Multimerization of Chimeric Antigen Receptor (CAR) binding domains: A solution to assess tissue specificity of low to medium affinity scFv

Poster P503

Claire Gendrin*[1], Stephanie Beghelli*[1], Irene Schneider[2], Yue Cui[1], Julie Gartner Dardenne[1], Maurus de la Rosa[1] and Tobias Abel[1]

[1] Sangamo Therapeutics France, Valbonne, France. [2] Schneider Consulting, Frankfurt, Germany.

Sangamo THERAPEUTICS

Introduction

In cell therapy, non-specific binding or specific binding of drugs to non-target tissues can result in adverse events and lack of efficacy. This is especially detrimental in the context of Chimeric Antigen Receptor (CAR) therapies. Hence, a careful assessment of a CAR's specificity is imperative prior to clinical application by determining the on- and off-target binding. ScFv affinities in CARs vary generally between low to medium, making this assessment challenging when detecting low target-expressing cells by flow cytometry, or when using standard immunohistochemical (IHC) protocols for tissues.

In the past, high-affinity surrogate antibodies have been used to circumvent this problem, which is time-consuming and requires comparability studies. Here, we demonstrate an avidity enhancement approach to allow rapid, sensitive detection and characterization of the CAR without the need for extensive comparability studies.

Concept

A Increase binding strength by avidity enhancement

Increasing the number of binders in a protein enhances the apparent binding strength (Avidity) to its target.

This concept is widely used in nature to increase the binding of antibodies before high-affinity clones can be selected.

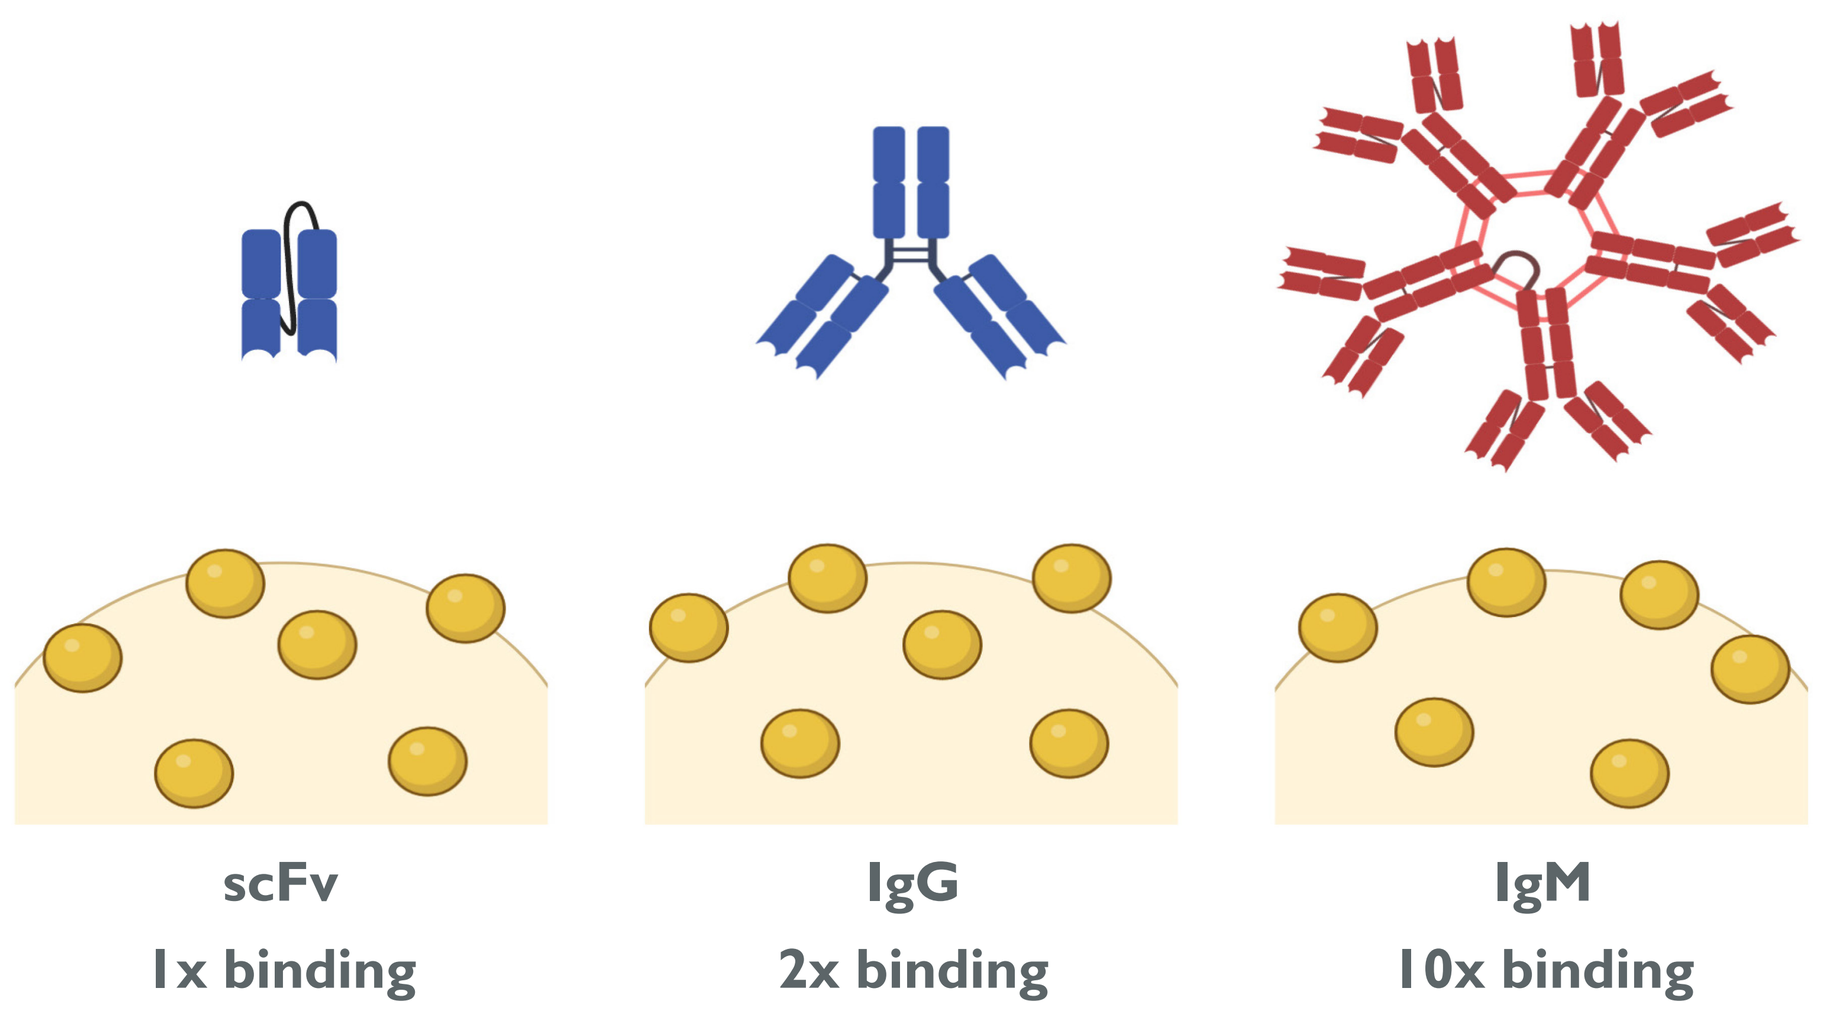
scFv
1x binding
IgG
2x binding
IgM
10x binding

B Avidity enhancement of CAR binding domain

Applying the same avidity concept to scFv, the binding domains of our CARs, we intend to increase their binding without changing specificity.

Monomeric scFvs were either tagged with a biotinylation domain, dimerized via an FC-tag, or multimerized by a dextran polymer. Detection was performed either with streptavidin, the respective tag or via a fluorophore coupled to the dextran-polymer.

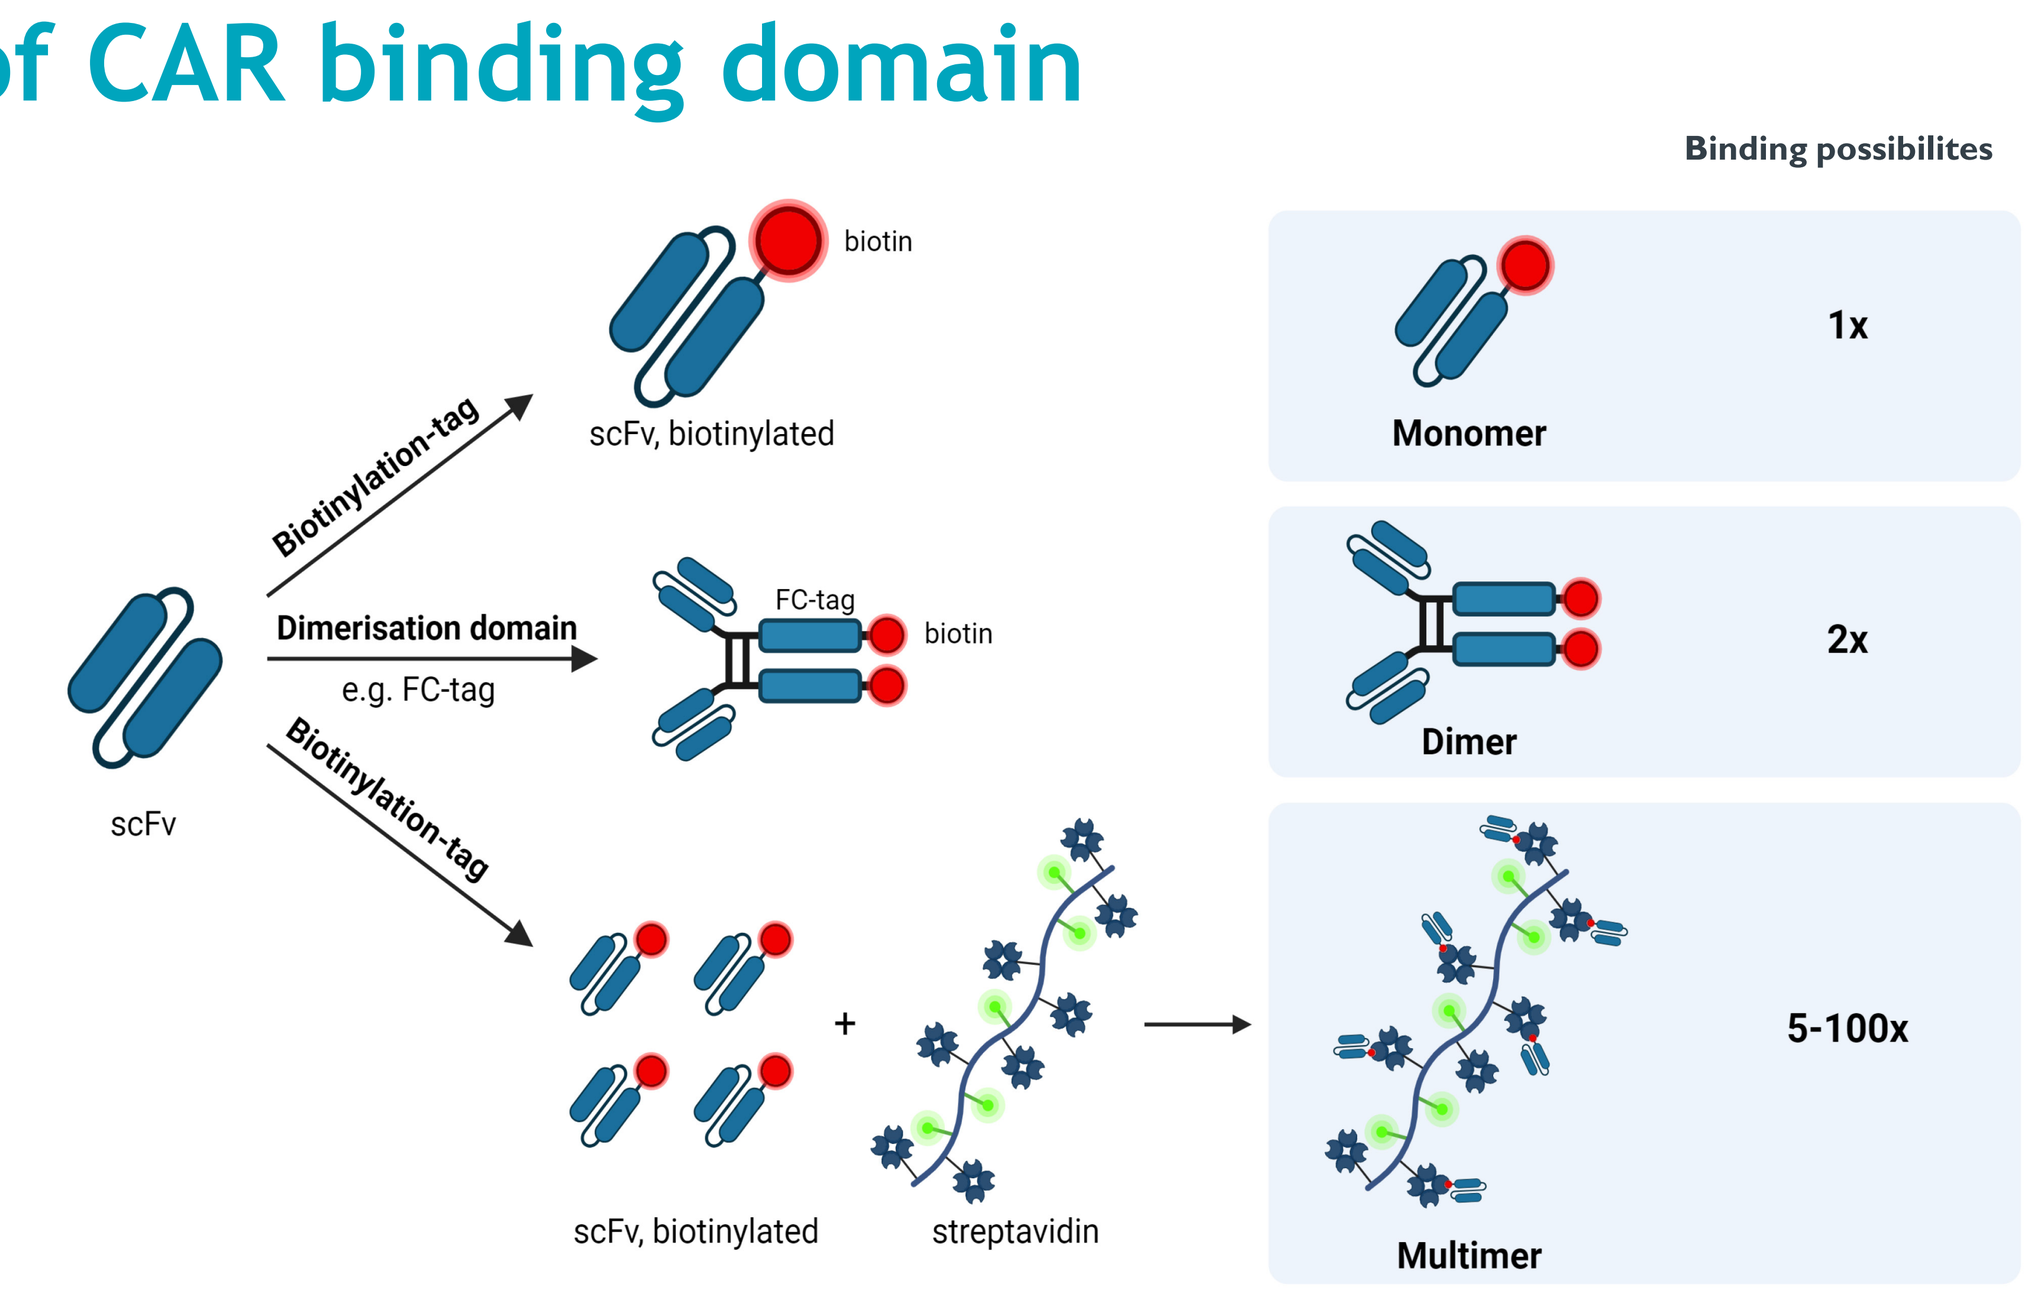
Binding possibilites
biotin
scFv, biotinylated
Biotinylation-tag
Dimerisation domain
e.g. FC-tag
FC-tag
biotin
Biotinylation-tag
scFv
scFv, biotinylated
streptavidin
Monomer
1x
Dimer
2x
Multimer
5-100x

Results

1 Ratio of binder to polymer is imperative for best activity

A dextran polymer can accommodate a certain number of binders. The ideal ratio needs to be assessed for every scFv/binder to achieve optimal results on their target.

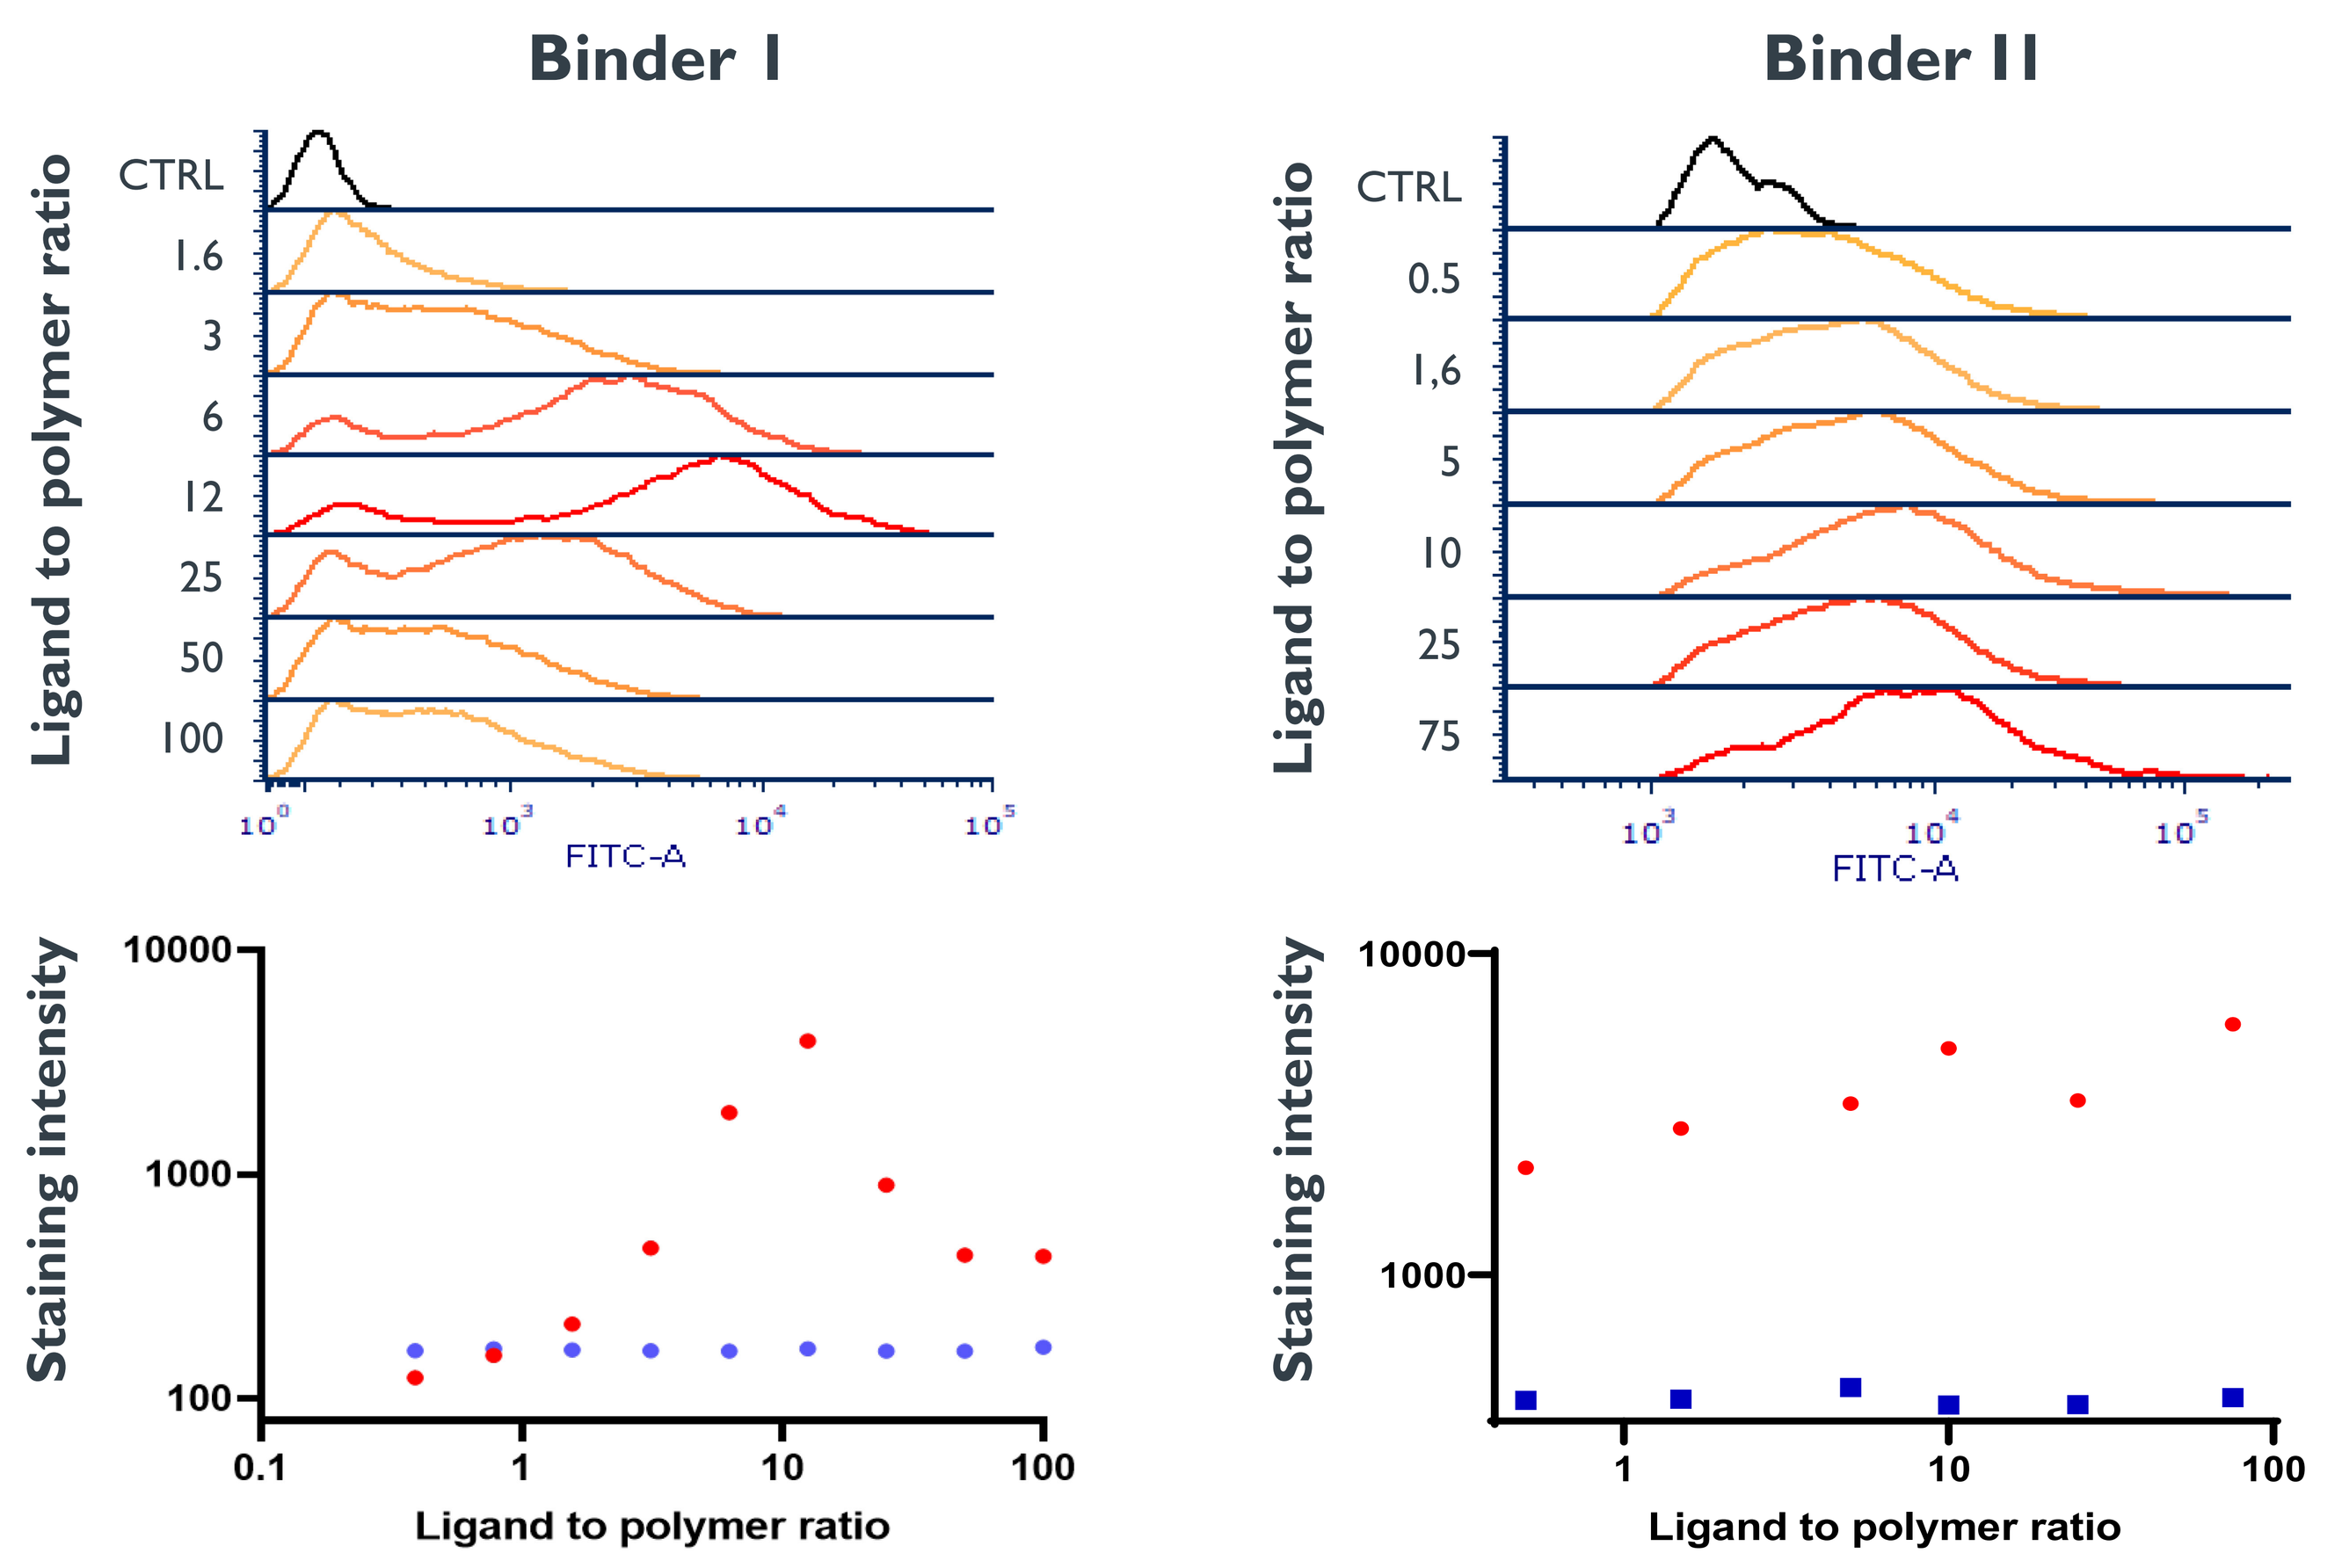
Binder I
Binder II
Ligand to polymer ratio
CTRL
1.6
3
6
12
25
50
100
CTRL
0.5
1,6
5
10
25
75
FITC-A
Staining intensity
10000
1000
100
Ligand to polymer ratio
0.1
1
10
100

Staining efficiency or different binder to polymer ratios were tested on transgenic cell lines expressing relevant levels of the respective antigen by Flow cytometry ***(upper panel).*** Median staining intensities are plotted in relation to B/P ratio on the respective target cell line (red), or the parental negative cell line (blue). ***(lower panel)***

Results demonstrate that Binder I obtains the highest signal to background a ratio of 12:1, while Binder 2 has its optimal performance at 75:1.

2 5-6x Increase of affinity after dimerization of scFv

Using a bead-based affinity assay, an increase in EC50 was observed for several cases of dimerization.

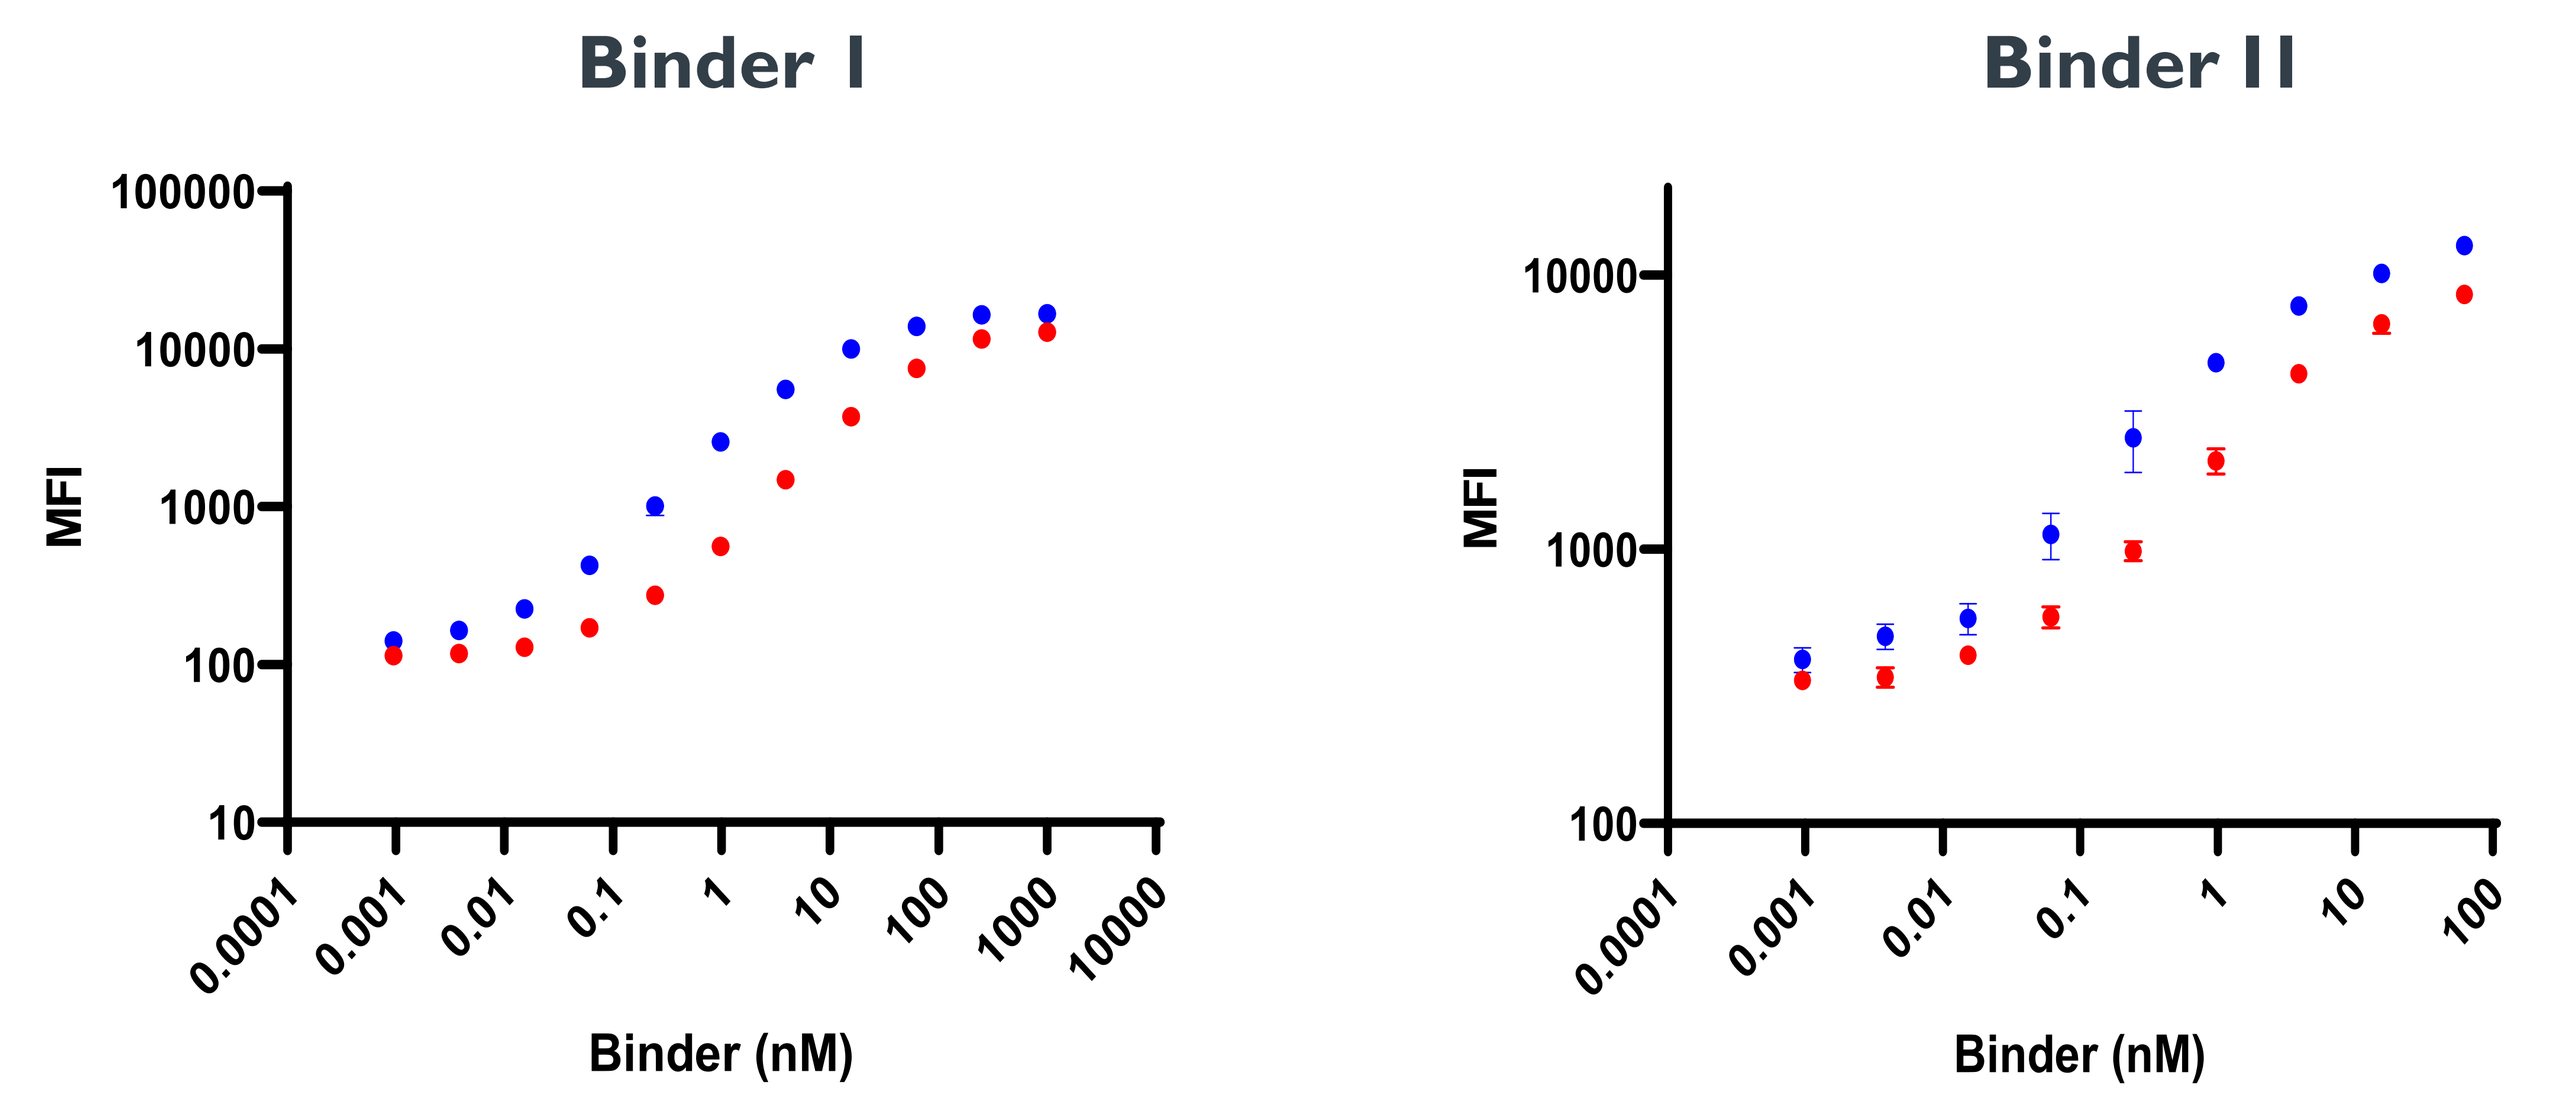
Binder I
Binder II
MFI
100000
10000
1000
100
10
0.0001
0.001
0.01
0.1
1
10
100
1000
10000
Binder (nM)

For both binders, an expected increase in avidity (EC50) was observed after dimerization (blue) of the monomeric scFv (red).
Depending on the respective binder and target, the increase in affinity was accompanied by a significant increase in maximum staining intensity (MFI) **(right)**.

3 Strongly increased sensitivity by multimerization

To demonstrate the increase in sensitivity, IL23R-multimer constructs were tested on transgenic Jurkat cell lines expressing either a low or a medium amount of IL23R (*see below*). For proof of IL23R expression a highly sensitive 3xFLAG-tag was genetically fused to the protein.

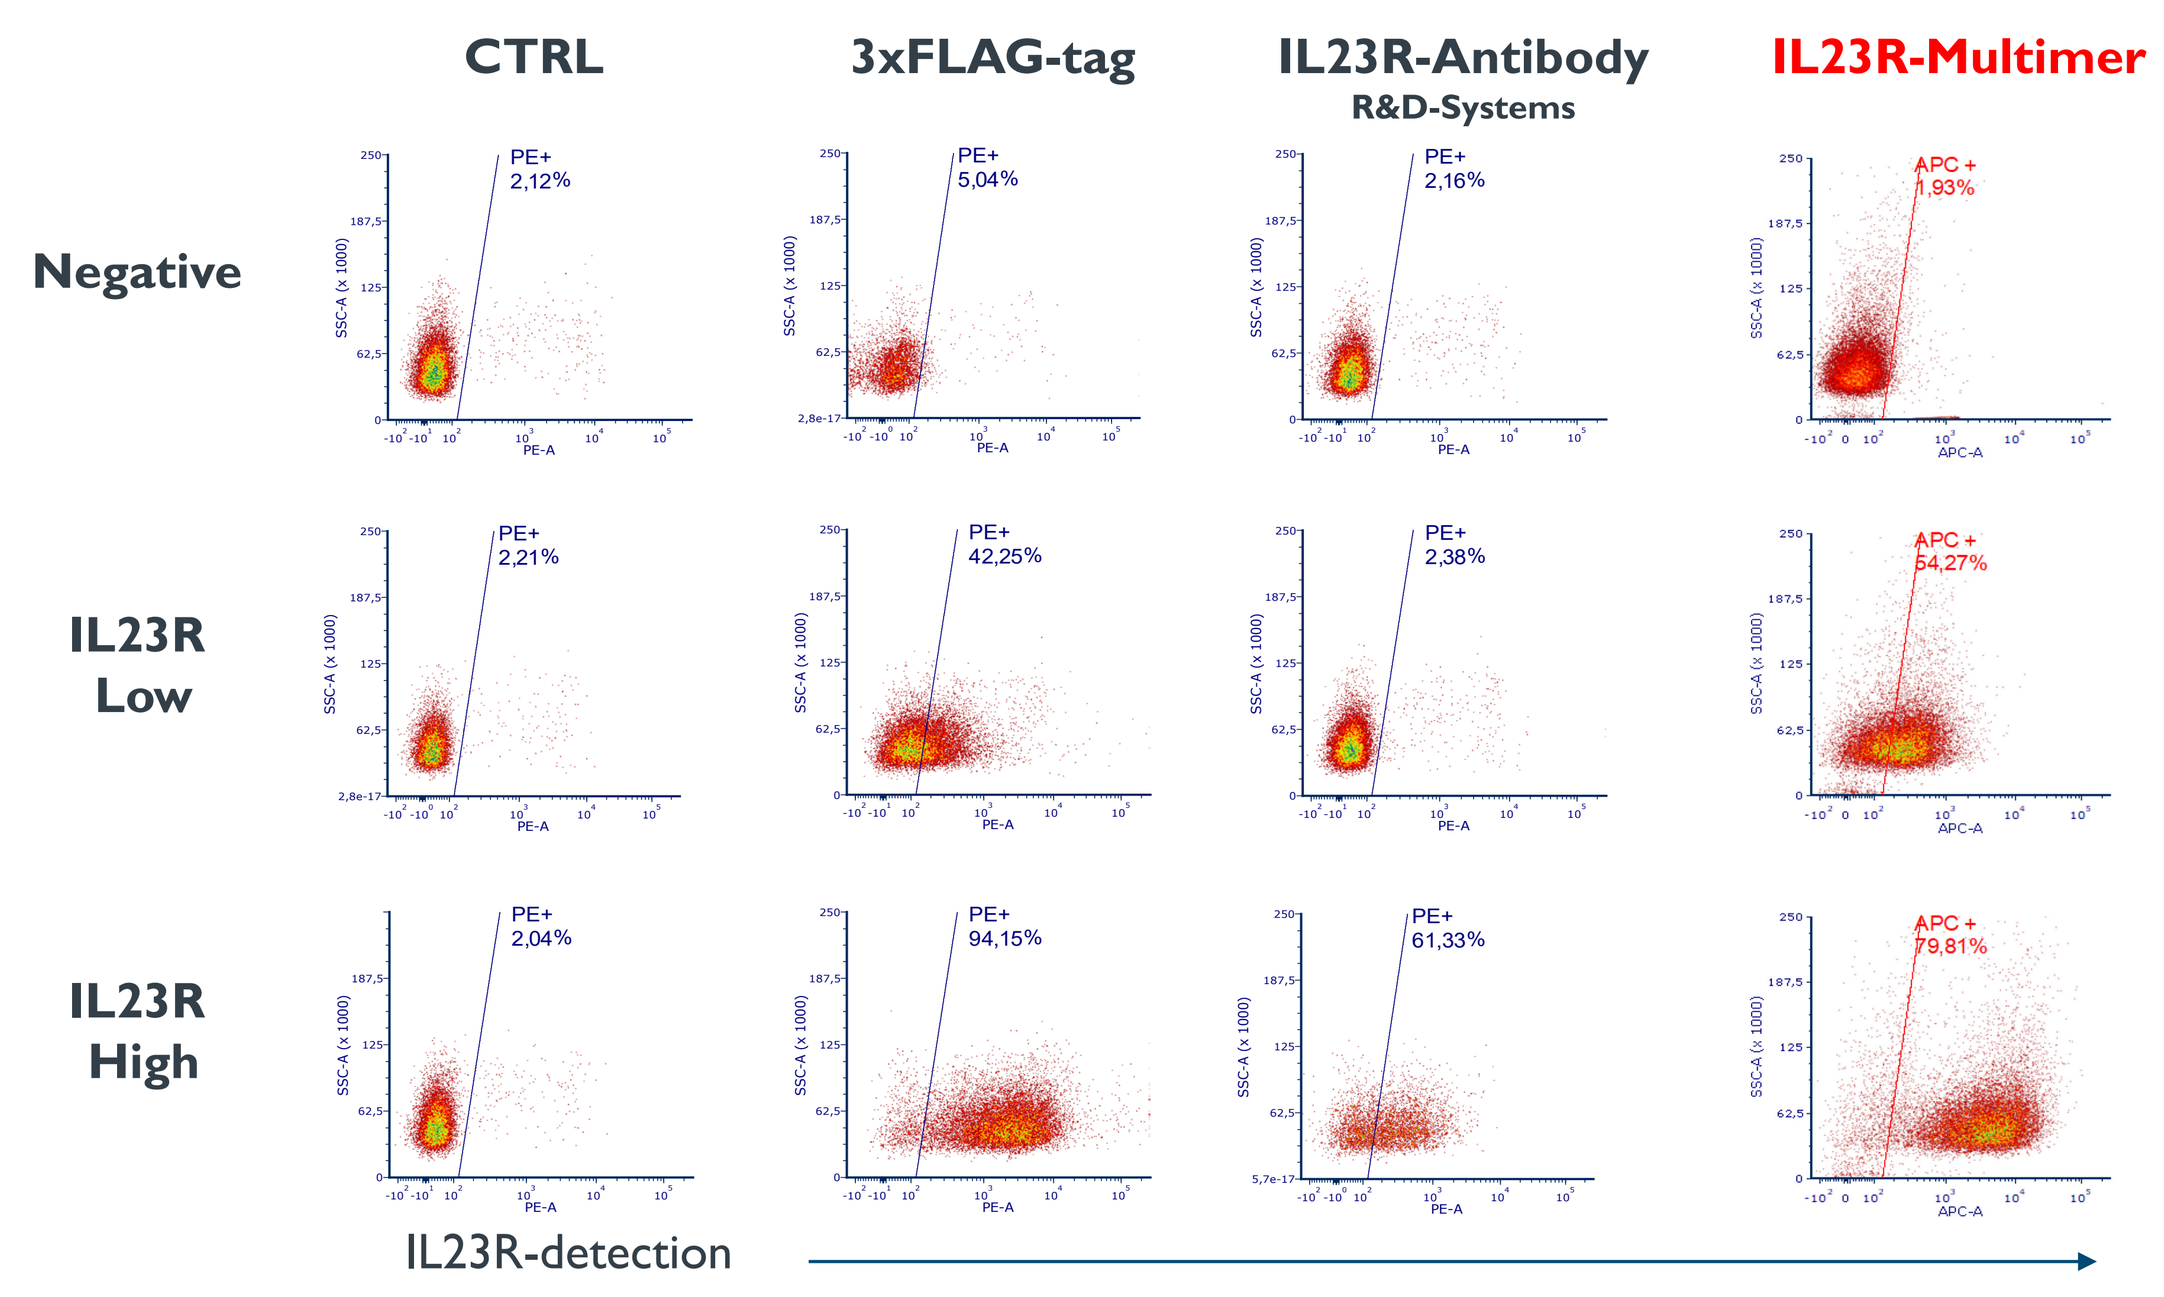
CTRL
3xFLAG-tag
IL23R-Antibody
R&D-Systems
IL23R-Multimer
Negative
IL23R Low
IL23R High
PE+ 2,12%
PE+ 5,04%
PE+ 2,16%
APC + 1,93%
PE+ 2,21%
PE+ 42,25%
PE+ 2,38%
APC + 54,27%
PE+ 2,04%
PE+ 94,15%
PE+ 61,33%
APC + 79,81%
IL23R-detection

The Multimerization of the IL23R scFv allows for a specific strong detection of low-expressed IL23R.

The sensitivity is similar or greater to what can be achieved by using a 3xFlag-tag.

The IL23R-scFv multimer even outperforms a commercially characterized IL23R antibody in terms of sensitivity.

4 Enabling IL23R CAR specificity detection in histology

One challenge in cell therapy is to assess the binding and specificity of novel CARs in tissues.
Both avidity-enhanced versions provide strong, specific staining in the cells expressing the target.

scFv-dimer | scFv-multimer

Colon
Crohns disease

Colon
Crohns disease
(high magnification)

Thymus
(negative control)

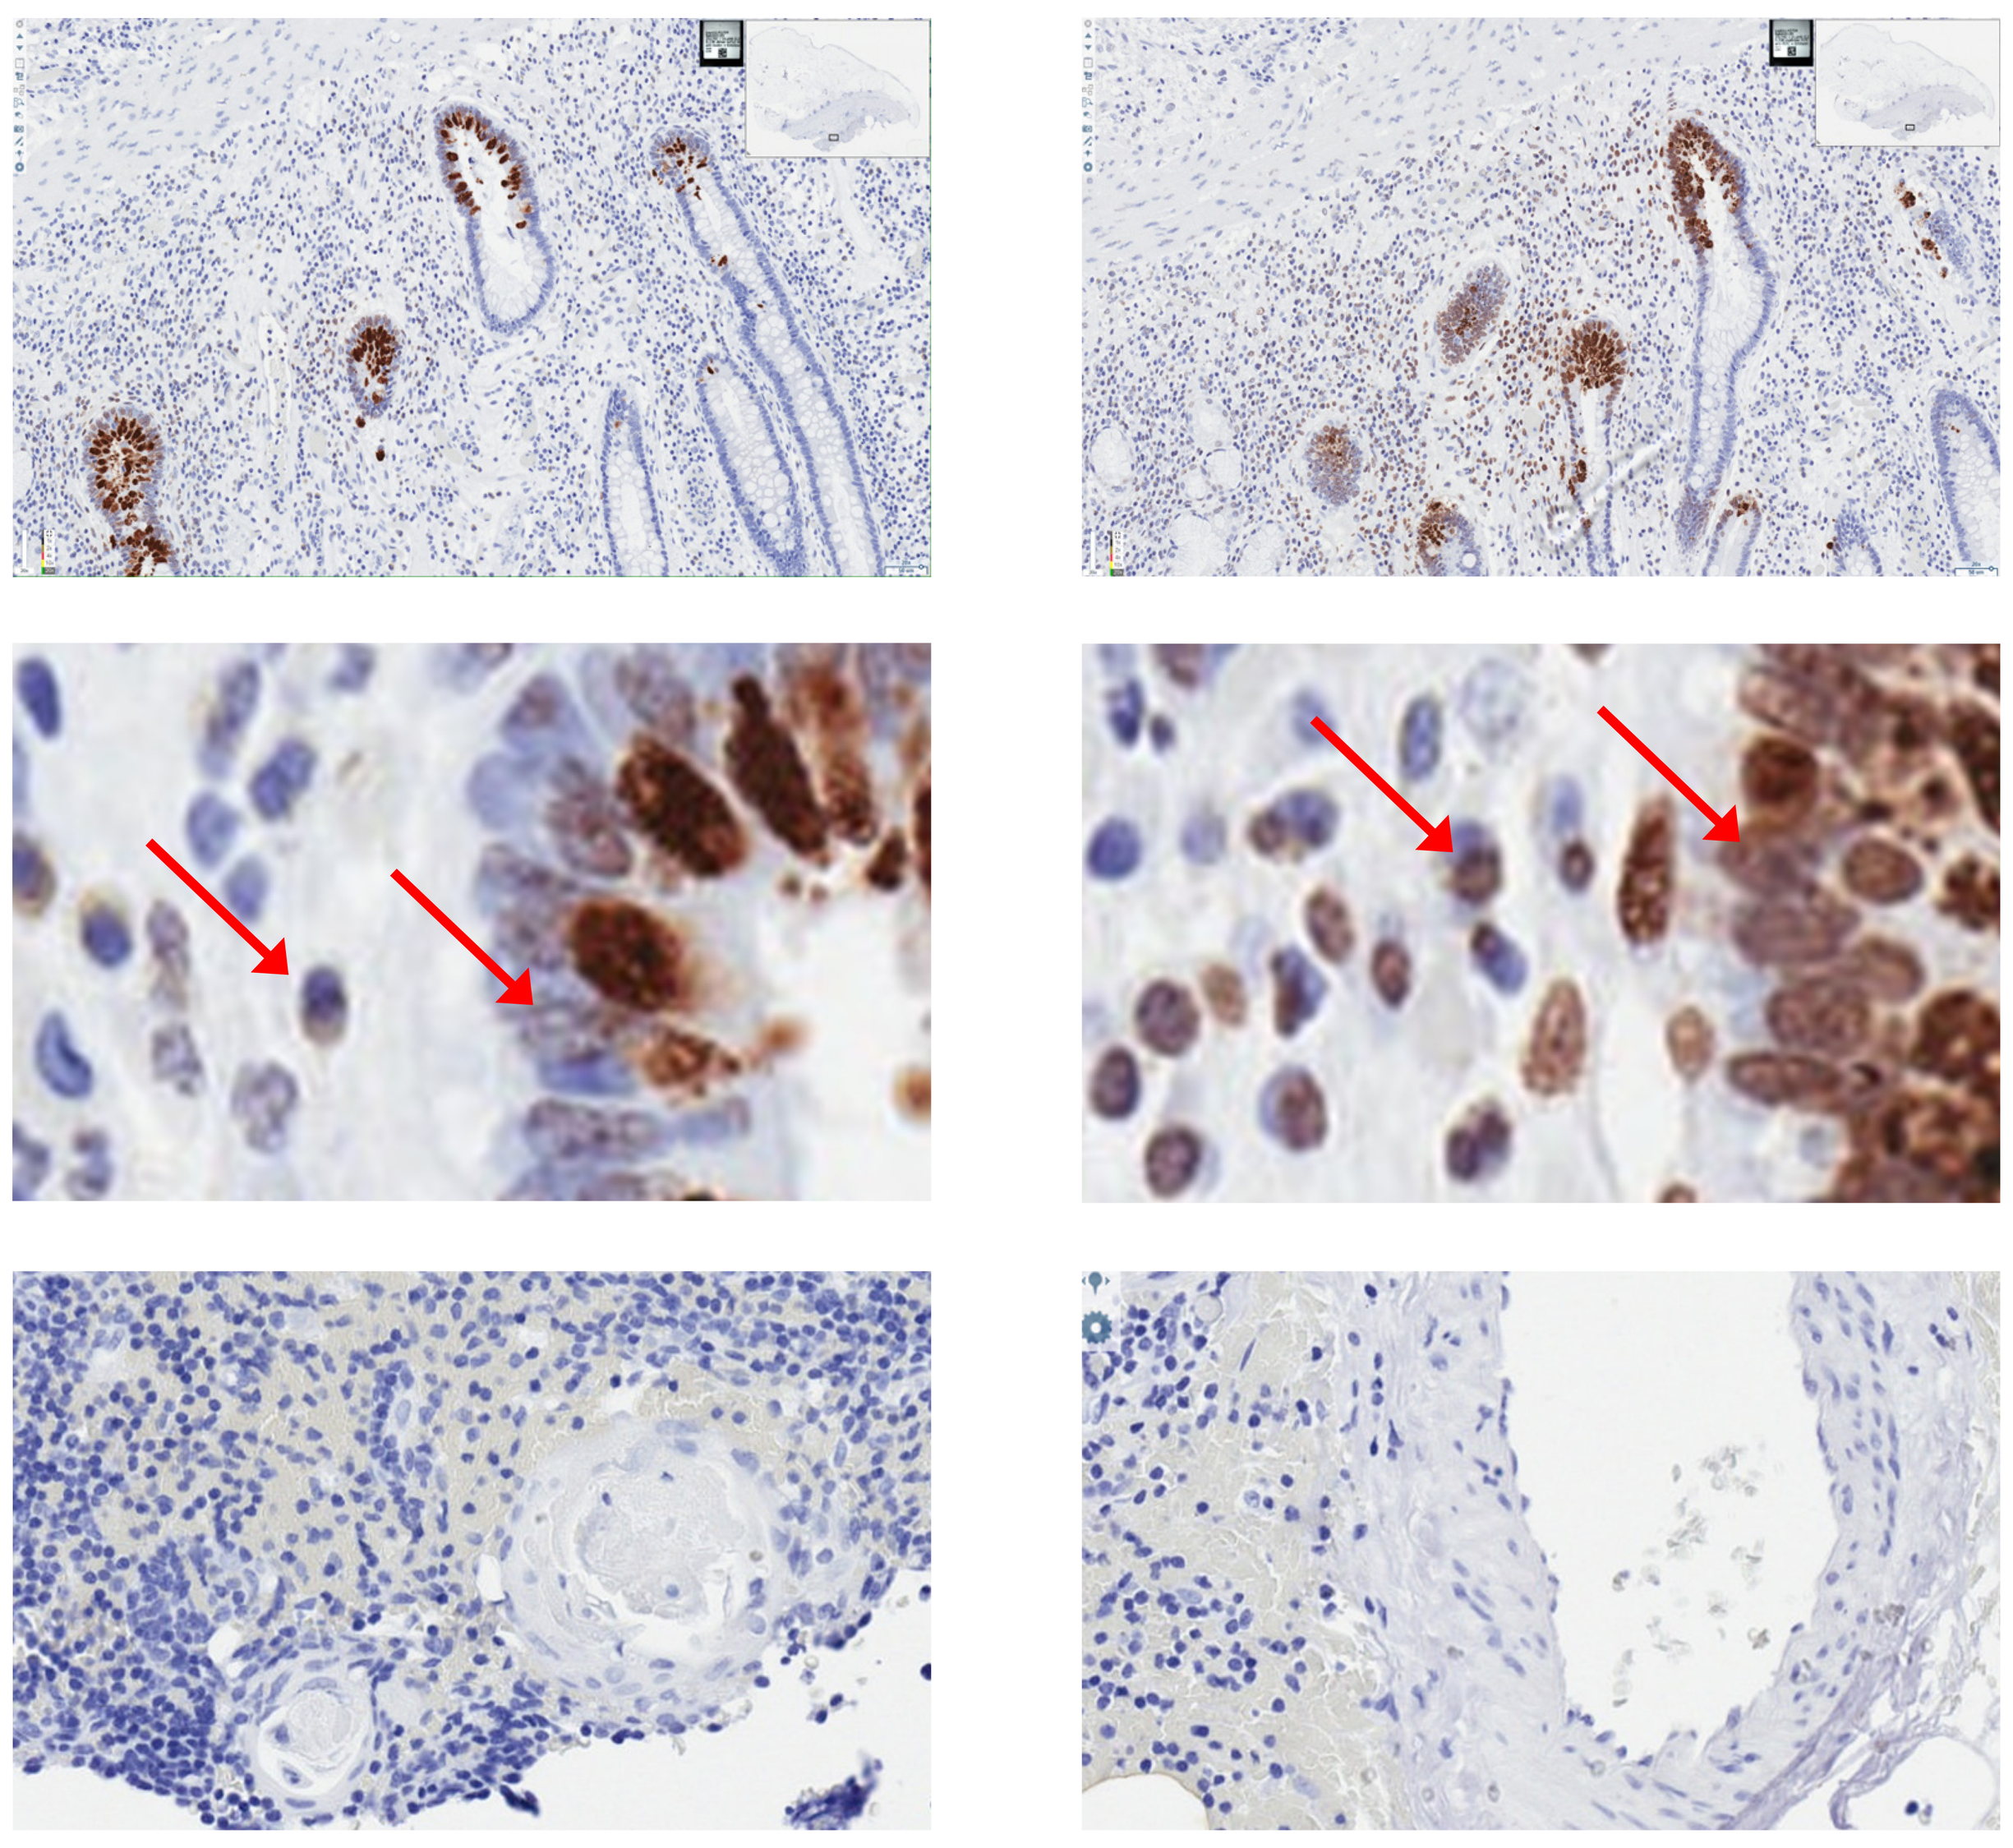

Arrows (red) highlight the difference in detection strength on target tissue using either dimeric version **(left)** of the IL23R scFv or a multimeric version **(right)**.

Conclusion

- Our approach allows the detection of low to high affinity binders like scFv by Flow Cytometry and Histology.
- Both multimerization approaches demonstrate a strong increase in sensitivity, making them ideal for the detection of weakly expressed or difficult cell therapy targets.
- This technique is suitable to accommodate various binders like scFv, Darpins, Nanobodies or other protein binders without long comparability studies.
- Great tool for sensitive preclinical specificity assessments like On/Off-target studies.
- Rapid and cost-effective way to characterize the binding and specificity of CARs for clinical translation.

Illustrations created with Biorender.com

Presented at the ESGCT 30th Annual Congress, 2023