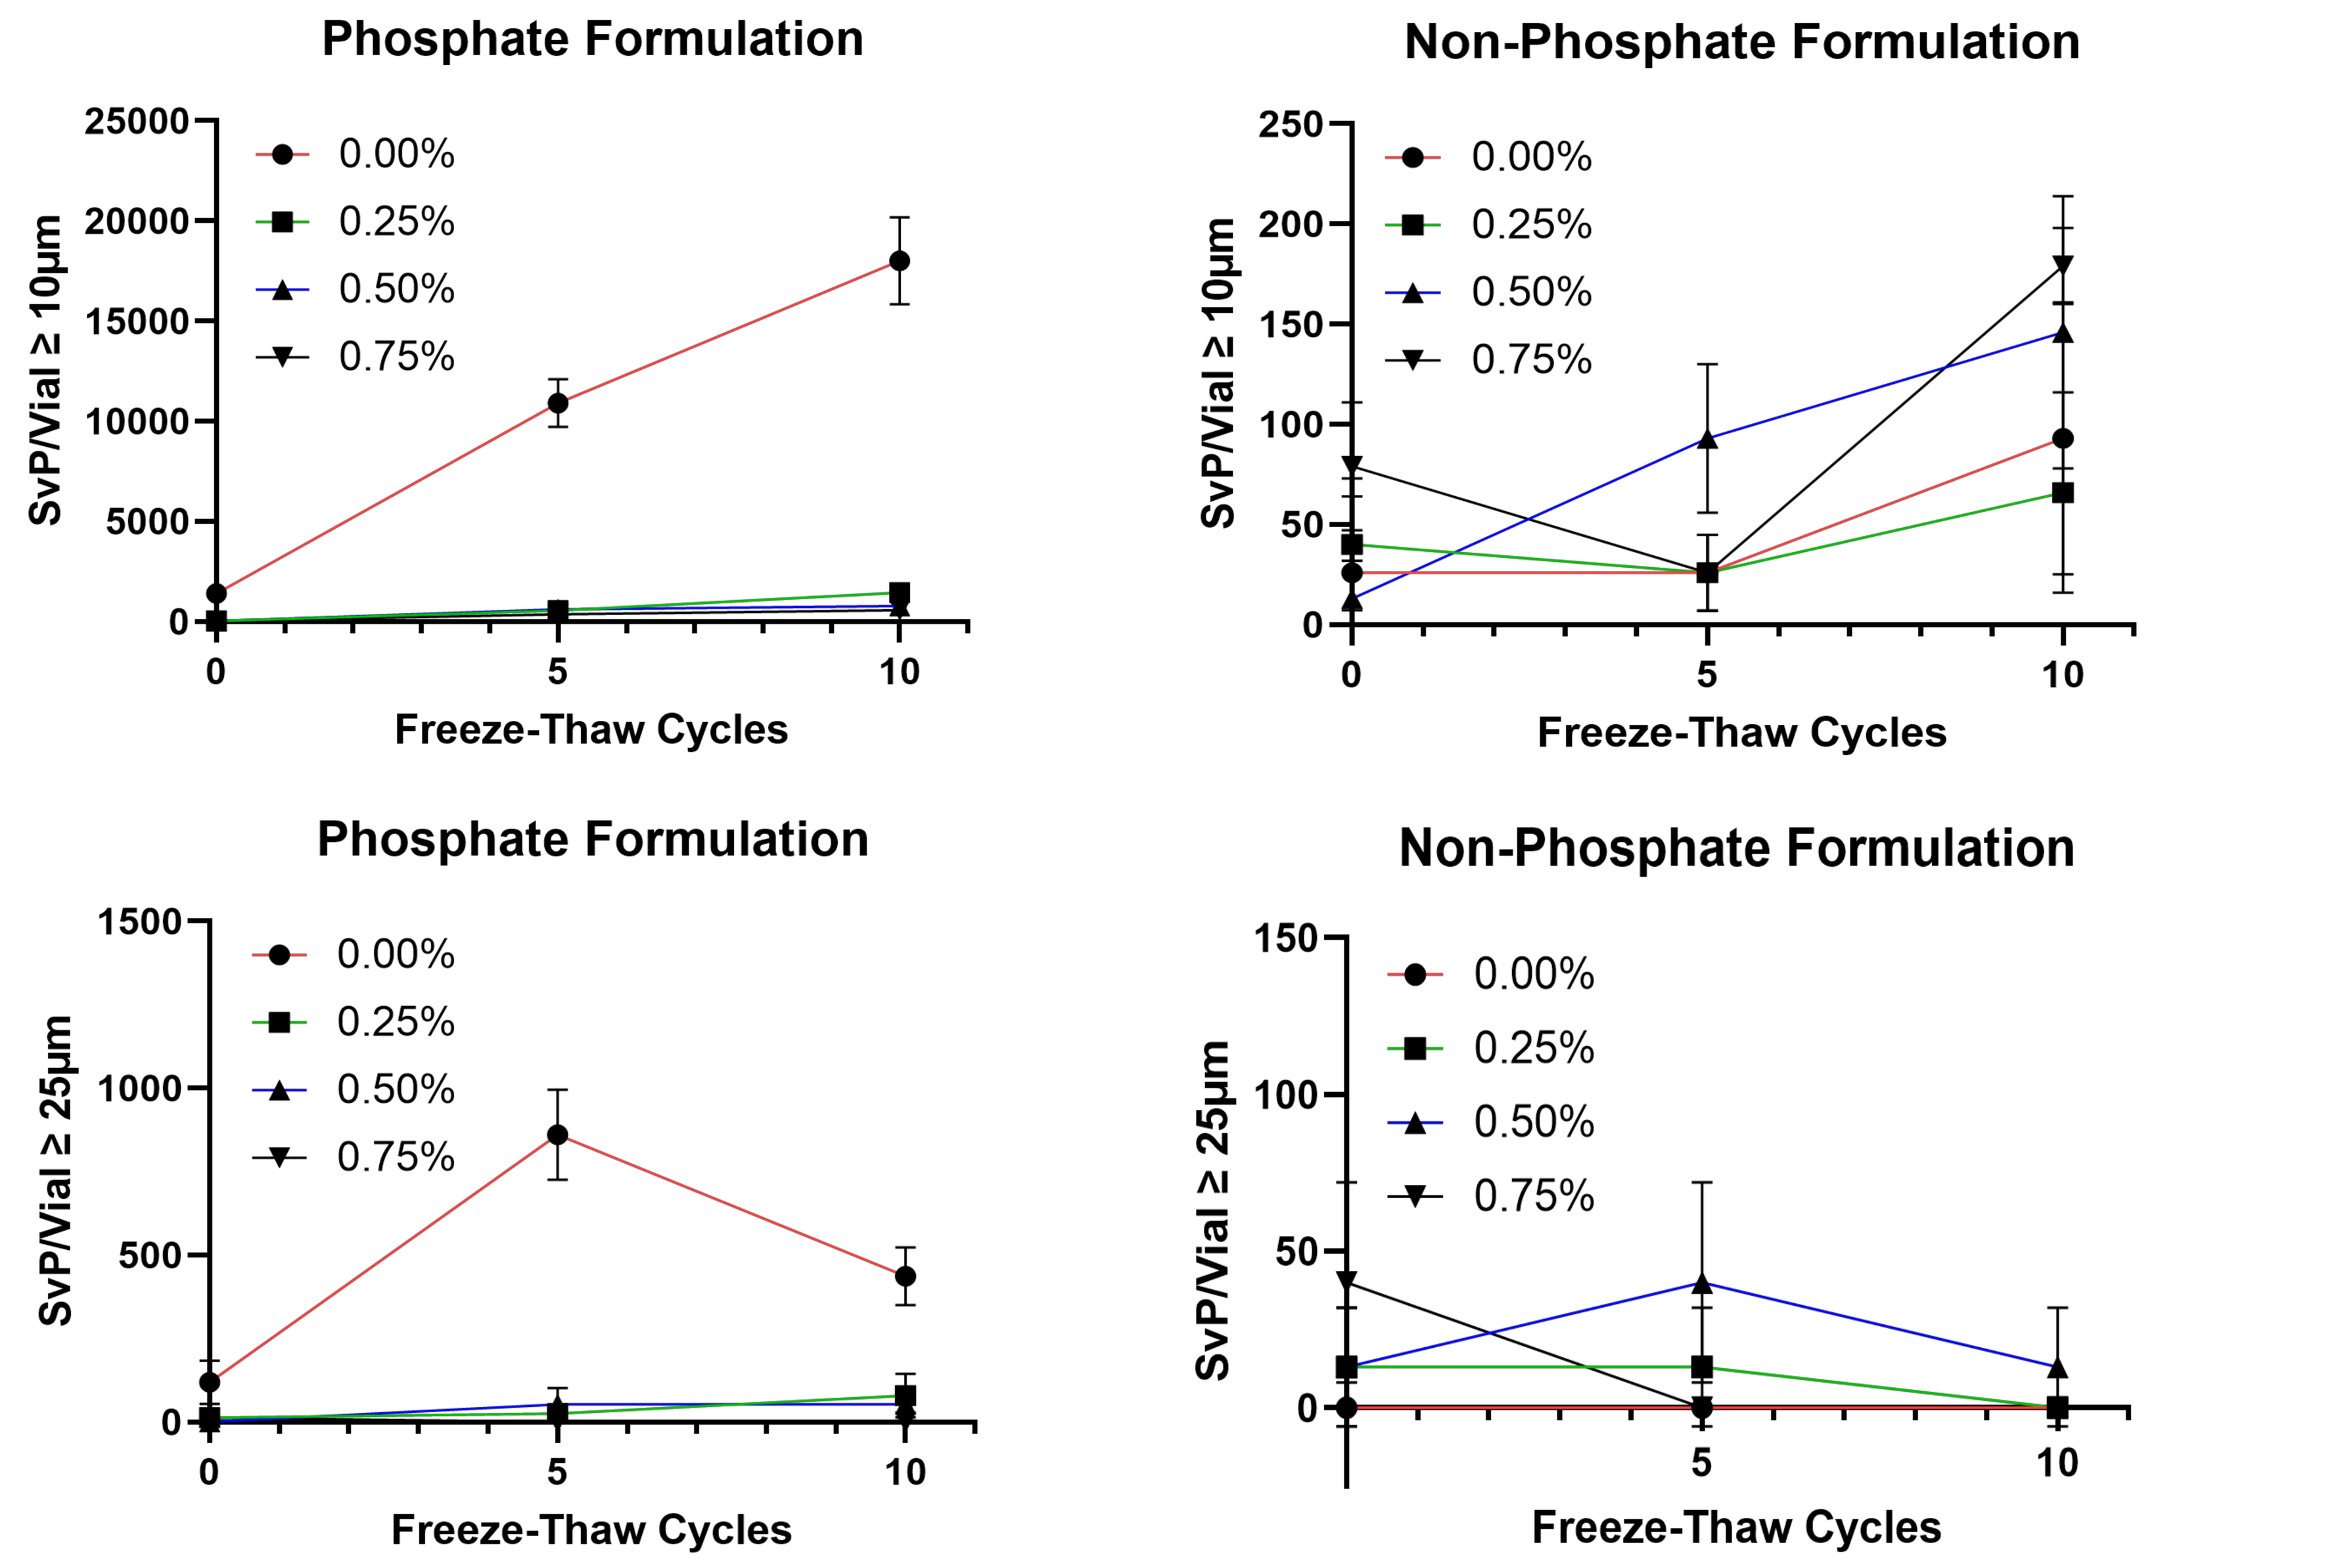

Phosphate Formulation
SvP/Vial ≥ 10μm
Non-Phosphate Formulation
SvP/Vial ≥ 10μm
Phosphate Formulation
SvP/Vial ≥ 25μm
Non-Phosphate Formulation
SvP/Vial ≥ 25μm
0.00%
0.25%
0.50%
0.75%
Freeze-Thaw Cycles

Figure 4. Number of SvP≥ 10μm **(top row)** and ≥ 25μm **(bottom row)** measured per vial with a BMI method are plotted for each formulation; Phosphate **(left)** and Non-phosphate **(right)** with various levels of sucrose 0% (w/v) to 0.75% (w/v), used as a cryoprotectant.

Conclusion

- Phosphate buffered formulations show a larger frozen pH drop than non-phosphate formulations within a 2-500 mL volume scale. Low pH exposure is known to be detrimental to AAV capsid integrity.
- We have presented two assays, free DNA and subvisible particulate analysis, which have shown that phosphate buffered formulations are more susceptible to AAV degradation when exposed to multi-cycle freeze-thaw stress compared to non-phosphate buffered formulations and require larger amounts of sucrose (cryoprotectant) to mitigate such effects.
- Sucrose is needed to protect against freeze-thaw stress. However, for sensitive routes of administration (RoA), e.g., eye and central nervous system, this may become a limiting factor thus necessitating buffers with lower amounts of excipients to conserve endotoxins, osmolality and density. Ultimately, the safety and biological/clinical compatibility of the buffer and any additional excipients for the RoA will drive the formulation choices for a specific application.

References

1. Croyle, M. A., Cheng, X. & Wilson, J. M. Development of formulations that enhance physical stability of viral vectors for gene therapy. Gene Ther **8**, 1281–1290 (2001).
2. Venkatakrishnan, B. *Et al.* Structure and Dynamics of Adeno-Associated Virus Serotype 1 VP1-Unique N-Terminal Domain and Its Role in Capsid Trafficking. *J Virol* **87**, 4974–4984 (2013).
3. Salganik, M. *Et al.* Evidence for ph-Dependent Protease Activity in the Adeno-Associated Virus Capsid. *J Virol* **86**, 11877–11885 (2012).
4. Lowell, J. A., Mah, K. M., Bixby, J. L. & Lemmon, V. P. AAV8 transduction capacity is reduced by prior exposure to endosome-like ph conditions. *Neural Regen Res* **16**, 851–855 (2021).

Presented at the ASGCT 26th Annual Meeting, 2023