Site-directed integration of large DNA sequences into endogenous sites in the human genome using engineered Modular Integrases (MINTs)

Friedrich Fauser, Sebastian Arangundy-Franklin, Jessica E Davis, Lifeng Liu, Nicholas A Scarlott, Nicola J Schmidt, Rakshaa Mureli, Danny F Xia, Luis Rodriguez, Mohammad Qasim, Vishvesha Vaidya, Sarah J Hinkley, Bhakti N Kadam, Nga Nguyen, Stephen Lam, Bryan Bourgeois, Adeline Chen, Andrew Nguyen, Patrick Li, David E Paschon, Gregory D Davis and Jeffrey C Miller

Sangamo THERAPEUTICS

Poster #1680

Introduction

- Despite much recent progress in the ability to manipulate the genomes of eukaryotic cells, there is still no effective and practical method to precisely integrate large synthetic DNA constructs into desirable chromosomal sites using a programable integrase.
- Serine integrases can perform the necessary molecular steps, but only after their natural target site is first installed into the recipient genome by other methods. A more elegant approach would be to directly reprogram the serine integrase itself to target a desired sequence already present in the genome.
- Here we describe the development of Modular Integrases (MINTs), a versatile protein-guided genome editing method that can facilitate site-directed targeted integration (TI) of synthetic DNA into endogenous chromosomal sites (native sites in the human genome that are different from the natural Bxb1 recognition site).
- Through a combination of structural modeling, directed evolution, and screening in human cells we have reprogramed the specificity of the serine integrase Bxb1. We then utilized these reprogrammed Bxb1 variants to enable precise integration of kilobase-sized constructs into multiple endogenous locations within the human genome with high activity and promising genome-wide specificity.
- We envision that our MINT platform could be used for correcting many disease-causing mutations in a diverse patient population by integrating a correct copy of the gene into it's natural locus.
- The MINT platform could be deployed for neurology-focused indications, but also provides potential partnering opportunities for the development of next-generation genomic medicines in ex vivo oncology, and hematology.
- We show the therapeutic potential of our MINT platform by retargeting Bxb1 to the human TRAC gene and AAVS1 safe harbor locus, and by demonstrating AAV-mediated donor delivery.

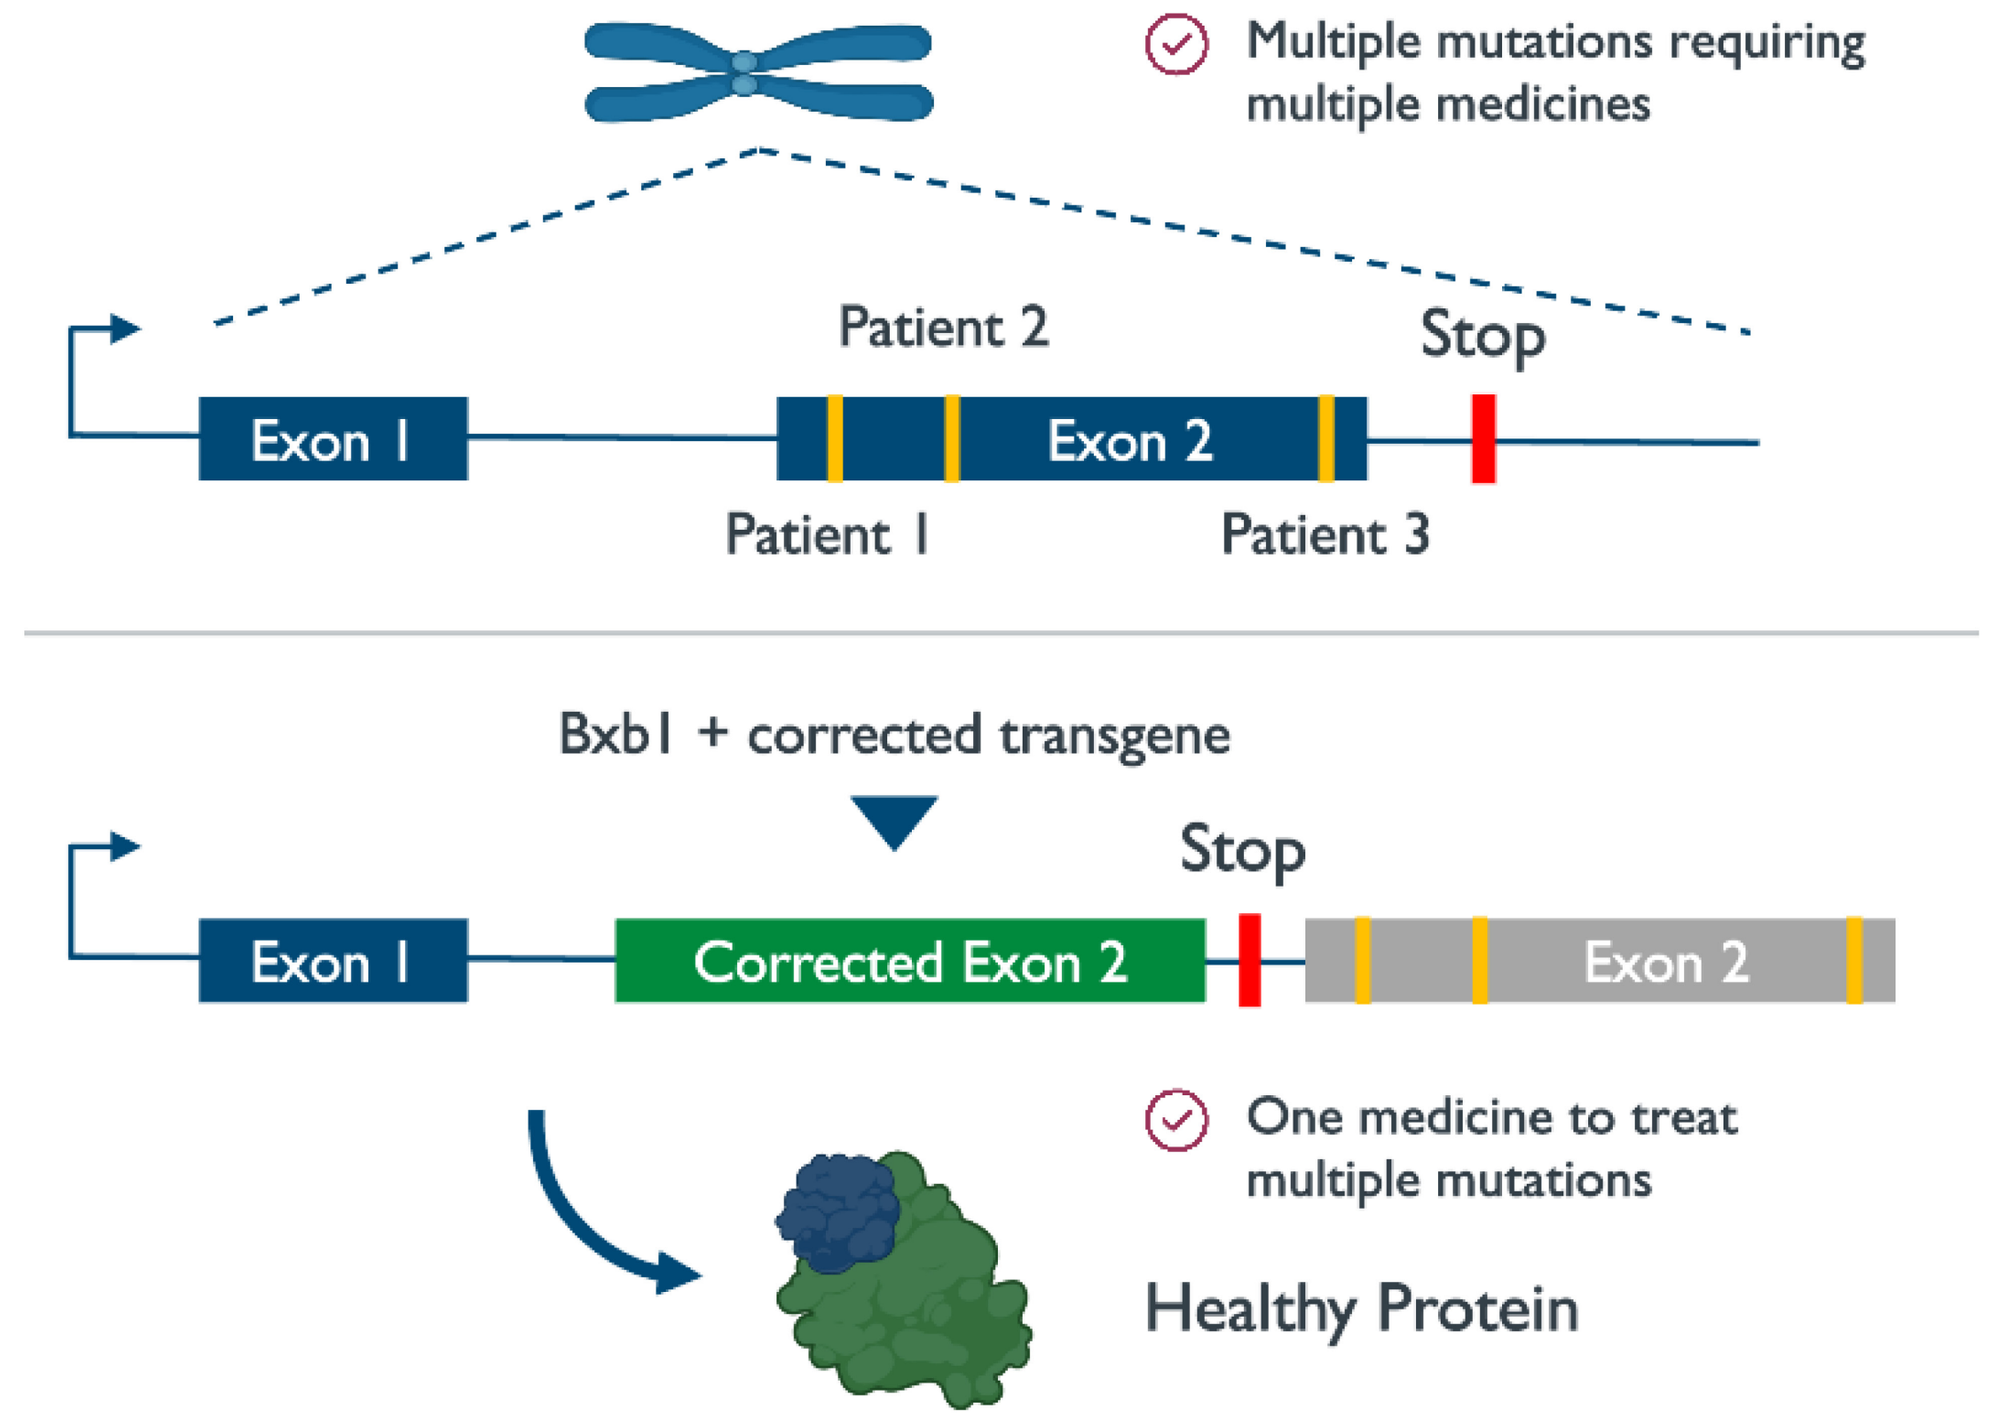
Multiple mutations requiring multiple medicines
Patient 2
Stop
Exon 1
Exon 2
Patient 1
Patient 3
Bxb1 + corrected transgene
Stop
Exon 1
Corrected Exon 2
Exon 2
One medicine to treat multiple mutations
Healthy Protein

Bxb1 efficiently facilitates targeted integration in human cells

- During the phage lifecycle, integration occurs when one Bxb1 dimer binds to a phage attP site, and a second dimer binds an attB site in the bacterial host's genome.
- As an initial test of our wild-type Bxb1 constructs, we tested them in a "landing pad" cell line where we had first integrated the natural attB or attP target in human K562 cells using nuclease-mediated HDR.
- We observed the highest targeted integration efficiency in human K562 cells when targeting an attB landing pad within the AAVS1 locus.

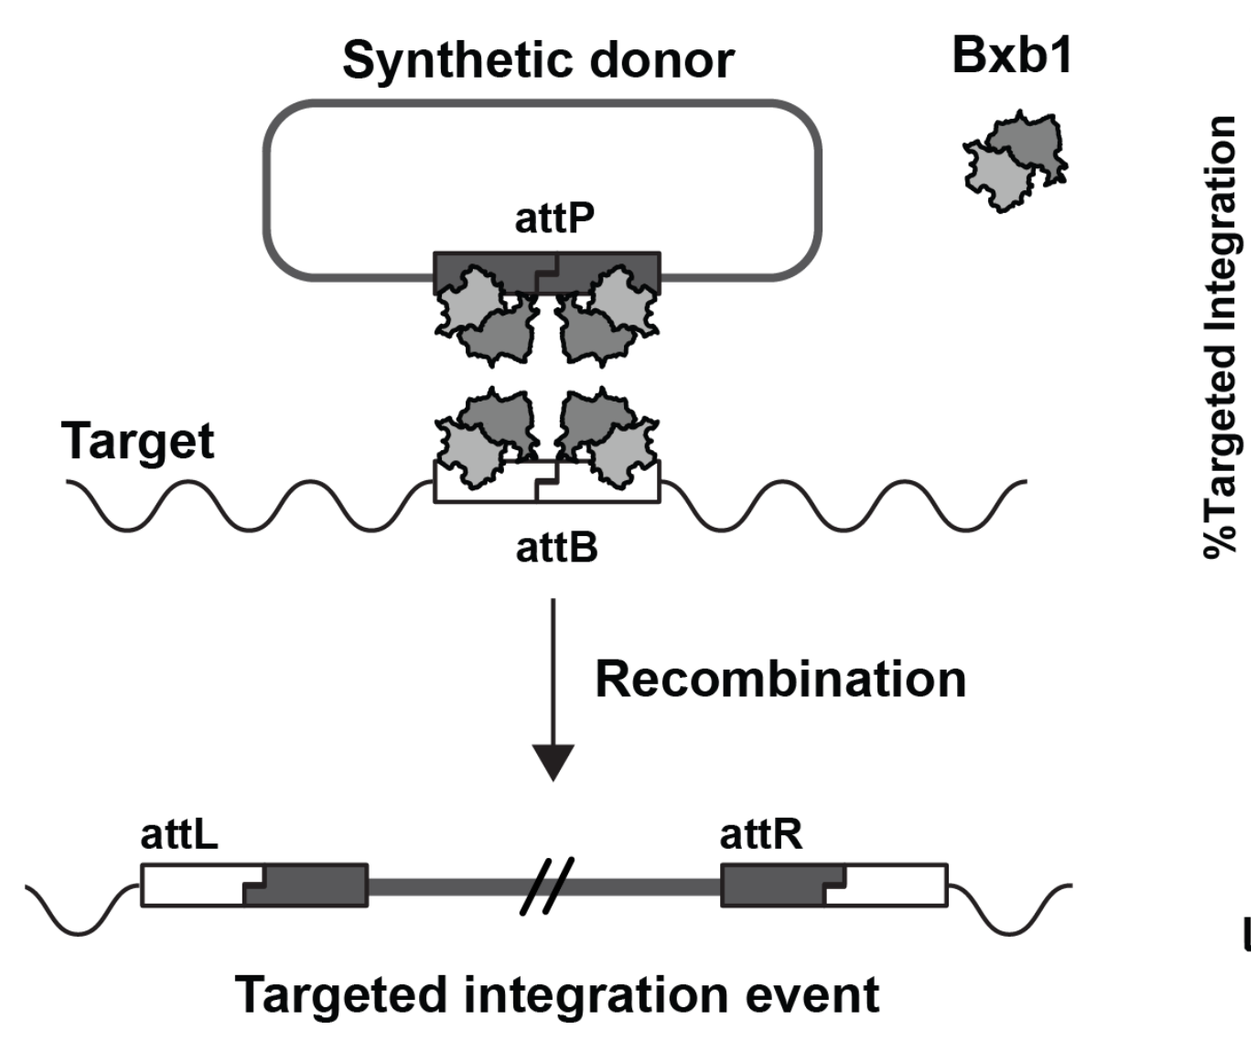
Synthetic donor
Bxb1
attP
Target
attB
Recombination
attL
attR
Targeted integration event

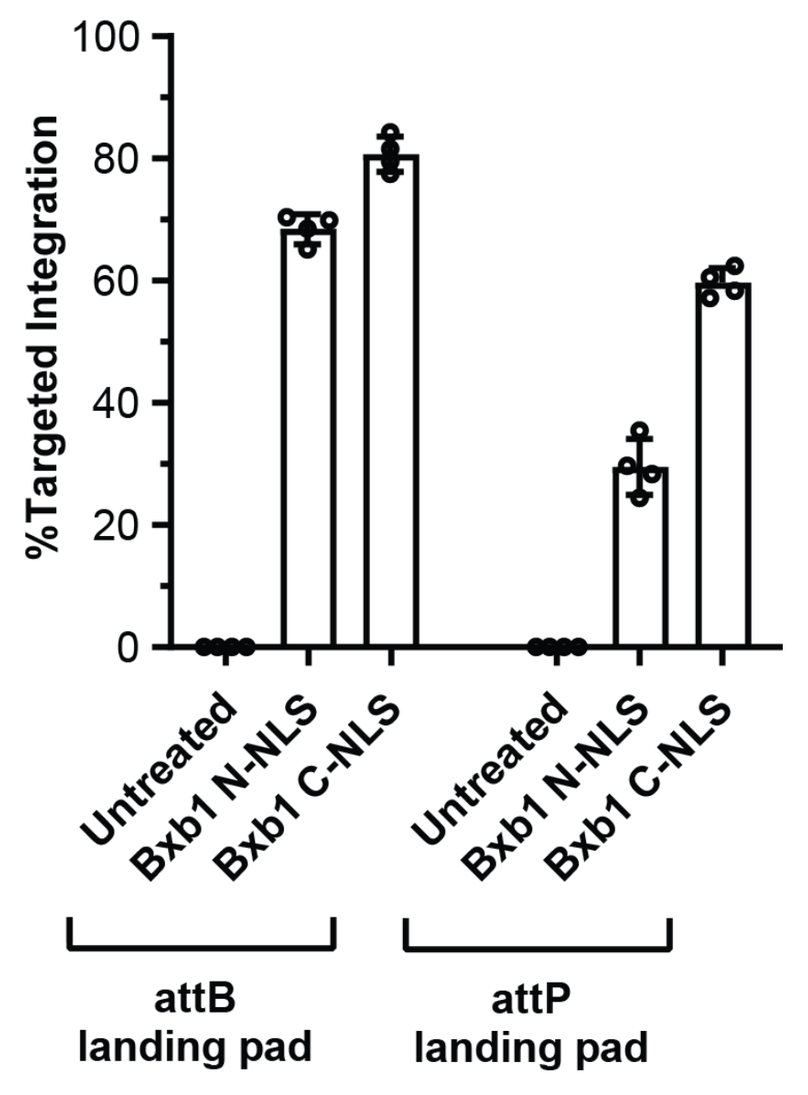
%Targeted Integration
100
80
60
40
20
0
Untreated
Bxb1 N-NLS
Bxb1 C-NLS
Untreated
Bxb1 N-NLS
Bxb1 C-NLS
attB landing pad
attP landing pad

Bxb1-DNA interaction mapping

- The use of serine integrases as genome editing reagents has been hampered by the difficulty in their reprogramming to non-cognate sites.
- We have used a combination of structural modeling and experimental characterization of a series of systematic mutations to both Bxb1 and its target site to map critical Bxb1-DNA interactions.
- We conjectured that, if Bxb1 recognized its target sites in a modular fashion, we could reprogram its DNA target specificity by performing directed evolution on each domain in parallel using Bxb1 libraries that randomize the residues that make sequence-specific DNA contacts.

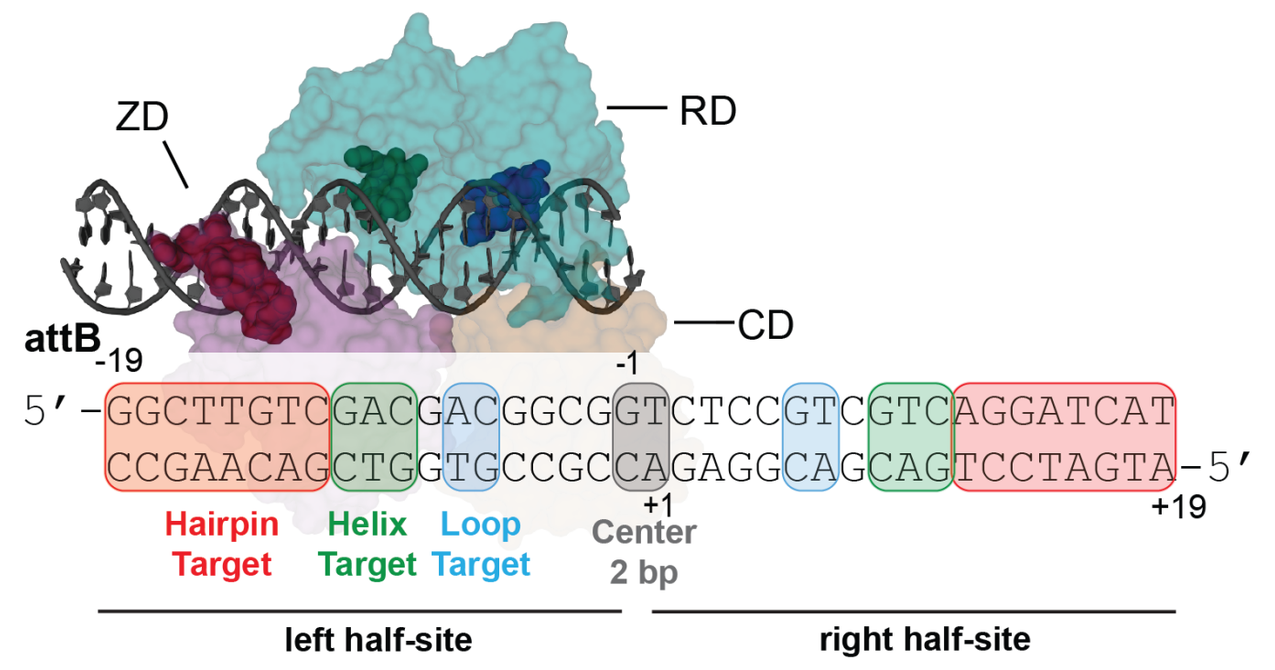
ZD
RD
CD
attB
-19
-1
5′-GGCTTGTCGACGACGGCGGTCTCCGTCGTCAGGATCAT
CCGAACAGCTGCTGCCGCCAGAGGCAGCAGTCCTAGTA-5′
+1
+19
Hairpin Target
Helix Target
Loop Target
Center 2 bp
left half-site
right half-site

Bxb1

```
                 154   159
NRSAAHFNIRAGKYRGSLPPWGYLPTRVDGEWRLVPDPVQ
                 Loop
RERILEVYHRVVDNHEPLHLVAHDLNRRGVLSPKDYFAQL
            231    237
QGREPQGREWSATALKRSMISEAMLGYATLNGKTVRDDDG
             Helix
APLVRAEPILTREQLEALRAELVKTSRAKPAVSTPSLLLR
              314         325
VLFCAVCGEPAYKFAGGGRKHPRYRCRSMGFPKHCGNGTV
                Hairpin
```

DNA recognition helix engineering changes Bxb1 specificity

- We developed a directed evolution system in which a recombinase variant present in a library is enriched when it can restore the open reading frame of an antibiotic-resistance gene if it acts on a new target sequence.
- We carried out 64 separate selections using our library of Bxb1 helix variants and each possible DNA triplet sequence:

 SATALKR wild-type Bxb1 residues 231-237
 XXXXLXX helix variant library

- We characterized the targeting specificity of the wild-type helix (SATALKR) which yielded a preference for NAC DNA binding motifs (N = G/A/T/C) consistent with natural target sites (attB: GAC; attP: AAC and CAC).
- Many of the selected helices were able to targeted alternative DNA triplets with high molecular specificity, and the data suggests interaction patterns.

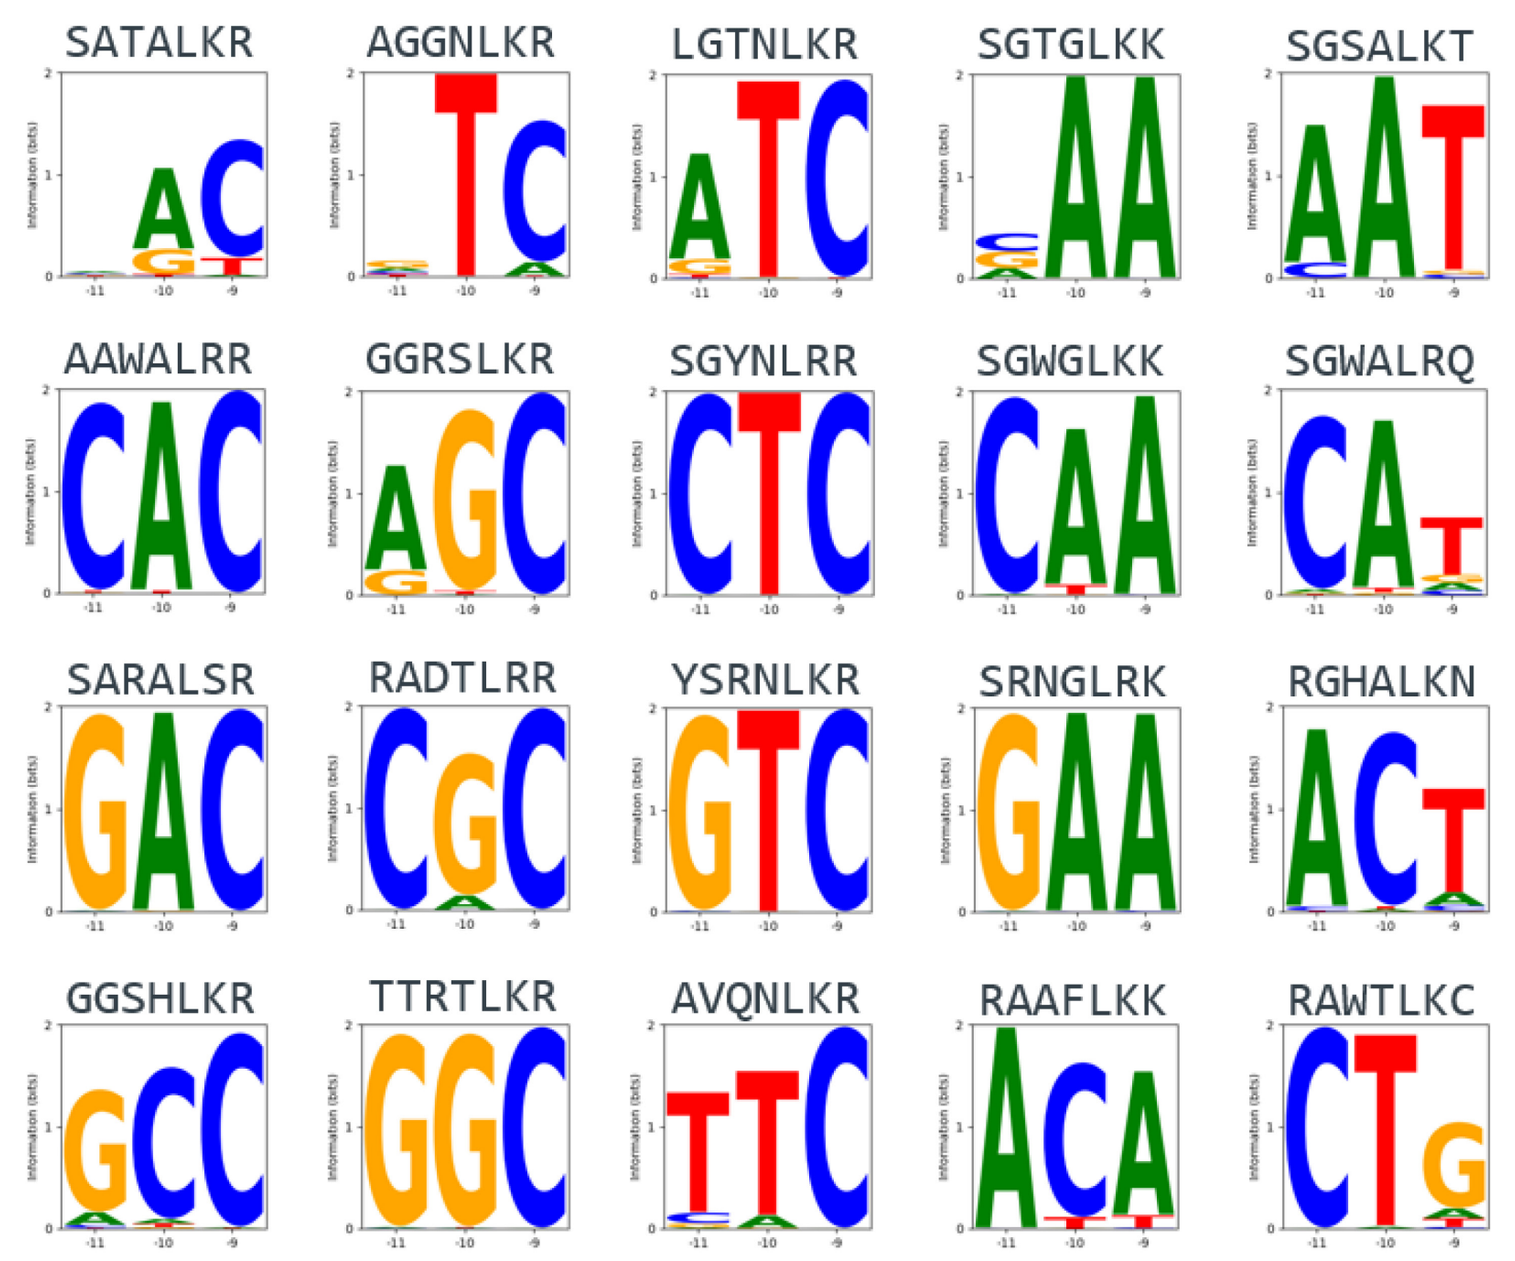
SATALKR
AGGNLKR
LGTNLKR
SGTGLKK
SGSALKT
AAWALRR
GGRSLKR
SGYNLRR
SGWGLKK
SGWALRQ
SARALSR
RADTLRR
YSRNLKR
SRNGLRK
RGHALKN
GGSHLKR
TTRTLKR
AVQNLKR
RAAFLKK
RAWTLKC

Details presented by Sebastian Arangundy-Franklin:
"192: DIRECTED EVOLUTION OF BXB1 FOR THE DEVELOPMENT OF MODULAR INTEGRASES (MINTs)"
Thursday, May 9th 4:00-4:15 pm – Ballroom 3

Performance of Bxb1 variants at endogenous human sites

- We identified Bxb1 pseudo-sites in human K562 cells using both computational and experimental methods. We then generated a plasmid target library consisting of corresponding RD and ZD regions and tested evolved Bxb1 variants against it.

Pseudo attB site: MACO1

```
5′-GCCCCTTCTCCTACAGAGCAAGCAGCAGGGTAAATTCT
   CGGGGAAGAGGATGTCTCGTTCGTCGTCCCATTTAAGA-5′
```

Pseudo attB site: GYS1

```
5′-GGGATTCCCATAACCGTGCACTCAGCTGCGGGAAGGCA
   CCCTAAGGGTATTGGCACGTGAGTCGACGCCCTTCCGT-5′
   Hairpin   Helix Loop      Loop Helix  Hairpin
     ZD         RD             RD        ZD
      left half-site          right half-site
```

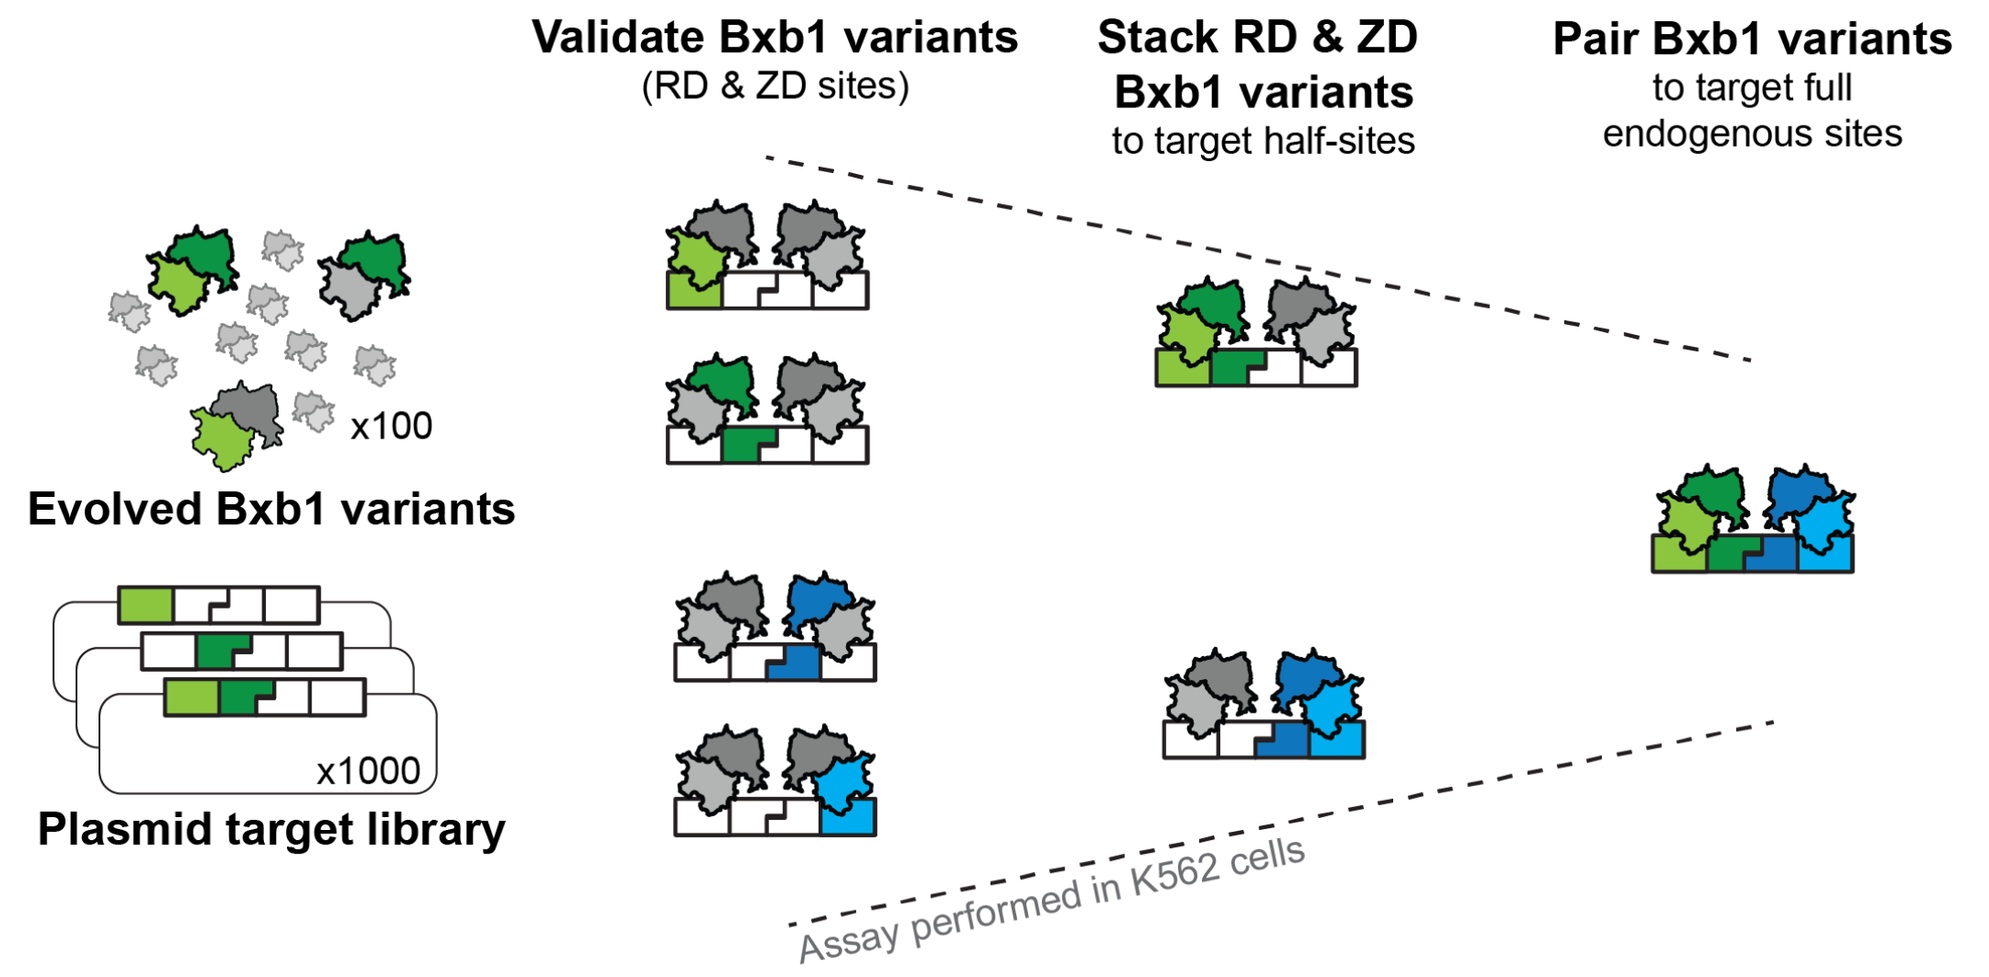
Validate Bxb1 variants
(RD & ZD sites)
Stack RD & ZD Bxb1 variants
to target half-sites
Pair Bxb1 variants
to target full endogenous sites
x100
Evolved Bxb1 variants
x1000
Plasmid target library
Assay performed in K562 cells

- Next, we tested Bxb1 variant pairs against their chromosomal target sites. This resulted in variant pairs that substantially outperformed wild-type Bxb1.

	Loop	Helix	Hairpin
		Bxb1 peptide sequence derived from directed evolution	
Wild-type Bxb1	YRGSLP	SATALKR	FAGGGRKHPRYR
MACO1-L	YRGSLP	SATALKR	MAGGHRKQALYR
MACO1-R	YRGSLP	RAWSLKR	MAGGPRKKGRYR
GYS1-L	YRGSLP	HGWSLKV	LARGSRKLALYR
GYS1-R	YRGSLP	HGCTLKR	NARGNRKRGRYR

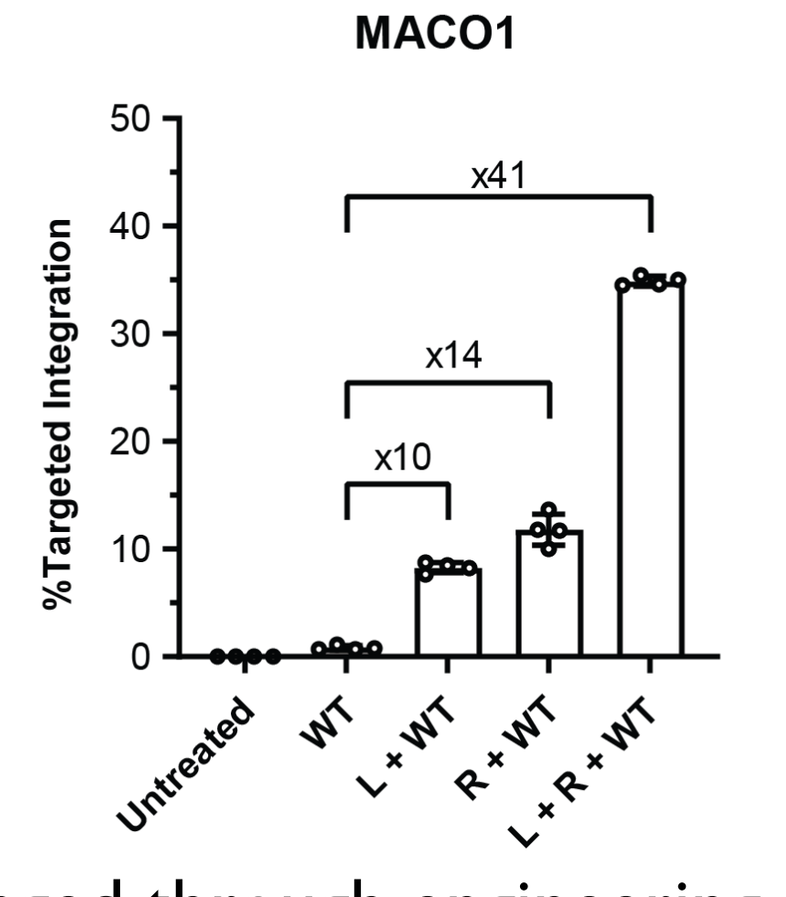
MACO1
%Targeted Integration
50
40
30
20
10
0
x41
x14
x10
Untreated
WT
L + WT
R + WT
L + R + WT

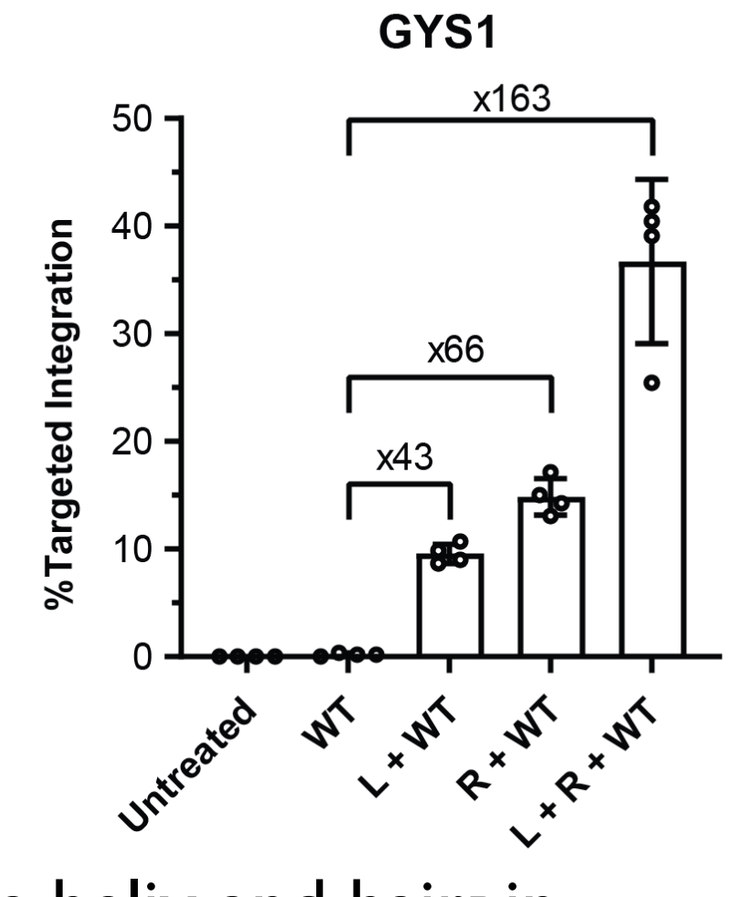
GYS1
%Targeted Integration
50
40
30
20
10
0
x163
x66
x43
Untreated
WT
L + WT
R + WT
L + R + WT

- This demonstrates that the specificity of Bxb1 can be changed through engineering of the helix and hairpin peptide regions, and that high levels of efficiency can be achieved at endogenous sites in the human genome.

Retargeting Bxb1 to therapeutically relevant sites

- Encouraged by our success targeting endogenous human sites where wild-type Bxb1 is already active, we proceeded to the more difficult challenge of targeting desired regions of the human genome where wild-type Bxb1 is inactive, such as the AAVS1 locus, a well-established safe harbor, and the TRAC locus.

TRAC: attB site

```
5′-TGGTGTCCAGGAGCCGAGGTATCGGTCCTGCCAGGGCC
   ACCACAGGTCCTCGGCTCCATAGCCAGGACGGTCCCGG-5′
```

AAVS1: attB site

```
5′-CTGAGCGCCTCTCCTGGGCTTGCCAAGGACTCAAACCC
   GACTCGCGGAGAGGACCCGAACGGTTCCTGAGTTTGGG-5′
     Hairpin   Helix Loop        Loop Helix  Hairpin
```

	Loop	Helix	Hairpin
		Bxb1 peptide sequence from directed evolution	
Wild-type Bxb1	YRGSLP	SATALKR	FAGGGRKHPRYR
TRAC-L*	YRGGLP	YGSALKQ	LARGPRKRAGYK
TRAC-R*	YRGGLP	SQWALKC	RAWGKRKYAYYQ
AAVS1-L	YRGGLP	YPWSLRR	KAWGSRKTRLYR
AAVS1-R	YRGGLP	AGGNLKR	MARGGRKSAIYY

*TRAC-L and TRAC-R variants include an additional D257K mutation

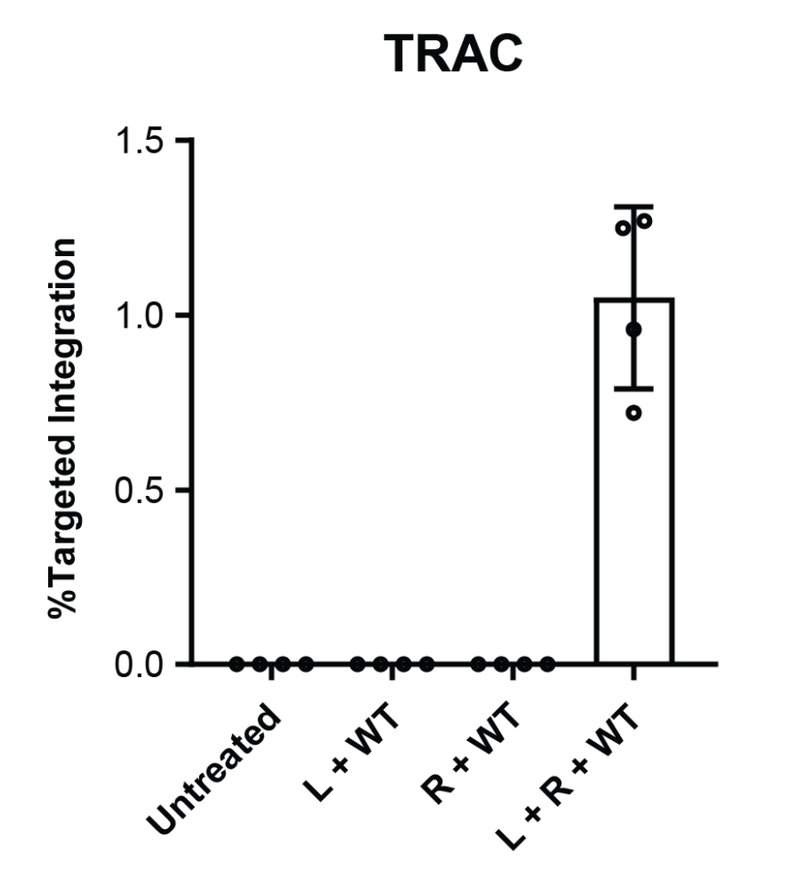
TRAC
%Targeted Integration
1.5
1.0
0.5
0.0
Untreated
L + WT
R + WT
L + R + WT

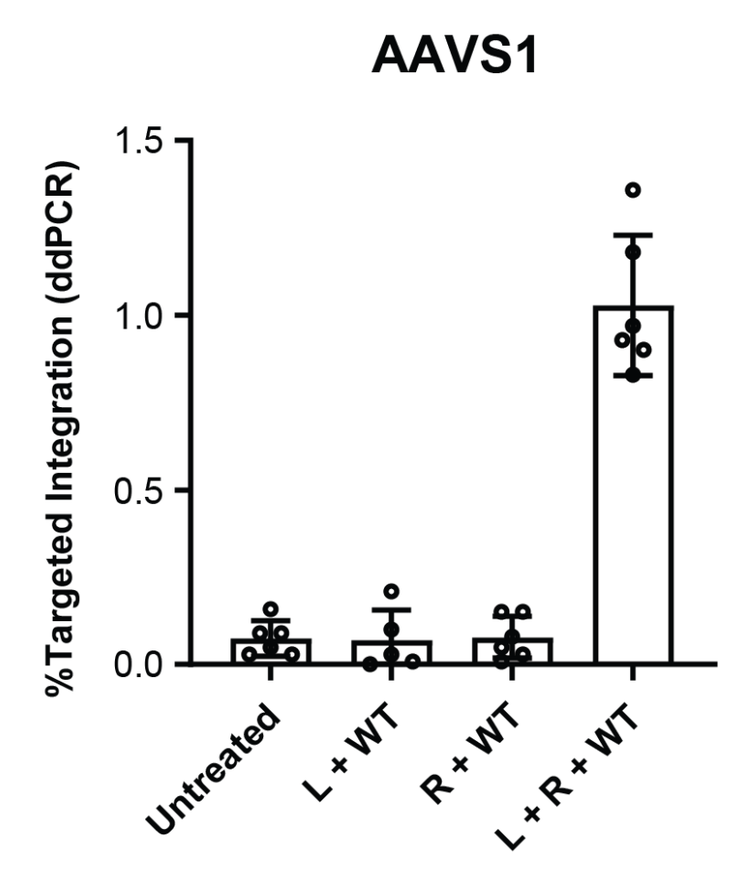
AAVS1
%Targeted Integration (ddPCR)
1.5
1.0
0.5
0.0
Untreated
L + WT
R + WT
L + R + WT

AAV-mediated donor delivery

- We established our MINT platform in human K562 cells using routine plasmid delivery protocols for the donor molecule, as well as plasmid or mRNA delivery for Bxb1.
- We envision that Bxb1 will be delivered as mRNA while the donor may be delivered via AAV, minicircle, or comparable methods.
- We successfully demonstrated donor delivery using single-stranded AAV (ssAAV), self-complementary AAV (scAAV) as well as minicircles (data not shown).

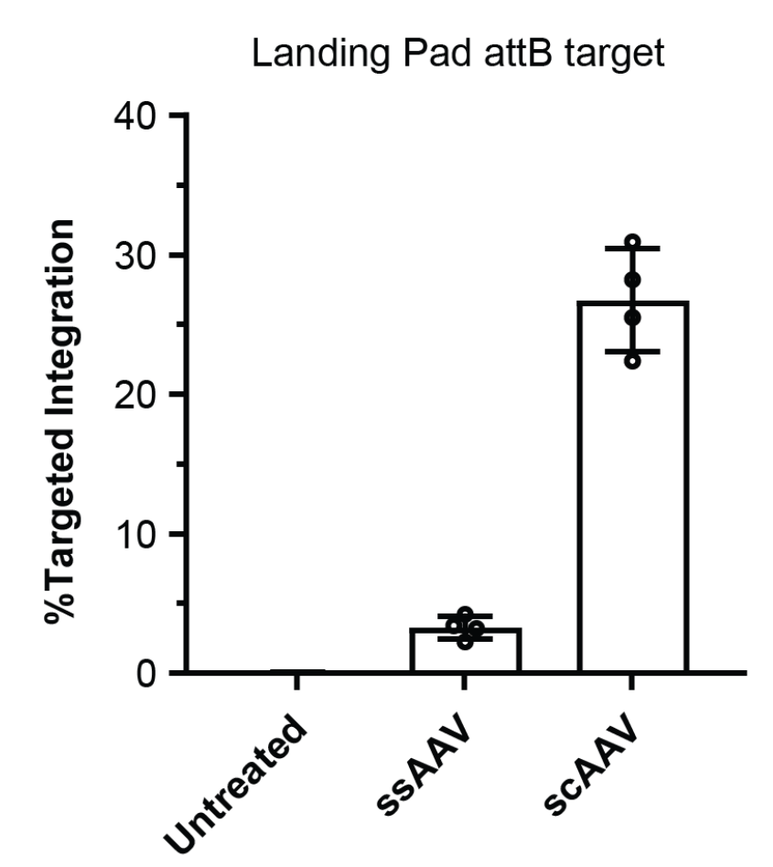
Landing Pad attB target
%Targeted Integration
40
30
20
10
0
Untreated
ssAAV
scAAV

Conclusion

- We have demonstrated the direct reprogramming of the site-specific serine integrase Bxb1.
- We showed the therapeutic potential of this approach by retargeting Bxb1 to clinically relevant sites and utilizing therapeutically relevant delivery modalities.
- In addition to human therapeutics, the MINT platform has a variety of potential applications in biomanufacturing, drug discovery, agricultural biotech settings, and basic research.

Presented at ASGCT 2024