Engineered zinc finger transcriptional regulators enable robust and reliable epigenetic regulation in the mouse brain

Abstract #682

Patrick Dunn, Jisoo Lee, Kim Marlen, Qi Yu, Mihika Jalan, Hoang-Oanh Nguyen, Sarah Hinkley, Jason Eshleman, David Paschon, Victoria Chou, Mohammad Samie, Asa Hatami, Amy Pooler, Bryan Zeitler

Sangamo Therapeutics Inc., 501 Canal Blvd, Richmond, CA 94804, USA

Introduction

Therapeutic gene modulation has the potential to treat numerous neurodegenerative and neurodevelopmental diseases.

We are creating genomic medicines for an array of neurological disorders using Zinc Finger Transcriptional Regulators (ZF-TRs), including ZF Repressors (ZF-Rs) and ZF Activators (ZF-As). ZF-TRs are obtained by attaching a Repressor or Activator domain to Zinc Finger Proteins (ZFPs), which are naturally occurring human proteins that can be engineered to recognize specific genomic DNA sequences.

Since uncovering novel regulatory sites for highly-specific epigenetic repression or activation can require extensive empirical testing, we leveraged our proprietary design algorithm to assemble hundreds of candidate ZF-TRs against multiple neurological disease targets to efficiently identify hot spots for epigenetic regulation in both mouse and human genomes.

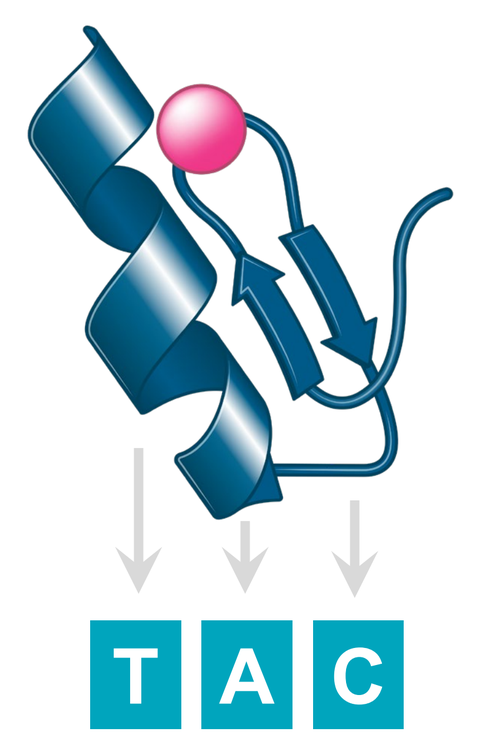
T A C

Key Features of Zinc Fingers

- Most abundant DNA binding domain in the human genome
- 28 amino acid peptide
- Compact structure
- Binds ~3 bp of DNA
- Specificity & affinity can be engineered

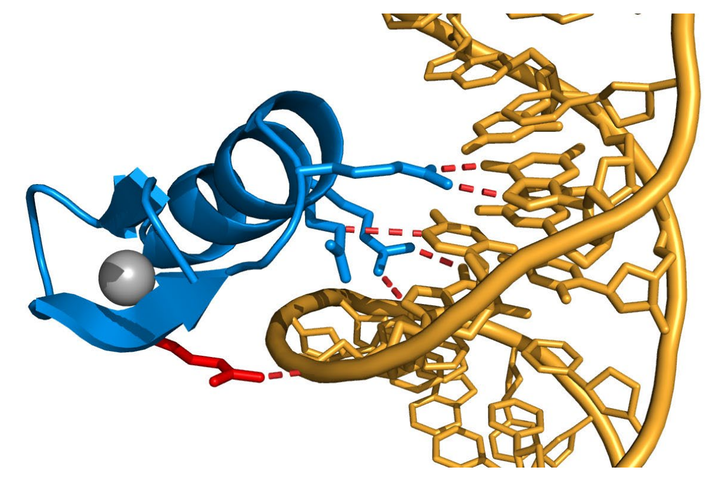

An optimized screening workflow for identifying potent and selective ZF Transcriptional Regulators

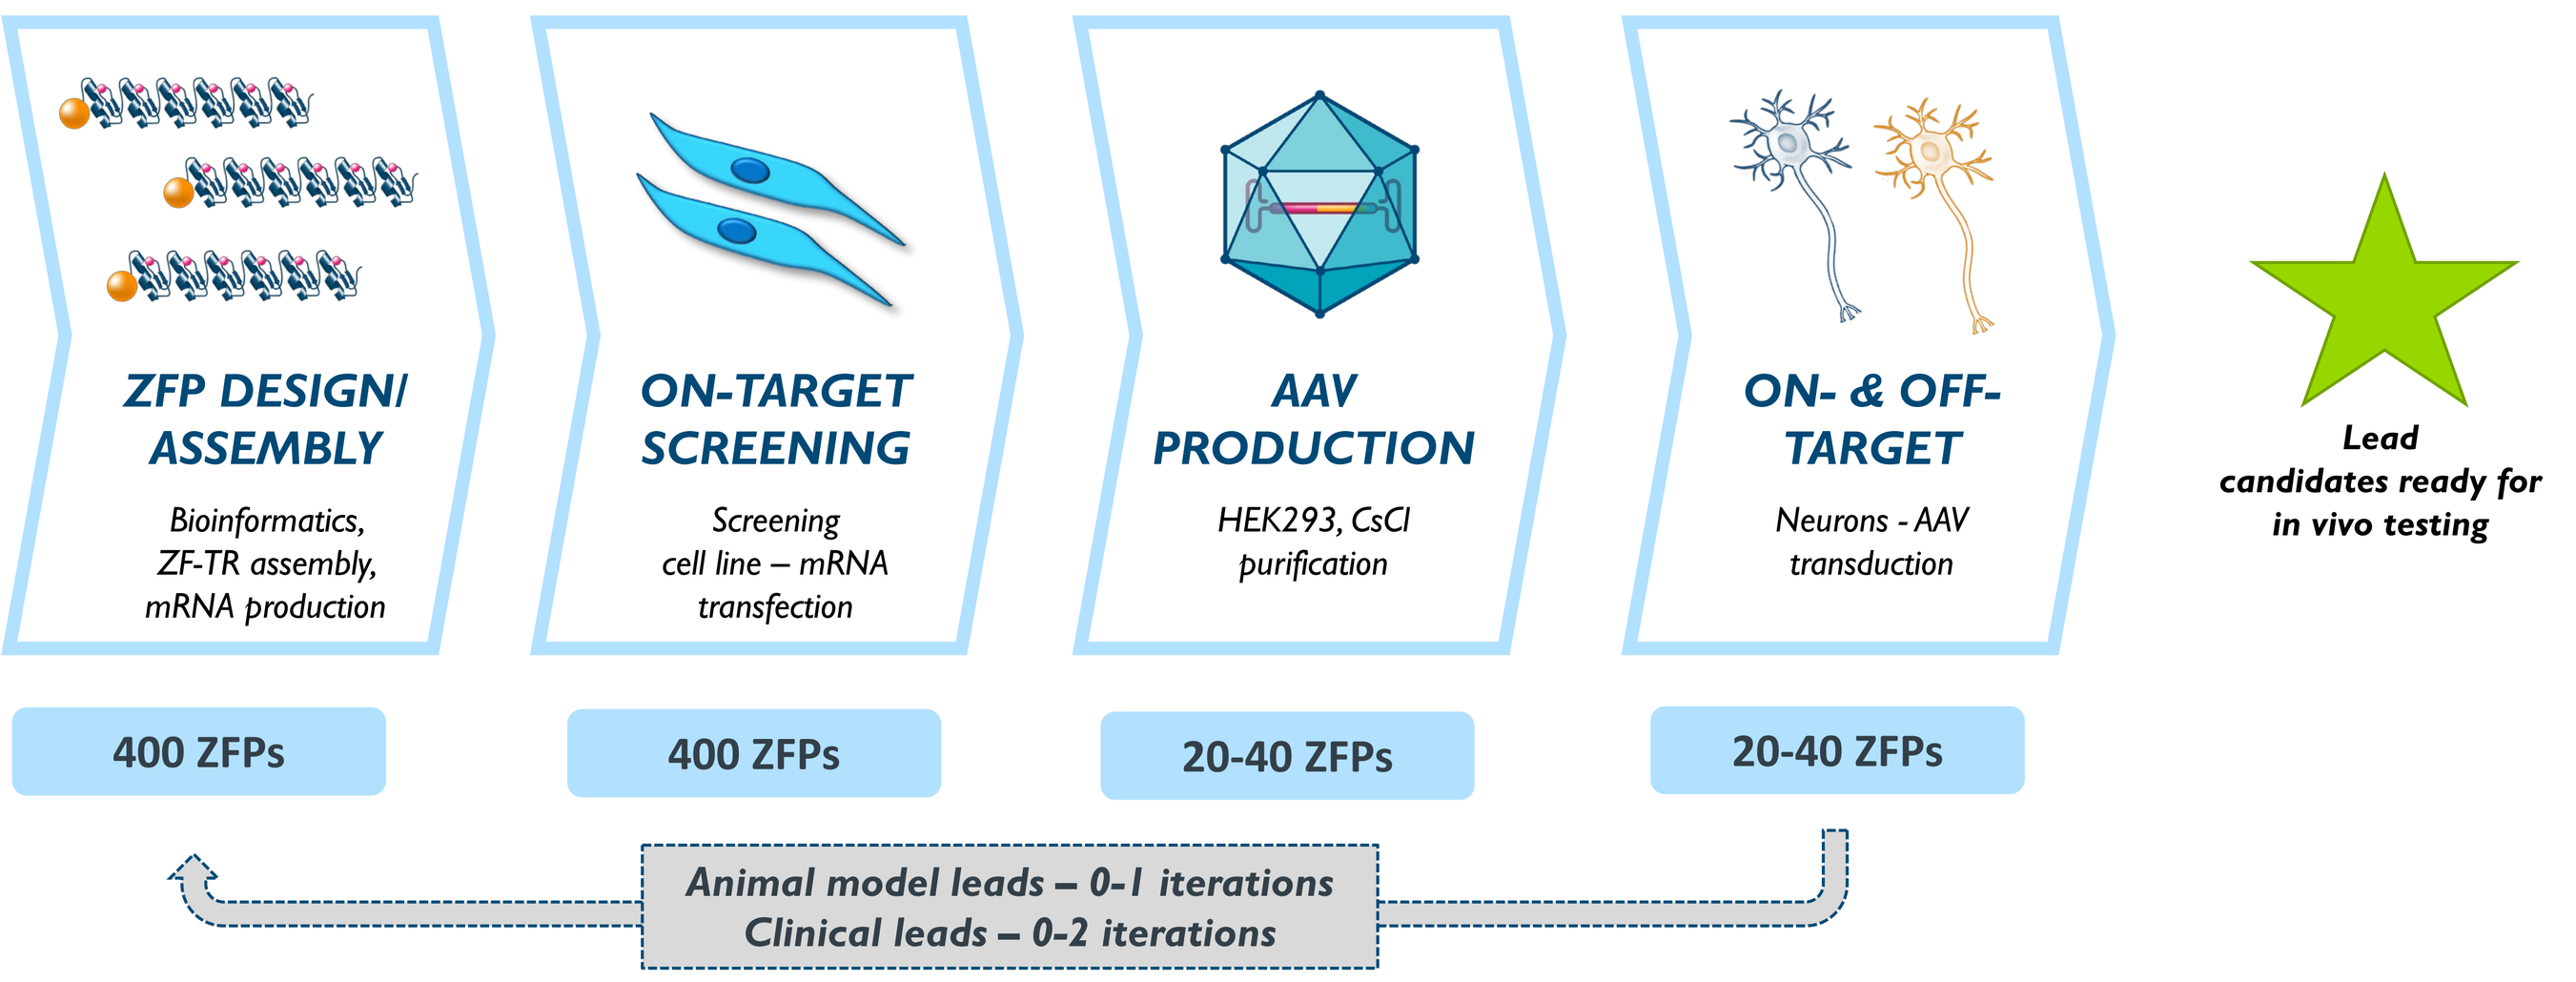
ZFP DESIGN/ ASSEMBLY
Bioinformatics, ZF-TR assembly, mRNA production
400 ZFPs
ON-TARGET SCREENING
Screening cell line – mRNA transfection
400 ZFPs
AAV PRODUCTION
HEK293, CsCl purification
20-40 ZFPs
ON- & OFF-TARGET
Neurons - AAV transduction
20-40 ZFPs
Lead candidates ready for in vivo testing
Animal model leads – 0-1 iterations
Clinical leads – 0-2 iterations

Figure 1. High-throughput workflow for identifying potent and selective ZF-TRs.

For each gene target, we use a proprietary design algorithm to generate hundreds of candidate ZF-TRs. These ZF-TRs are then assembled, transcribed into mRNA and screened in cell lines for on-target activity.

For a subset of ZF-TRs with desirable on-target activity, we then manufacture AAV-ZF-TR vectors and perform on- and off-target screening in primary and/or iPSC-derived neurons to identify lead candidates for in vivo testing.

If necessary, we perform additional iterations of this process to identify animal model (1 iteration) or clinical (2 iterations) leads.

High-throughput screening in cell lines identifies ZF Repressors with desirable on-target activity

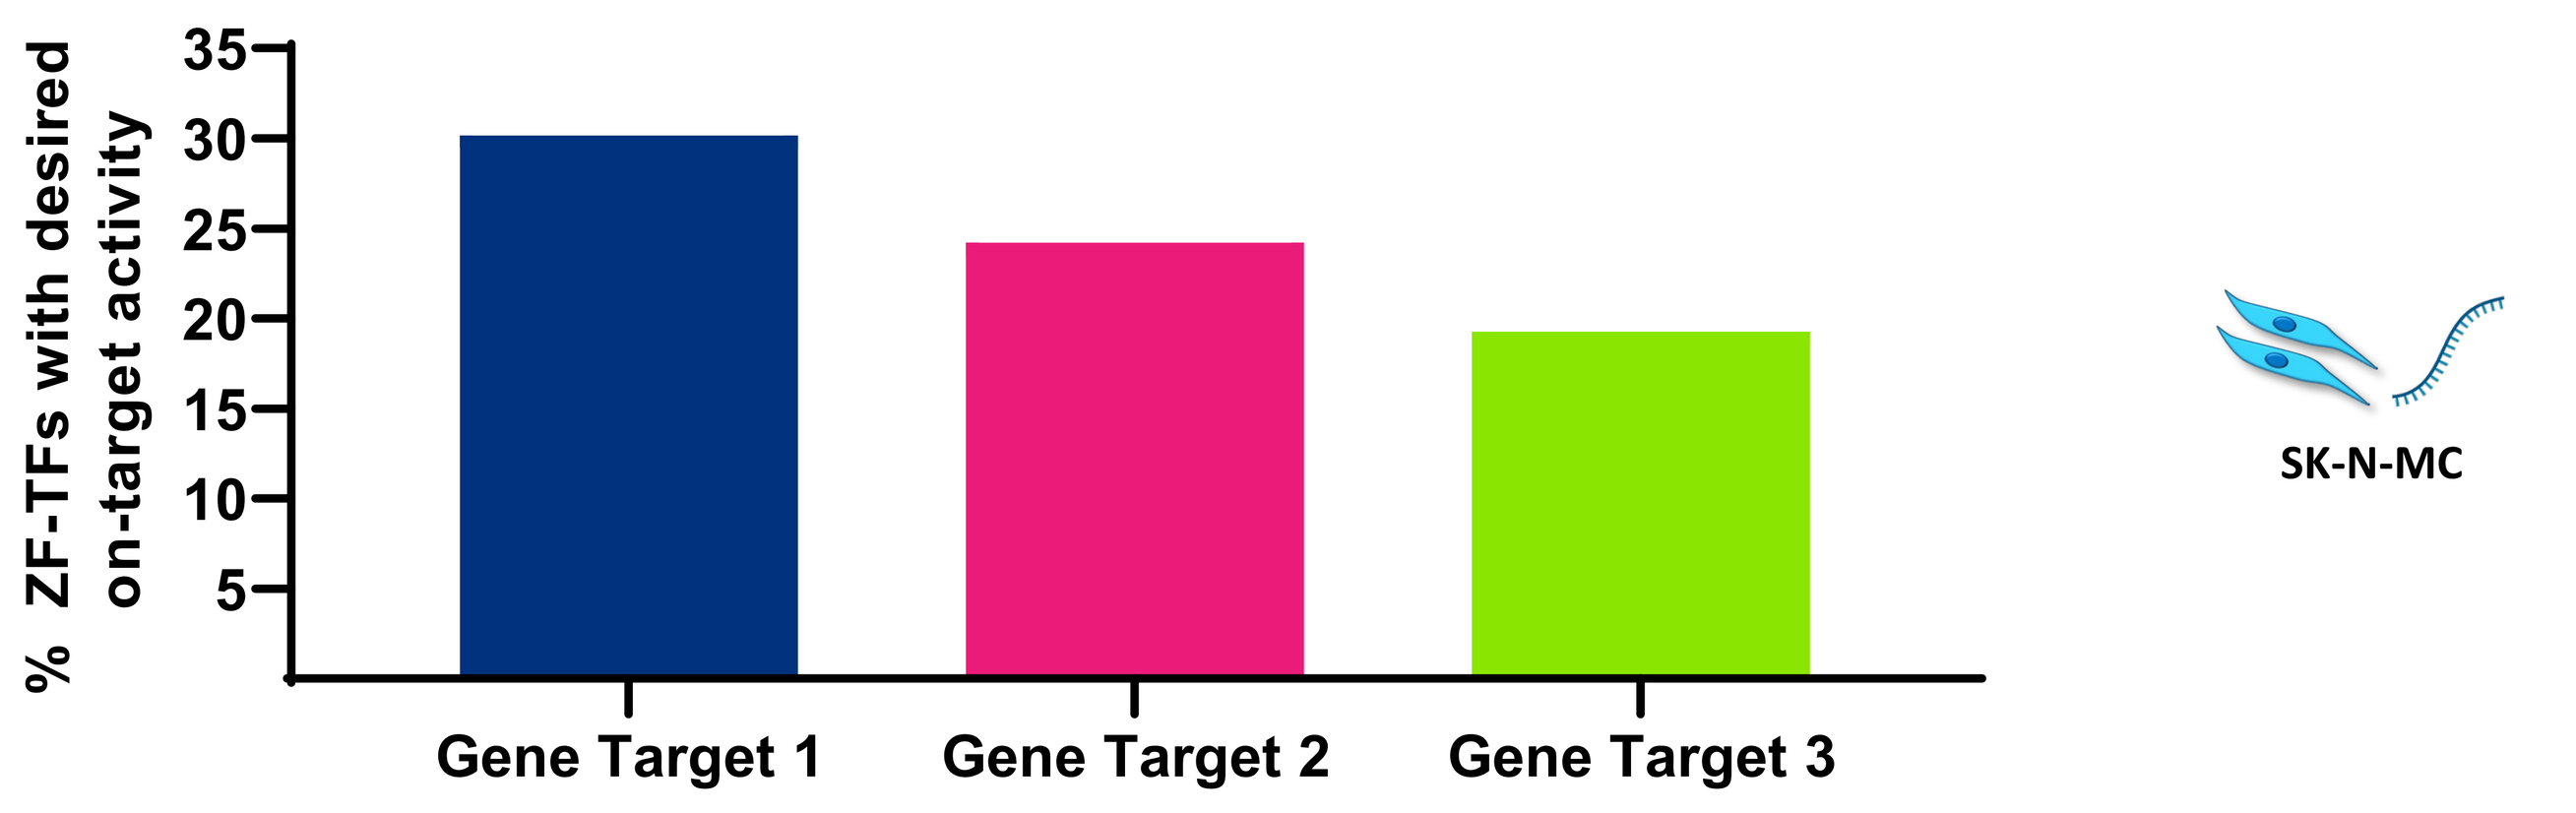
% ZF-TFs with desired on-target activity
35
30
25
20
15
10
5
Gene Target 1
Gene Target 2
Gene Target 3
SK-N-MC

Figure 2. High-throughput screening in cell lines identifies active ZF Repressors.

Hundreds of candidate ZF-TRs targeting multiple neurological disease targets were designed and assembled into plasmids for in vitro transcription.

Using automated methods, transcribed mRNA was transiently transfected into neuroblastoma (SK-N-MC) cells and expressed for ~24 hours prior to analysis by RT-qPCR.

Active ZF Repressors cluster in genomic "hot spots"

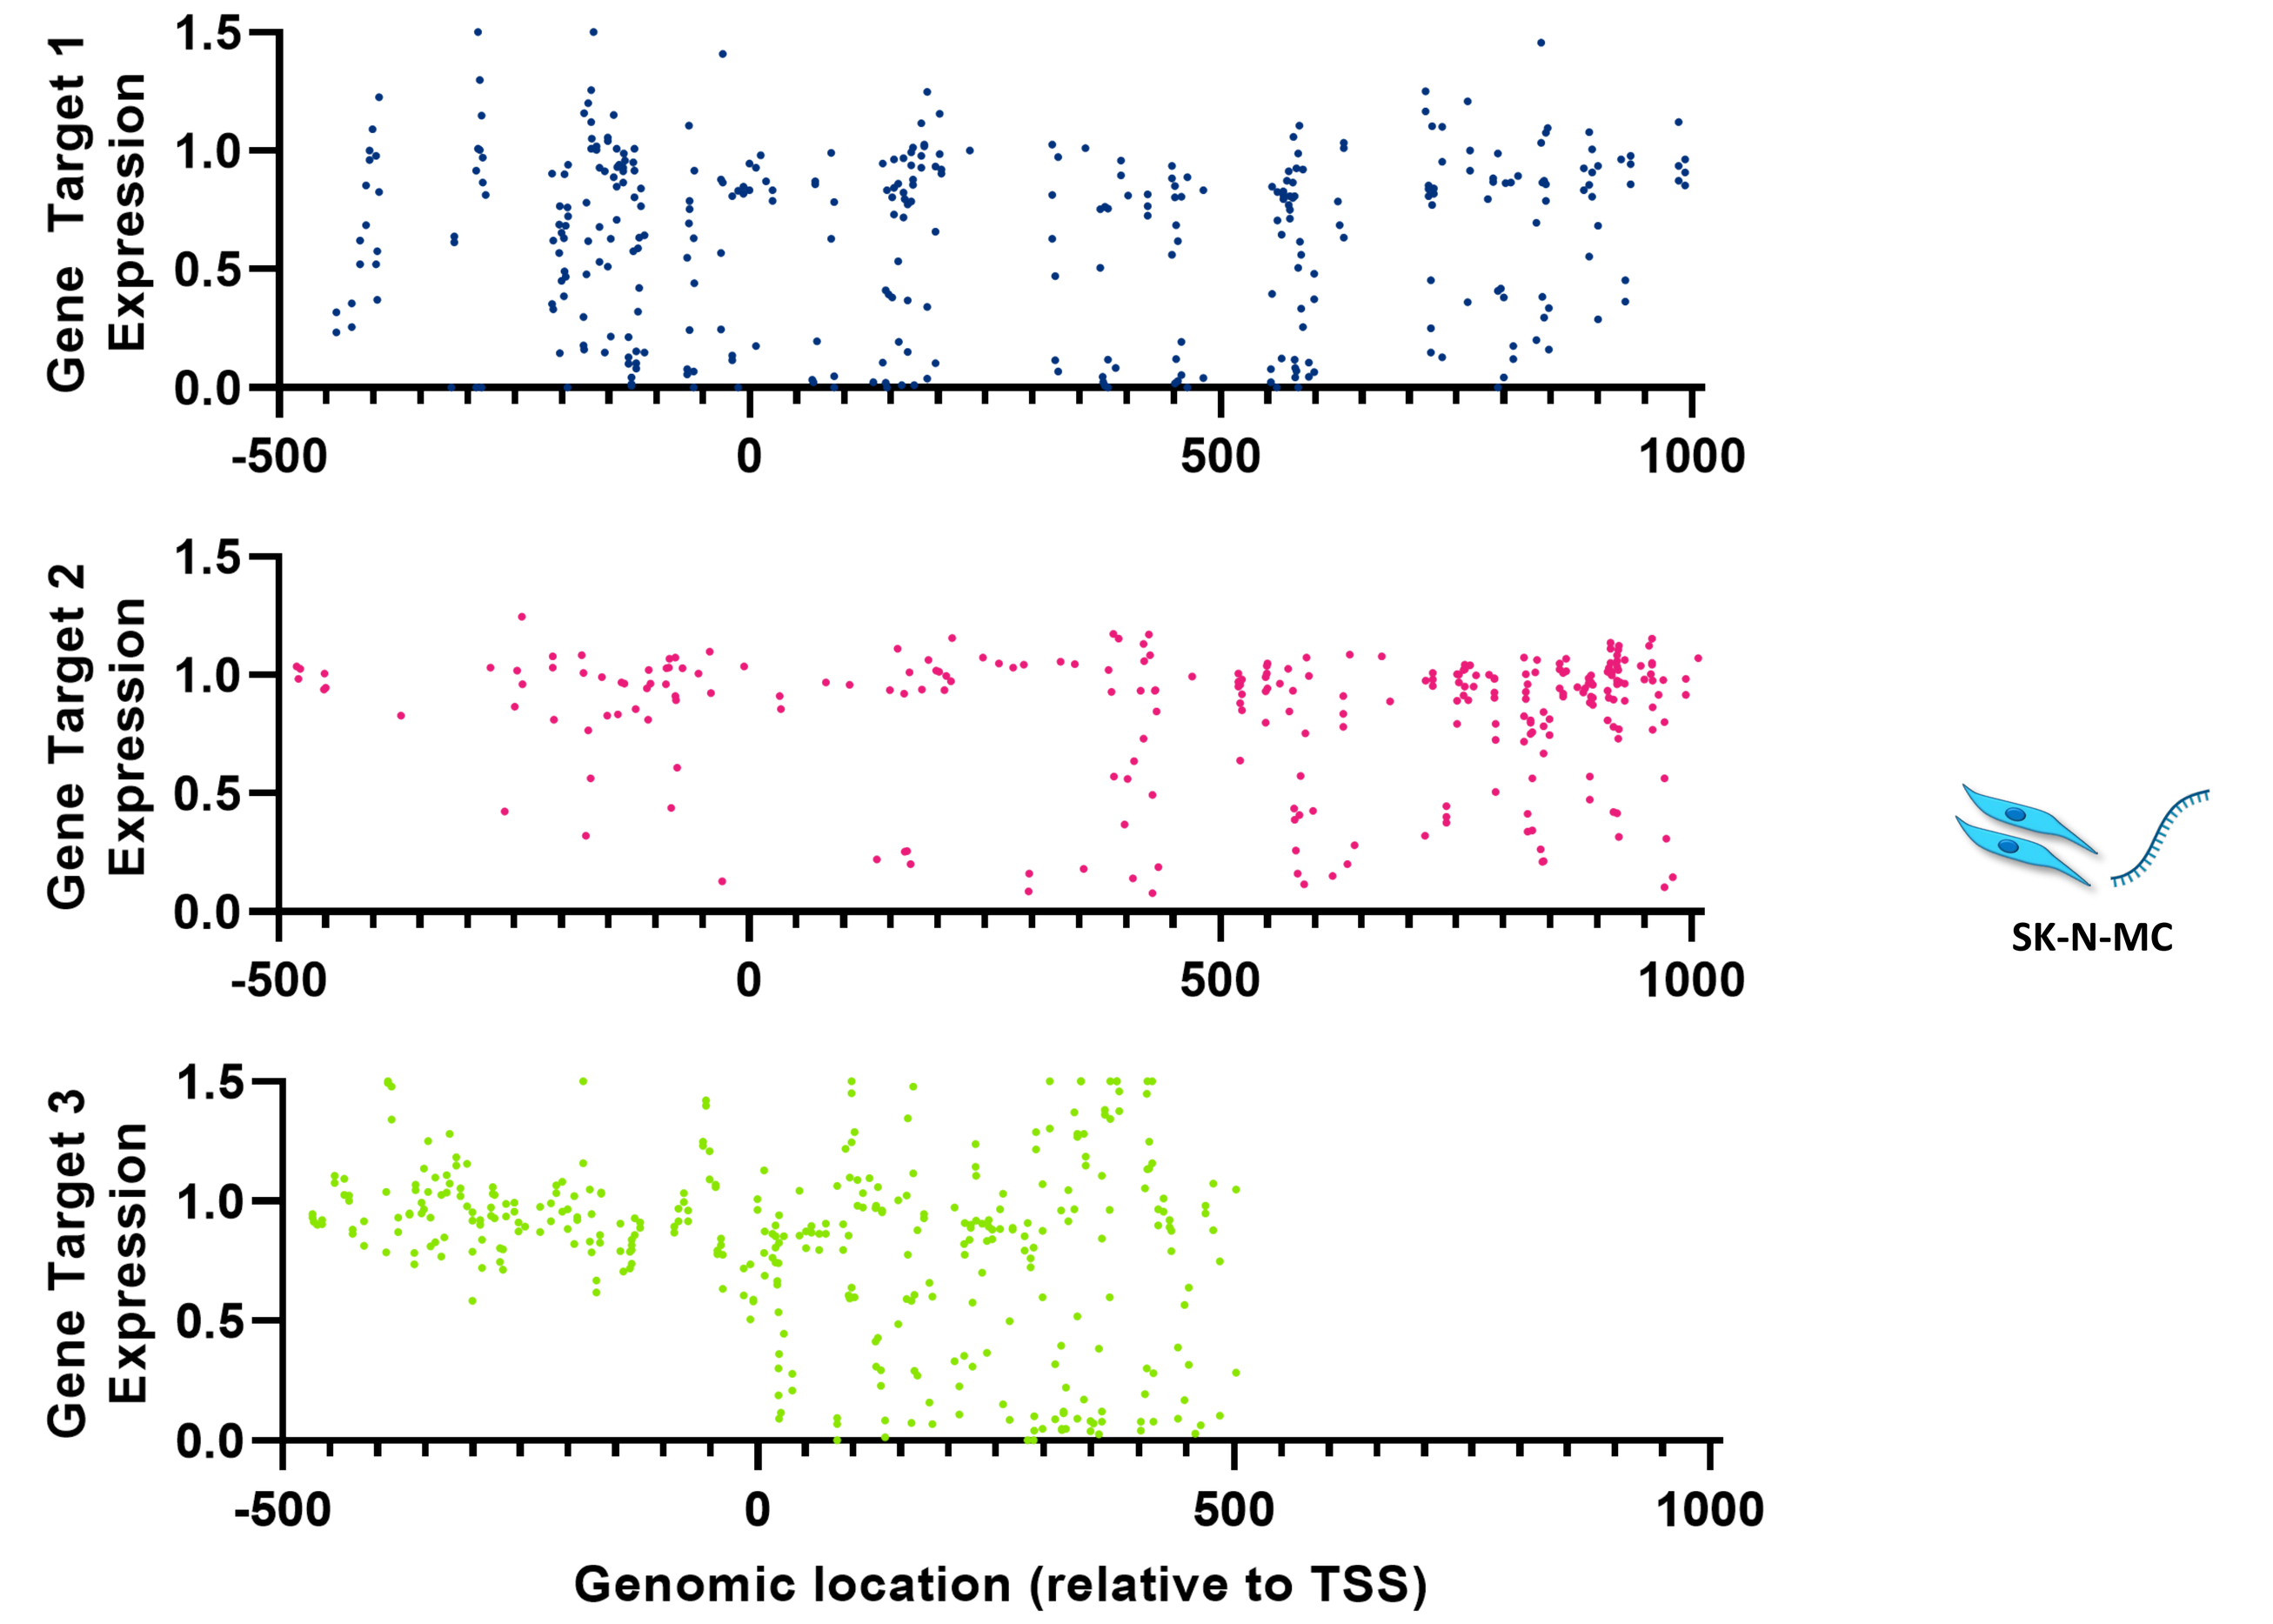
Gene Target 1 Expression
Gene Target 2 Expression
Gene Target 3 Expression
1.5
1.0
0.5
0.0
-500
0
500
1000
SK-N-MC
Genomic location (relative to TSS)

Figure 3. Genomic location for active ZF-TRs relative to transcriptional start sites.

Target gene expression was assessed by RT-qPCR ~24 hours after transfecting neuroblastoma (SK-N-MC) cells with mRNA encoding ZF-TRs targeted immediately up- or down-stream of the major transcriptional start site for each target.

Strong correlation between ZF Repressor activity in screening cells and neurons

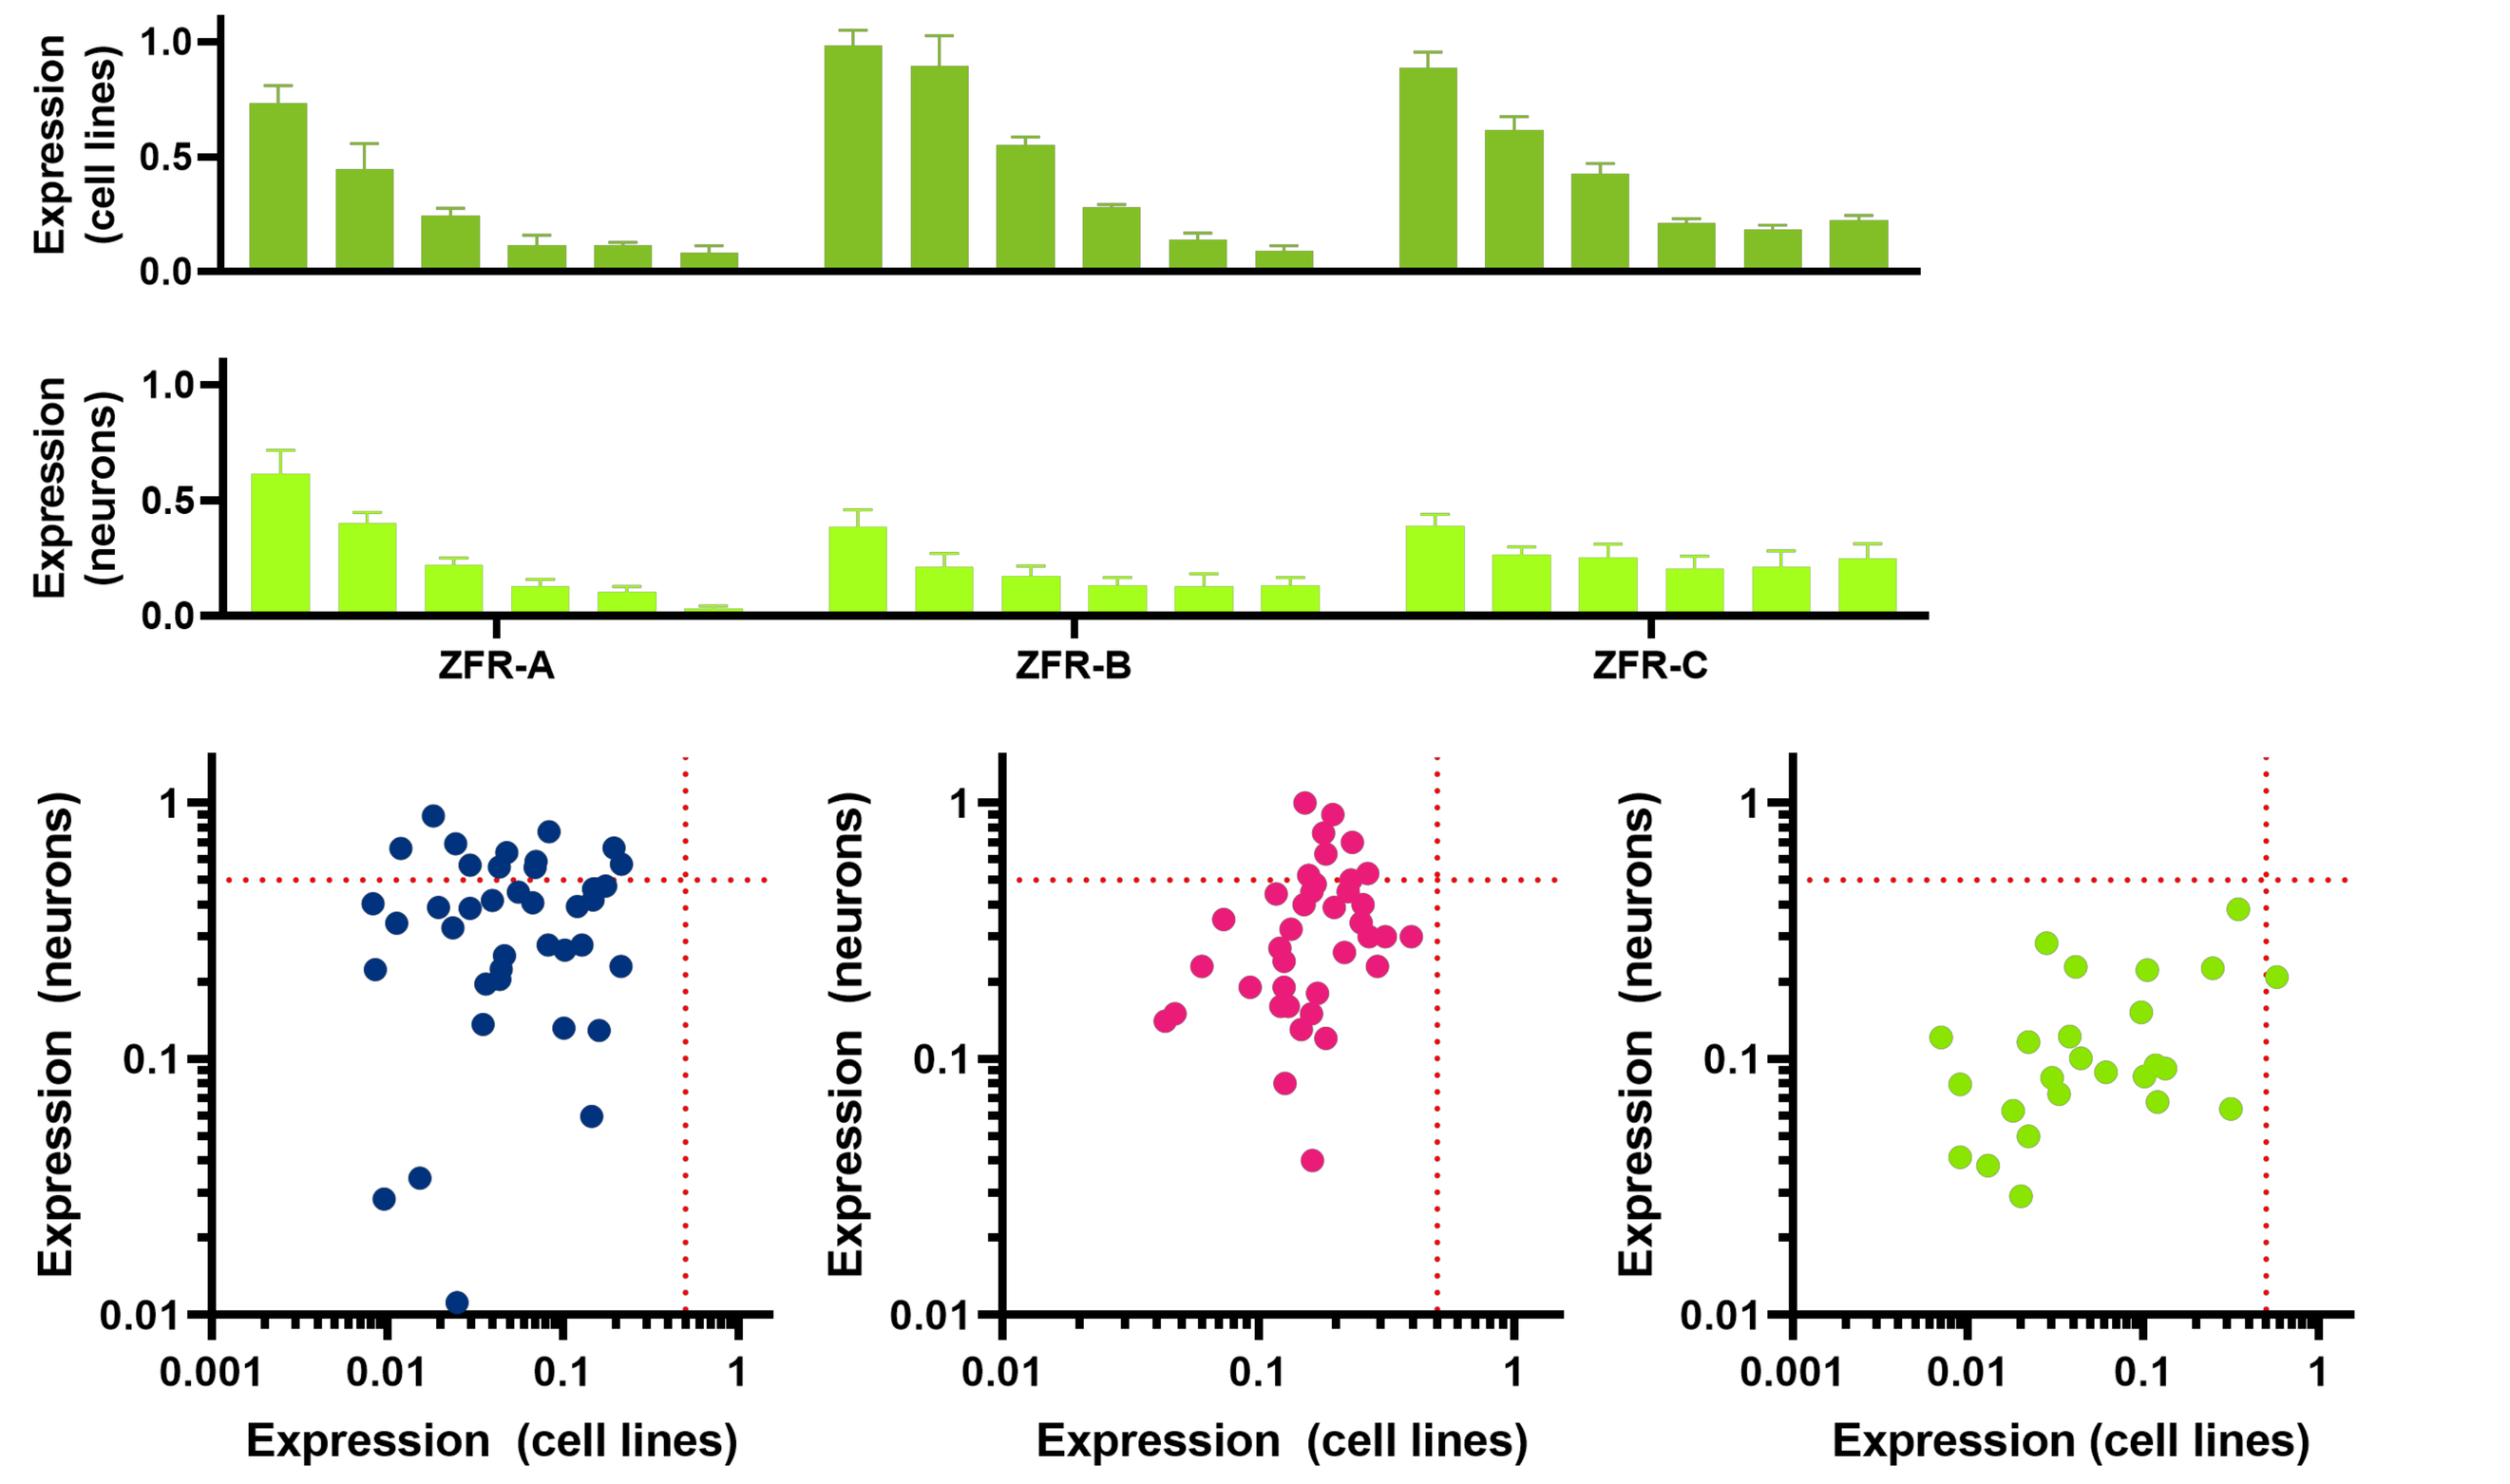
Expression (cell lines)
Expression (neurons)
1.0
0.5
0.0
ZFR-A
ZFR-B
ZFR-C
Expression (neurons)
Expression (cell lines)
1
0.1
0.01
0.001

Figure 4. ZF-TRs repress target expression in screening cells and neurons.

A subset of active ZF-TRs identified in screening cells were cloned into AAV expression vectors, transduced into iPSC-derived neurons, then analyzed by RT-qPCR between 21-31 days post-transduction.

(Top) Dose-response curves showing robust target gene repression after transfection (cell lines) or transduction (neurons) with ZF-TR expression vectors (mean +/- SD).

(Bottom) Correlation plots comparing ZF-TR activity in screening cells and neurons for each target.

First pass screening identifies active ZF Repressors with few to no off-targets

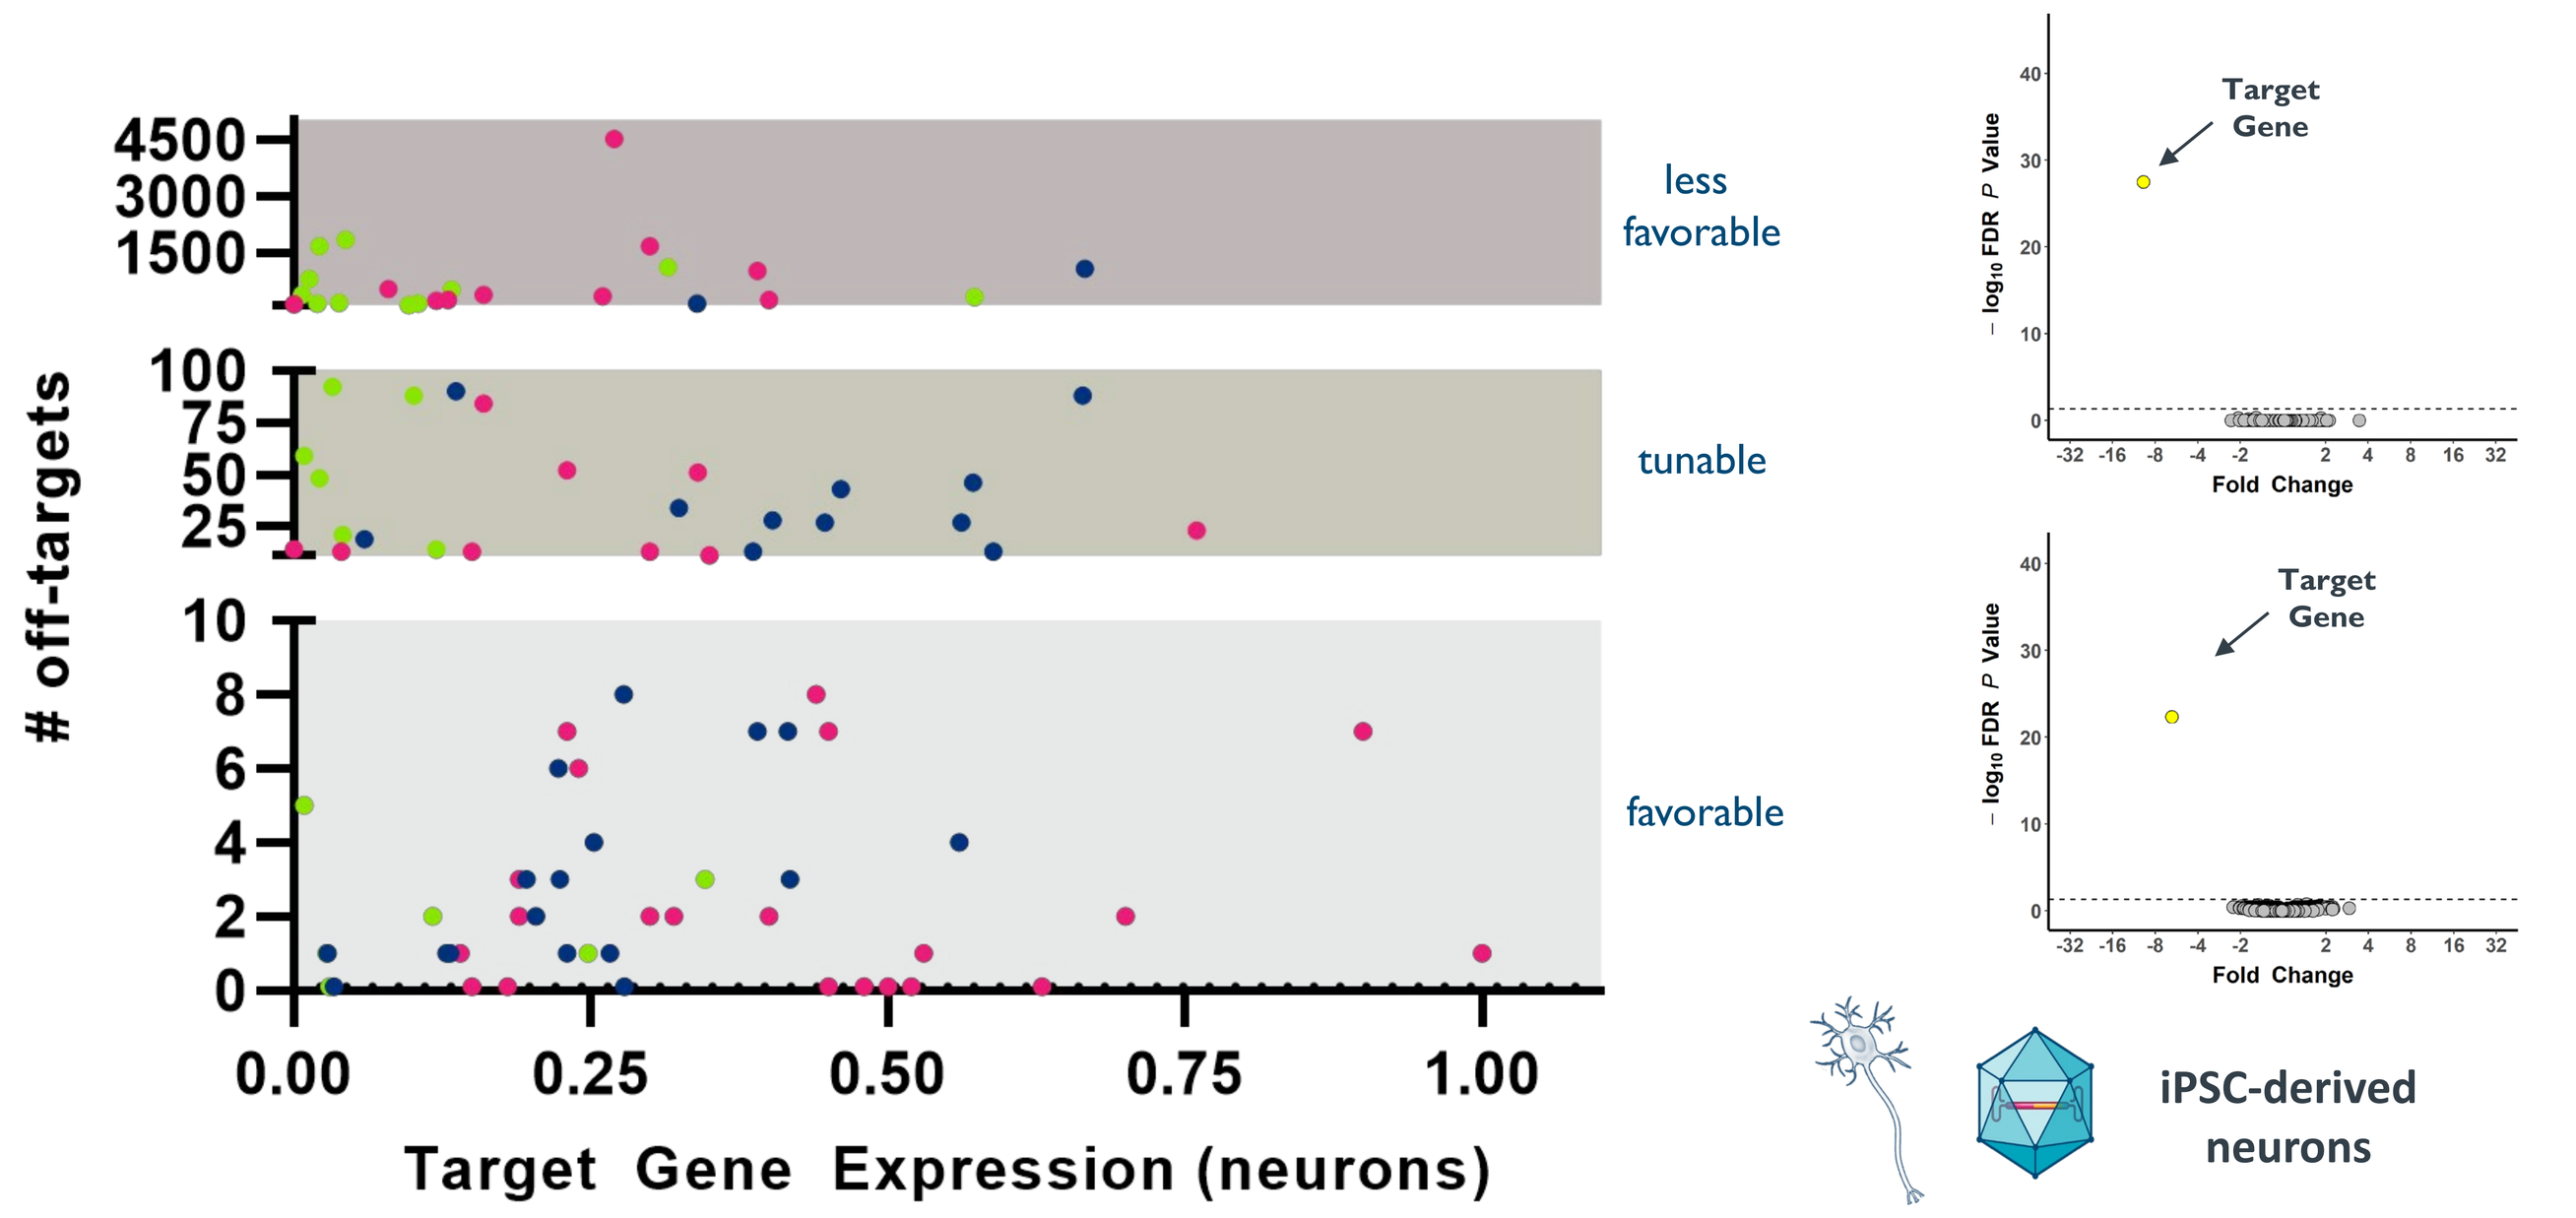
off-targets
4500
3000
1500
100
75
50
25
10
8
6
4
2
0
less favorable
tunable
favorable
0.00
0.25
0.50
0.75
1.00
Target Gene Expression (neurons)
Target Gene
Fold Change
iPSC-derived neurons

Figure 5. ZF-TRs repress target gene expression with minimal off-target activity.

AAV-ZF-TRs were transduced into human iPSC-derived neurons and cultured for 19 days before transcriptomic changes were assessed using high-throughput Affymetrix peg-based microarrays processed on a GeneTitan instrument.

(Left) Correlation plot showing the number of differentially expressed genes (FDR-adjusted p-value > 0.05) between control- and ZF-TR-treated samples (n=4-6) and target gene expression.

(Right) Example volcano plots for AAV-ZF-TRs designed against Gene Target 3.

ZF Repressors enable robust and reliable epigenetic regulation in the mouse brain

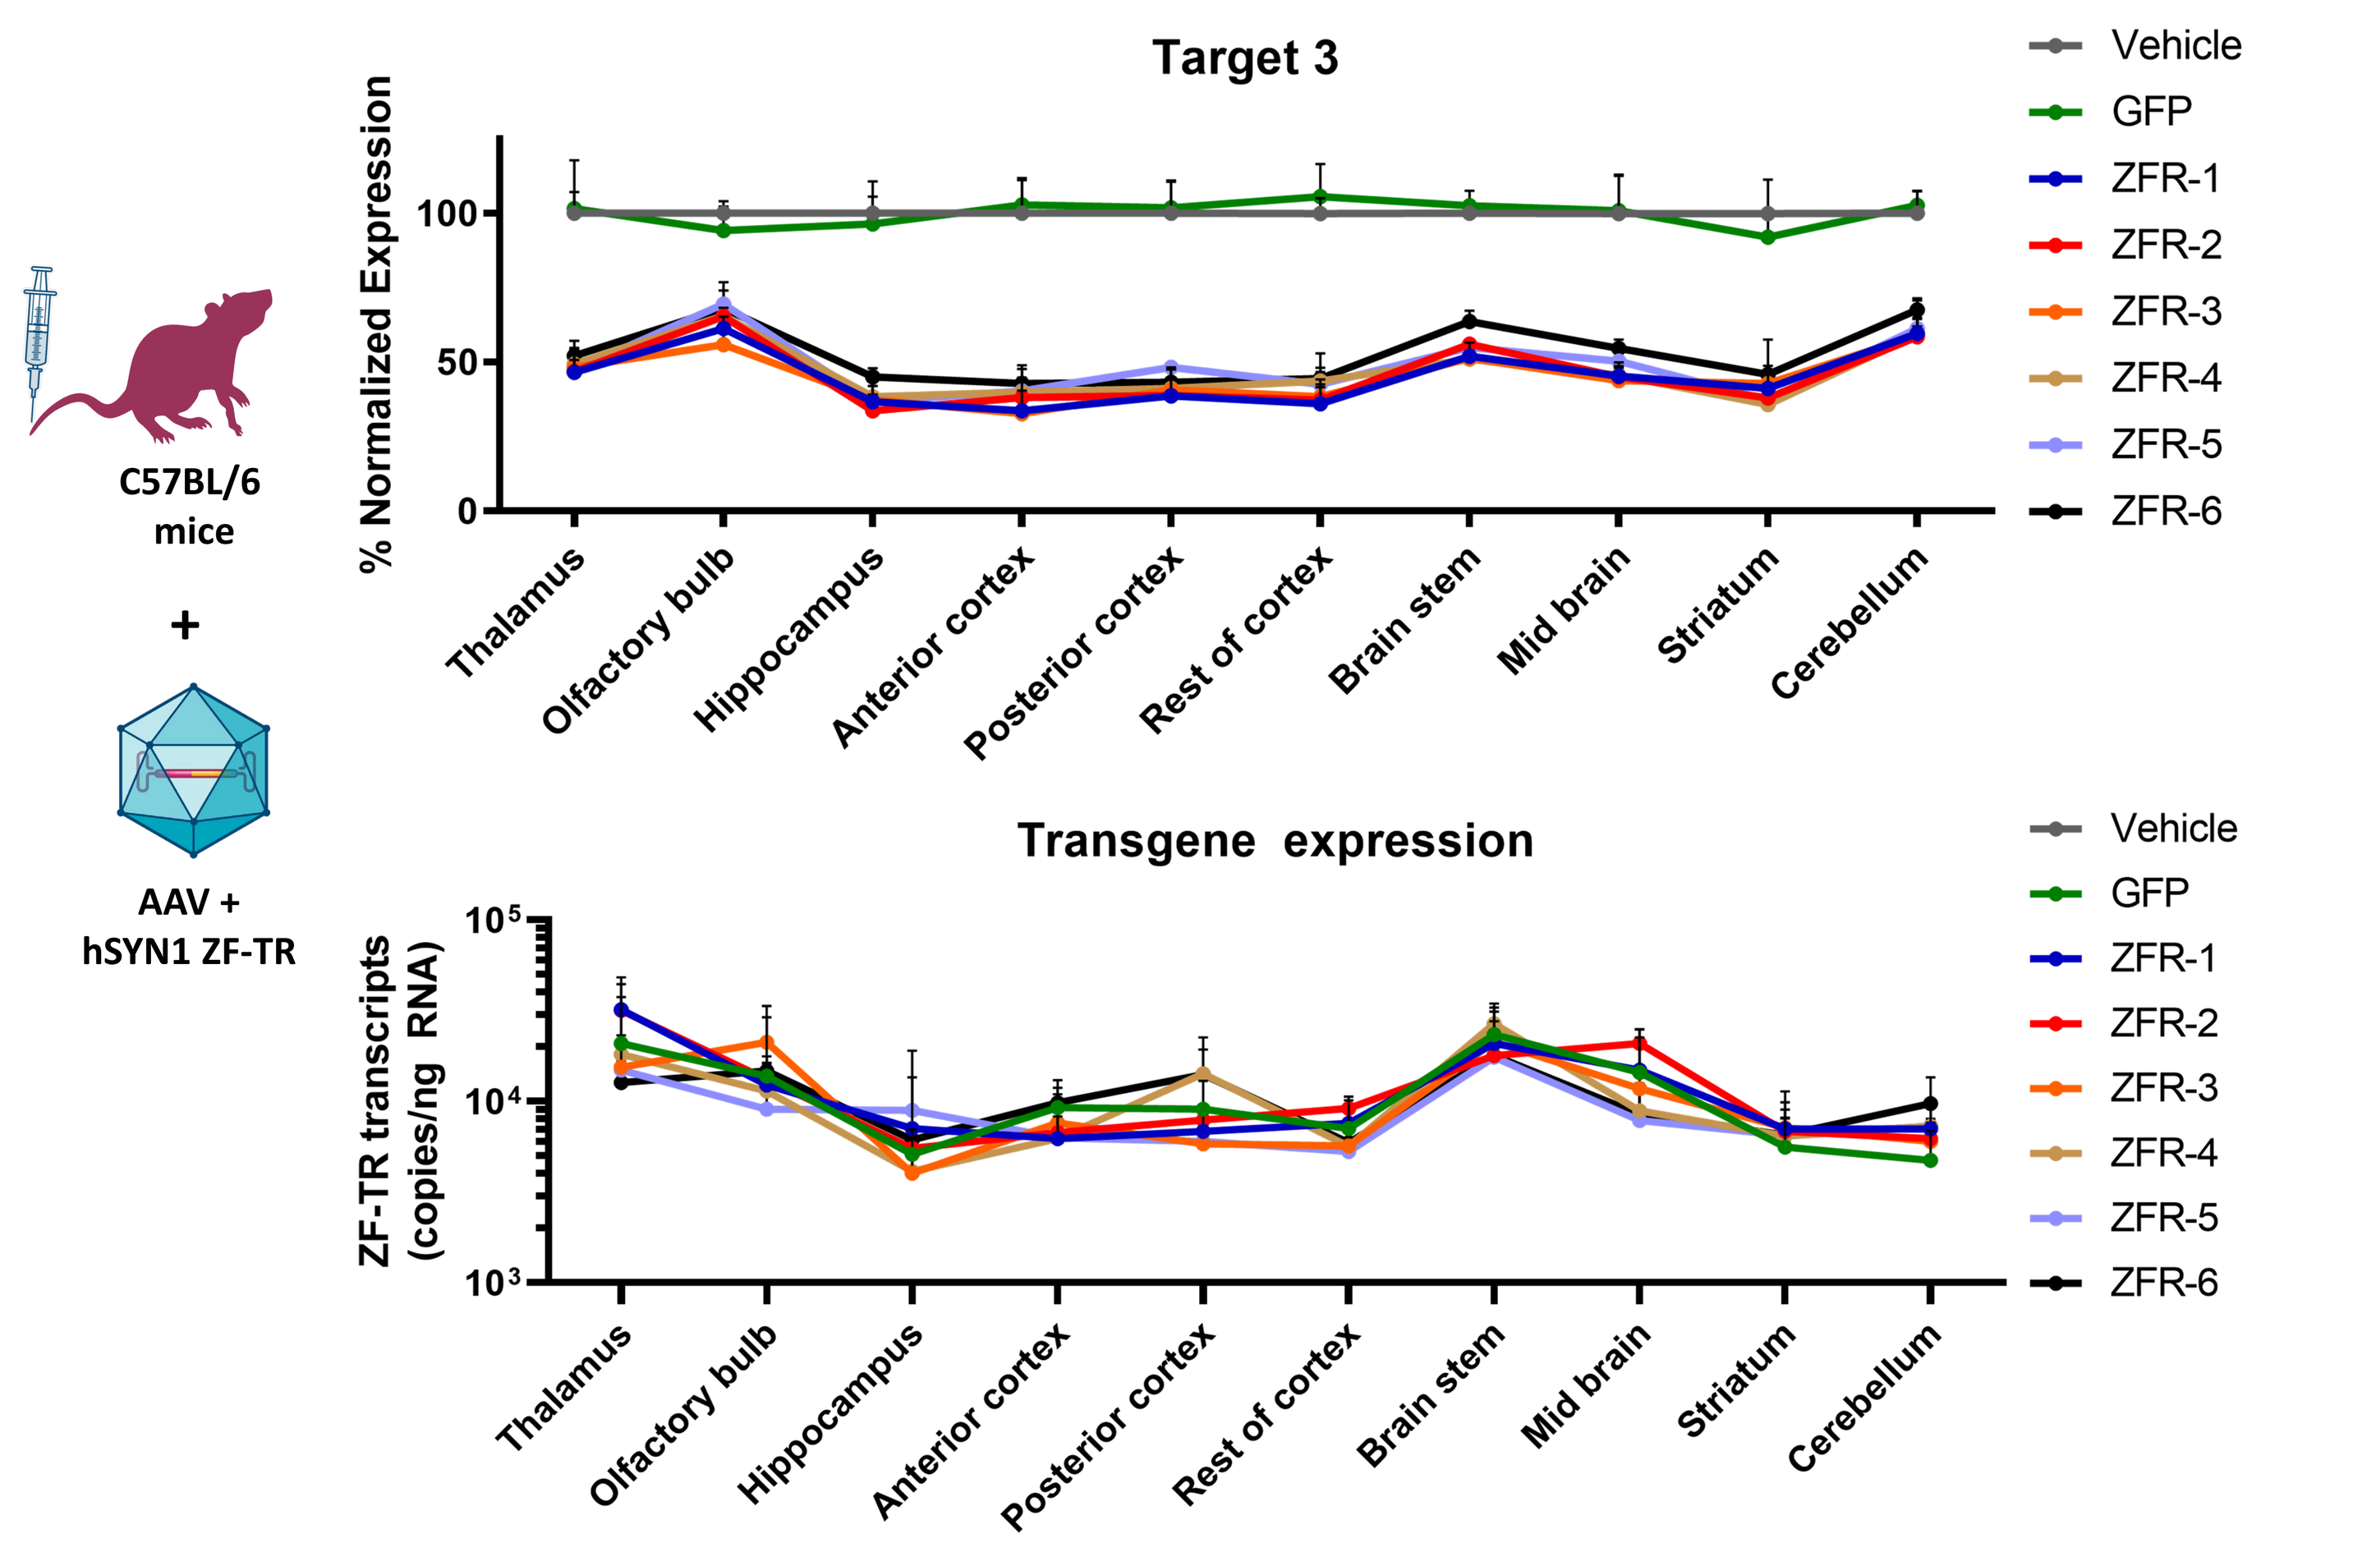
C57BL/6 mice
+
AAV + hSYN1 ZF-TR
Target 3
% Normalized Expression
100
50
0
Transgene expression
ZF-TR transcripts (copies/ng RNA)
10^5
10^4
10^3
Thalamus
Olfactory bulb
Hippocampus
Anterior cortex
Posterior cortex
Rest of cortex
Brain stem
Mid brain
Striatum
Cerebellum
Vehicle
GFP
ZFR-1
ZFR-2
ZFR-3
ZFR-4
ZFR-5
ZFR-6

Figure 6. ZF-TRs enable robust and reliable epigenetic regulation in the mouse brain.

Select AAV-ZF-TRs with potent on-target activity and minimal to no off-targets in vitro and 40-70% expected bulk target repression in vivo (based on known target gene expression patterns and transduction efficiencies) were administered intravenously into C57BL/6 mice (n=4-8 mice per group) under the control of a neuron-specific promoter (human synapsin I). Five weeks post-injection, total RNA was isolated from several brain regions and analyzed by RT-qPCR.

(Top) Target3 gene expression normalized to the mean of Atp5b, Eif4a2 and Gapdh.

(Bottom) Transgene expression normalized to total RNA input. All data are shown as mean +/- SD.

Conclusion

- We created an optimized screening platform that efficiently identified potent and selective ZF Transcriptional Regulators that target novel regulatory sites for multiple genes implicated in neurodegenerative and neurodevelopmental diseases.
- Our results highlight the potential of the ZF Transcriptional Regulator platform to develop potential genomic medicines for additional devastating neurological diseases.

Presented at the ASGCT 26th Annual Meeting, 2023