SOD1 Gene Repression Mediated by Zinc Finger Repressors (ZFRs) as a Therapeutic Approach for SOD1-Mediated ALS

Andrew Young, Kenneth Kim, Jill Cifrese, Asa Hatami, Jisoo Lee, Patrick Dunn, David Paschon, Marian Glynn, Cecelia Paculba, Toufan Parman, Emily Tait, Stephen Lam, Sarah Hinkley, Alicia Goodwin, Alex M. Ward, Kathleen Meyer, Bryan Zeitler, Amy M. Pooler

Sangamo THERAPEUTICS

Poster #1597

Introduction

- Amyotrophic lateral sclerosis (ALS) is a progressive neurodegenerative disease that leads to loss of motor neurons in the brain and spinal cord.
- Superoxide dismutase 1 (*SOD1*) is a gene that codes for an enzyme that acts to protect cells from free radical damage.
- Mutations in *SOD1* account for 10-20% of inherited forms of ALS and thought to manifest symptoms through a mechanism whereby toxic aggregates of misfolded SOD1 protein accumulate in motor neurons leading to dysfunction and loss.
- We designed a library containing 100s of ZFRs targeting human sequences near the *SOD1* TSS.
- We identified ZFRs that repressed *SOD1* mRNA in neurons from 38%-84% with minimal detectable off-target activity.
- Select ZFRs repressed *SOD1* within key spinal cord and brain regions *in vivo* in a humanized mouse model of SOD1 ALS.

SOD1 ALS disease introduction and therapeutic strategy

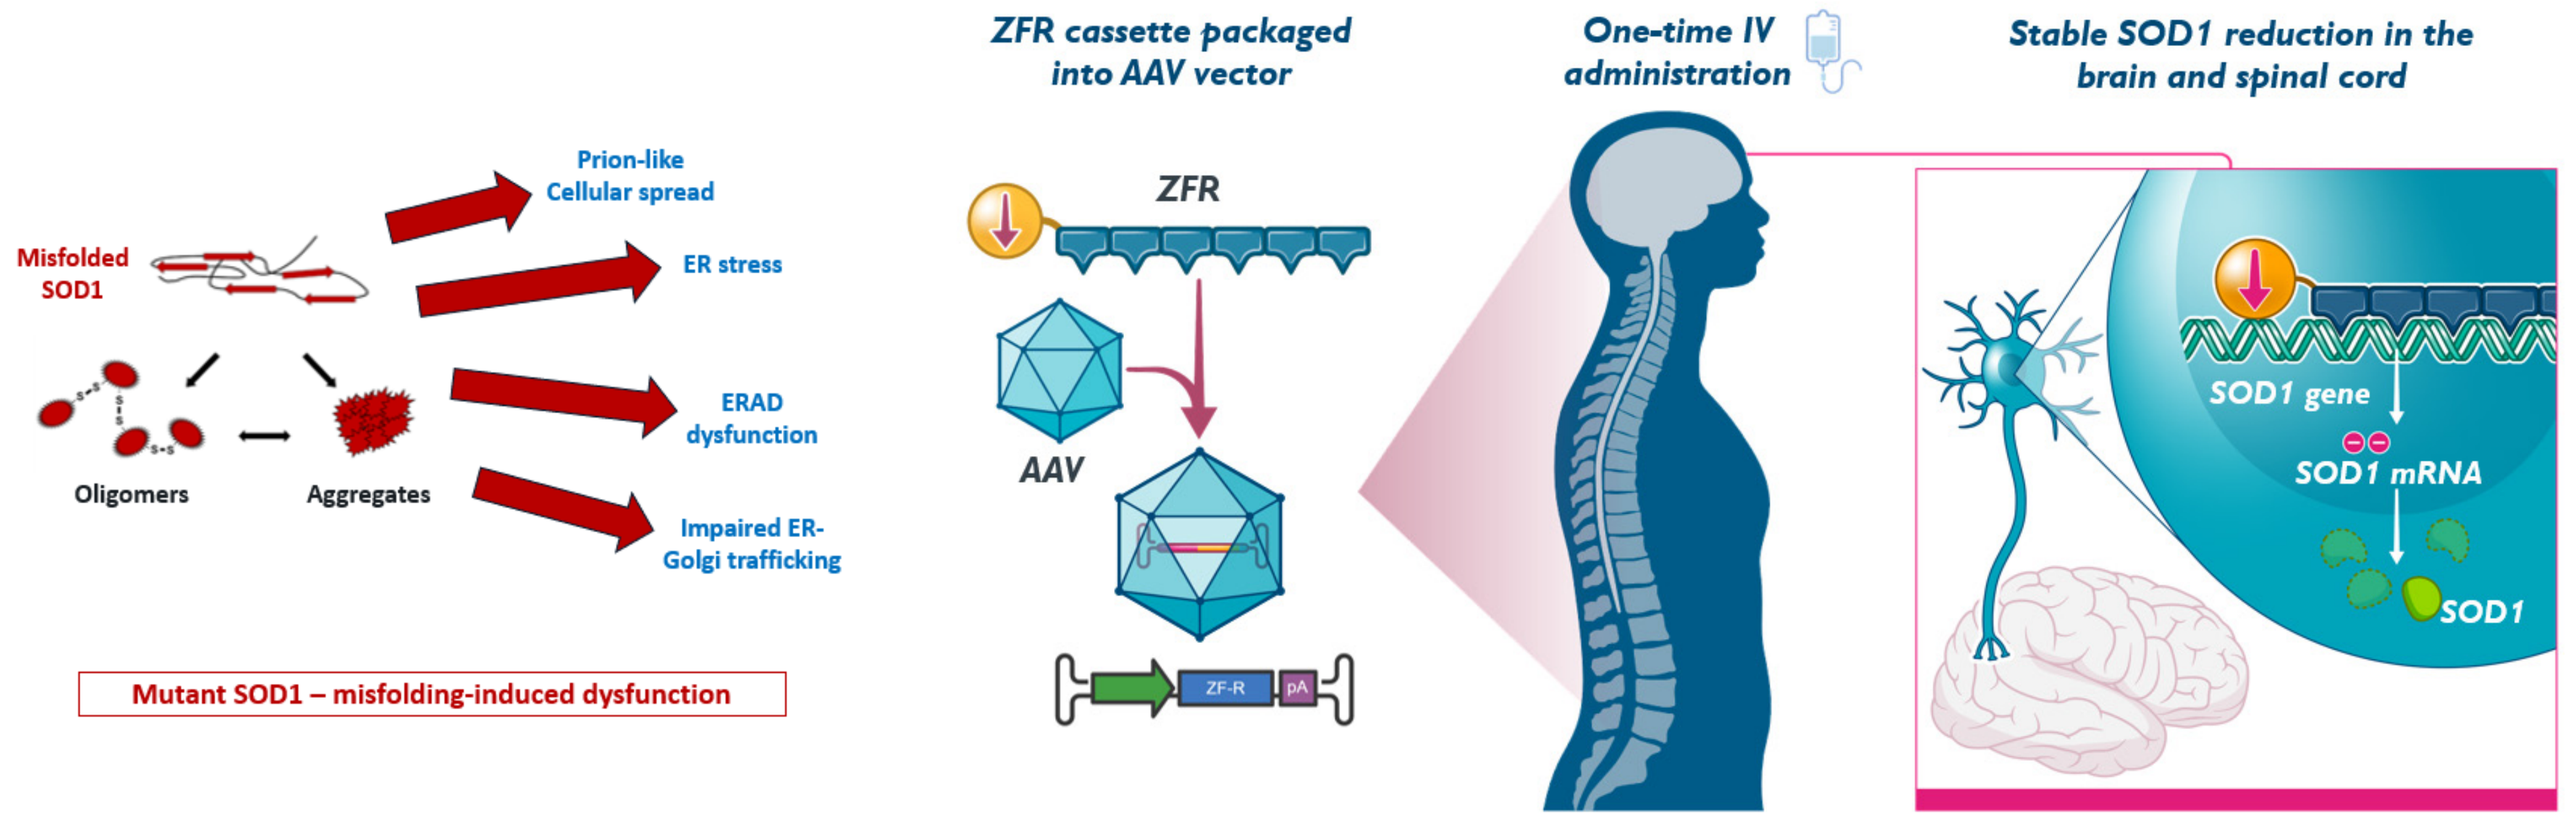
ZFR cassette packaged into AAV vector
One-time IV administration
Stable SOD1 reduction in the brain and spinal cord
Misfolded SOD1
Prion-like Cellular spread
ER stress
ERAD dysfunction
Impaired ER-Golgi trafficking
Oligomers
Aggregates
Mutant SOD1 – misfolding-induced dysfunction
ZFR
AAV
ZF-R
pA
SOD1 gene
SOD1 mRNA
SOD1

Knowledge of the pathological mechanisms underlying SOD1 ALS makes it an attractive therapeutic target for a genomic medicine. Therapeutic approach targeting motor neurons and astrocytes in the brain and spinal cord can be achieved following a single administration of a ZFR packaged in a novel intravenous capsid (STAC-BBB).

Screening strategy to identify SOD1-targeting ZFRs

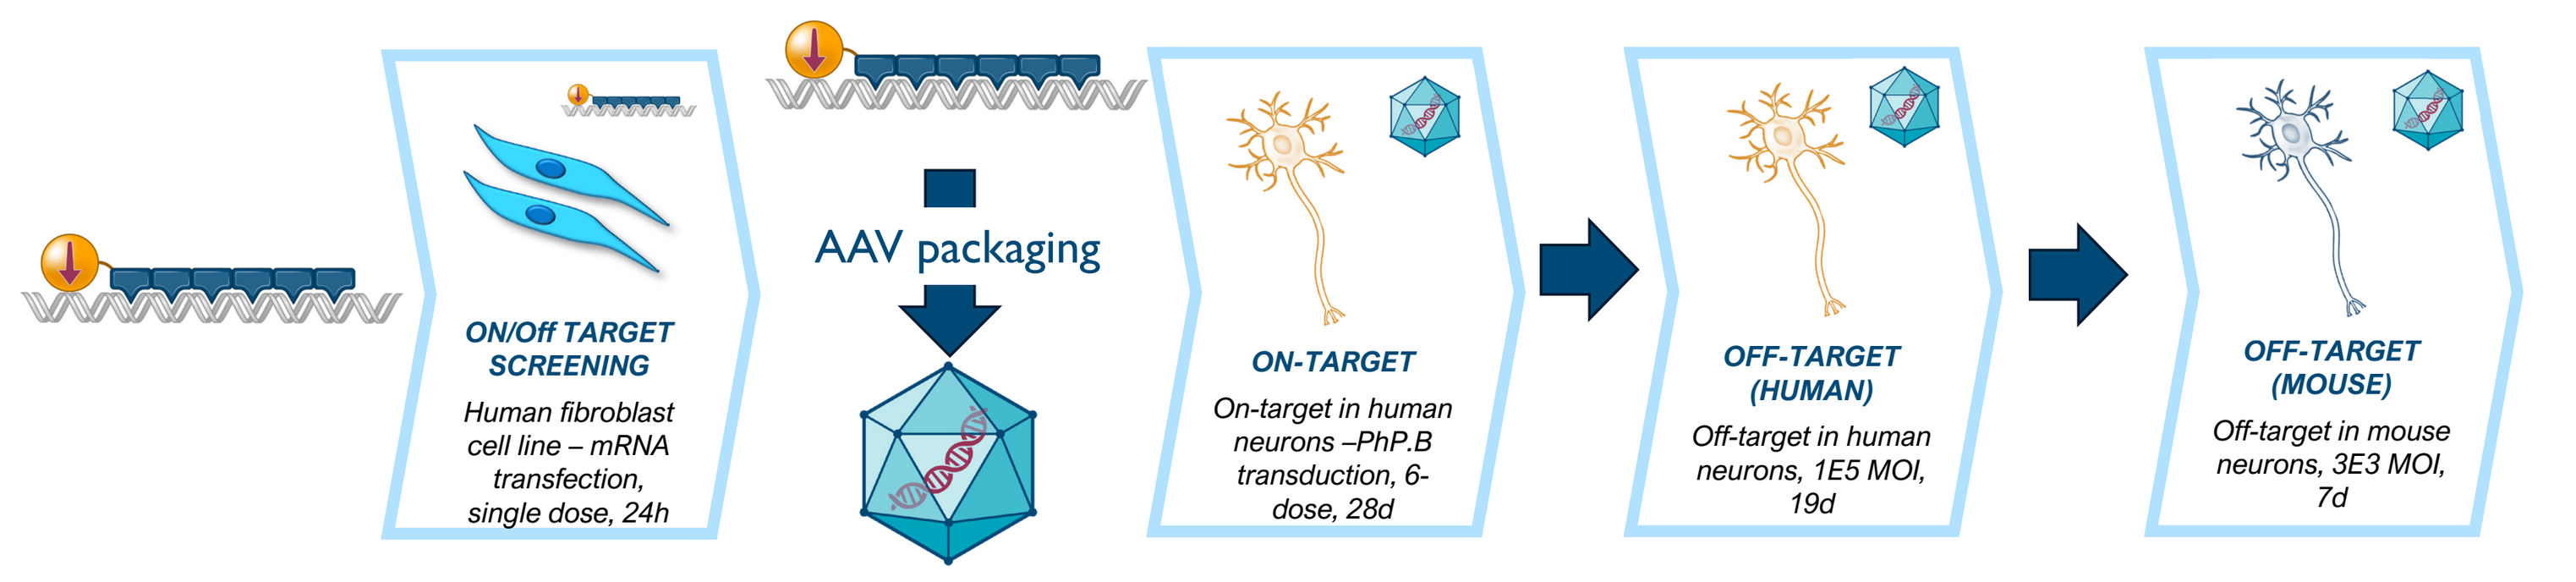
ON/Off TARGET SCREENING
Human fibroblast cell line – mRNA transfection, single dose, 24h
AAV packaging
ON-TARGET
On-target in human neurons –PhP.B transduction, 6-dose, 28d
OFF-TARGET (HUMAN)
Off-target in human neurons, 1E5 MOI, 19d
OFF-TARGET (MOUSE)
Off-target in mouse neurons, 3E3 MOI, 7d

Screening for ZFRs involved an iterative *in vitro* process; starting with human fibroblasts and then moving into human and mouse neurons.

Leveraging human fibroblasts to rapidly identify potent and specific ZFRs targeting human *SOD1*

Human fibroblasts allow for rapid screening of ZFR RNA and avoids the difficulty in nucleofecting neurons and the need to manufacture AAV vector. Once a smaller set of ZFRs are selected AAV vector can be manufactured, and the on/off target profile determined in neurons.

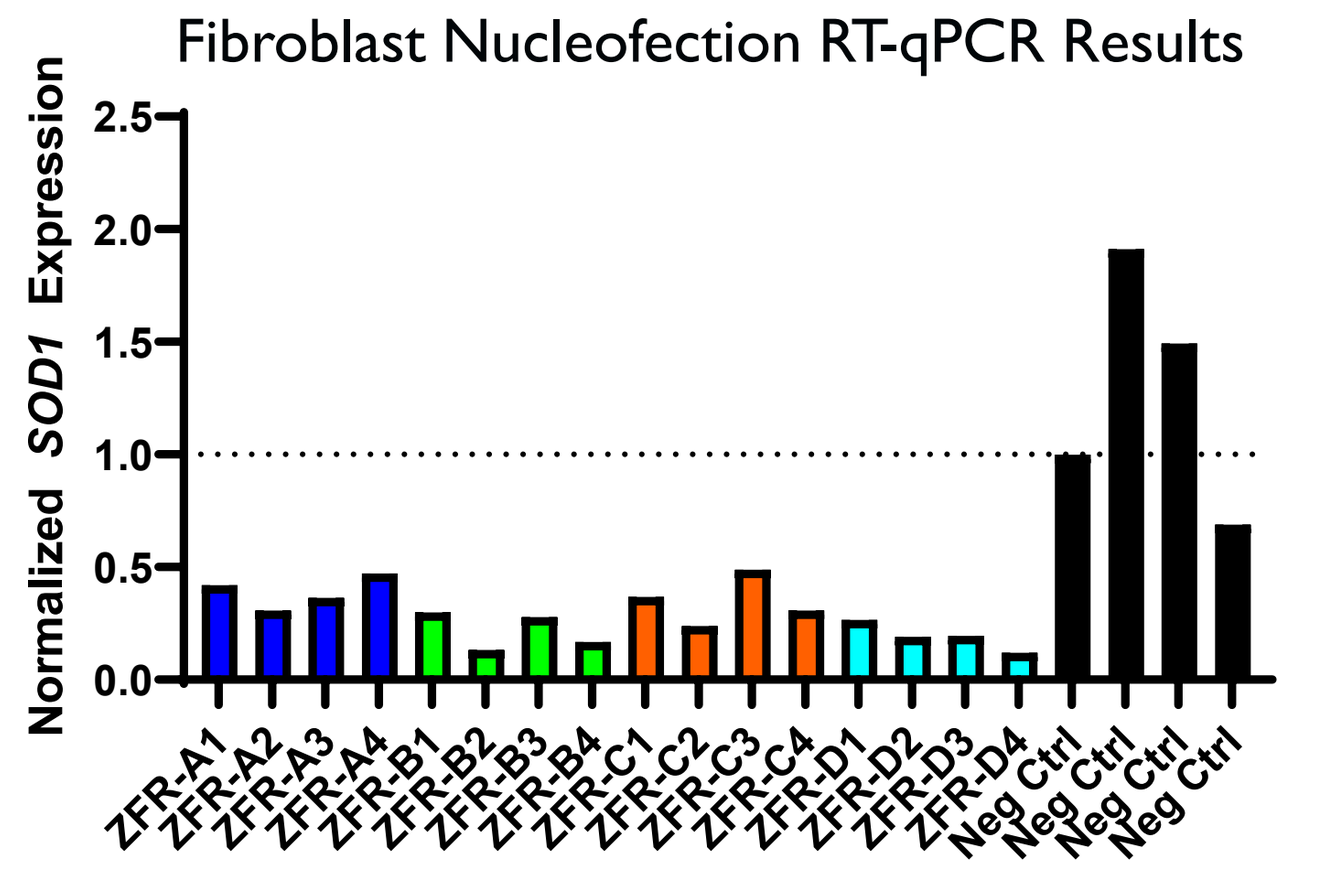
Fibroblast Nucleofection RT-qPCR Results
Normalized SOD1 Expression
ZFR-A1
ZFR-A2
ZFR-A3
ZFR-A4
ZFR-B1
ZFR-B2
ZFR-B3
ZFR-B4
ZFR-C1
ZFR-C2
ZFR-C3
ZFR-C4
ZFR-D1
ZFR-D2
ZFR-D3
ZFR-D4
Neg Ctrl

Nucleofection of 300 ng RNA per well; 4 replicates
RT-qPCR run at 22-24 hours post-nucleofection

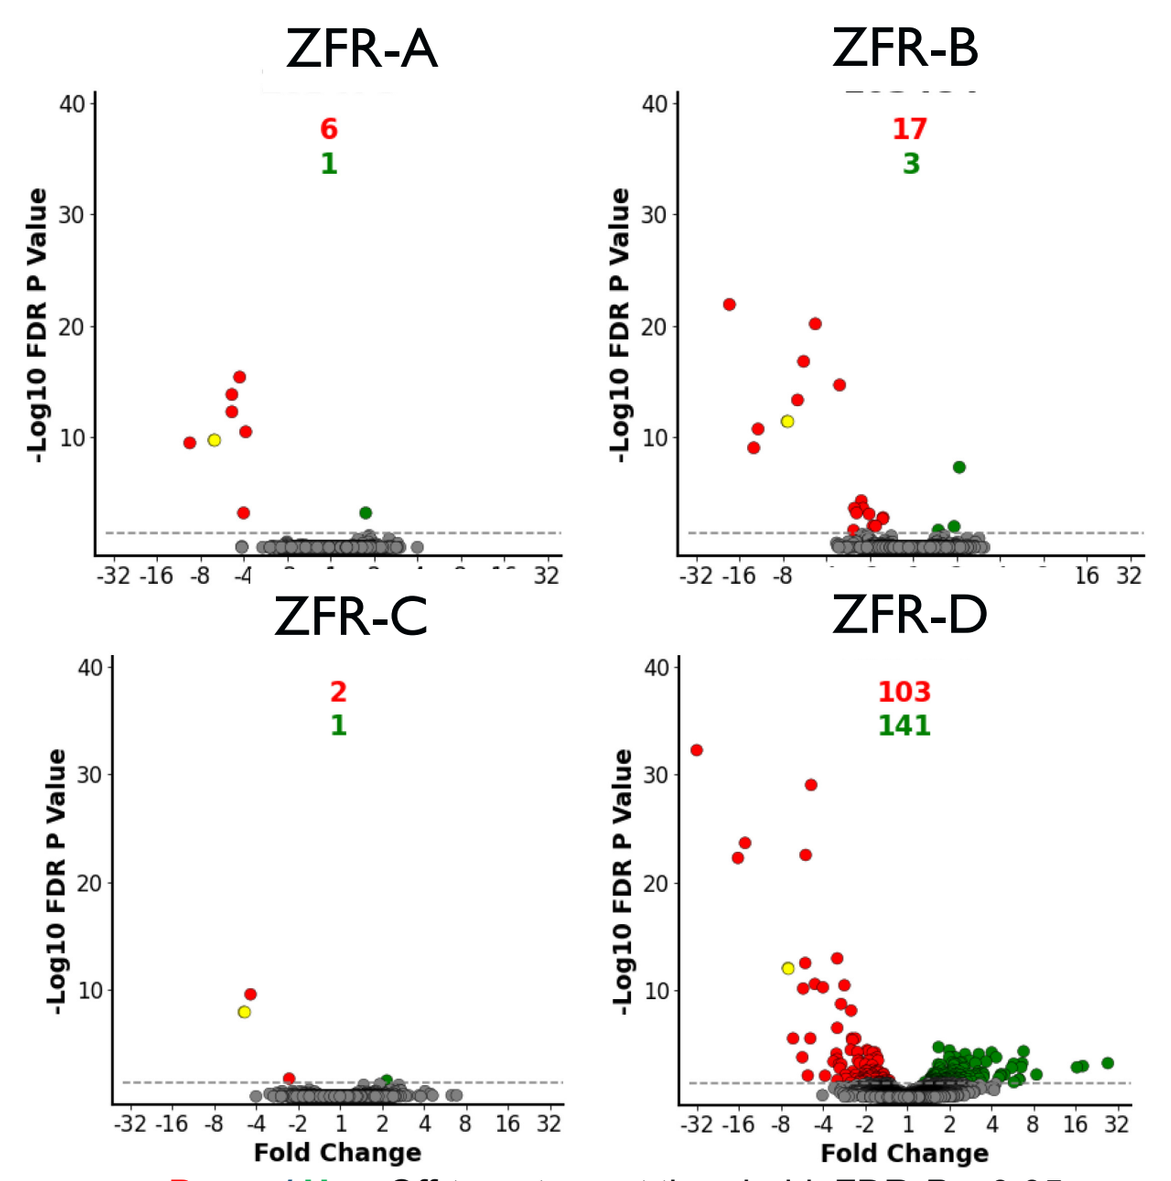
ZFR-A
ZFR-B
ZFR-C
ZFR-D
-Log10 FDR P Value
Fold Change

Down / Up : Off-target count threshold; FDR *P* < 0.05
Yellow icon indicates *SCNA* transcript

Select ZFRs show robust repression and high specificity *in vitro*

ZFR	*SOD1* Repression (max MOI)	Human DEGs	Mouse DEGs
ZFR-1	84%	3/0	4/0
ZFR-2	77%	1/0	3/1
ZFR-3	58%	1/0	0/0
ZFR-4	65%	1/0	1/1
ZFR-5	68%	4/0	0/0
ZFR-6	38%	0/0	0/0

Above: a table of select *SOD1*-targeting ZFRs that show robust on target activation and high specificity as seen in the # of differentially expressed genes (DEGs). On the left is the on-target dose response for ZFR-2 (A) and its DEG expression in volcano plots generated from microarrays following AAV transduction in human (B) and mouse neurons (C).

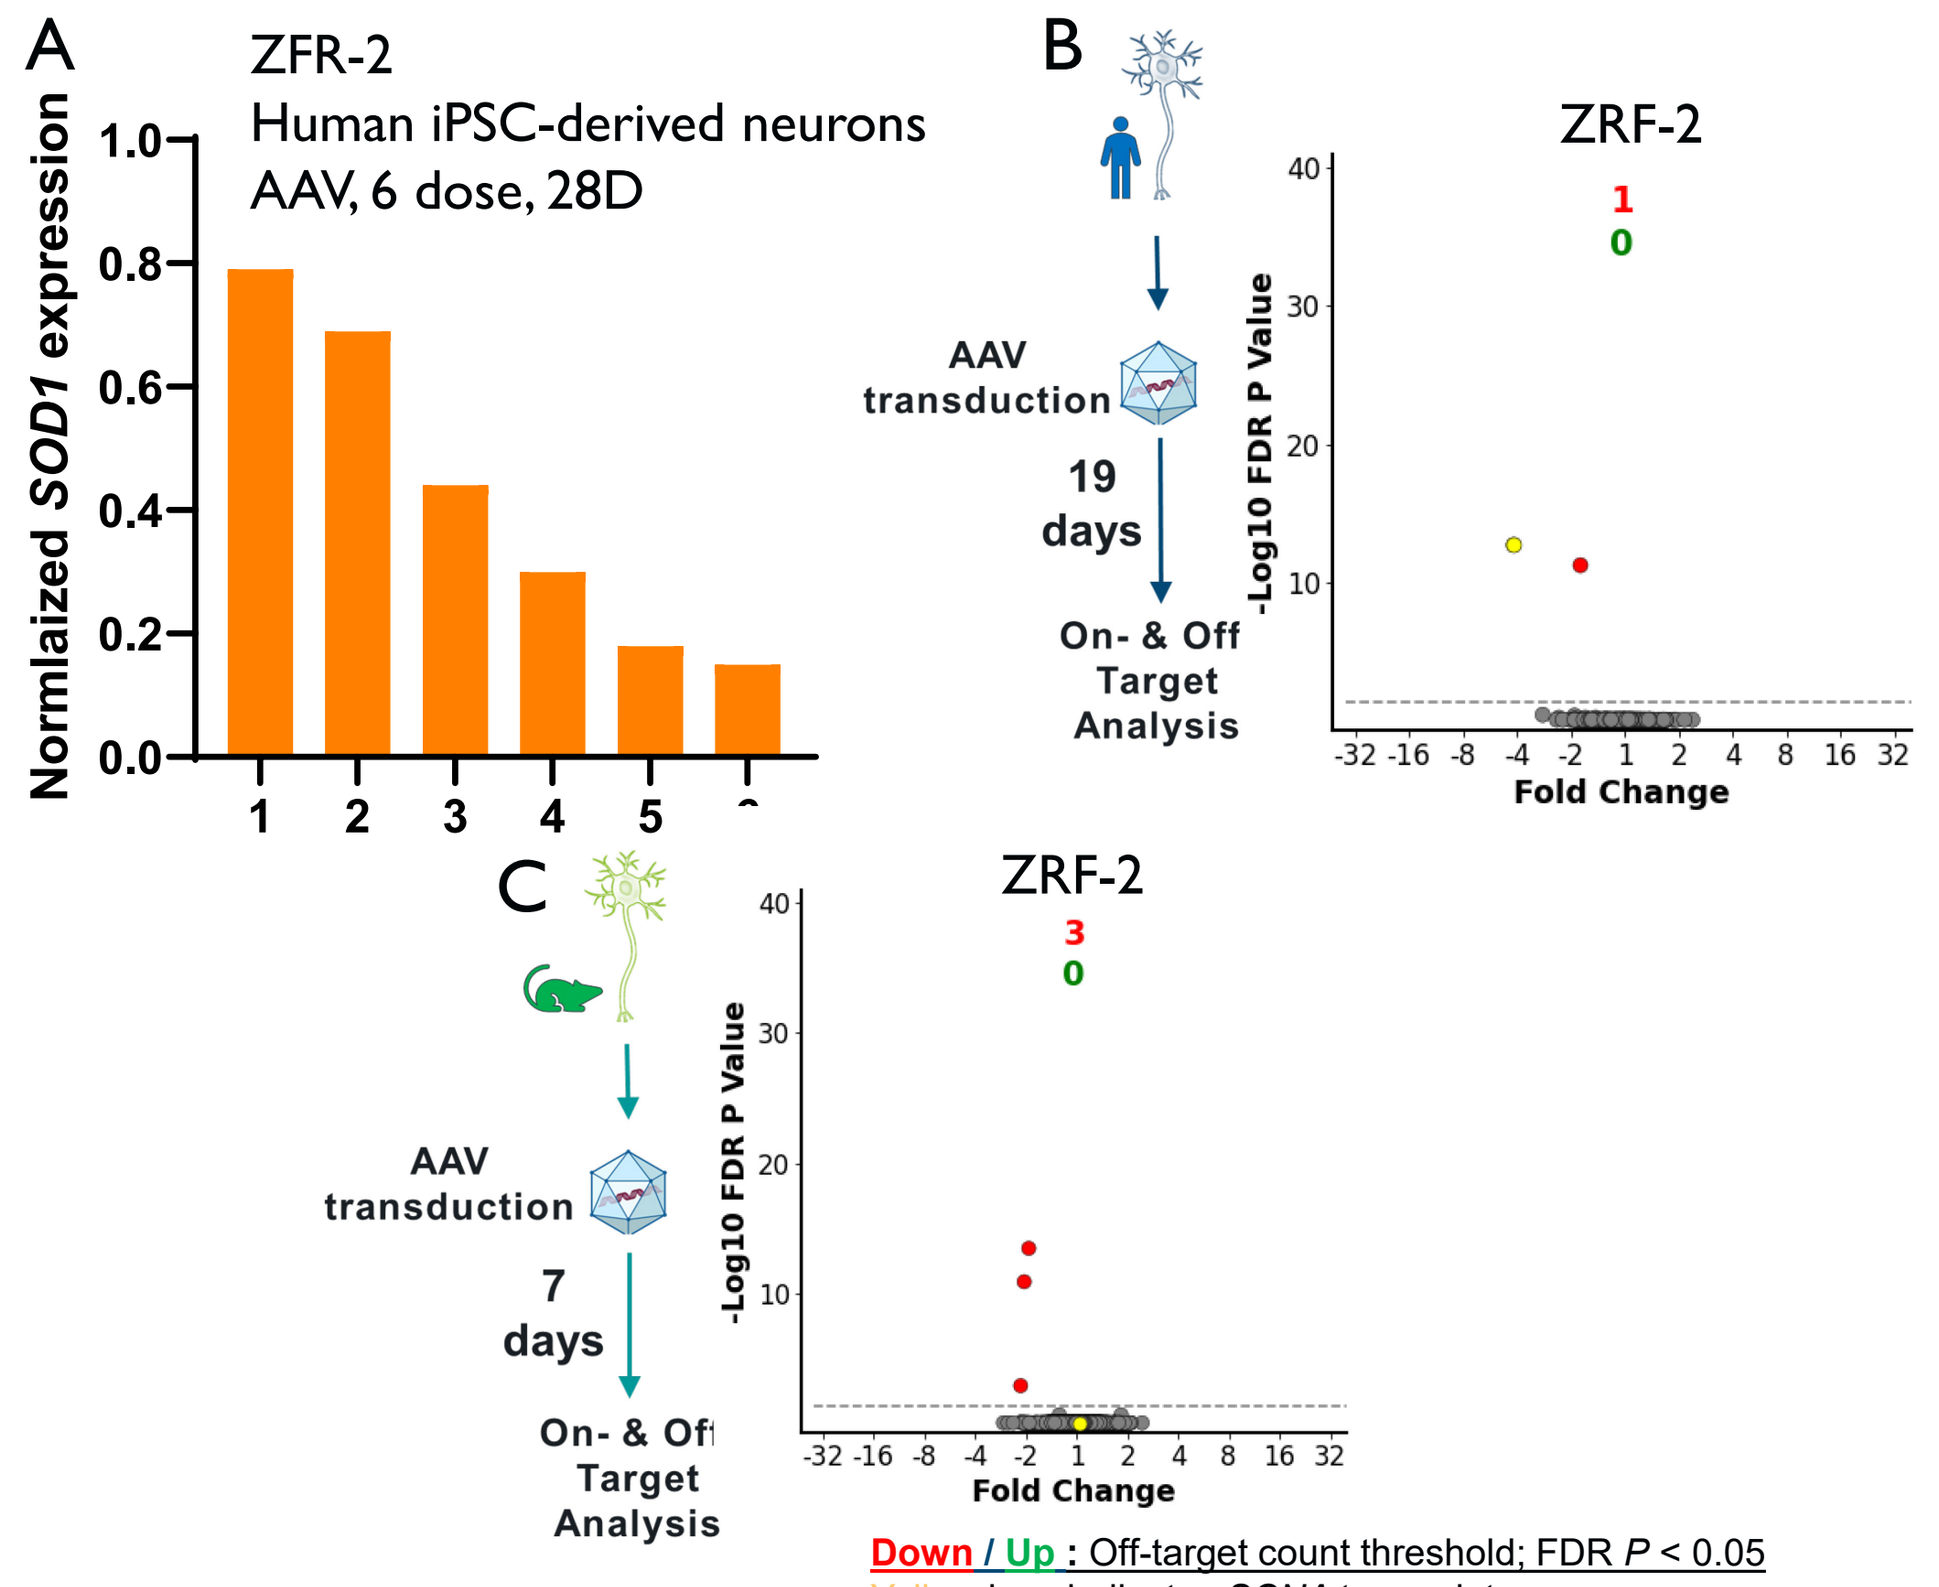
A
ZFR-2
Human iPSC-derived neurons
AAV, 6 dose, 28D
Normlaized SOD1 expression
B
ZRF-2
AAV transduction
19 days
On- & Off Target Analysis
C
ZRF-2
AAV transduction
7 days
On- & Off Target Analysis
-Log10 FDR P Value
Fold Change

Down / Up : Off-target count threshold; FDR *P* < 0.05
Yellow icon indicates *SCNA* transcript

Approach for *in vivo* testing of ZFRs in a humanized mouse model

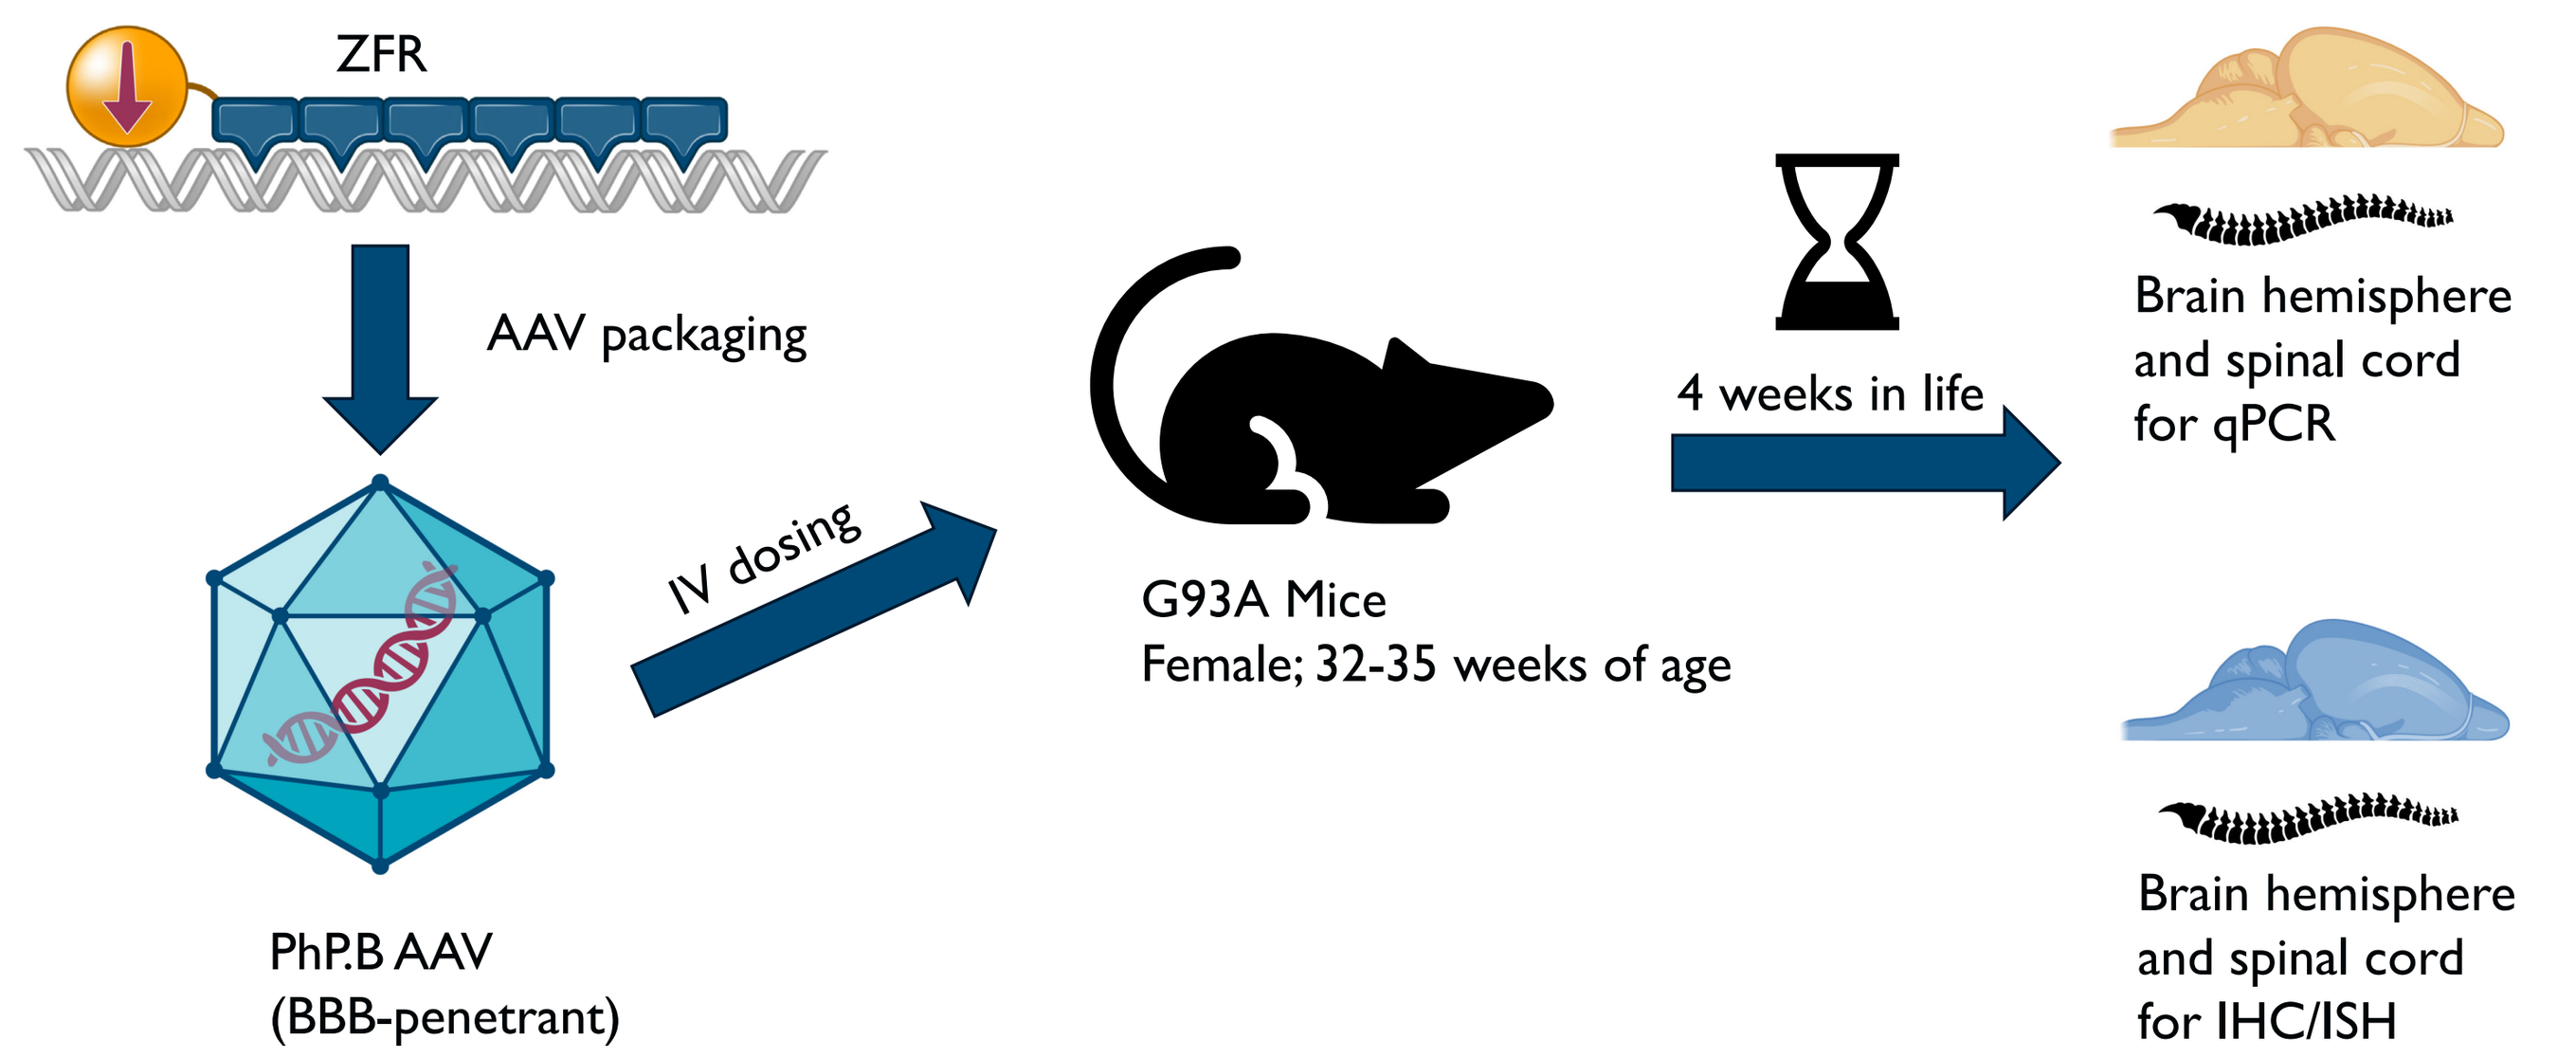
ZFR
AAV packaging
PhP.B AAV
(BBB-penetrant)
IV dosing
G93A Mice
Female; 32-35 weeks of age
4 weeks in life
Brain hemisphere and spinal cord for qPCR
Brain hemisphere and spinal cord for IHC/ISH

Following *in vitro* characterization, 5 ZFRs were packaged into PhP.B AAV with a human synapsin promoter. AAV was administered via tail vein injection to G93A mice at a dose of 1E14 vg/kg. After 4 weeks brain and spinal cord were processed individually for molecular and histological analysis.

ZFRs repress human *SOD1* transcript in key brain regions

Tissue collected from G93A mice and analyzed by RT-qPCR showed a significant decrease in *SOD1* expression in all the ZFR treated groups when compared with a vehicle treated control group. Cervical spinal cord is a critical center of neurodegeneration in SOD1 ALS patients and an important target for a genomic medicine therapeutic approach.

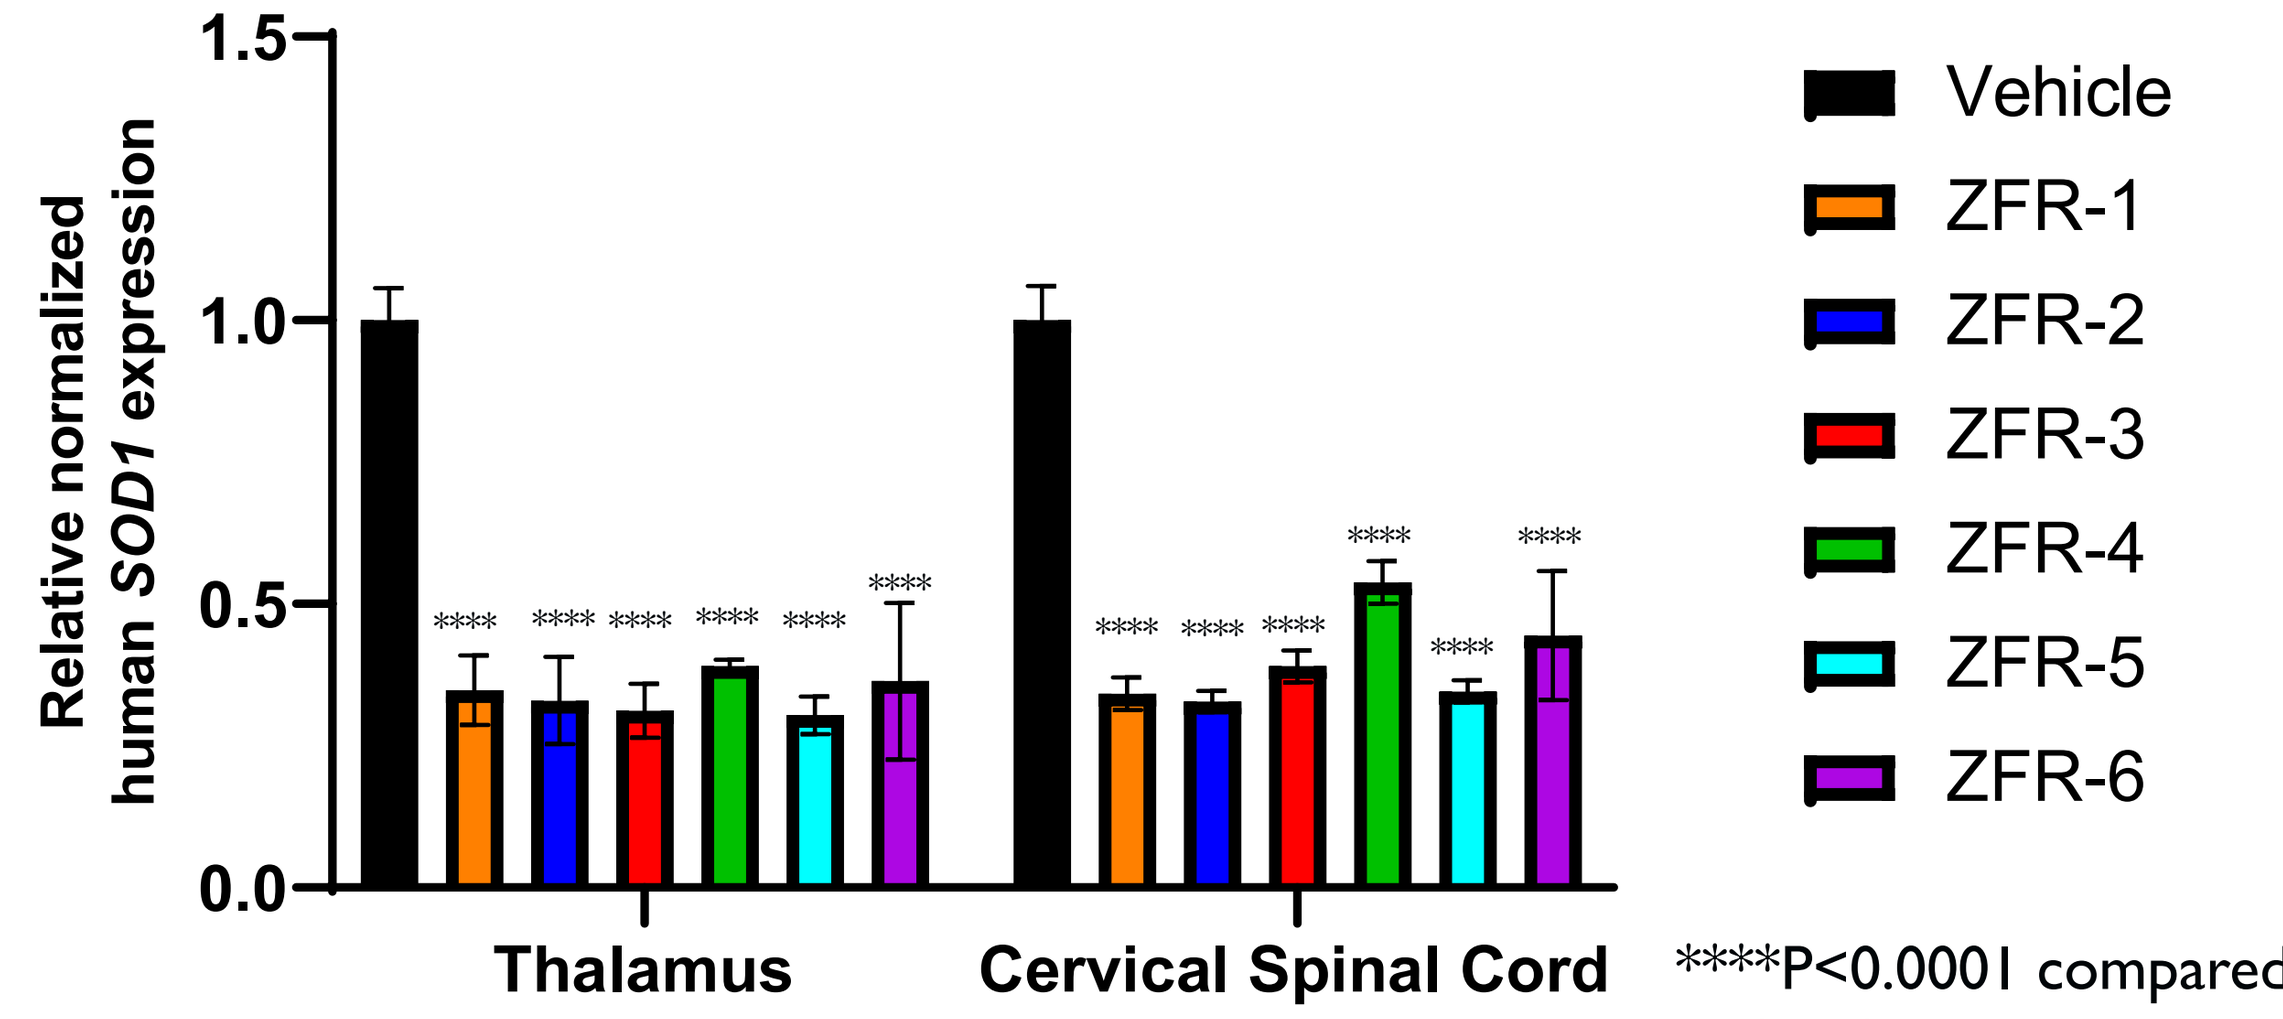
Relative normalized human SOD1 expression
Thalamus
Cervical Spinal Cord
Vehicle
ZFR-1
ZFR-2
ZFR-3
ZFR-4
ZFR-5
ZFR-6

****P<0.0001 compared with vehicle

In vivo screening for dual neuron/astrocyte promoters

Novel dual promoters to target neurons and astrocytes were designed and tested *in vitro*. Top performers and human synapsin were selected and packaged with ZFR-X targeting a mouse gene into PhP.B AAV. Following 8 weeks in life, repression of a mouse gene was analyzed in thalamus via RT-qPCR. Two novel promoters demonstrated higher repression than human synapsin, suggesting broadened transgene expression. Black circles indicate individual tissue samples.

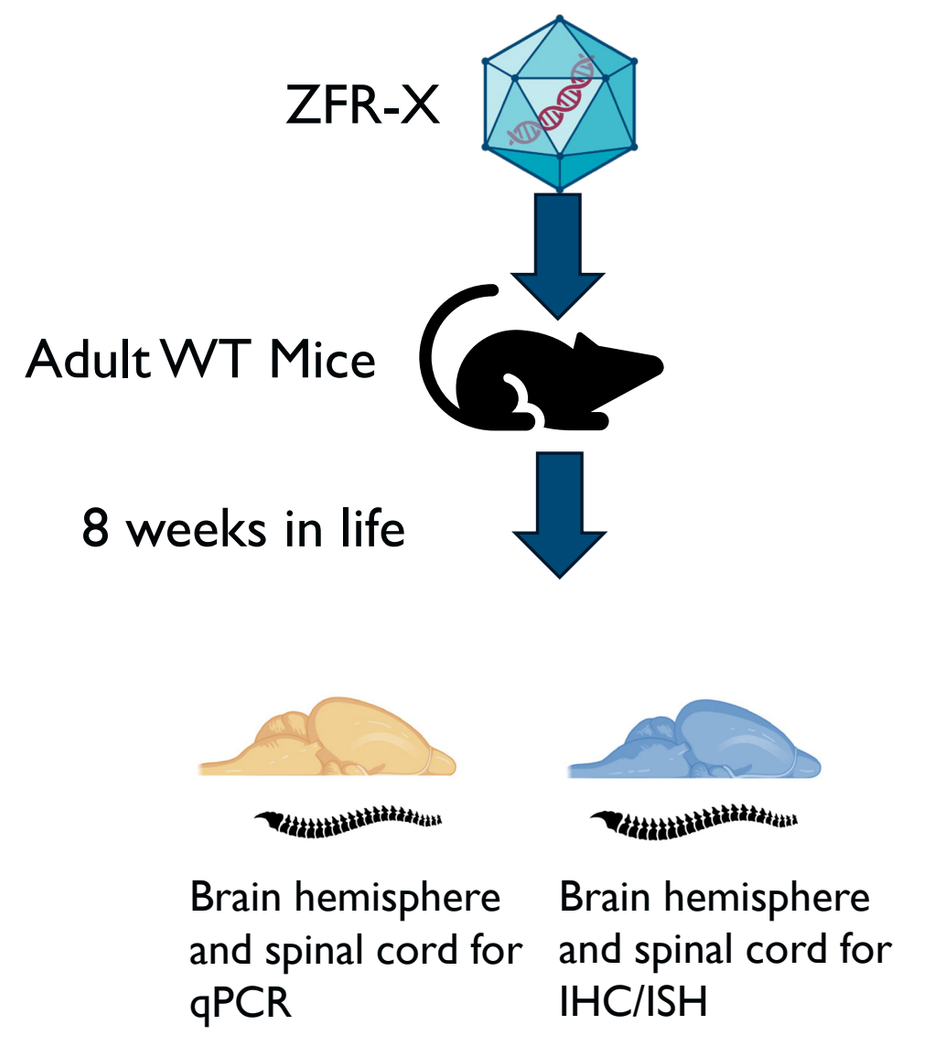
ZFR-X
Adult WT Mice
8 weeks in life
Brain hemisphere and spinal cord for qPCR
Brain hemisphere and spinal cord for IHC/ISH

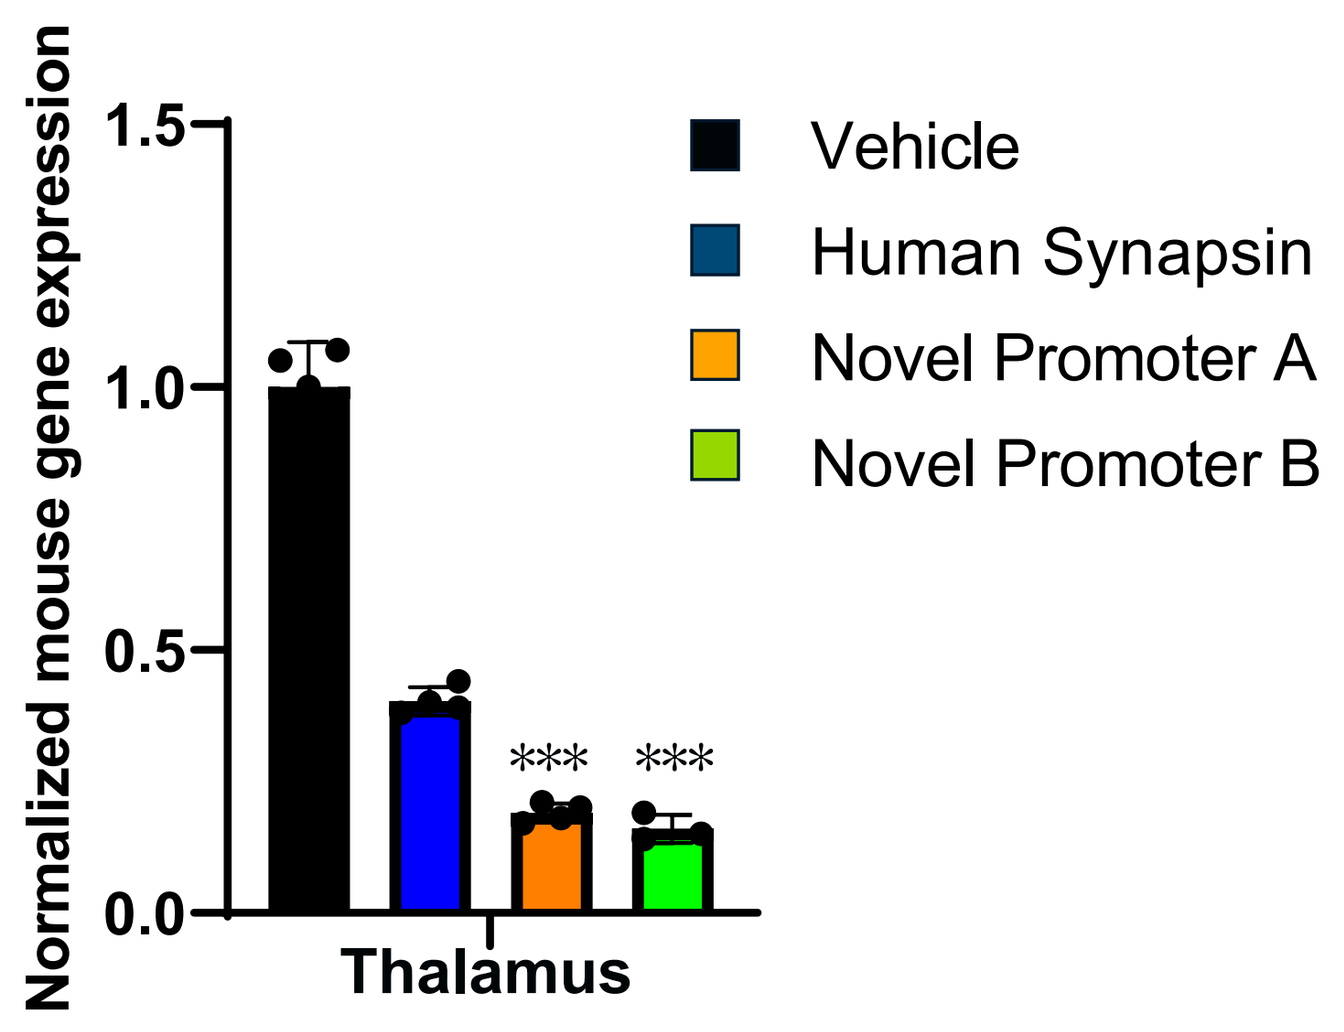
Normalized mouse gene expression
Thalamus
Vehicle
Human Synapsin
Novel Promoter A
Novel Promoter B

***P<0.001 compared with human synapsin

Conclusions and next steps

- We have developed ZFRs capable of robust SOD1 repression both *in vitro* and *in vivo*.
- In addition, we have generated initial evidence of a novel promoter driving a more robust repression profile than human synapsin, indicating the possibility of targeting astrocytes as well as neurons with a novel promoter.
- For next steps, proceed to a phenotypic rescue study in G93A mice using top ZFRs driven by human synapsin and novel regulatory elements.
- Continue to utilize SIFTER platform to identify spinal cord motor neuron-targeting AAV capsids.
- Proceed with IHC/ISH of dual promoter study tissue for single cell analysis and confirmation of targeting to both neurons and astrocytes.

Disclosures

All authors are current or former employees of Sangamo Therapeutics.

Presented at ASGCT 2024