Epigenetic Regulation of Human Prion Expression as a Potential One-Time Treatment for Prion Disease

Shih-Wei Chou[1], Kimberly Marlen[1], David Ojala[1], Patrick Dunn[1], Giulia Cisbani[2], Finn Peters[2], Chiara Melis[2], Jing Hu[1], Qi Yu[1], Sarah Hinkley[1], Mohad Mehrabian[1], Toufan Parman[1], Marina Falaleeva[1], Alaric Falcon[1], Marian Glynn[1], Kathleen Meyer[1], Amy M Pooler[1], Bryan Zeitler[1]

[1]Sangamo Therapeutics Inc., 501 Canal Blvd, Richmond, CA 94804, USA [2]Evotec SE, Hamburg, Germany

Poster #1616

Introduction

- Prion disease is a rapidly progressing, invariably fatal neurodegenerative disorder caused by misfolding of the cellular prion protein, PrP^C, encoded by the *PRNP* gene.
- Most cases are sporadic or caused by inherited dominant mutations in *PRNP*.
- There are currently no approved or clinical-stage disease-modifying therapies for the prevention or treatment of prion disease.
- Here, we investigated a single-administration epigenetic regulation approach using AAV-delivered zinc finger repressors (ZFRs) to achieve sustained and widespread reduction of PrP in the brain and rapid pharmacological effect.

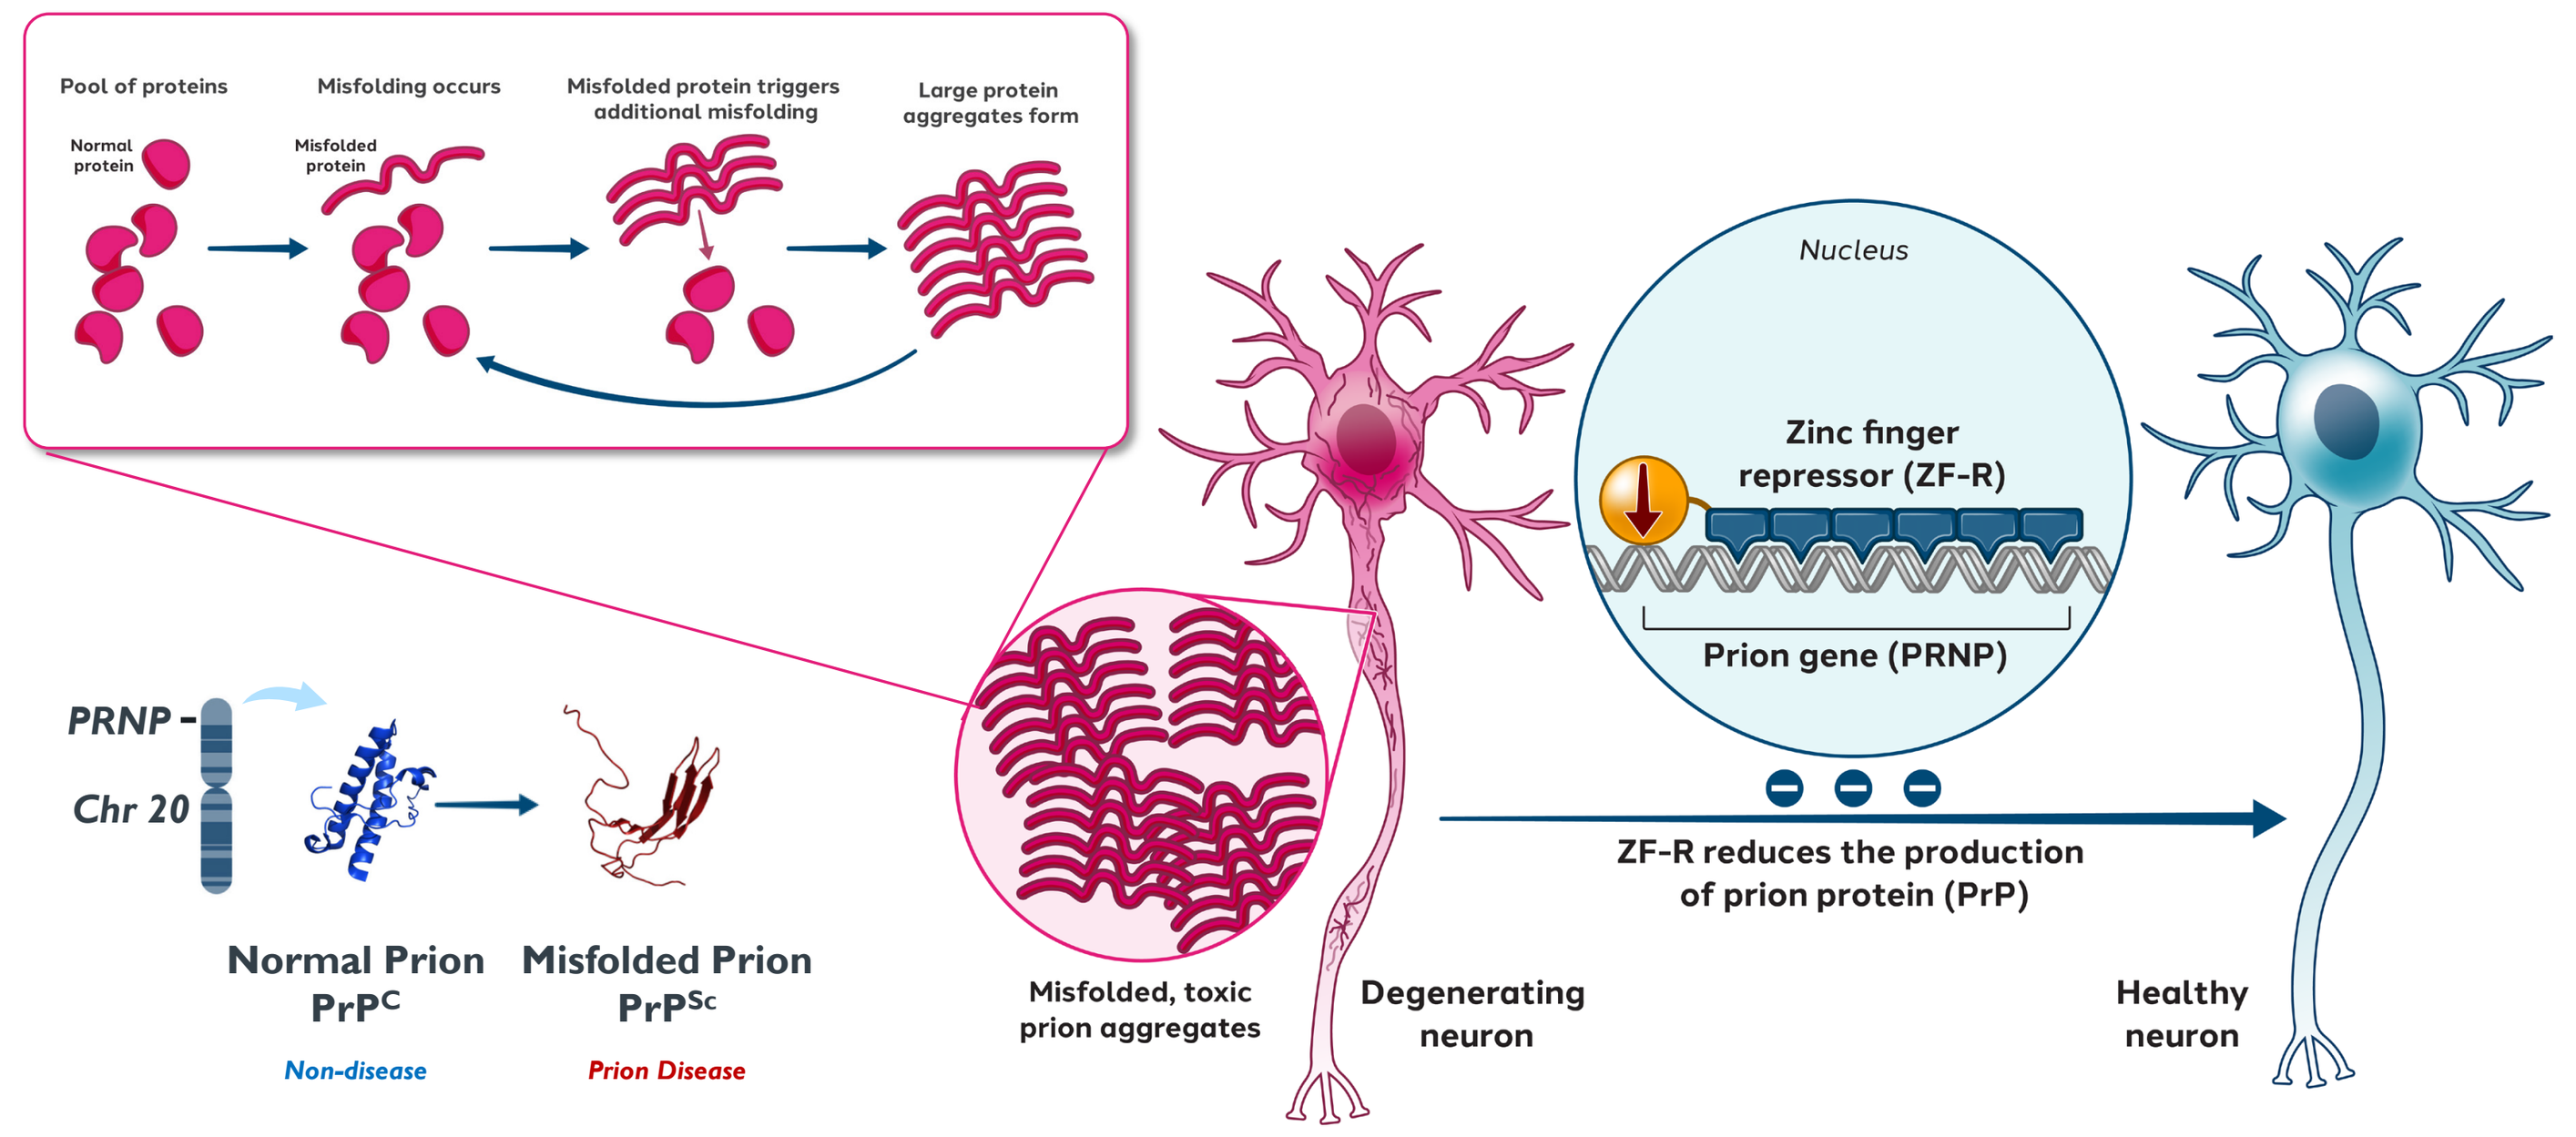
Pool of proteins
Normal protein
Misfolding occurs
Misfolded protein
Misfolded protein triggers additional misfolding
Large protein aggregates form
Nucleus
Zinc finger repressor (ZF-R)
Prion gene (PRNP)
ZF-R reduces the production of prion protein (PrP)
PRNP
Chr 20
Normal Prion PrPC
Non-disease
Misfolded Prion PrPSc
Prion Disease
Misfolded, toxic prion aggregates
Degenerating neuron
Healthy neuron

Figure 1. Schematic diagram of prion disease and PrP-lowing approach

ZF-Repressor genomic medicine for prion disease

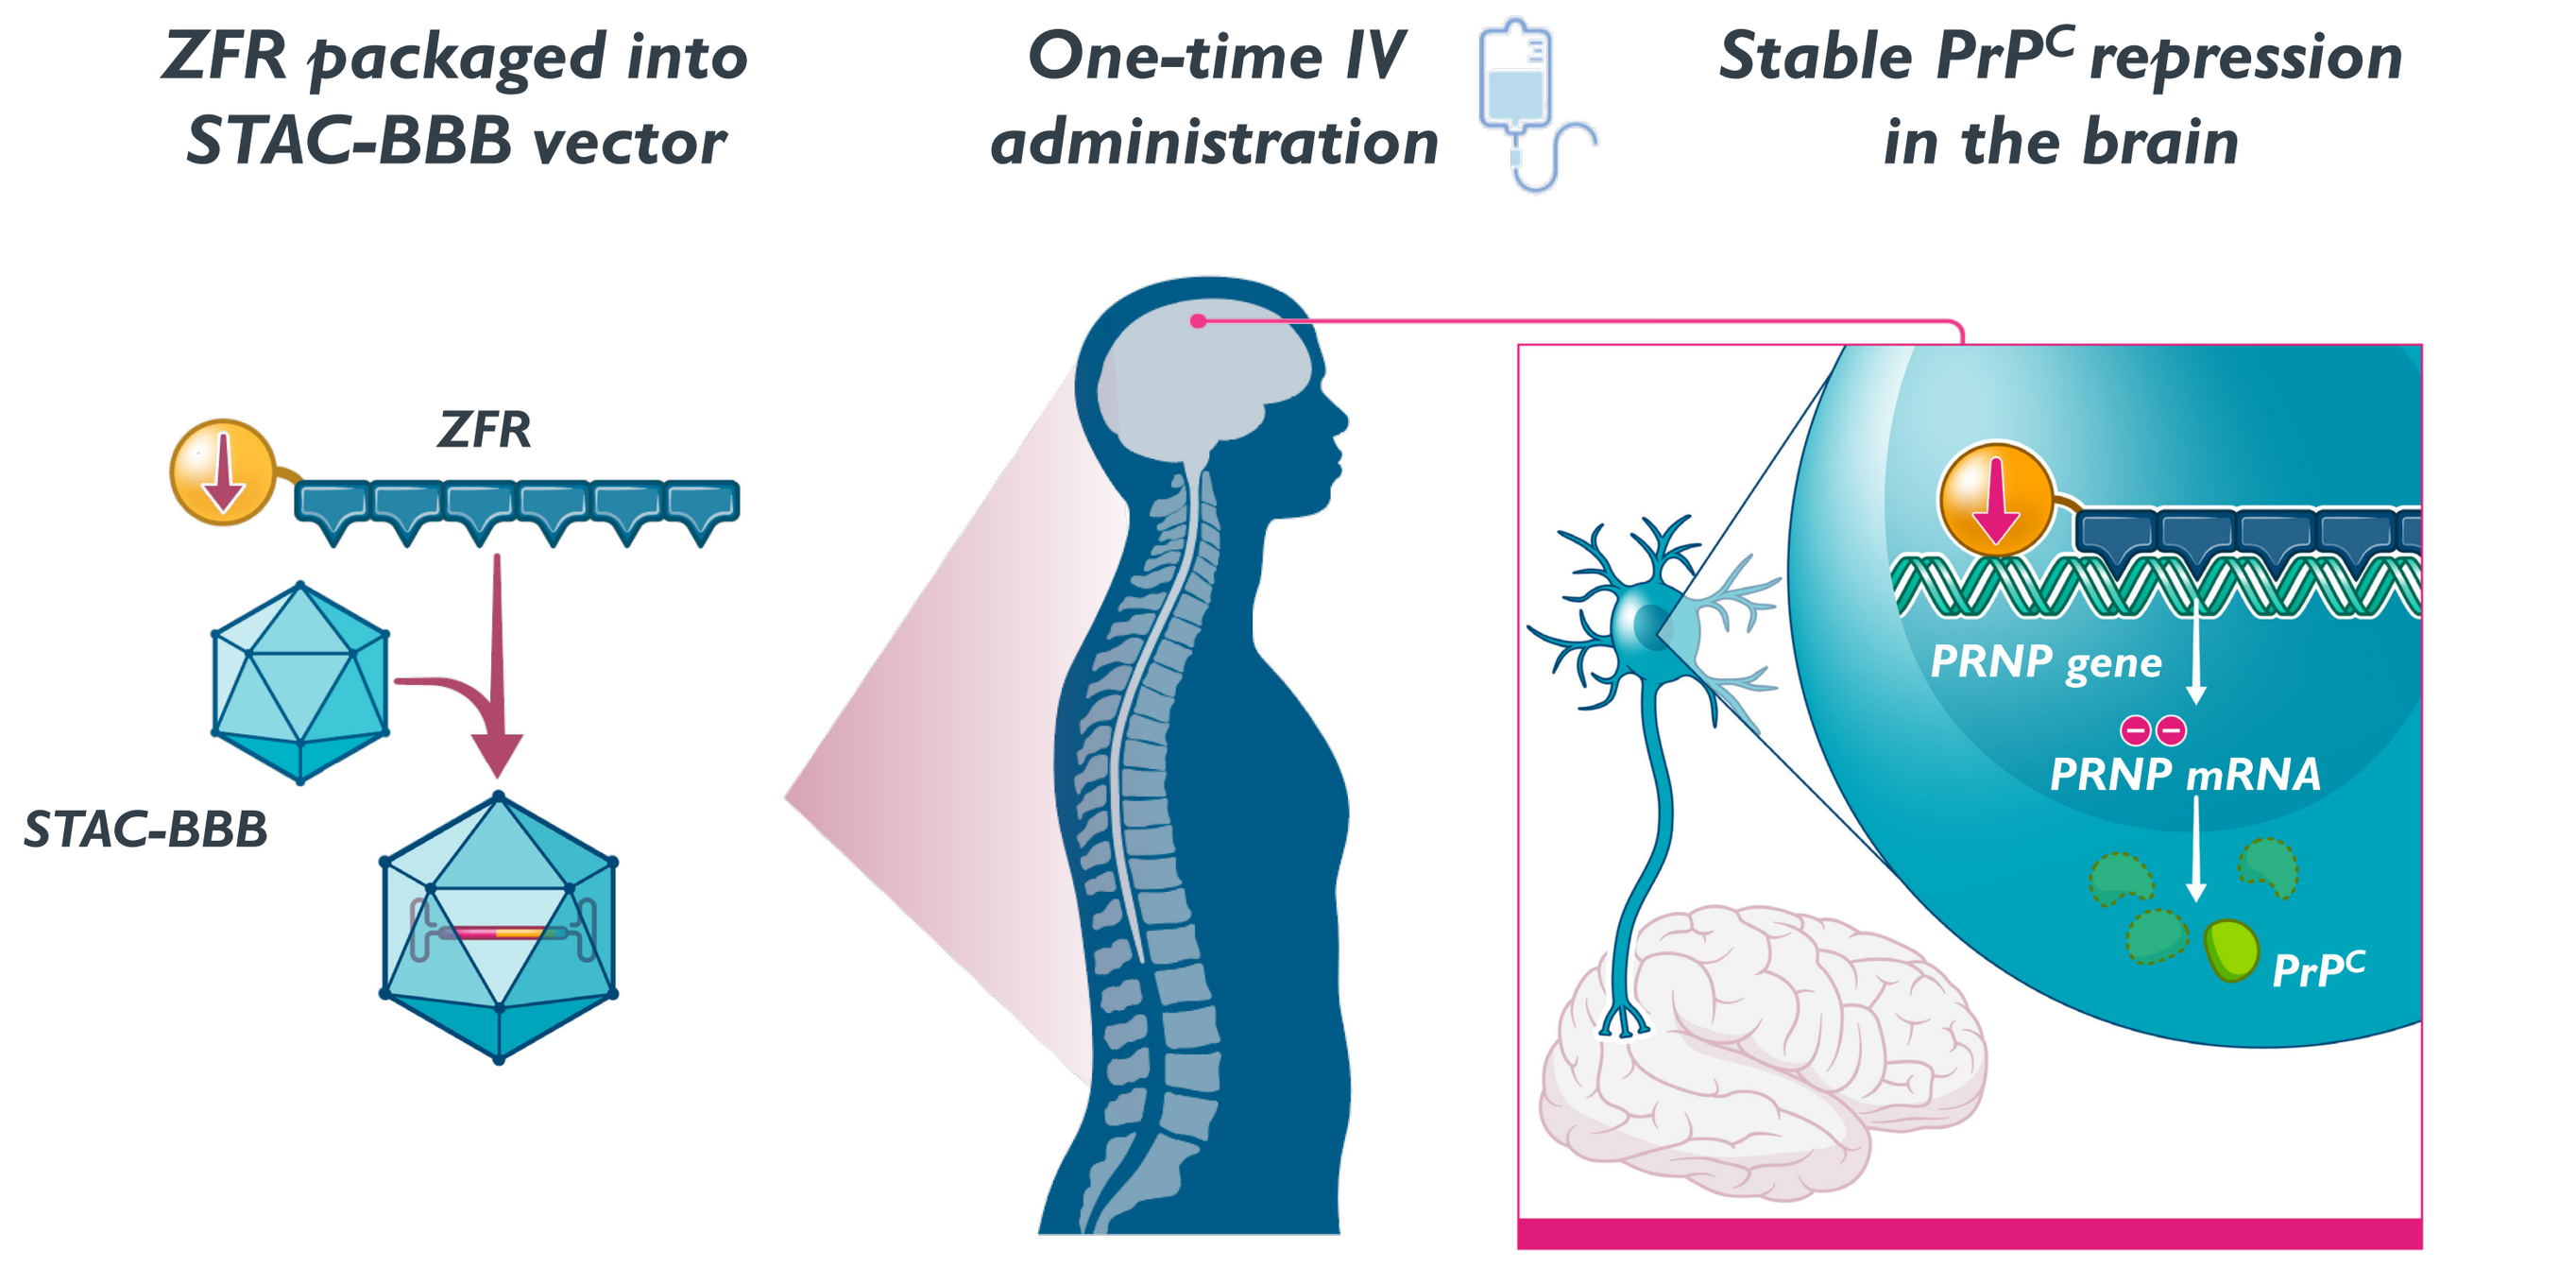
ZFR packaged into STAC-BBB vector
One-time IV administration
Stable PrPC repression in the brain
ZFR
STAC-BBB
PRNP gene
PRNP mRNA
PrPC

Figure 2. AAV ZFR genomic medicine for the treatment of prion disease

- A ZFR targeting the prion gene is packaged into STAC-BBB, which crosses the blood brain barrier (BBB) following a one-time IV administration.
- The ZFR represses *PRNP* transcription, resulting in specific depletion of neuronal PrP^C protein.

Disclosures

This work was funded by Sangamo Therapeutics. All listed Sangamo authors are current or former employees of Sangamo Therapeutic. Tg25109 mice were kindly provided by Dr. E. Minikel and Dr. S. Vallabh from the Broad Institute of MIT and Harvard.

Highly potent and specific ZFRs targeting *PRNP*

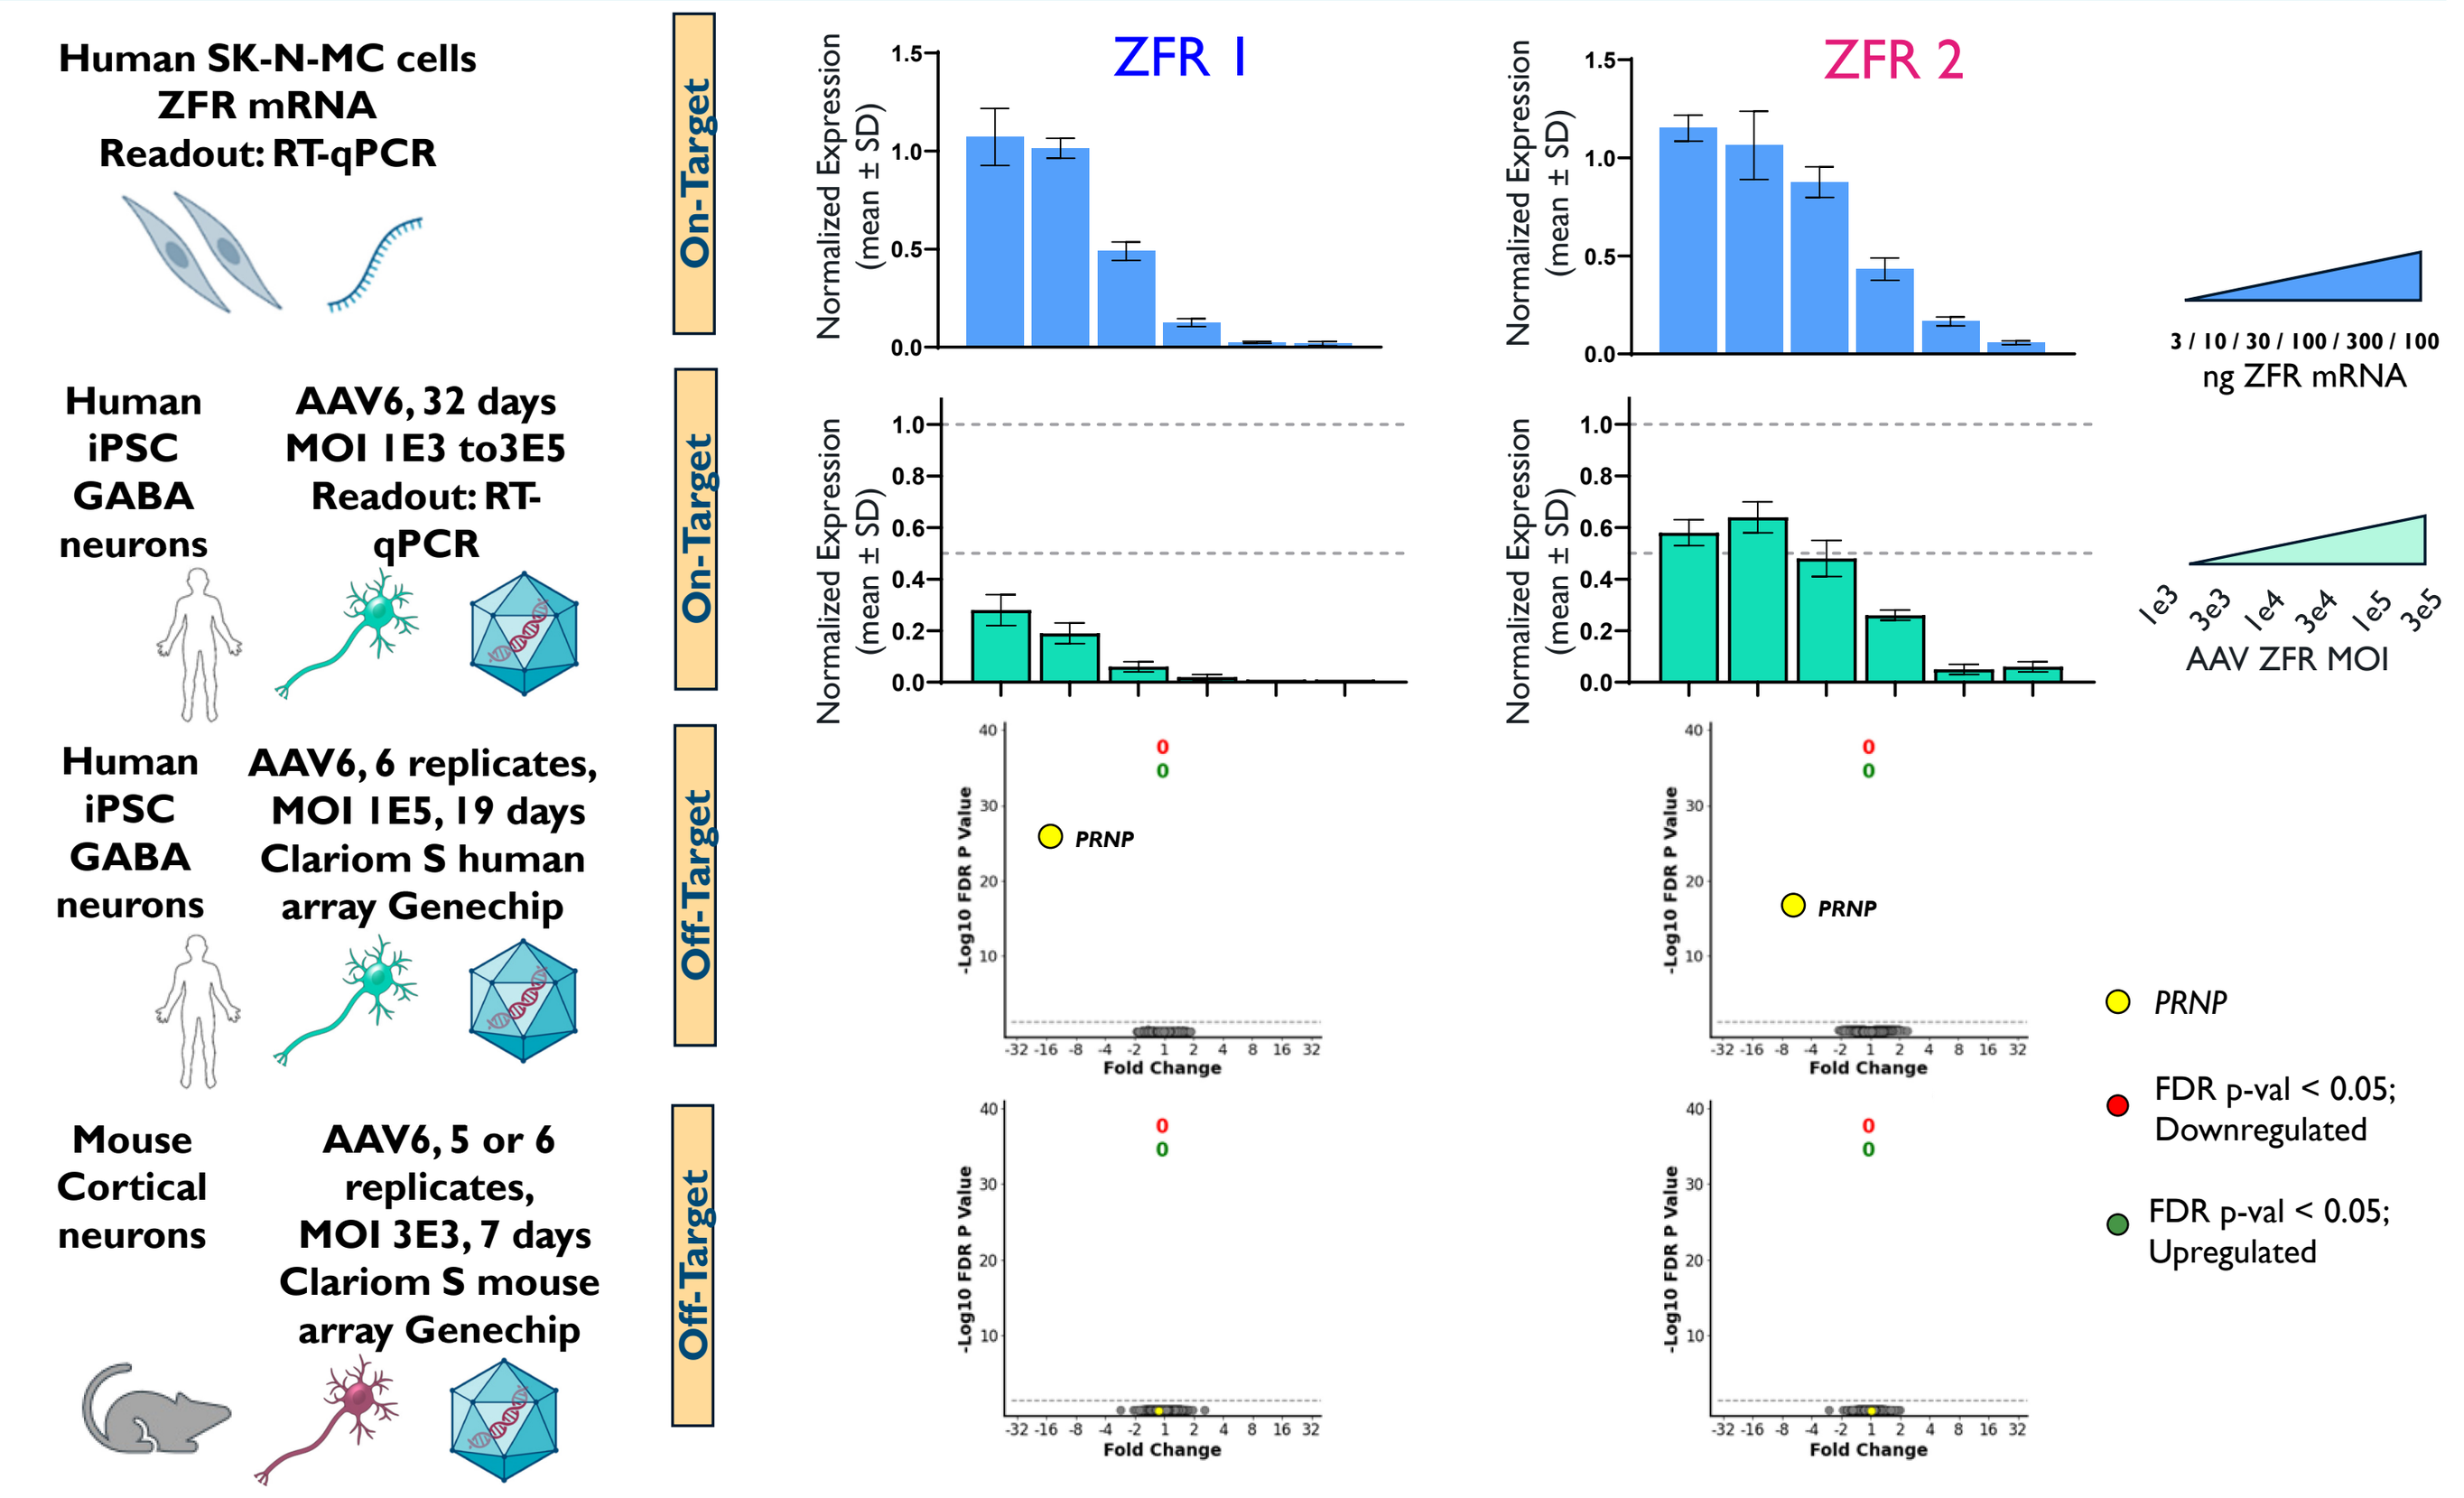
Human SK-N-MC cells ZFR mRNA Readout: RT-qPCR
Human iPSC GABA neurons
AAV6, 32 days MOI 1E3 to3E5 Readout: RT-qPCR
Human iPSC GABA neurons
AAV6, 6 replicates, MOI 1E5, 19 days Clariom S human array Genechip
Mouse Cortical neurons
AAV6, 5 or 6 replicates, MOI 3E3, 7 days Clariom S mouse array Genechip
On-Target
Off-Target
ZFR 1
ZFR 2
Normalized Expression (mean ± SD)
3 / 10 / 30 / 100 / 300 / 100 ng ZFR mRNA
1e3 3e3 1e4 3e4 1e5 3e5
AAV ZFR MOI
-Log10 FDR P Value
Fold Change
PRNP
FDR p-val < 0.05; Downregulated
FDR p-val < 0.05; Upregulated

Figure 3. Characterization of potent and specific ZFRs targeting human *PRNP*

- Candidates ZFR 1 and 2 potently and specifically reduced *PRNP* in human neuroblastoma (SK-N-MC) cells and iPSC-derived GABA neuron culture.
- No off-target activity was detected in human iGABA nor mouse cortical neuron culture.

ZFRs achieve 50-90% prion reduction in the mouse brain, spinal cord and CSF following IV delivery

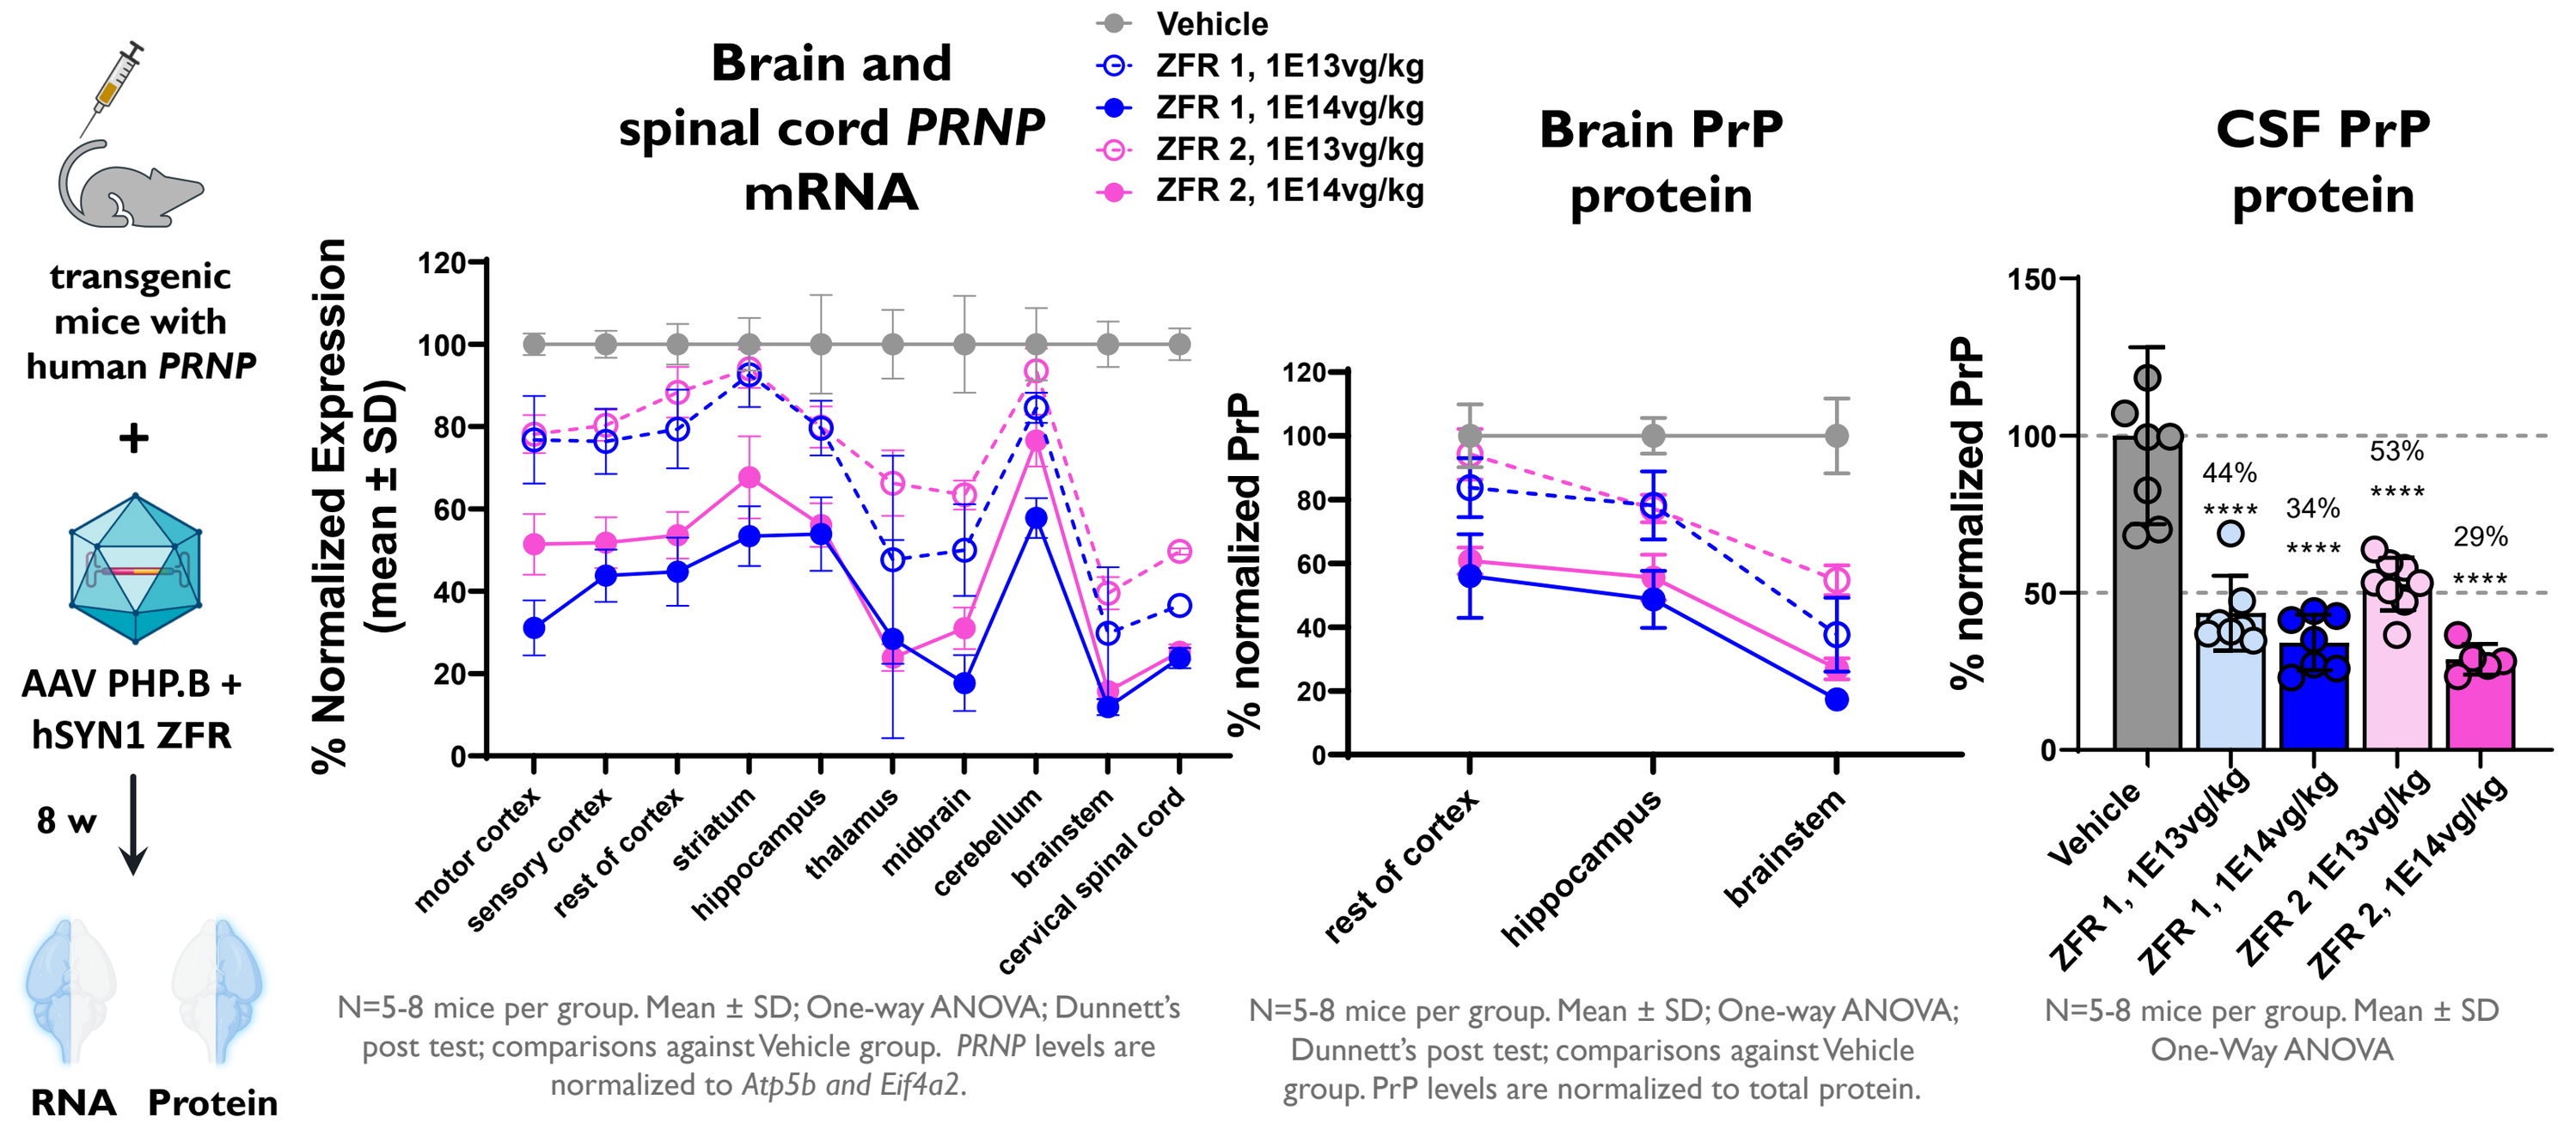
transgenic mice with human PRNP
+
AAV PHP.B + hSYN1 ZFR
8 w
RNA
Protein
Brain and spinal cord PRNP mRNA
Vehicle
ZFR 1, 1E13vg/kg
ZFR 1, 1E14vg/kg
ZFR 2, 1E13vg/kg
ZFR 2, 1E14vg/kg
% Normalized Expression (mean ± SD)
motor cortex
sensory cortex
rest of cortex
striatum
hippocampus
thalamus
midbrain
cerebellum
brainstem
cervical spinal cord
N=5-8 mice per group. Mean ± SD; One-way ANOVA; Dunnett's post test; comparisons against Vehicle group. PRNP levels are normalized to Atp5b and Eif4a2.
Brain PrP protein
% normalized PrP
rest of cortex
hippocampus
brainstem
N=5-8 mice per group. Mean ± SD; One-way ANOVA; Dunnett's post test; comparisons against Vehicle group. PrP levels are normalized to total protein.
CSF PrP protein
% normalized PrP
44%
34%
53%
29%
Vehicle
ZFR 1, 1E13vg/kg
ZFR 1, 1E14vg/kg
ZFR 2 1E13vg/kg
ZFR 2, 1E14vg/kg
N=5-8 mice per group. Mean ± SD One-Way ANOVA

Figure 4. Widespread reduction of prion mRNA and protein in the CNS of a humanized *PRNP*-expressing transgenic mouse model (Tg25109)

- Dose-dependent prion repression was observed for both ZFRs. At 1E14 vg/kg, AAV hSYN1-ZFR 1 or ZFR 2 delivered to Tg mice resulted in 50-90% repression of bulk *Prnp* mRNA levels across ten Central Nervous System (CNS) regions compared to vehicle controls.
- Both constructs led to >70% bulk PrP protein knockdown in the brainstem.
- In the CSF, ~70% PrP protein knockdown was observed, suggesting the majority of CSF PrP is derived from neurons.

Potent single-neuron *PRNP* repression in the brain

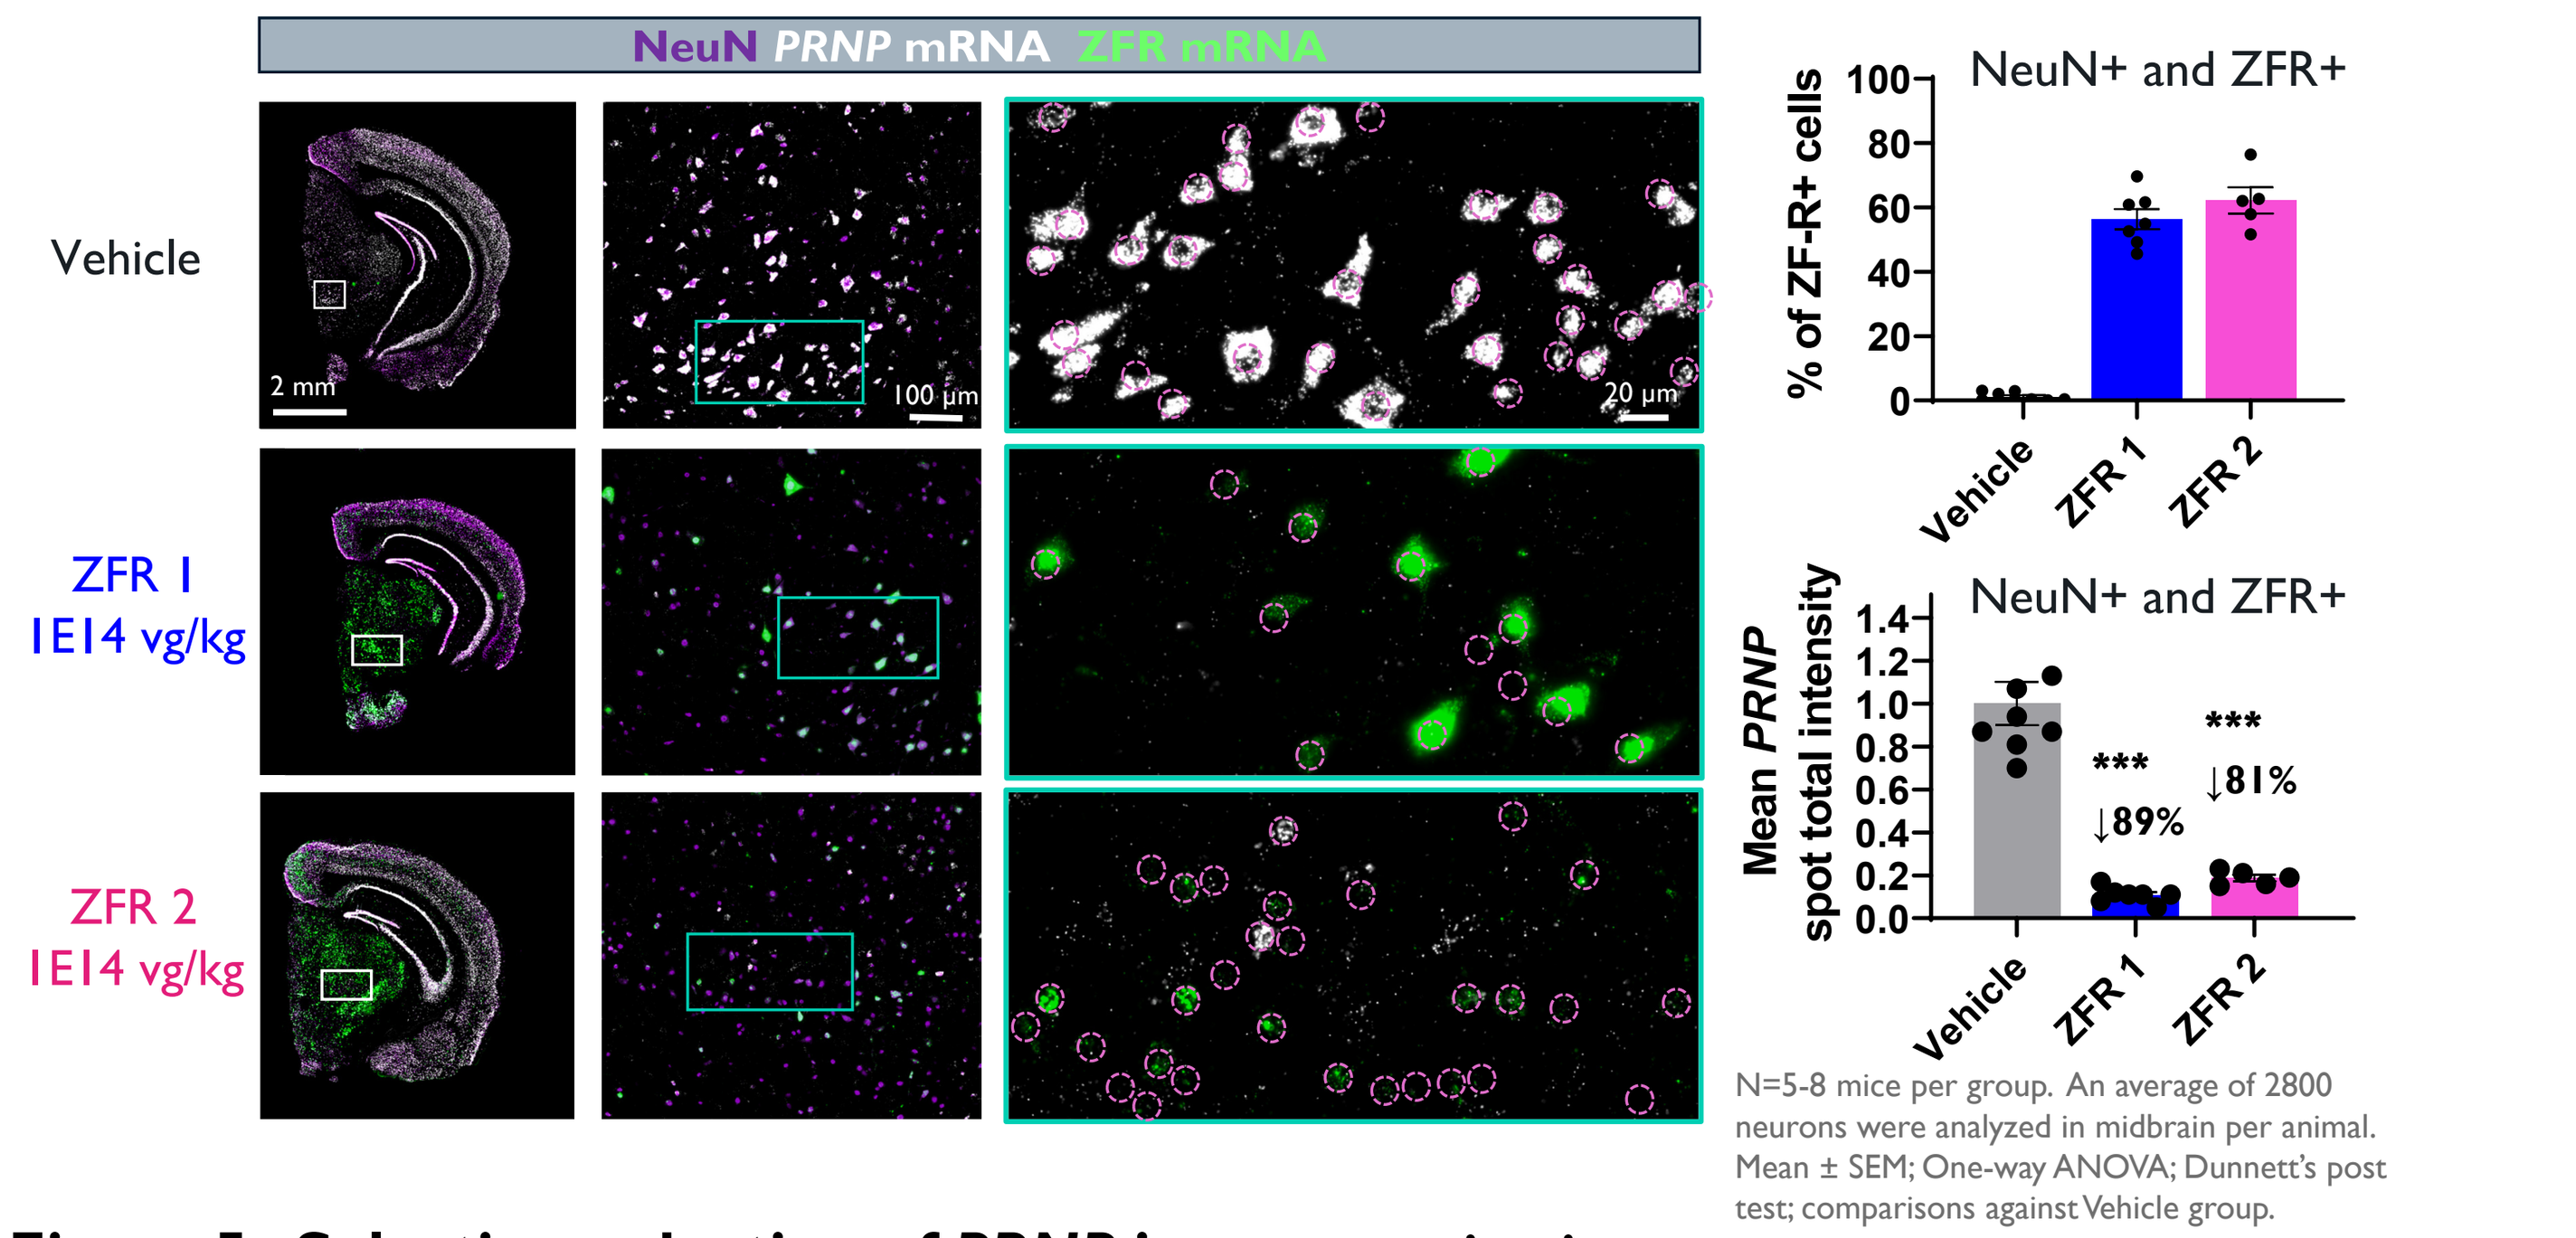
NeuN PRNP mRNA ZFR mRNA
Vehicle
ZFR 1 1E14 vg/kg
ZFR 2 1E14 vg/kg
2 mm
100 µm
20 µm
NeuN+ and ZFR+
% of ZF-R+ cells
Mean PRNP spot total intensity
↓89%
↓81%
Vehicle
ZFR 1
ZFR 2
N=5-8 mice per group. An average of 2800 neurons were analyzed in midbrain per animal. Mean ± SEM; One-way ANOVA; Dunnett's post test; comparisons against Vehicle group.

Figure 5. Selective reduction of *PRNP* in neurons *in vivo*

- Multiplexed RNAscope (*PRNP, ZFR*) + IHC (NeuN) revealed potent and selective prion transcript repression in Neun+ neuron through out the brain.
- In the midbrain region, efficient ZFR expression was detected in ~60% of NeuN+ cells. For both test articles, ZFR+ midbrain neurons showed significantly lower *PRNP* transcript expression (~90% repression at single-cell level) compared to abundant expression observed in vehicle-treated mice.

STAC-BBB efficiently delivers a prion-targeted ZFR to the adult NHP brain following IV delivery

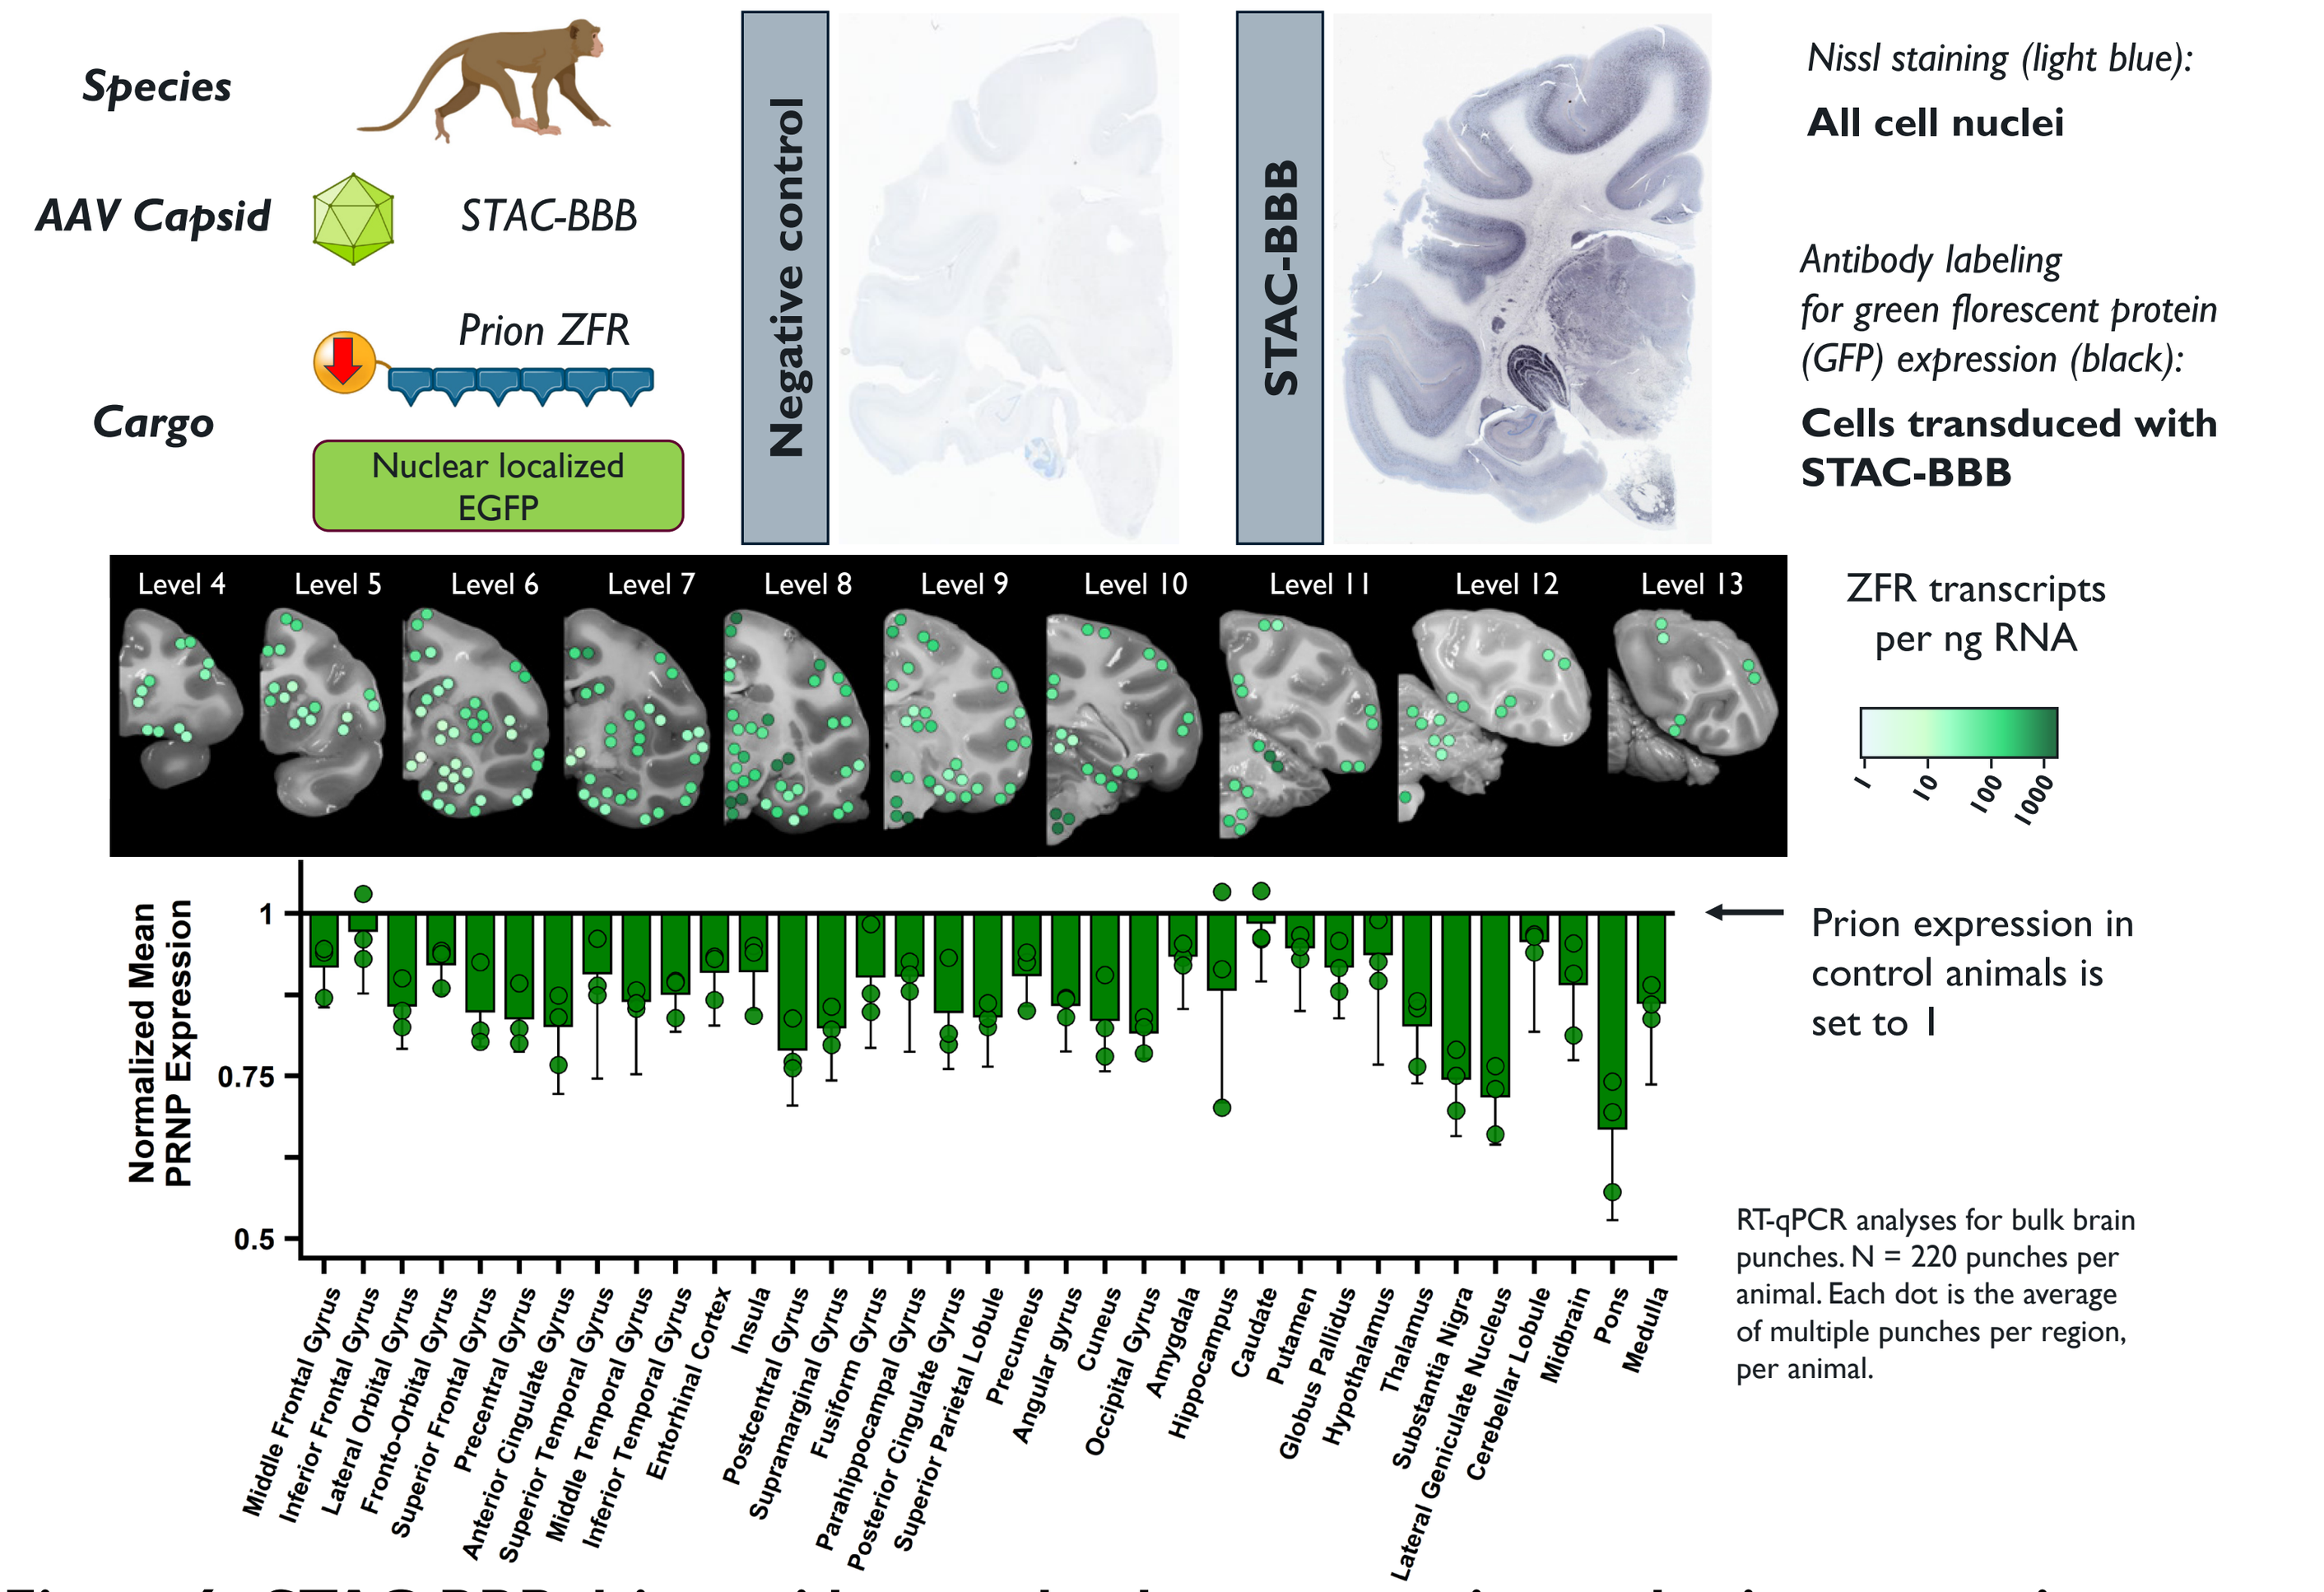
Species
AAV Capsid
STAC-BBB
Prion ZFR
Cargo
Nuclear localized EGFP
Negative control
STAC-BBB
Nissl staining (light blue):
All cell nuclei
Antibody labeling for green florescent protein (GFP) expression (black):
Cells transduced with STAC-BBB
Level 4
Level 5
Level 6
Level 7
Level 8
Level 9
Level 10
Level 11
Level 12
Level 13
ZFR transcripts per ng RNA
Normalized Mean PRNP Expression
Prion expression in control animals is set to 1
RT-qPCR analyses for bulk brain punches. N = 220 punches per animal. Each dot is the average of multiple punches per region, per animal.
Middle Frontal Gyrus
Inferior Frontal Gyrus
Lateral Orbital Gyrus
Fronto-Orbital Gyrus
Superior Frontal Gyrus
Precentral Gyrus
Anterior Cingulate Gyrus
Superior Temporal Gyrus
Middle Temporal Gyrus
Inferior Temporal Gyrus
Entorhinal Cortex
Insula
Postcentral Gyrus
Supramarginal Gyrus
Fusiform Gyrus
Parahippocampal Gyrus
Posterior Cingulate Gyrus
Superior Parietal Lobule
Precuneus
Angular gyrus
Cuneus
Occipital Gyrus
Amygdala
Hippocampus
Caudate
Putamen
Globus Pallidus
Hypothalamus
Thalamus
Substantia Nigra
Lateral Geniculate Nucleus
Cerebellar Lobule
Midbrain
Pons
Medulla

Figure 6. STAC-BBB drives widespread, robust expression and prion repression throughout the adult NHP brain

- IHC (Anti-GFP) demonstrated broad transgene expression in the NHP brain 19 days after IV administration of STAC-BBB at a 2E13 vg/kg dose.
- RT-qPCR analyses revealed ZFR expression and *PRNP* repression in all 35 brain regions tested, with the greatest reduction observed in the pons, lateral geniculate nucleus, and substantia nigra.

Effective *PRNP* repression in ZFR transduced neurons of adult non-human primates

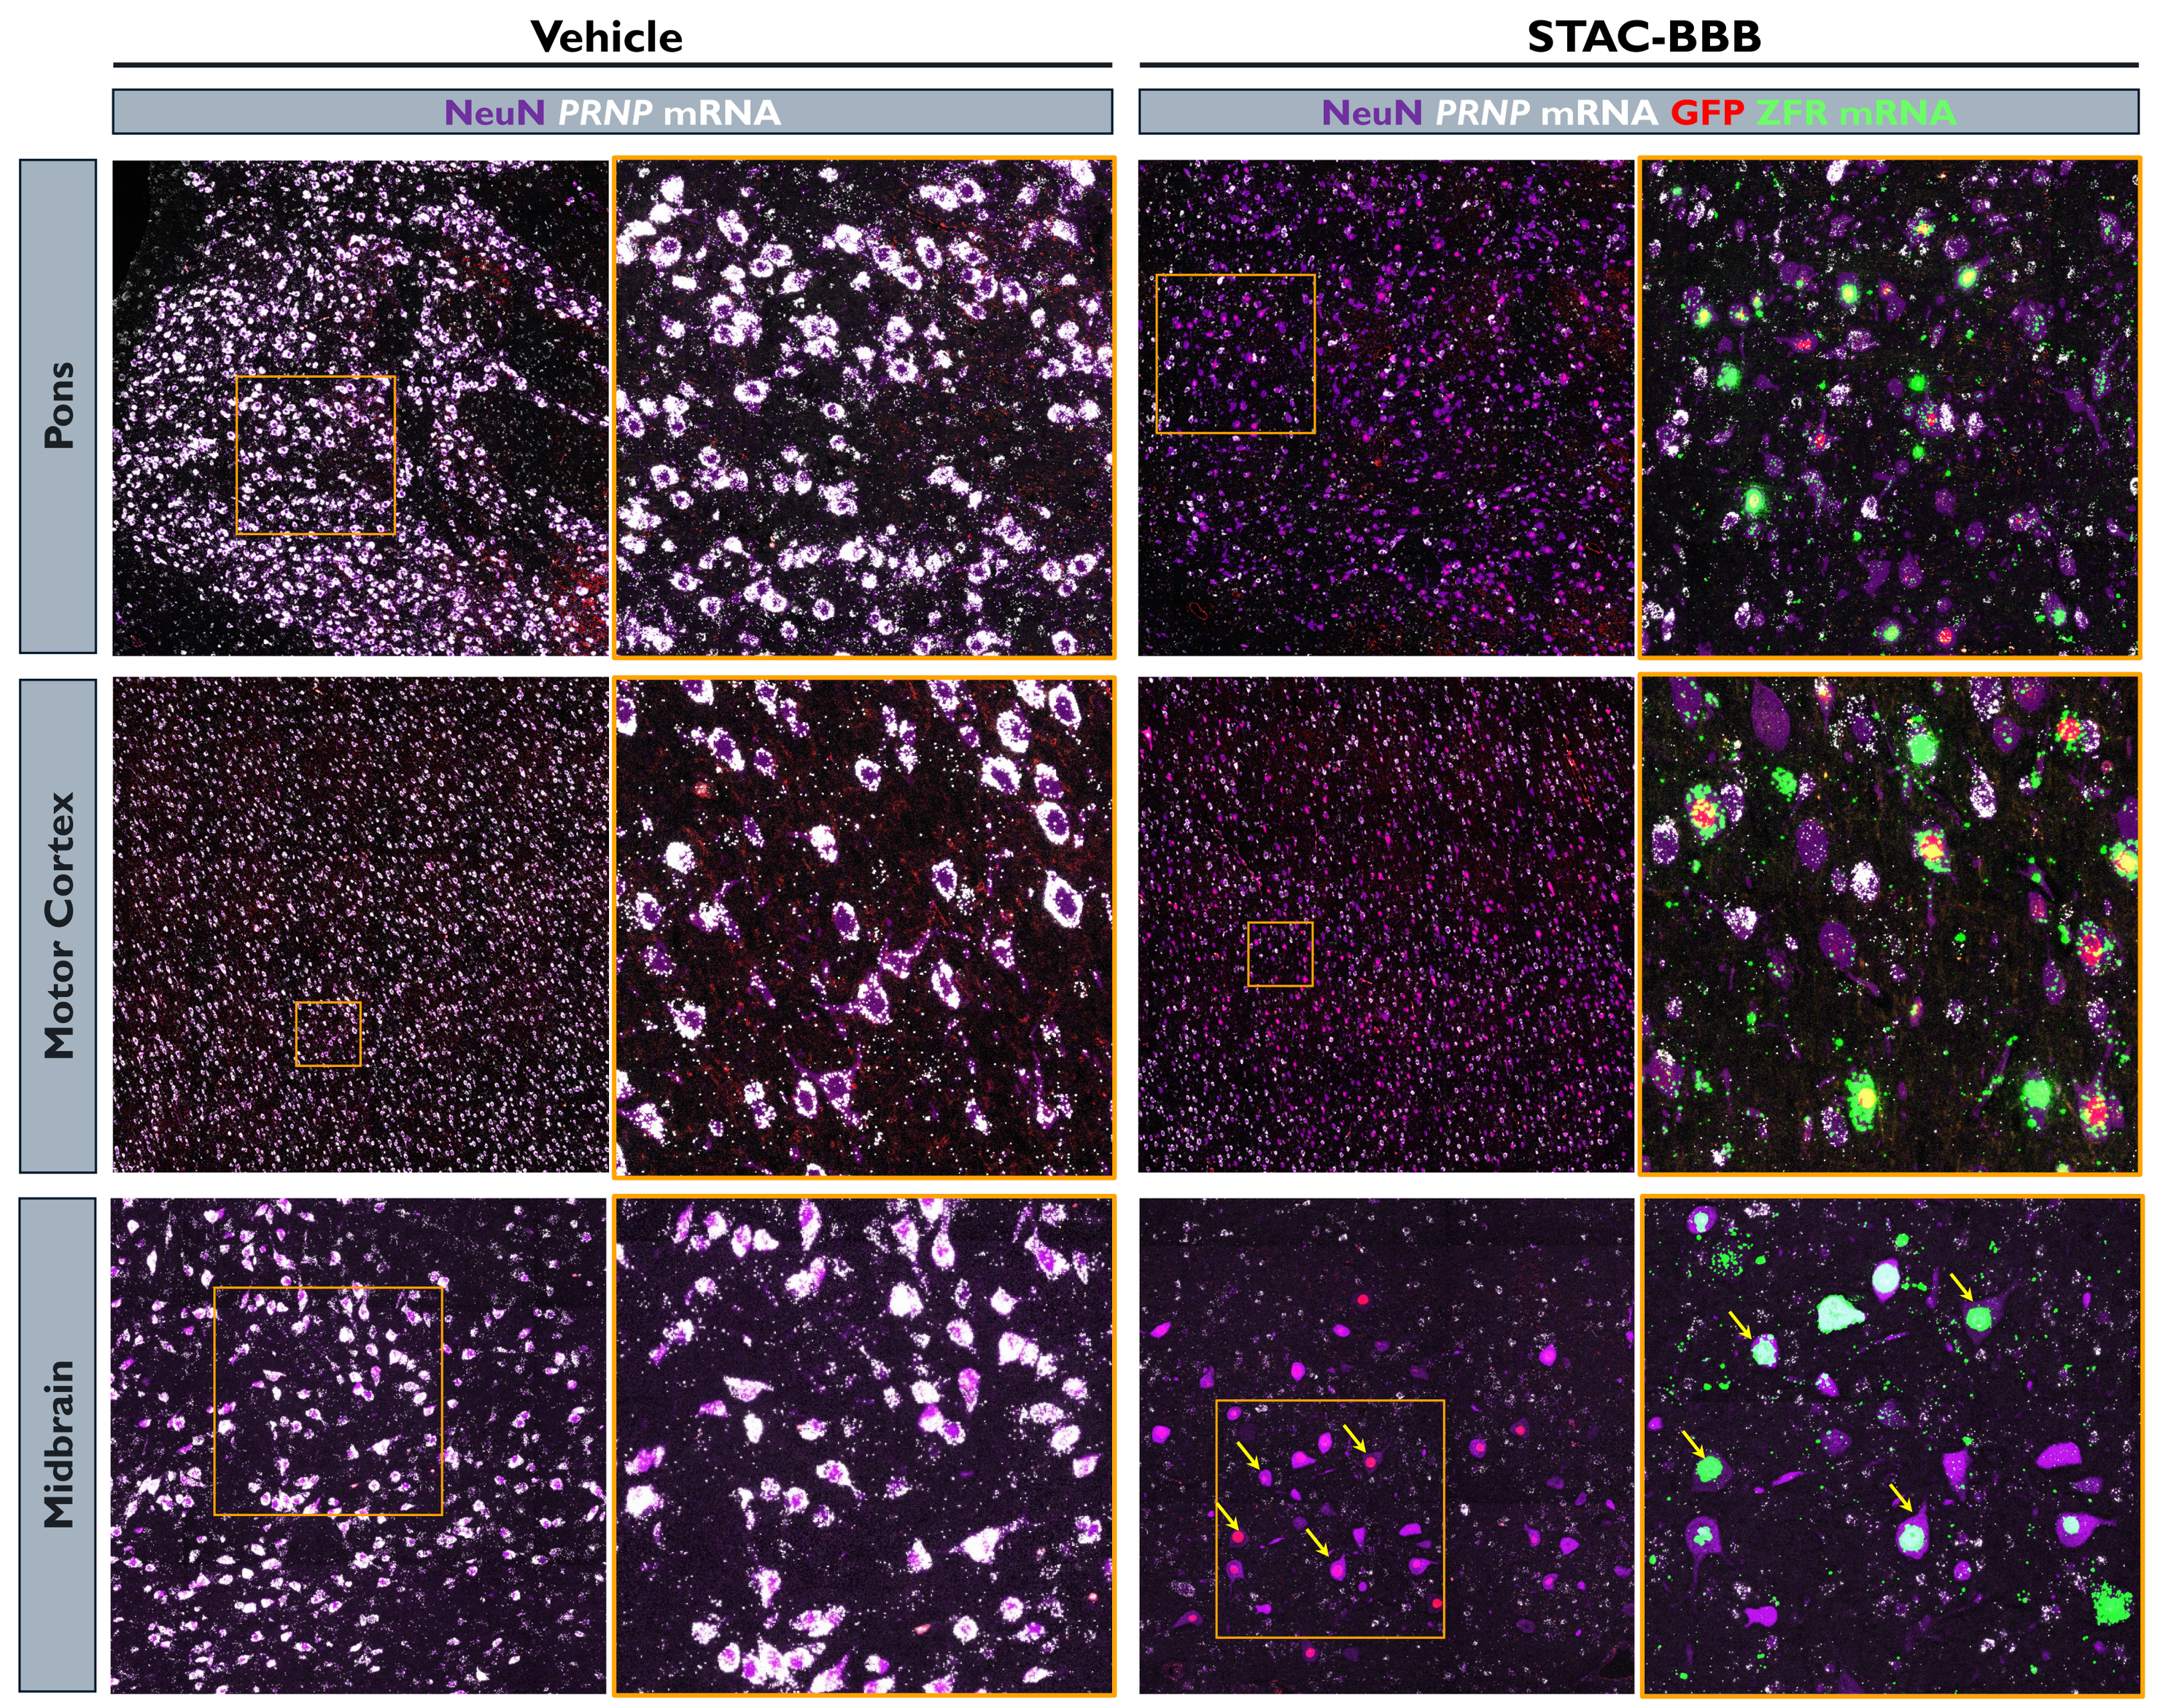
Vehicle
STAC-BBB
NeuN PRNP mRNA
NeuN PRNP mRNA GFP ZFR mRNA
Pons
Motor Cortex
Midbrain

Figure 7. STAC-BBB efficiently transduces the NHP CNS following IV administration and enables ZFR-mediated *PRNP* repression in neurons throughout the brain

- Multiplexed RNAscope (*PRNP, ZFR*) with IHC (NeuN, GFP) revealed that prion transcripts were repressed in transduced neurons across multiple brain regions in animals treated with STAC-BBB encoding a *PRNP*-targeted ZFR, but not the vehicle treated animals.
- Examples of transduced neurons (yellow arrows) in representative images of the midbrain.
- Despite the use of a non-specific promoter (CAG) in this study, *PRNP* repression and ZFR expression was predominantly observed in Neun+ cells, suggesting that STAC-BBB is primarily neurotropic.

Conclusion and next steps

- ZFRs potently and specifically repress human *PRNP in vitro* in human iPSC-derived neurons.
- Neuronal promoter driven ZFRs potently reduce prion mRNA and protein expression in human *PRNP* transgenic mice. The ZFRs were well tolerated following systemic administration and showed no signs of pathology or toxicity in brain.
- Intravenous administration of STAC-BBB delivering a prion-targeted ZFR to adult NHPs resulted in widespread neuronal ZFR expression. Repression of *PRNP* mRNA was detected at the bulk tissue and single-cell level throughout the brain.
- Ongoing quantification will characterize % transduction of neuronal vs non-neuronal cells and *PRNP* repression at the single-cell level in different brain regions.
- This work supports the initiation of an IND-enabling toxicology study for a STAC-BBB delivered ZFR for the potential treatment of prion disease.

Presented at ASGCT 2024